Phase 1 Study of KT-333, a Targeted Protein Degrader of STAT3, in Patients with Relapsed or Refractory Lymphomas, Large Granular Lymphocytic Leukemia, and Solid Tumors

Starodub, Alexander N[1]; Ewesuedo, Reginald[2]; De Savi, Chris[2]; Dey, Joyoti[2]; Agarwal, Sagar[2]; Donohue, Sean[2]; Perea, Rachelle[2]; Klaus, Christine[2]; Gollob, Jared[2]
[1]The Christ Hospital Cancer Center, Cincinnati OH; [2]Kymera Therapeutics, Watertown MA

BACKGROUND

- The signal transducer and activator of transcription 3 (STAT3) protein is aberrantly activated by oncogenic mutations or upstream signaling through cytokines and growth factors, resulting in tumor growth and promotion and hindering antitumor immunity.
- Approximately 70% of human hematological malignancies and solid tumors exhibit increased STAT3 signaling.
- There is evidence of constitutive activation of STAT3 in large granular lymphocytic leukemia (LGL-L), peripheral T-cell lymphoma (PTCL), and cutaneous T-cell lymphomas (CTCL). Hyperactivation of STAT3 has been reported in a variety of solid tumors.
- Selective inhibition of STAT3 has been proven to be difficult with conventional therapeutic approaches.
- Targeted protein degraders represent a new therapeutic class of compounds that utilize the ubiquitin proteasome system to target the degradation of specific proteins.

KT-333

- KT-333 is a potent, highly selective, heterobifunctional small molecule degrader of STAT3.
- In preclinical studies, durable tumor regressions were seen with weekly intravenous (IV) dosing of KT-333 in STAT3-dependent T-cell lymphomas (ALK + ALCL).

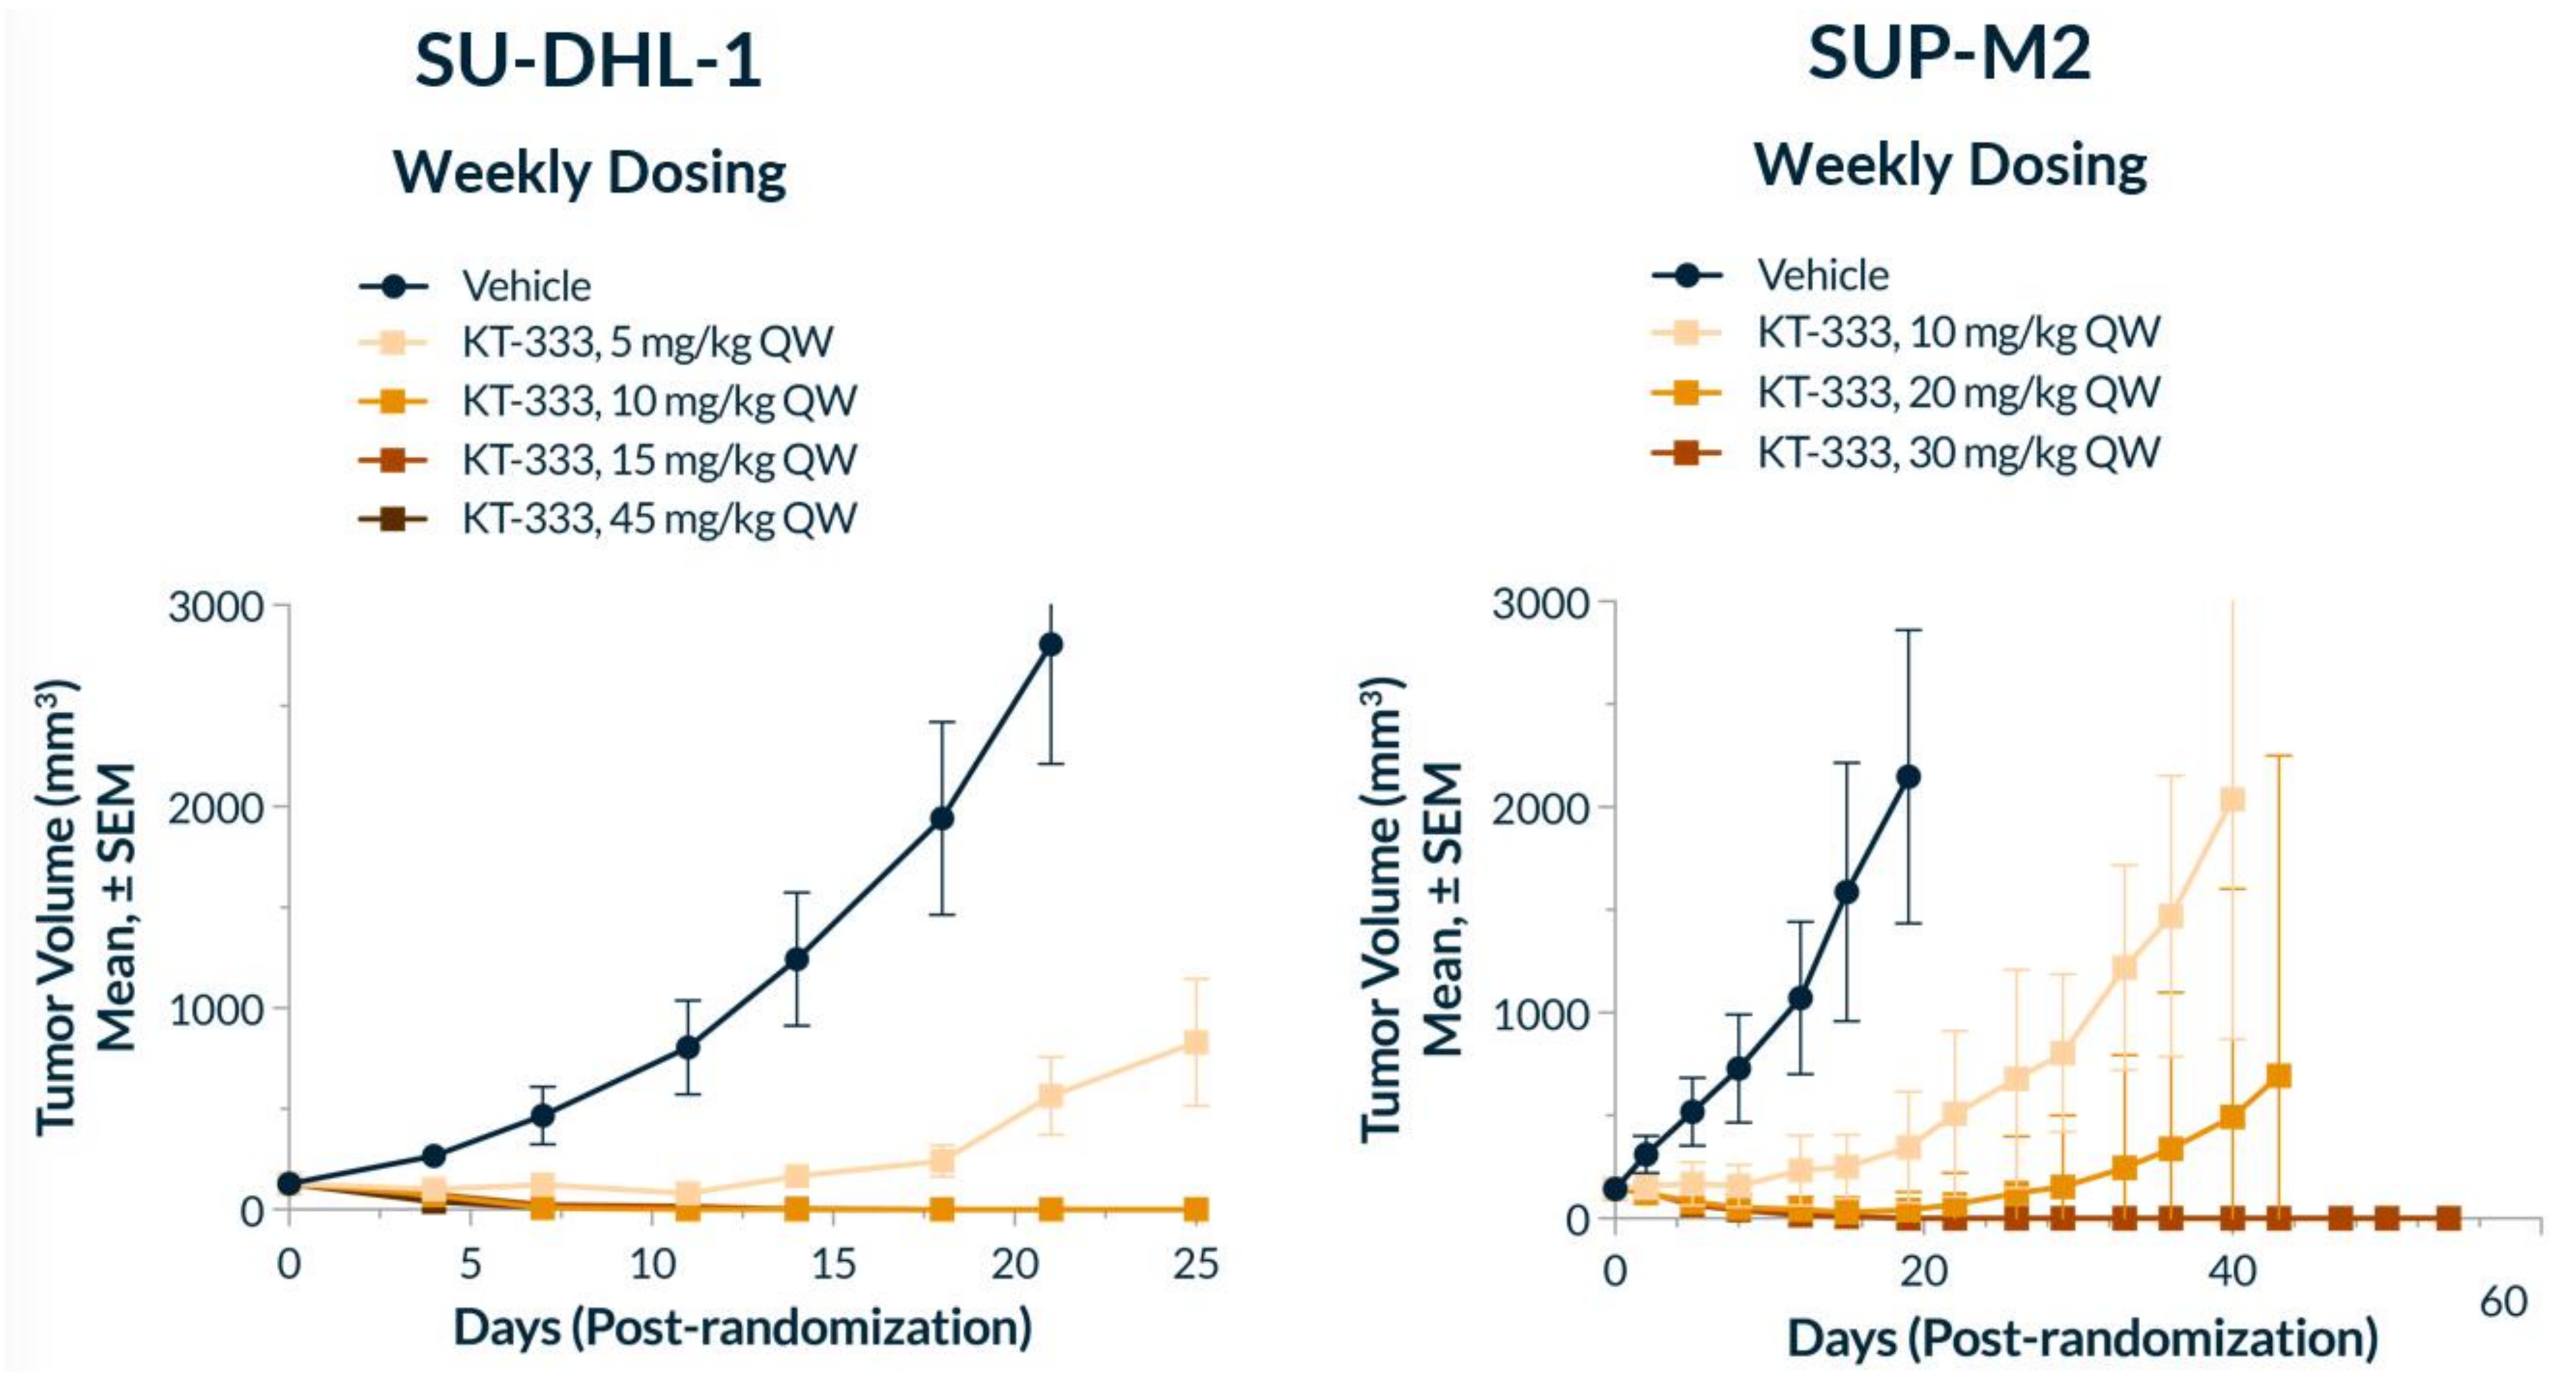
SU-DHL-1
Weekly Dosing
Vehicle
KT-333, 5 mg/kg QW
KT-333, 10 mg/kg QW
KT-333, 15 mg/kg QW
KT-333, 45 mg/kg QW
Tumor Volume (mm³) Mean, ± SEM
0
1000
2000
3000
0
5
10
15
20
25
Days (Post-randomization)
SUP-M2
Weekly Dosing
Vehicle
KT-333, 10 mg/kg QW
KT-333, 20 mg/kg QW
KT-333, 30 mg/kg QW
Tumor Volume (mm³) Mean, ± SEM
0
1000
2000
3000
0
20
40
60
Days (Post-randomization)

METHODS

- This is an open-label, Phase 1a (dose escalation) and Phase 1b (expansion) first-in-human study in patients with relapsed/ refractory (R/R) lymphomas and advanced solid tumors.
- The Phase 1 RP2D determined in R/R lymphoma and solid tumor patients will be evaluated further during Phase 1b in patients with PTCL, CTCL, LGL- L or advanced solid tumors.
- In Phase 1a, ascending doses of IV KT-333 given once weekly in 28-day cycles will be evaluated in an accelerated dose titration design followed by the 3+3 design.
- In Phase 1b, KT-333 will be administered at the RP2D determined during Phase 1a.
- Treatment with KT-333 will continue until disease progression, unacceptable toxicity, or patient refusal.

TRIAL DESIGN

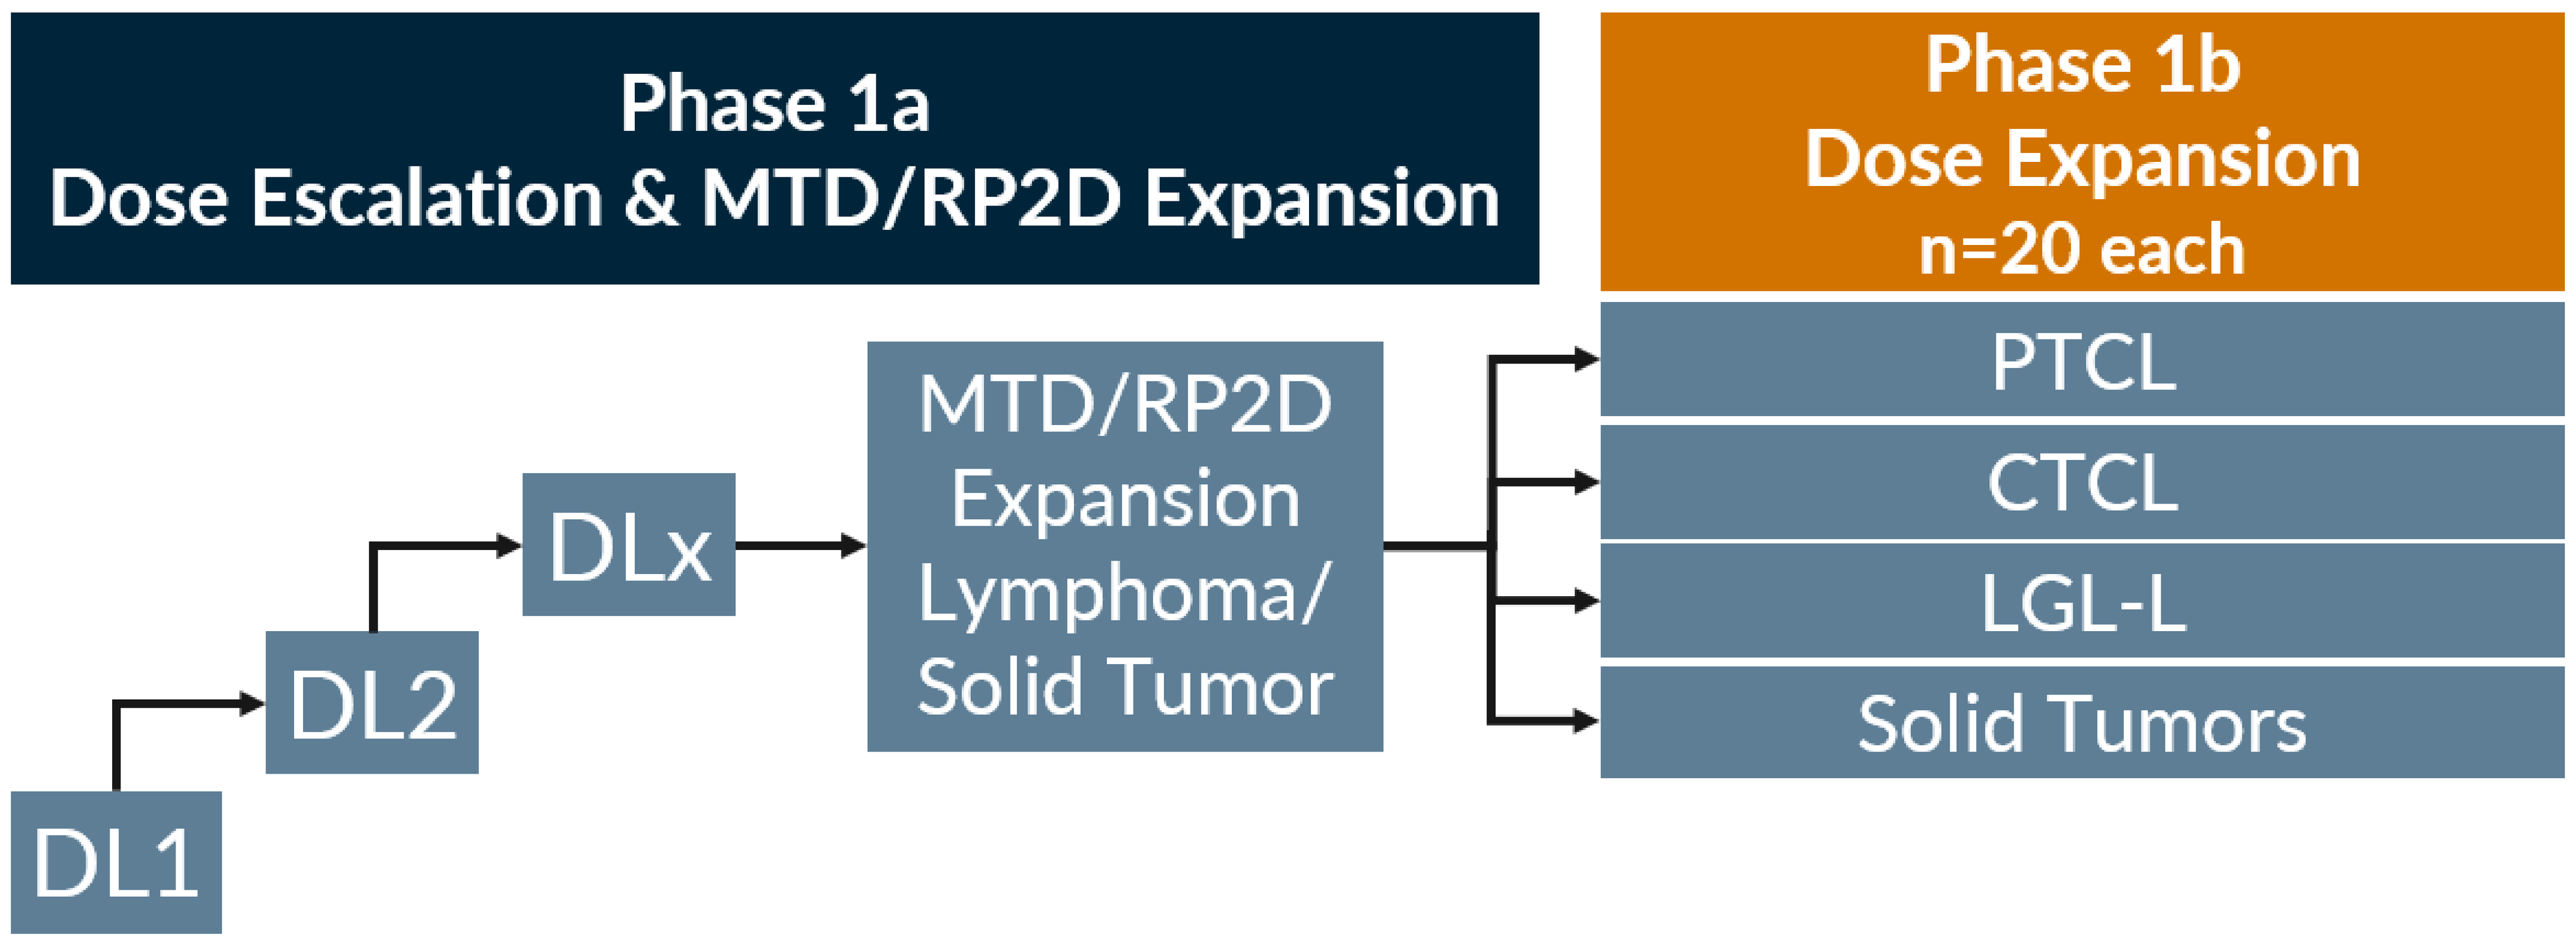
Phase 1a
Dose Escalation & MTD/RP2D Expansion
Phase 1b
Dose Expansion
n=20 each
DL1
DL2
DLx
MTD/RP2D Expansion Lymphoma/ Solid Tumor
PTCL
CTCL
LGL-L
Solid Tumors

Primary Objective: To evaluate safety, tolerability and PK/PD of KT-333 in patients with relapsed or refractory lymphomas, solid tumors, PTCL, CTCL, and LGL-L (Phase 1b only).

Study Endpoints:
Primary: Safety, tolerability, MTD/RP2D (Phase 1a).
Secondary: PK, preliminary efficacy.
Exploratory: Tumor genotyping including STAT3 mutational status; STAT3 pathway gene expression including pSTAT3 expression at baseline, immune TME profiling & correlations thereof with anti-tumor activity.

KEY ELIGIBILITY CRITERIA

Inclusion Criteria

- Phase 1a: Lymphomas (including Hodgkin's, B-cell, T-cell, Small Lymphocytic, or NK-cell Lymphomas) or solid tumors R/R to at least 2 prior systemic treatments or in cases where there is no standard therapy available.
- Phase 1b: PTCL, CTCL, LGL-L (T-cell LGL-L or CLPD-NK) or solid tumors R/R to at least 1 prior systemic treatment or in cases where there is no standard therapy available.
- LGL-L (Phase 1b only): Severe neutropenia $<500/mm^3$, or neutropenia associated with recurrent infection, or symptomatic anemia and/or transfusion-dependent anemia.
- PTCL and Solid Tumors: Measurable disease (Phase 1a allows non-measurable disease for solid tumor patients).
- Eastern Cooperative Oncology Group performance status of 0-2.
- Adequate bone marrow function (except for patients with LGL-L), adequate liver/kidney organ function for all patients.

Exclusion Criteria

- History or suspicion of central nervous system (CNS) metastases.
- Not recovered from clinically significant AEs of previous treatments to a pretreatment baseline or Grade 1.
- Radiation treatment or major surgery requiring general anesthesia within 4 weeks prior to first dose of KT-333.
- Autologous hematopoietic stem cell transplant less than 3 months prior to first dose of study drug.
- Prior allogenic hematopoietic or bone marrow transplant.

Acknowledgments

The study is sponsored by Kymera Therapeutics, which would like to thank the clinical sites as well as the patients and their families for participation in the study.
NCT# NCT05225584

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com