Development of KT-253, a Highly Potent and Selective Heterobifunctional MDM2 Degrader for the treatment of Acute Myeloid Leukemia

Poster #5

Yogesh Chutake, Nancy Dumont, Michele Mayo, Jessica Filiatrault, Dapeng Chen, Vaishali Dixit, William Proctor, Susanne Breitkopf, Joyoti Dey, Alice McDonald, Frank Qi, Kan-Nian Hu, Kirti Sharma, Rahul Karnik, Reggie Ewesuedo, Stefanie Schalm, Joseph Growney, Ashwin Gollerkeri, Matt Weiss, Juliet Williams

INTRODUCTION

The murine double minute 2 (MDM2) oncoprotein is a key E3 ubiquitin ligase that degrades the tumor-suppressor p53. Targeting of the MDM2/p53 interaction with reversible small molecule inhibitors (SMI) to stabilize p53 and induce apoptosis in wildtype (WT) p53 tumors has gained momentum as emerging therapeutic approach for p53 WT malignancies. However, recent clinical trials with MDM2 SMIs, including in R/R AML, have resulted in suboptimal clinical activity, highlighting the need for more effective approaches to target MDM2.

KT-253 is a novel, highly potent and selective heterobifunctional MDM2 degrader that has demonstrated the ability to overcome the MDM2 feedback loop that is known to be triggered upon p53 upregulation and thereby limiting clinical activity of small molecule inhibitors. We have shown >200-fold higher growth inhibition potency in vitro compared with MDM2 SMIs, and a favorable pharmacological profile where a single dose of KT-253 was sufficient to induce rapid apoptosis in vivo and sustained tumor regression in the MV4;11 AML and RS4;11 ALL cell line-derived xenograft (CDX) models, supporting an intermittent dosing schedule for KT-253. In addition, when administered once every 3 weeks, KT-253 led to tumor regressions in several AML patient-derived xenograft models. Moreover, in AML CDX models such as MOLM13, with known resistance to standard of care (SOC) venetoclax, acute administration of KT-253 in combination with clinical and sub-clinical doses of venetoclax resulted in durable tumor regressions.

KT-253, currently being studied in Phase 1 clinical trial (NCT05775406) in high grade myeloid malignancies including AML, solid tumors, and lymphomas, has the potential to benefit AML patients both as a monotherapy and in combination with SOC agents such as venetoclax.

Proteome Editing with Targeted Protein Degradation

Co-opting a Naturally Occurring Process to Regulate Protein Levels

1. E3 ligase recognizes protein
2. Ubiquitin chain transferred
3. Protein is marked for elimination

KYMERA

Small Molecule Protein Degrader

Broad Opportunity
Only binding site required

Highly Potent
Catalytic

Highly Specific
Targeted protein degradation
Genetic knock-down-like effects

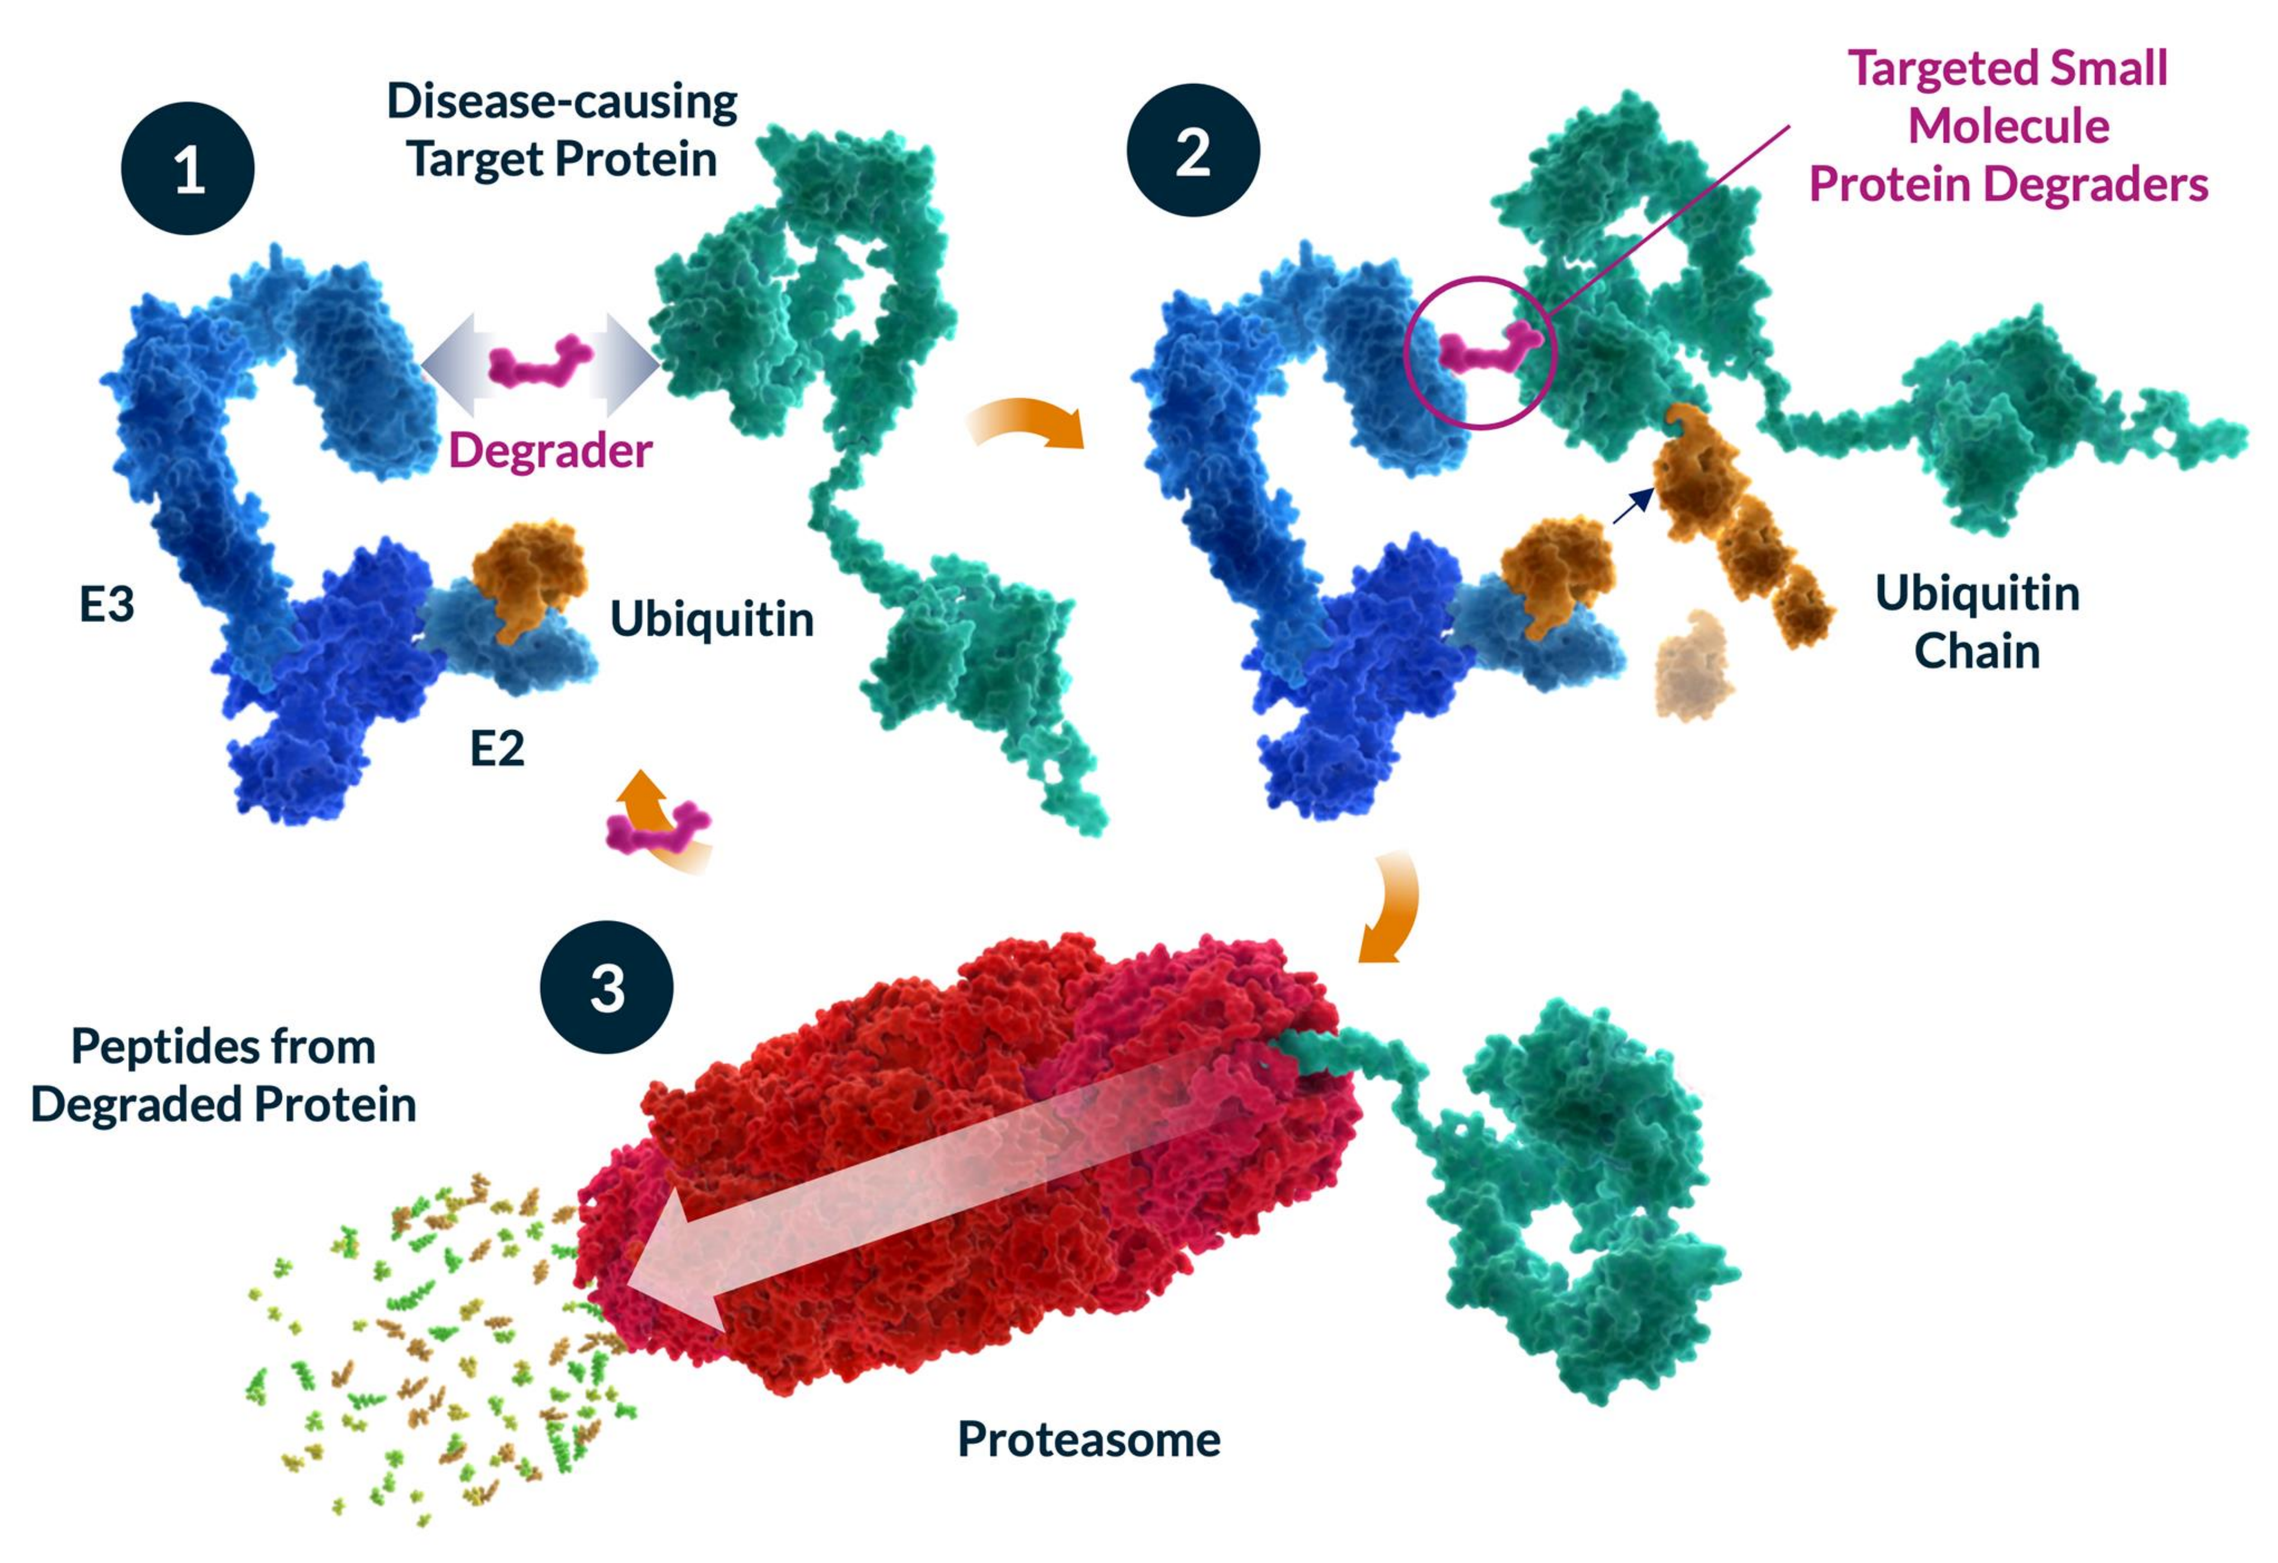
1
Disease-causing Target Protein
Degrader
E3
Ubiquitin
E2
2
Targeted Small Molecule Protein Degraders
Ubiquitin Chain
3
Peptides from Degraded Protein
Proteasome

MDM2 Degradation Effectively Induces p53 Signaling

Clinical Validation

- **Small molecule inhibitors** of the MDM2/p53 are active but are limited due to narrow therapeutic index

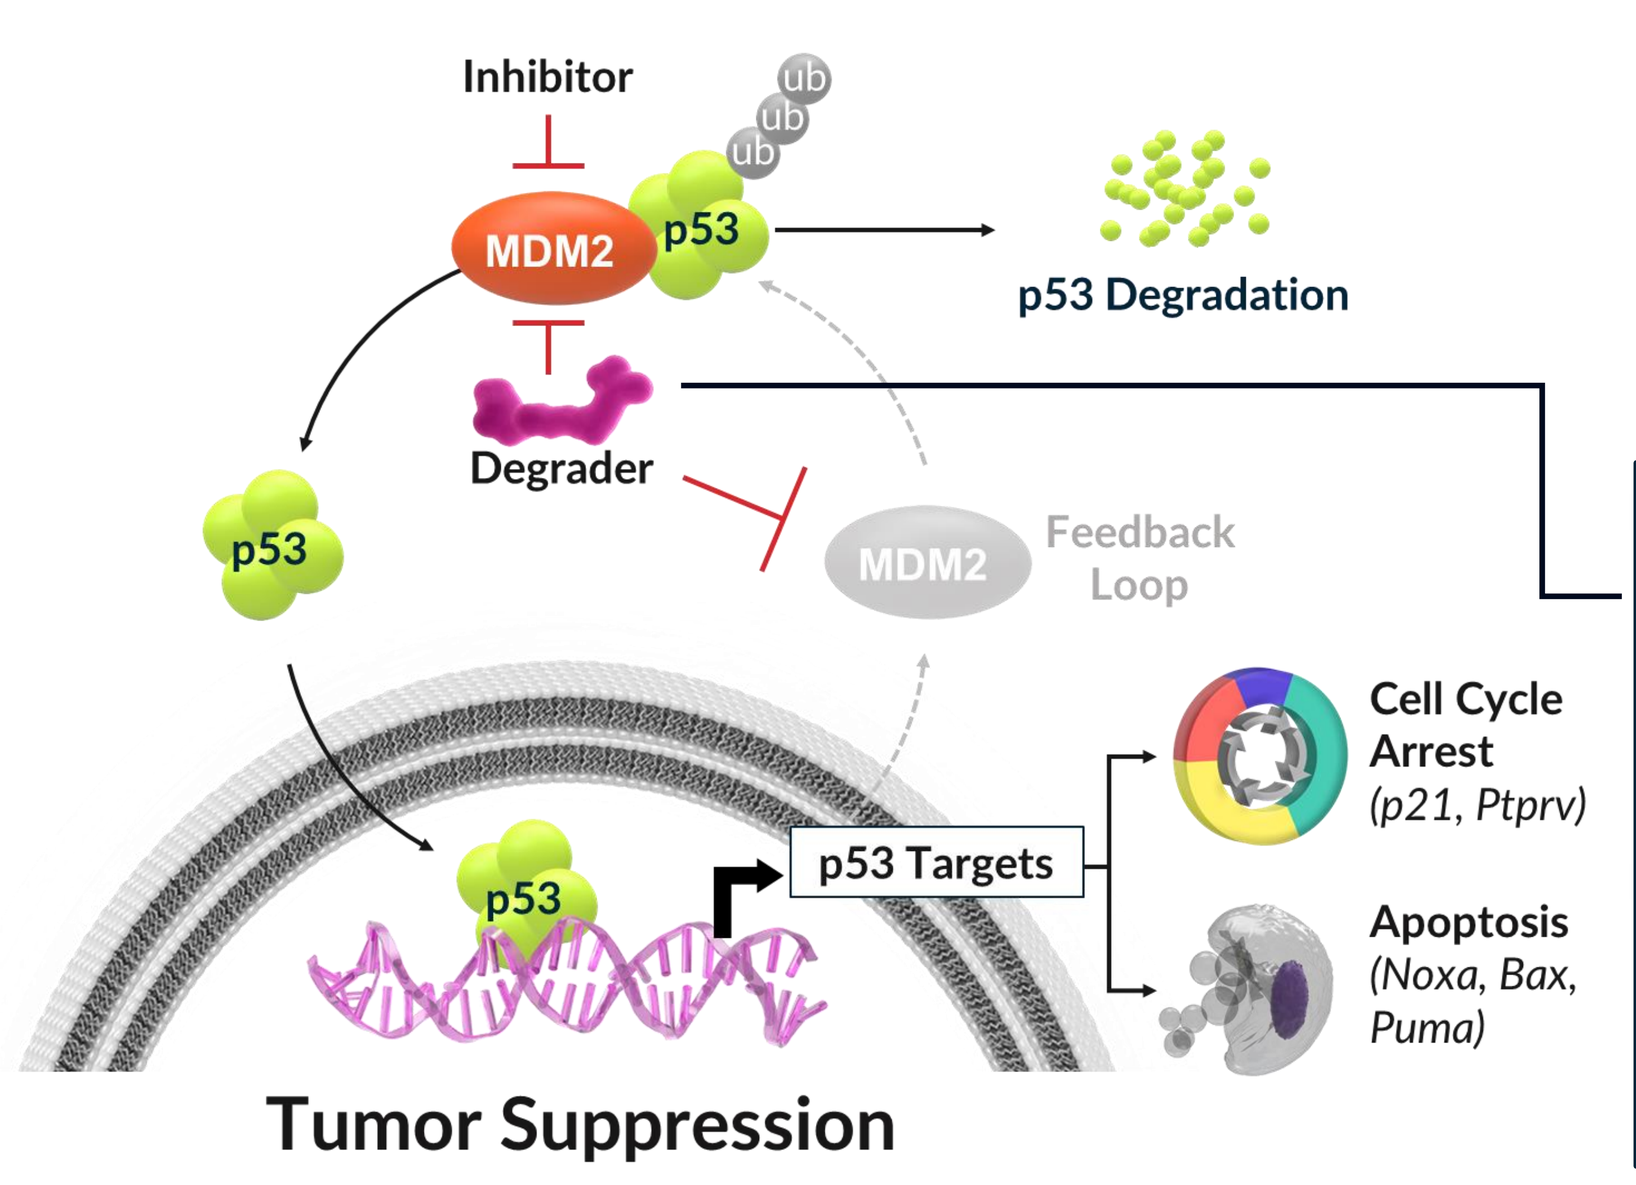
Inhibitor
ub
MDM2
p53
p53 Degradation
Degrader
MDM2
Feedback Loop
Cell Cycle Arrest
(p21, Ptprv)
Apoptosis
(Noxa, Bax, Puma)
p53 Targets
Tumor Suppression

Cancer Genetics

- p53 is **NOT mutated** in almost 50% of tumors
- MDM2 overexpression or amplification can inactivate p53
- Large opportunity in wide variety of cancers

Degrader Advantage

- **MDM2 degraders**, because of their catalytic and not occupancy driven mechanism can lead to more efficient p53 stabilization and induction of an acute apoptotic response in tumor cells
- The distinct degrader pharmacology enables an intermittent dosing schedule that gives normal cells more time to recover and may increase the therapeutic index vs a small molecule inhibitor

Figure 1: KT-253 is a highly potent MDM2 degrader and p53 stabilizer compared to MDM2/p53 small molecule inhibitors

KT-253 is a potent MDM2 degrader → **KT-253, unlike SMI's, potently stabilizes p53…** → **… which leads to superior tumor cell killing (pM range)**

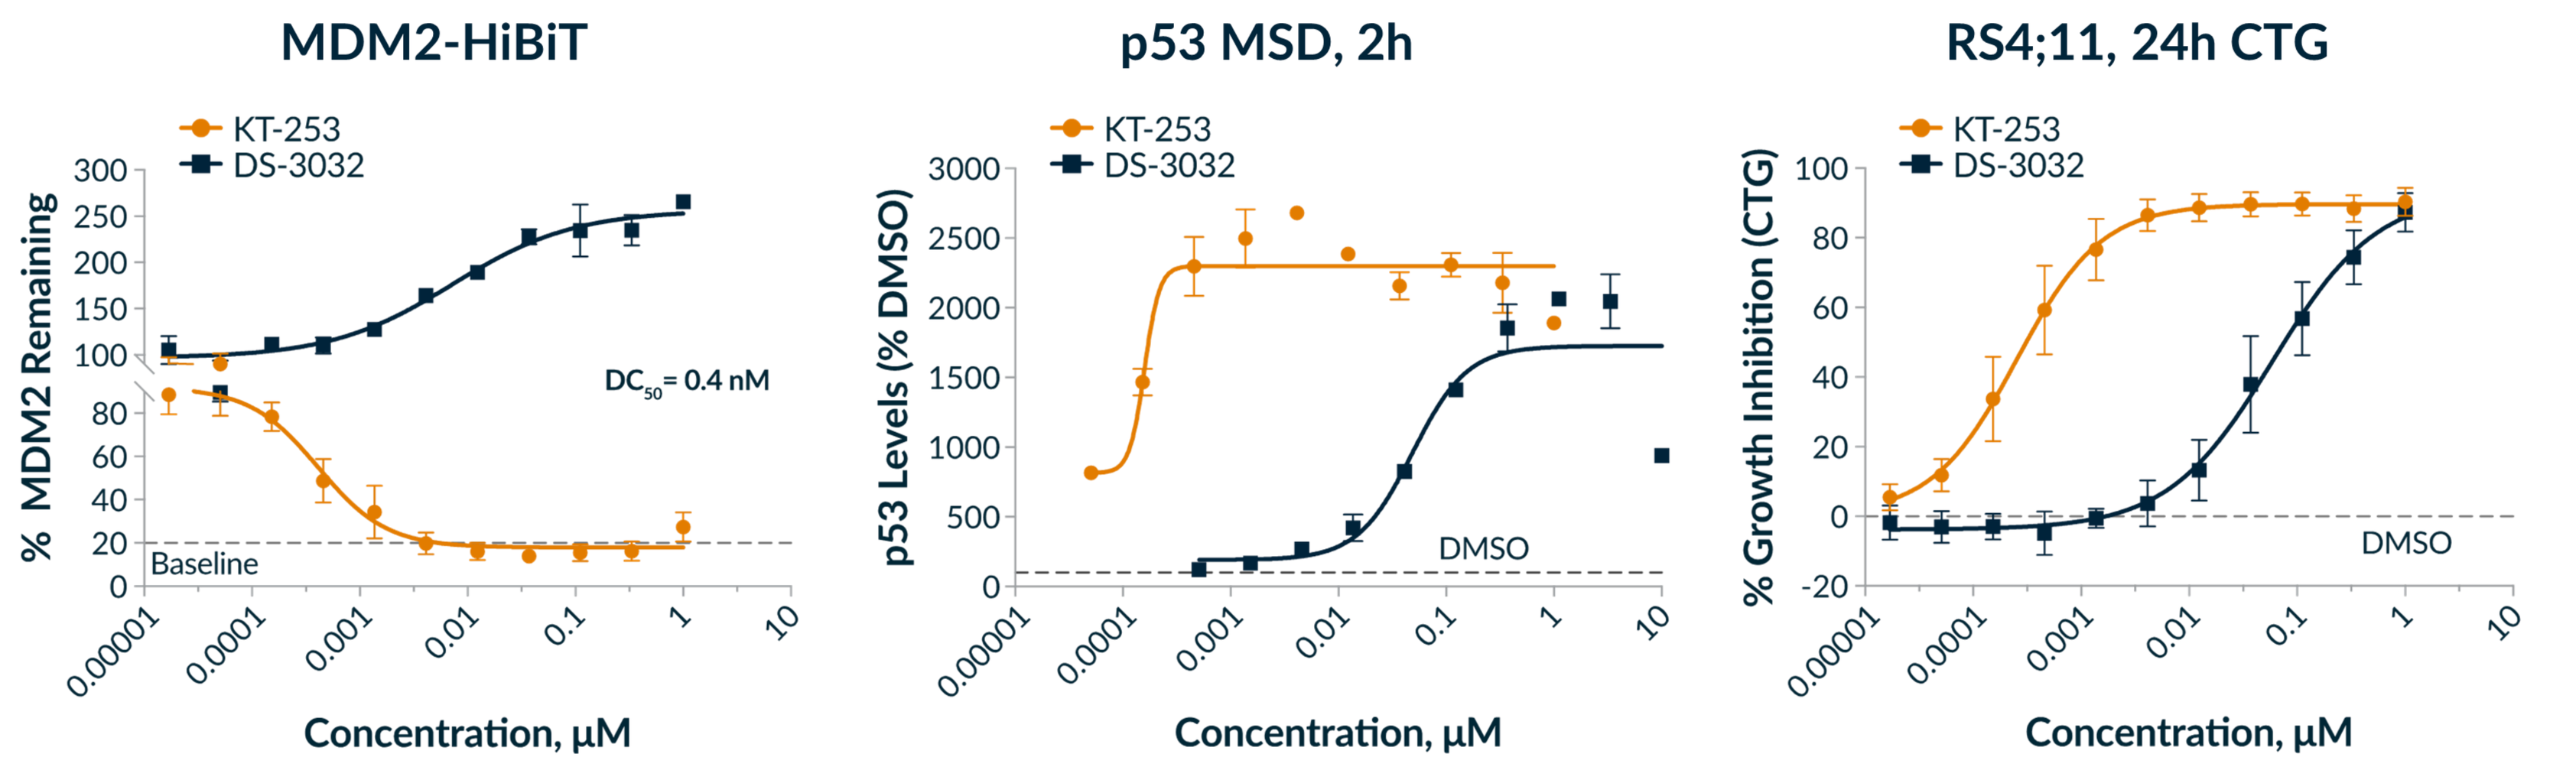
MDM2-HiBiT
KT-253
DS-3032
DC50 = 0.4 nM
Baseline
% MDM2 Remaining
Concentration, µM
p53 MSD, 2h
p53 Levels (% DMSO)
DMSO
RS4;11, 24h CTG
% Growth Inhibition (CTG)

Compound	KT-253	DS-3032	RG7388	SAR405838	HDM201	AMG-232
Company	Kymera	Sankyo/Rain	Roche	Sanofi	Novartis	Amgen/ Kartos
RS4;11 IC_{50} (nM) (ALL Cell Killing)	0.3	67	220	620	163	280
MDM2-HiBiT, DC_{50} (nM) (Degradation)	0.4	-	-	-	-	-

- KT-253 is **>200-fold more potent** in tumor cell killing assays than SMI's due to its mechanism of action

Figure 2: A single dose of KT-253 drives sustained tumor regression in ALL and AML xenograft models

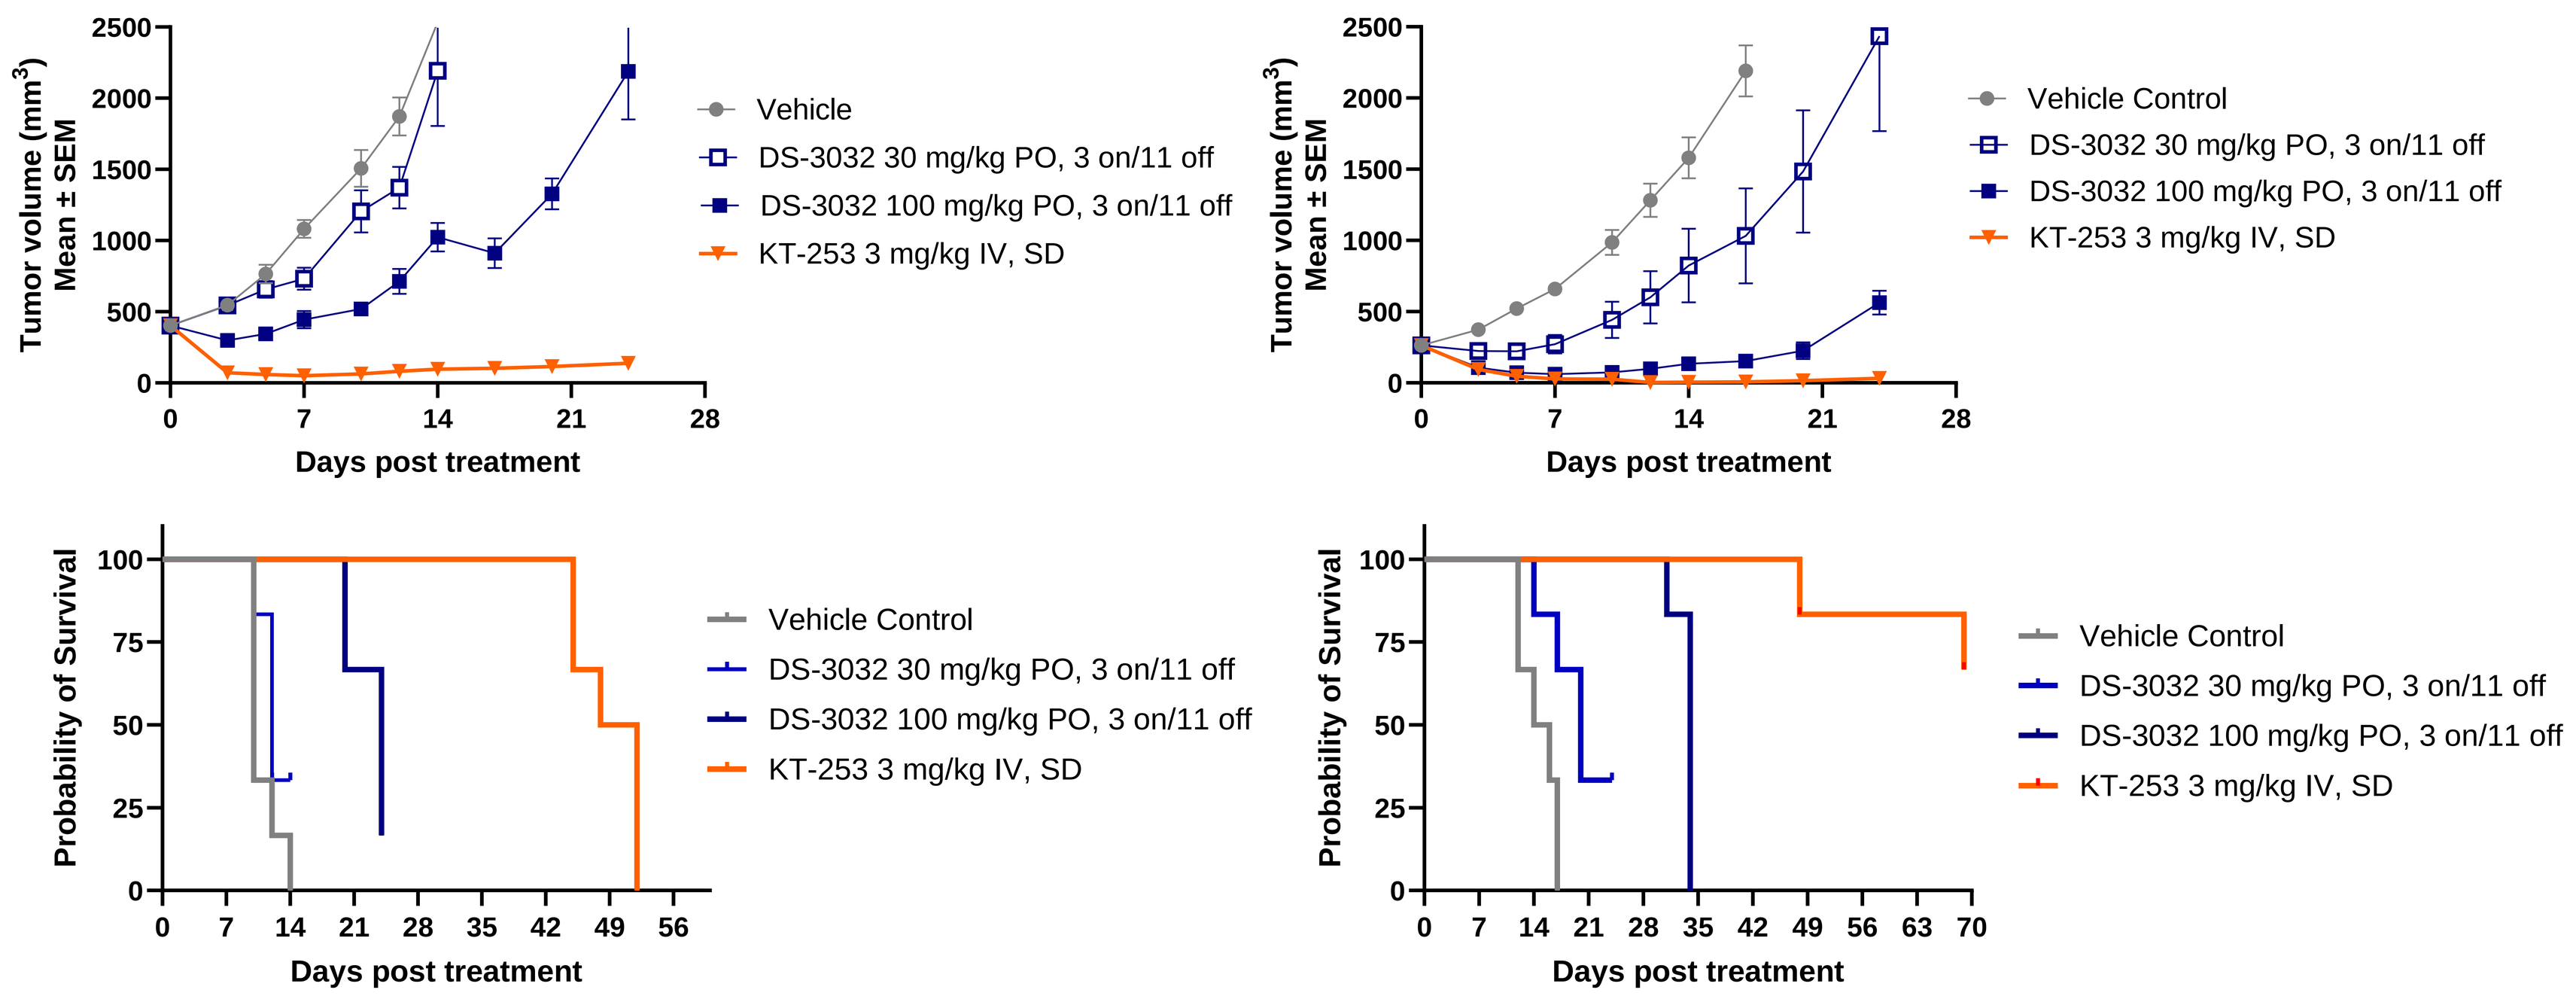
RS4;11
Tumor volume (mm³) Mean ± SEM
Vehicle
DS-3032 30 mg/kg PO, 3 on/11 off
DS-3032 100 mg/kg PO, 3 on/11 off
KT-253 3 mg/kg IV, SD
Days post treatment
Probability of Survival
Vehicle Control
MV4;11

- In the RS4;11 ALL model, the median survival after a single dose of KT-253 at 3 mg/kg was 50 days vs 12 days for the clinically equivalent dosing regimen of DS-3032
- In the MV4;11 AML model, a single dose of KT-253 at 3 mg/kg led to complete responses in 5 of 6 animals, and 4 of 6 remain tumor-free on study 80 days post dosing
- No complete responses were observed following treatment with DS-3032
- n=6 animals/group

Figure 3: A single dose of KT-253 leads to robust activation of the p53 Pathway and apoptosis in RS4;11 (ALL) and MV4;11(AML) models while MDM2 SMIs do not

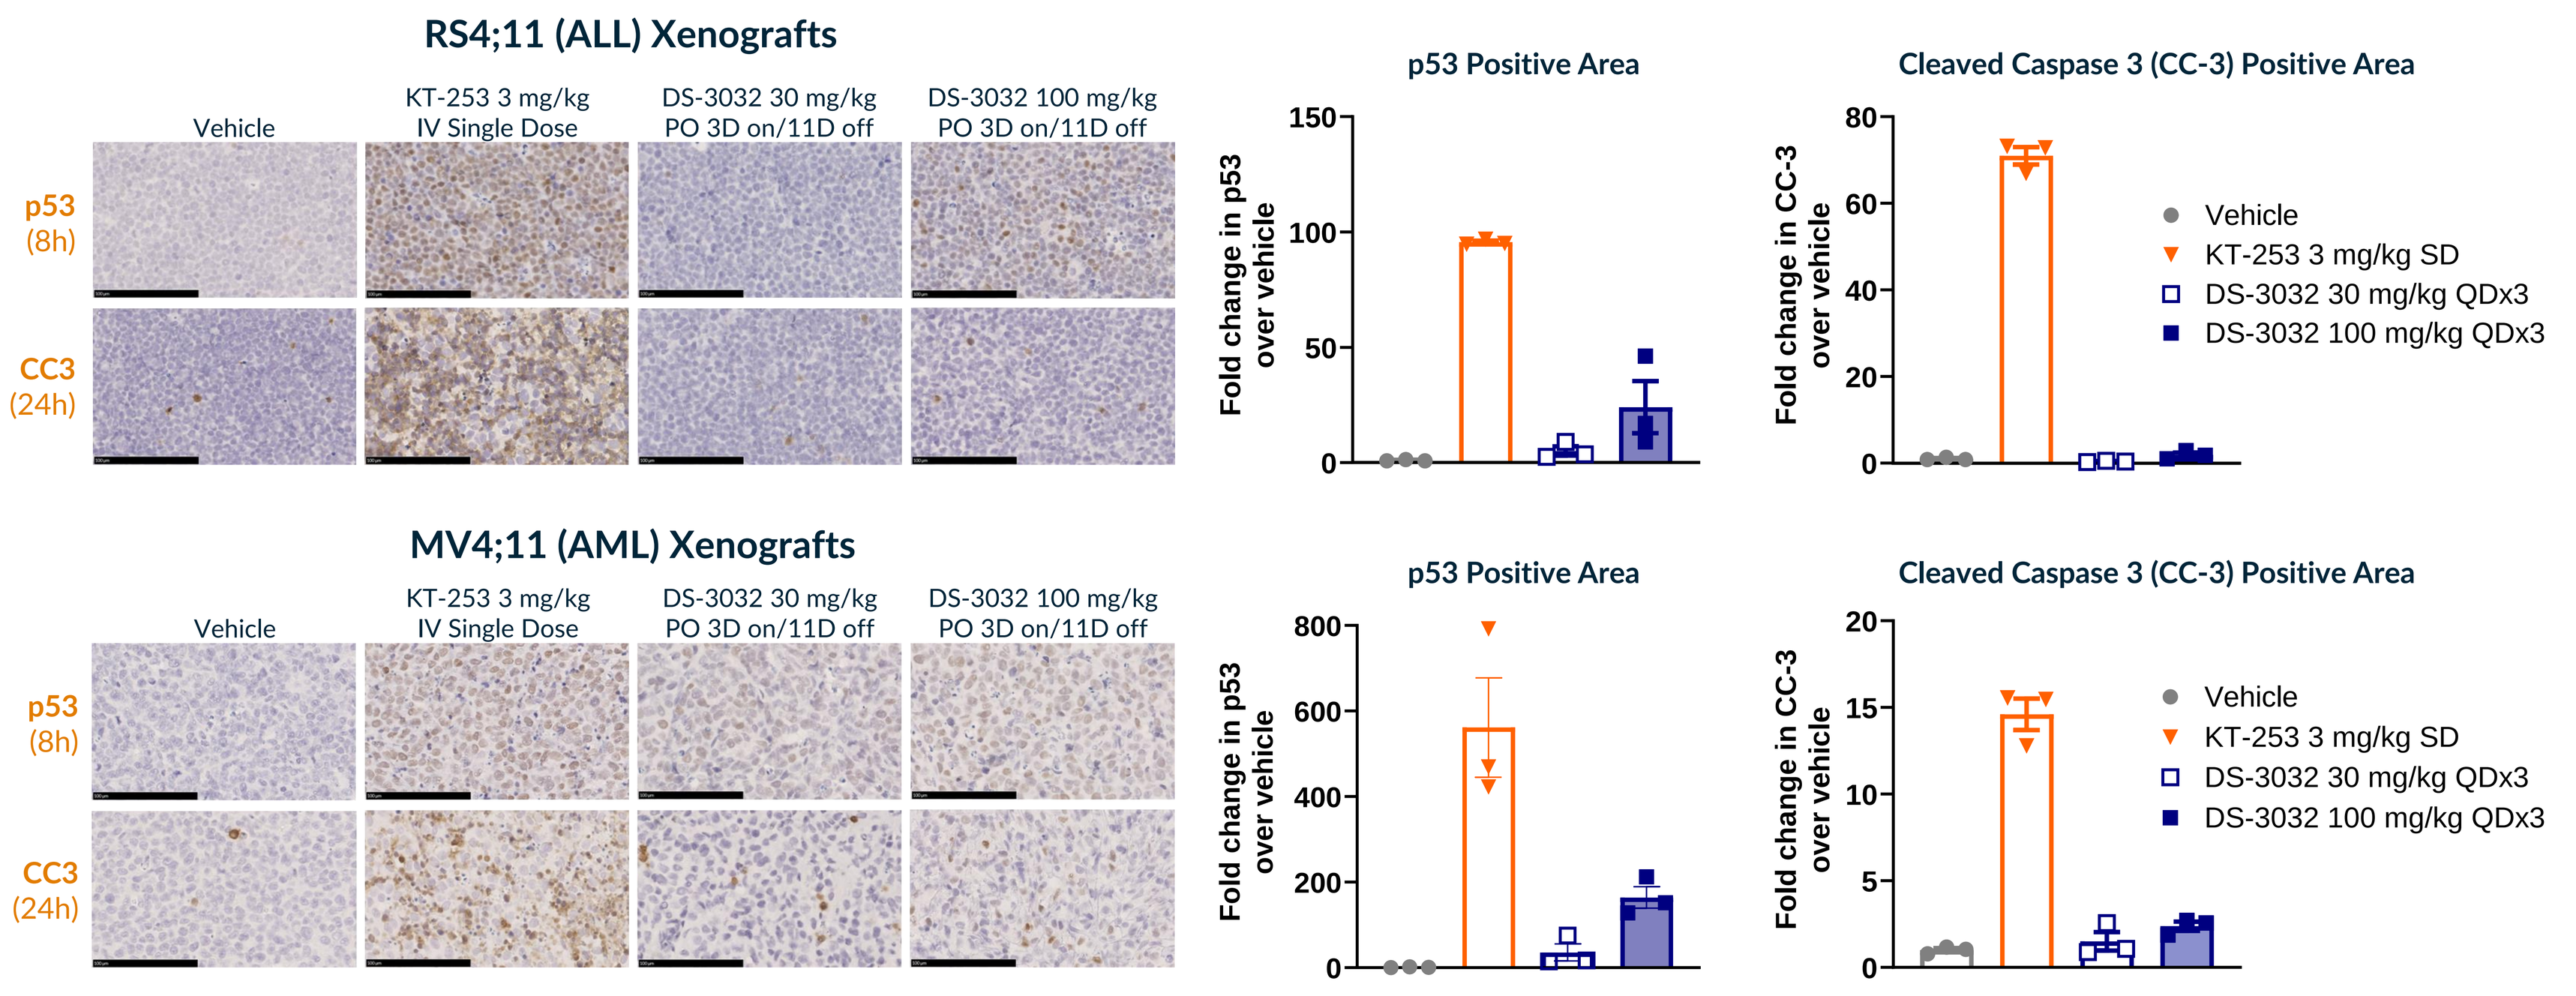
RS4;11 (ALL) Xenografts
Vehicle
KT-253 3 mg/kg IV Single Dose
DS-3032 30 mg/kg PO 3D on/11D off
DS-3032 100 mg/kg PO 3D on/11D off
p53 (8h)
CC3 (24h)
p53 Positive Area
Cleaved Caspase 3 (CC-3) Positive Area
Fold change in p53 over vehicle
Fold change in CC-3 over vehicle
Vehicle
KT-253 3 mg/kg SD
DS-3032 30 mg/kg QDx3
DS-3032 100 mg/kg QDx3
MV4;11 (AML) Xenografts

- IHC analysis of RS4;11 and MV4;11 tumors demonstrates robust activation of the p53 pathway and induction of cleaved caspase-3 (CC-3) following a single dose of KT-253
- In comparison, induction of CC-3 following treatment with the MDM2 SMI, DS-3032, is modest

Figure 4: KT-253 demonstrates strong anti-leukemic activity in patient-derived xenograft models of systemic AML

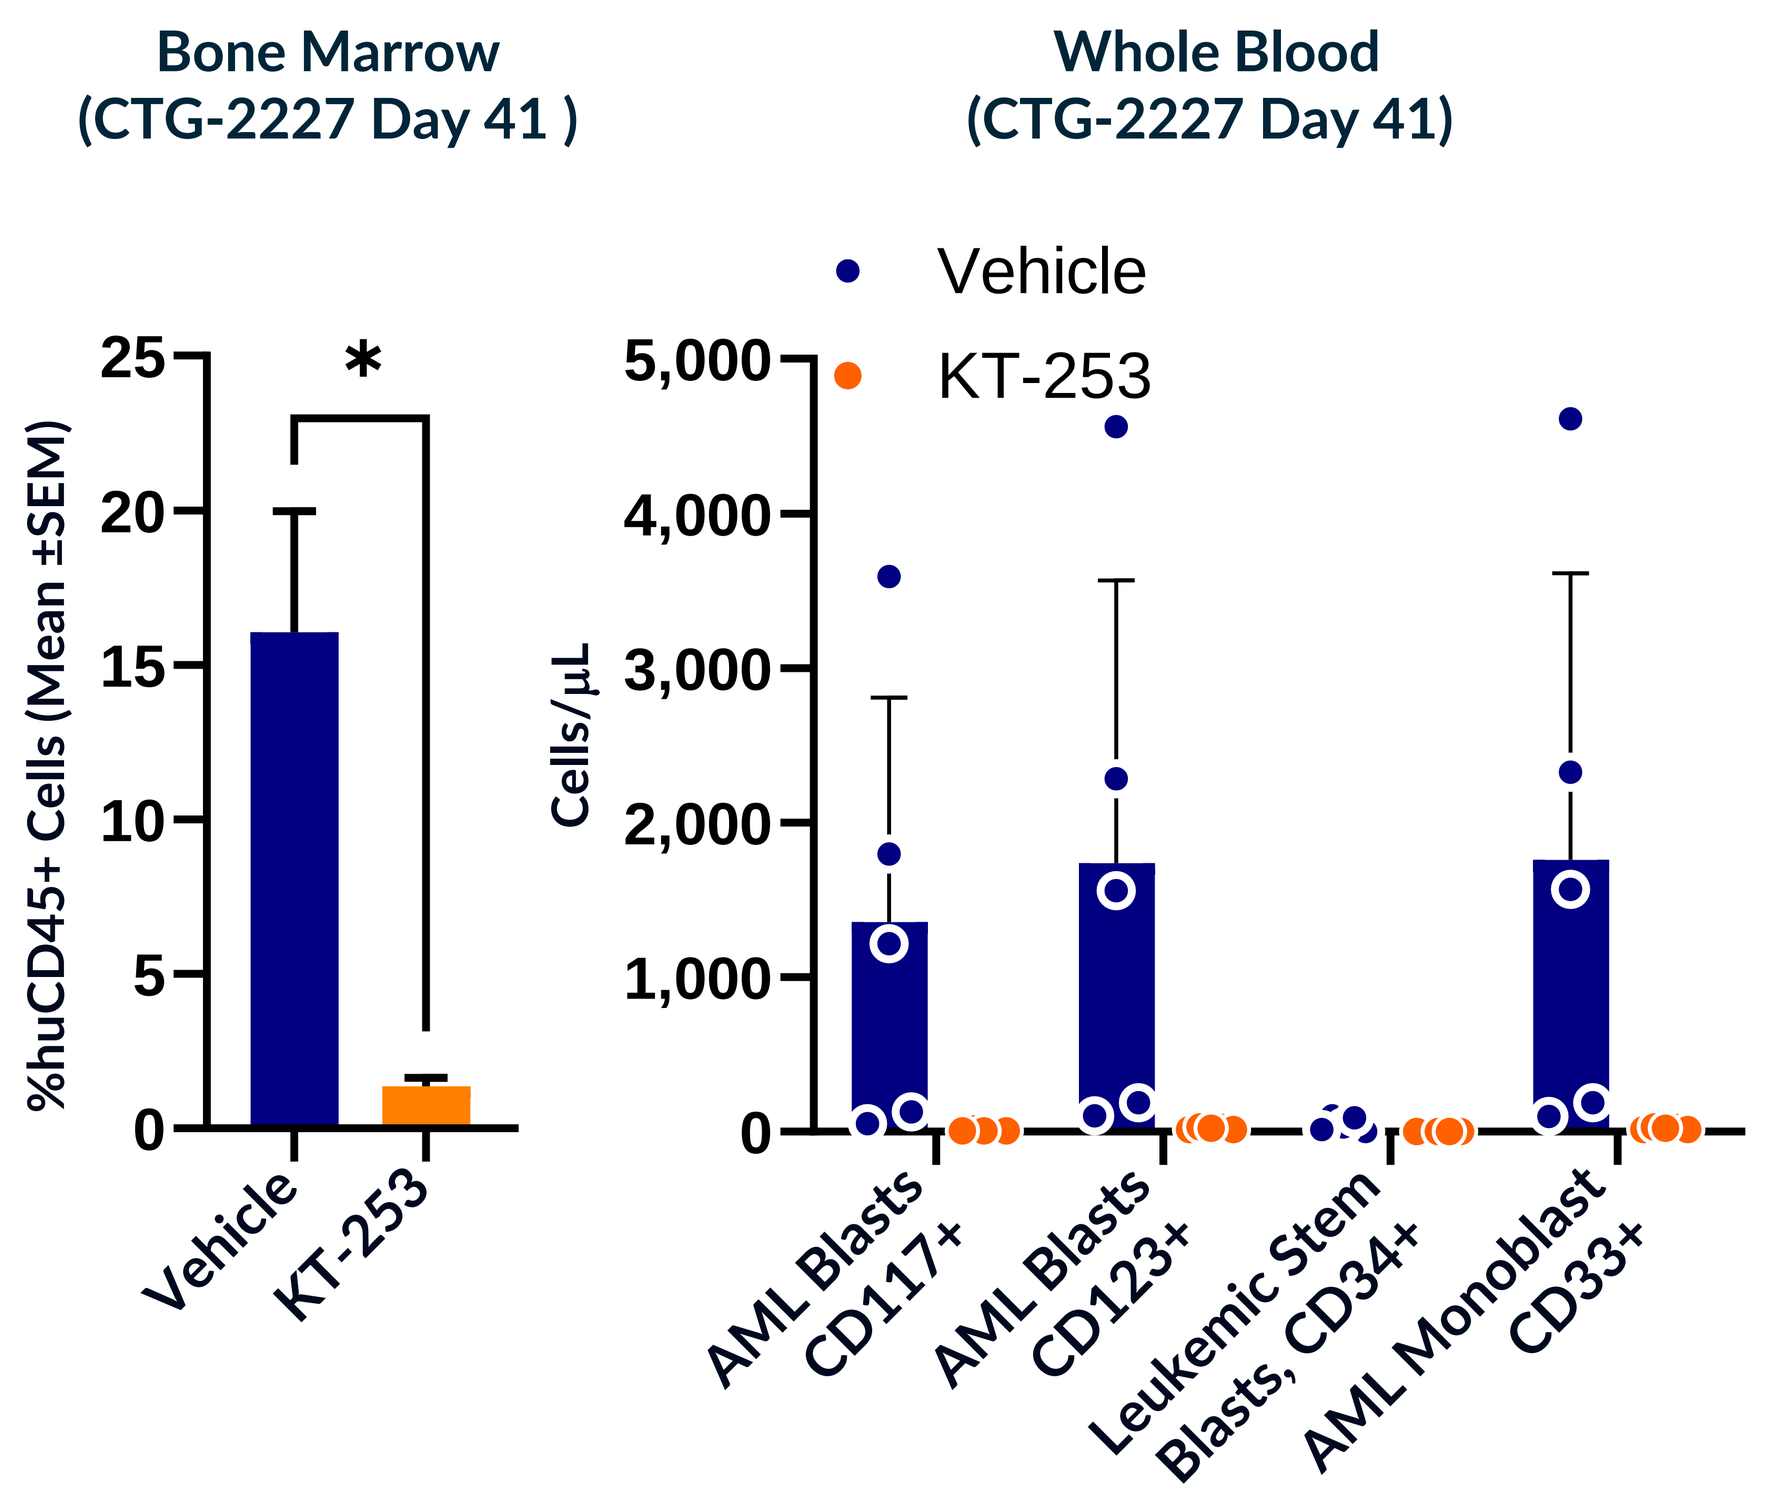
Bone Marrow
(CTG-2227 Day 41)
Whole Blood
(CTG-2227 Day 41)
%huCD45+ Cells (Mean ±SEM)
Vehicle
KT-253
Cells/µL
AML Blasts CD117+
AML Blasts CD123+
Leukemic Stem Blasts, CD34+
AML Monoblast CD33+

Model	FAB/WHO Subtype	Treatment History	Response* in Peripheral Blood
CTG-2227	M4	Pretreated	**Complete Response**
CTG-2240	AML	Naïve	**Partial Response**
CTG-2700	M2	Pretreated	**Partial Response**
CTG-2235	AML-MLD	-	No Response

- Animals received two doses; takedown and tumor burden assessment at day 41 post implant

*Response Criteria, by Champions Oncology

<25% decrease or increase	No response
25-50% decrease	Partial response
>50% decrease	Complete response

CTG-2227 Characteristics:

- R/R AML, M4 (myelomonocytic)
- Mutations: IDH1, FLT3 ITD, DNMT3A, NPM1, ASXL1

- KT-253 1 mg/ kg, Q3W dosing significantly reduces hCD45+ leukemic cells in bone marrow and peripheral blood
- Three out of 4 AML PDX models show appreciable tumor response in peripheral blood

Figure 5: Significant combination benefit is observed with KT-253 at subclinical doses of venetoclax in a venetoclax-resistant AML model

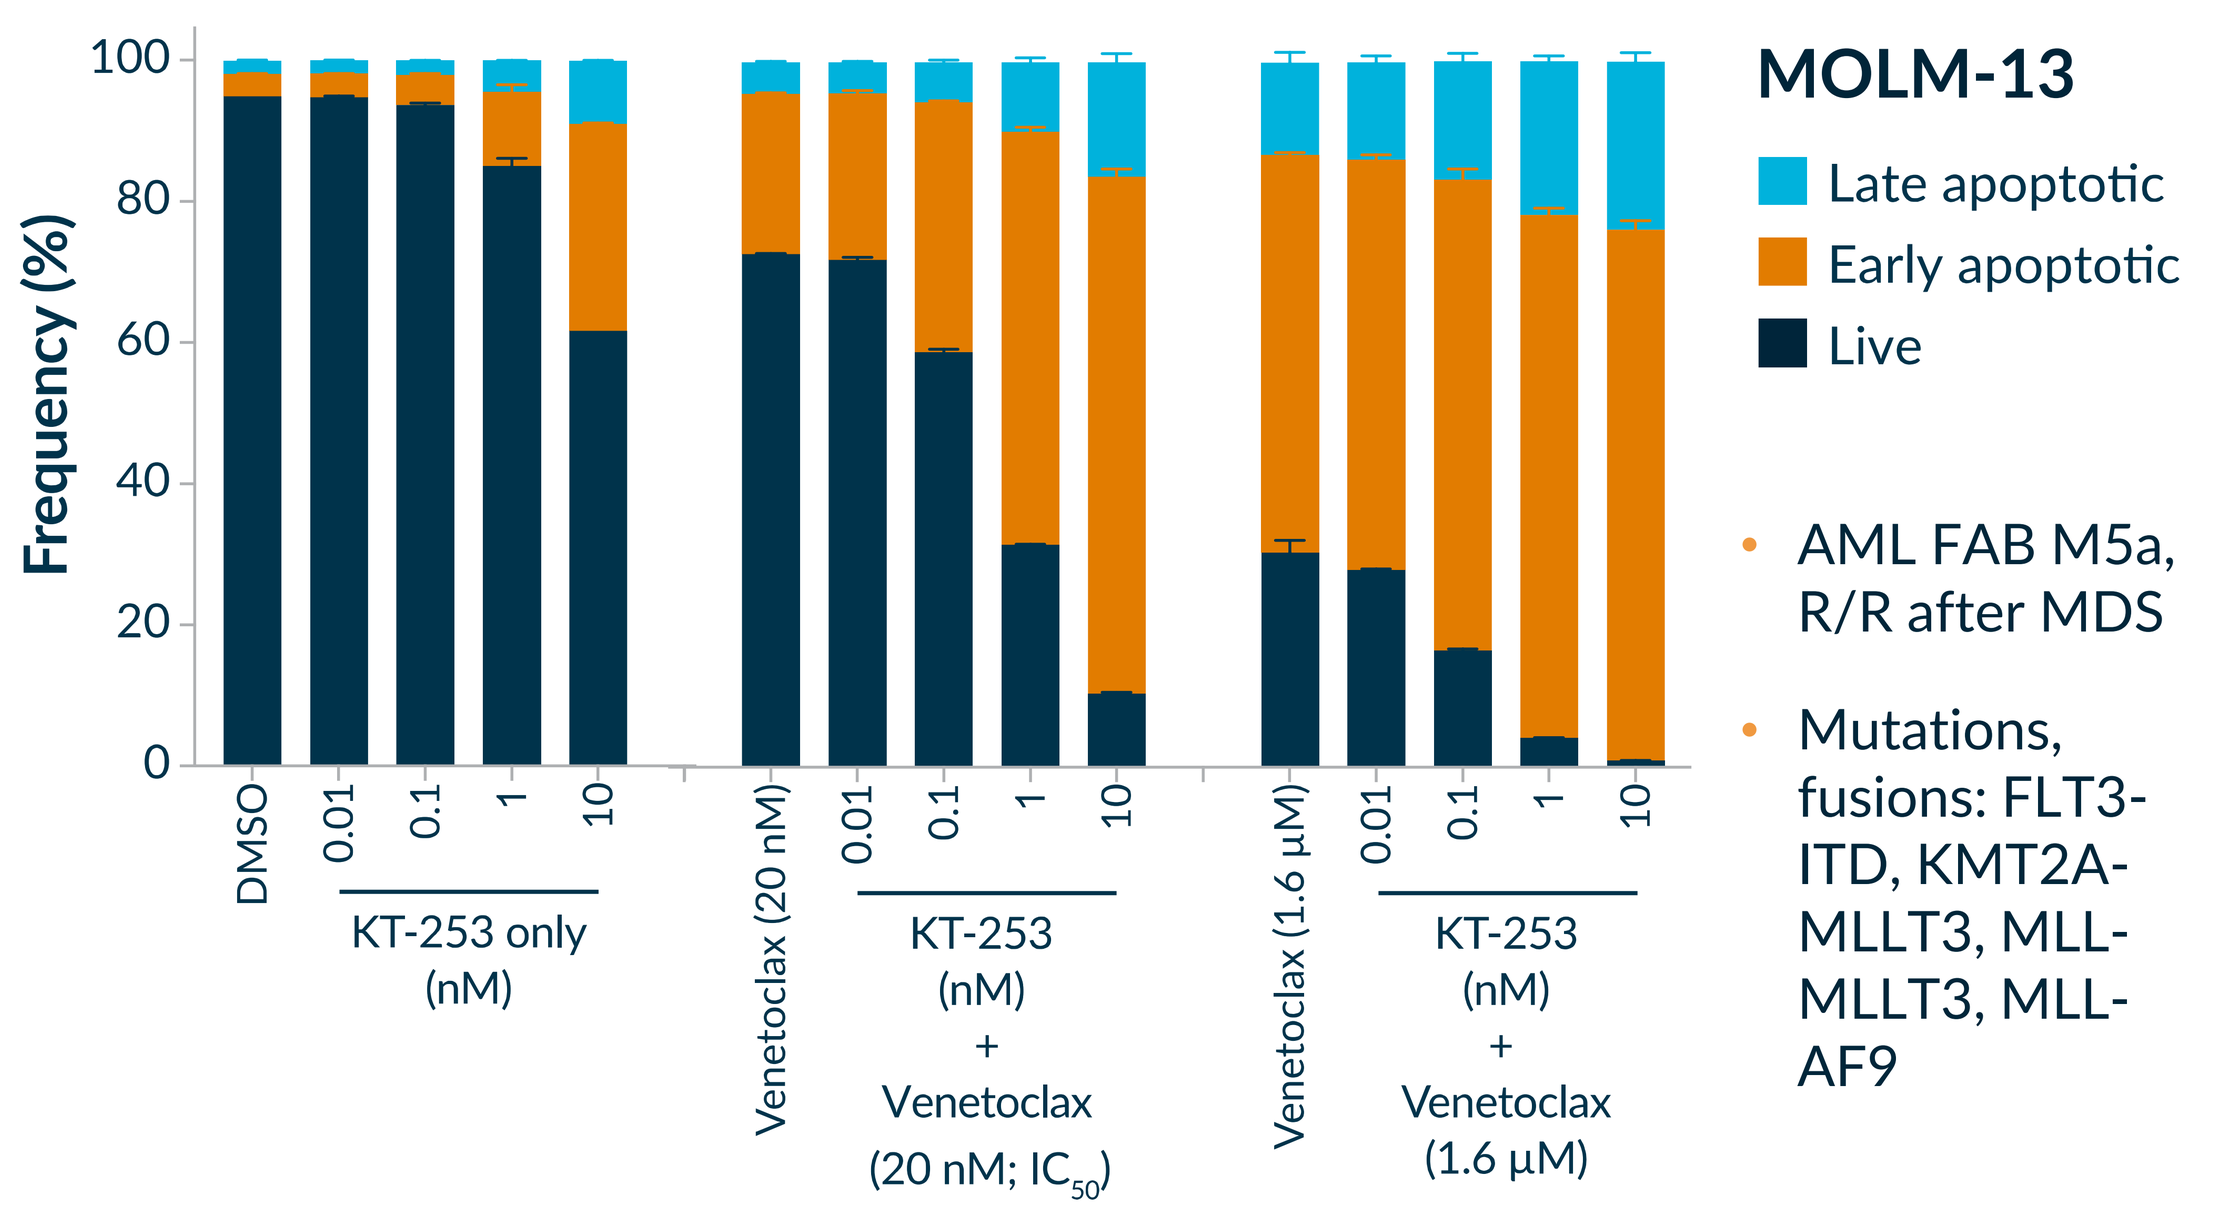
Frequency (%)
DMSO
KT-253 only (nM)
Venetoclax (20 nM)
KT-253 (nM) + Venetoclax (20 nM; IC50)
Venetoclax (1.6 µM)
KT-253 (nM) + Venetoclax (1.6 µM)
MOLM-13
Late apoptotic
Early apoptotic
Live

- AML FAB M5a, R/R after MDS
- Mutations, fusions: FLT3-ITD, KMT2A-MLLT3, MLL-MLLT3, MLL-AF9

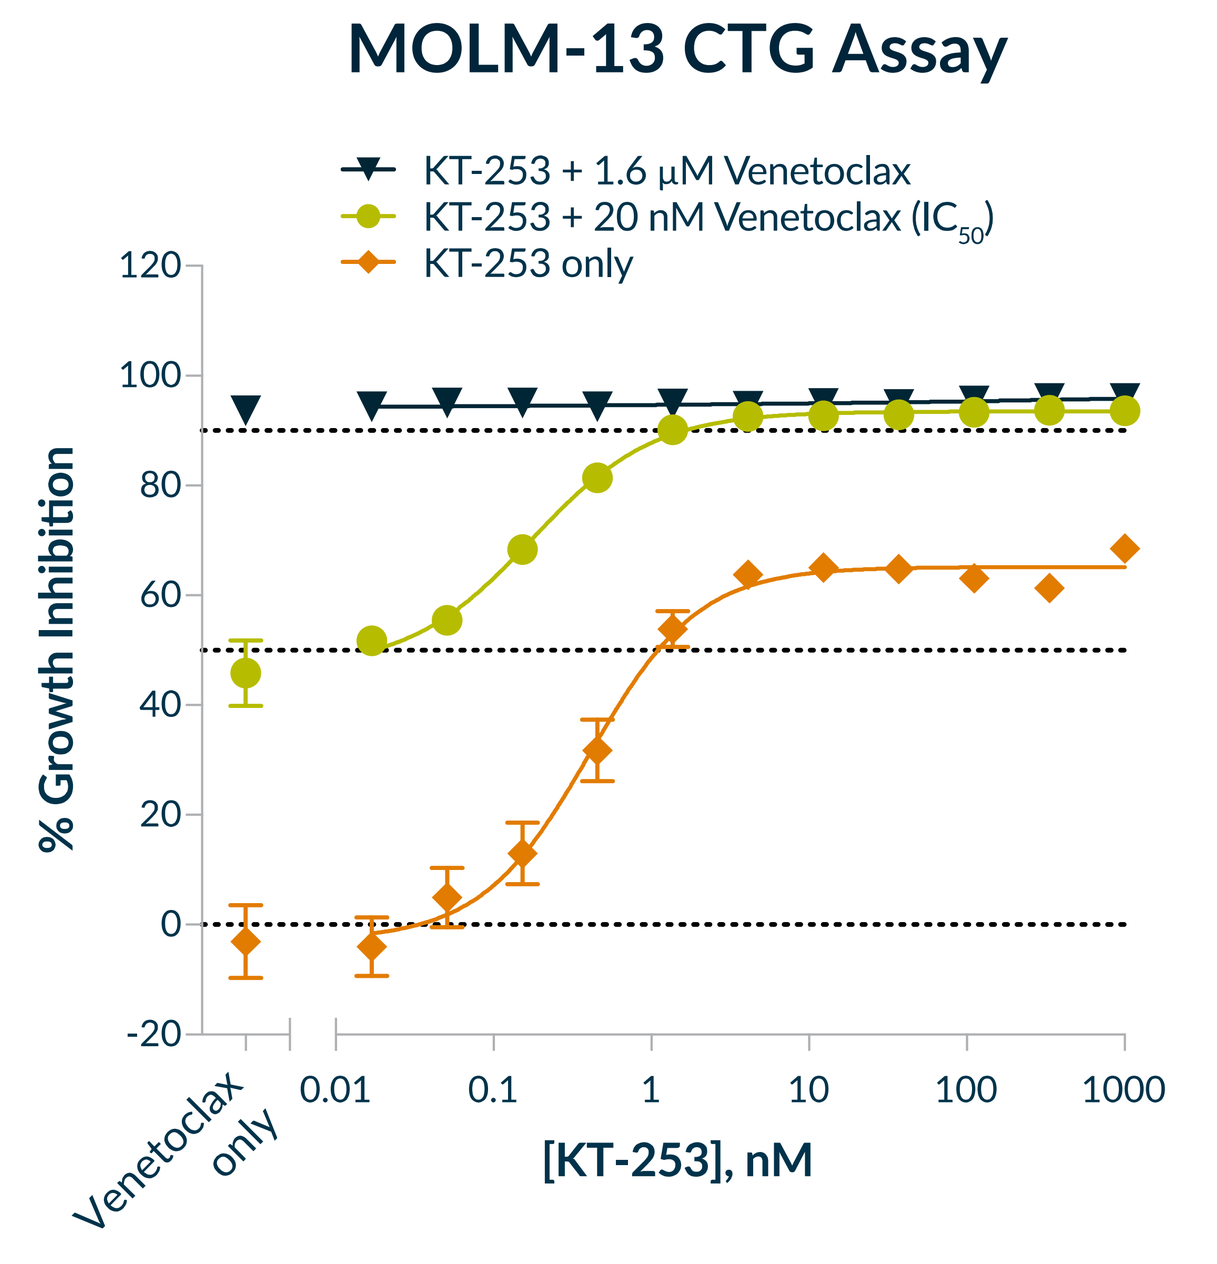
MOLM-13 CTG Assay
KT-253 + 1.6 µM Venetoclax
KT-253 + 20 nM Venetoclax (IC50)
KT-253 only
% Growth Inhibition
Venetoclax only
[KT-253], nM

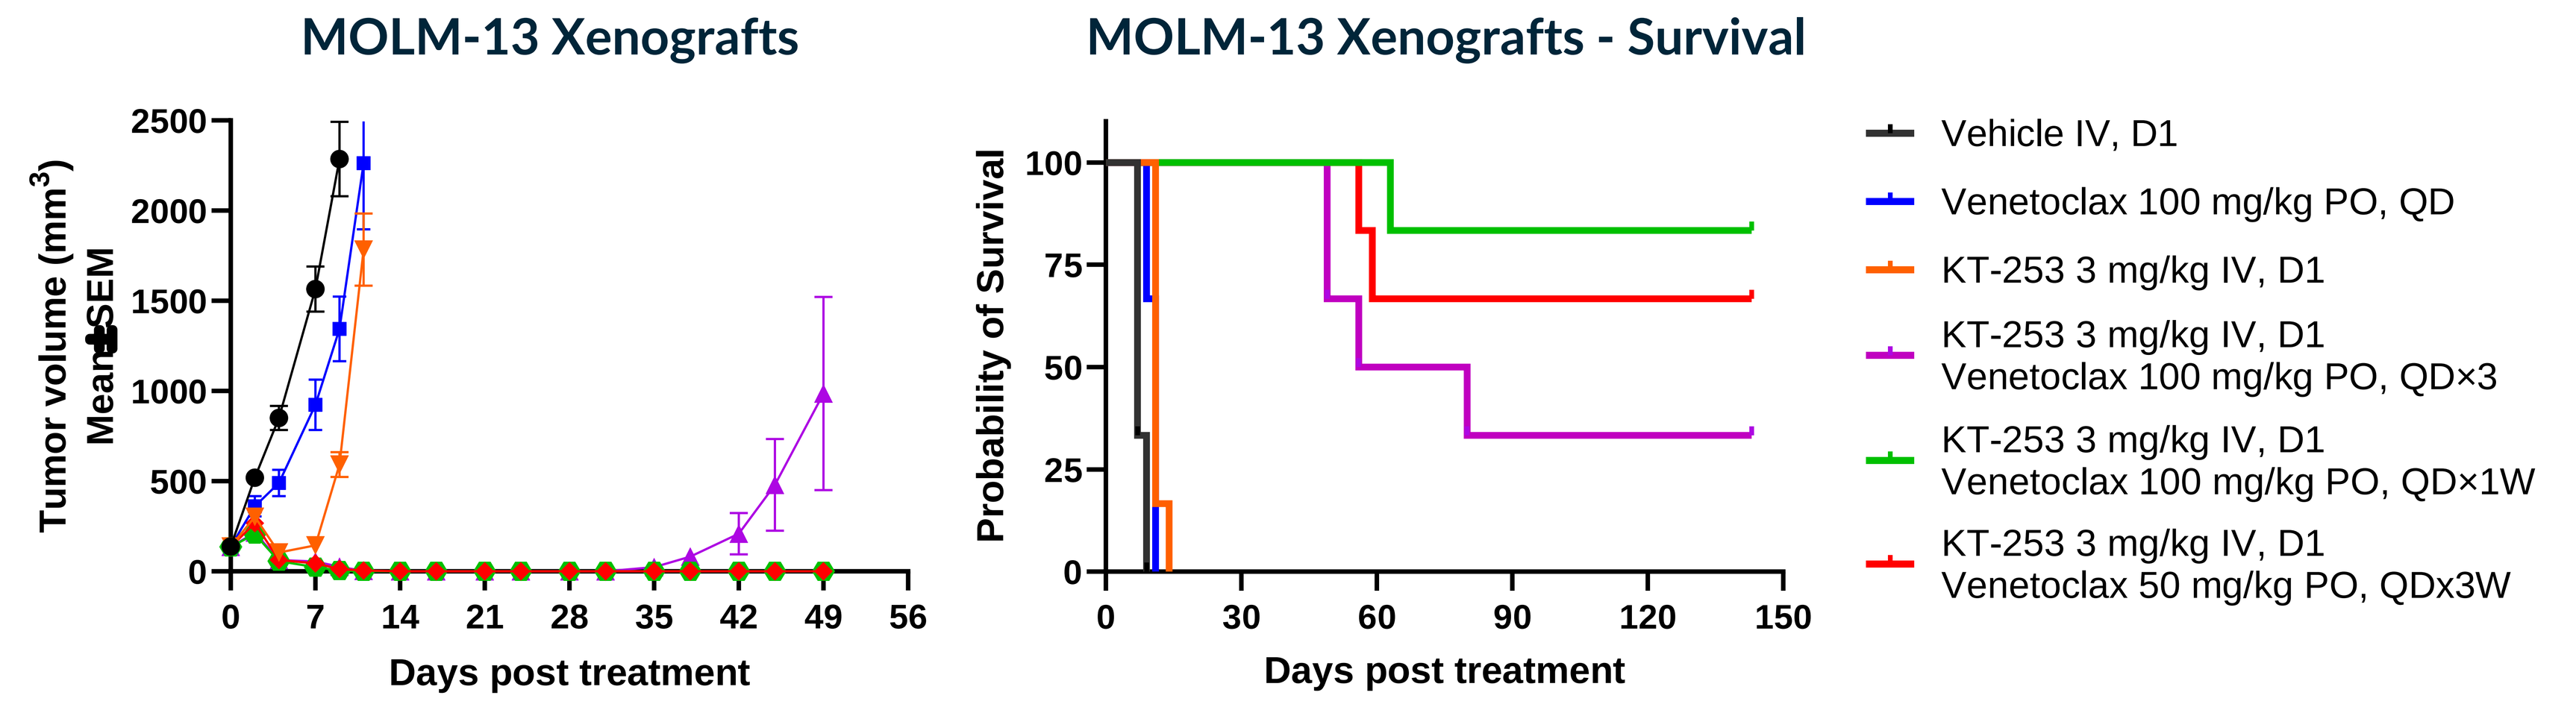
MOLM-13 Xenografts
Tumor volume (mm³) Mean ± SEM
Days post treatment
MOLM-13 Xenografts - Survival
Probability of Survival
Vehicle IV, D1
Venetoclax 100 mg/kg PO, QD
KT-253 3 mg/kg IV, D1
KT-253 3 mg/kg IV, D1 Venetoclax 100 mg/kg PO, QD×3
KT-253 3 mg/kg IV, D1 Venetoclax 100 mg/kg PO, QD×1W
KT-253 3 mg/kg IV, D1 Venetoclax 50 mg/kg PO, QDx3W

- KT-253 in combination with venetoclax enhances apoptotic cell fate commitment in MOLM-13 AML cells
- A single 3 mg/kg dose of KT-253 administered in combination with subclinical 50 mg/kg dose of venetoclax administered daily for three weeks resulted in durable complete responses and 4 of 6 animals remained tumor-free for 150 days

CONCLUSIONS

- KT-253 is a potent MDM2 degrader and a best-in-class p53 stabilizer that inhibits tumor cell growth with picomolar potency and is >200-fold more potent than clinically active MDM2 small molecule inhibitors.
- KT-253 monotherapy results in potent MDM2 degradation leading to robust activation of the p53 pathway, apoptosis and sustained tumor regression in AML and ALL xenograft models
- KT-253 results in anti-leukemic activity in PDX models of systemic AML
- KT-253 shows in vivo combinatorial benefit with SoC agent venetoclax, therefore highlighting potential to address venetoclax resistance in AML
- The 'hit and run' approach with intermittent dosing regimen of KT-253, by allowing time for recovery of normal cells, has the potential to result in improved efficacy and safety profiles compared to the more frequent dosing of MDM2/p53 small molecule inhibitors in the clinic
- KT-253, currently in Phase 1 (NCT05775406), has the potential to provide therapeutic benefit in p53 WT high-grade myeloid malignancies including AML, as well as lymphomas and solid tumor indications that are sensitive to MDM2 degrader mechanism

REFERENCES

1. Hassin and Oren. Drugging p53 in cancer: one protein, many targets. *Nature Reviews Drug Discovery* 2022; 2:127-144.
2. Liu et al. p53 modifications: exquisite decorations of the powerful guardian. *J Mol Cell Biol.* 2019; 7:564–567.
3. McWade and Fischer. TP53. *Encyclopedia of Cancer (3rd edition).* 2019; 483-495.
4. Wade *et al*. MDM2, MDMX and p53 in oncogenesis and cancer therapy. *Nat Rev Cancer* 2013; 13: 83-96.
5. Burgess *et al*. Clinical overview of MDM2/X-targeted therapies. *Front. Oncol.* 2016; 6: 7
6. Jeay *et al*. Dose and schedule determine distinct molecular mechanisms underlying the efficacy of the p53–MDM2 inhibitor HDM201. Cancer Res. 78(21): 6257-6267

DISCLOSURES

All authors are Kymera Therapeutics employees and equity owners.
Mayo, Dixit, Filiatrault, Proctor, Ewesuedo, Schalm are former Kymera employees

Kymera Therapeutics • 200 Arsenal Yards Blvd., Suite 230, Watertown, MA 02472, USA
www.kymeratx.com • Contact: Yogesh Chutake | ychutake@kymeratx.com

10th International MDM2 Workshop
October 15-18, 2023 • Tokyo, Japan