Discovery and targeted mass spectrometry-based proteomics: Enabling technologies advancing IRAK4 protein degrader along the drug discovery pipeline into the clinic

***Eric Kuhn**, Cait Feeney, Susanne Breitkopf, Chris Browne, Yatao Shi, Dirk Walther, Dapeng Chen, Xiaozhang Zheng, Stephanie Skouras, Veronica Campbell, Haojing Rong, Sagar Agarwal, Alice McDonald, Jared Gollob, Anthony Slavin, Matthew Weiss, Chris De Savi, Nello Mainolfi and Kirti Sharma*

KYMERA

MP 386

INTRODUCTION

Targeted Protein Degradation (TPD) is an emerging field that utilizes heterobifunctional compounds to facilitate the degradation of deleterious proteins using the ubiquitin-proteasome system (UPS). Pharmacodynamic analysis using mass spectrometry alleviates the barriers of reagent specificity of antibody-based assays which can be affected by species and matrices measured during preclinical drug development.

Selective degradation of Interleukin-1 receptor associated kinase 4 (IRAK4), which plays a central role in myddosome signaling is being explored for treatment of TLR- and IL-1R-driven inflammatory diseases.

Here, we outline discovery and targeted mass spectrometry workflows to evaluate the selectivity, potency and degradation effectiveness of an IRAK4 degrader *in vitro* and *in vivo* and highlight its impact on key decision-making studies and on monitoring dose effects in on-going clinical trials.

Proteome Editing with Targeted Protein Degradation

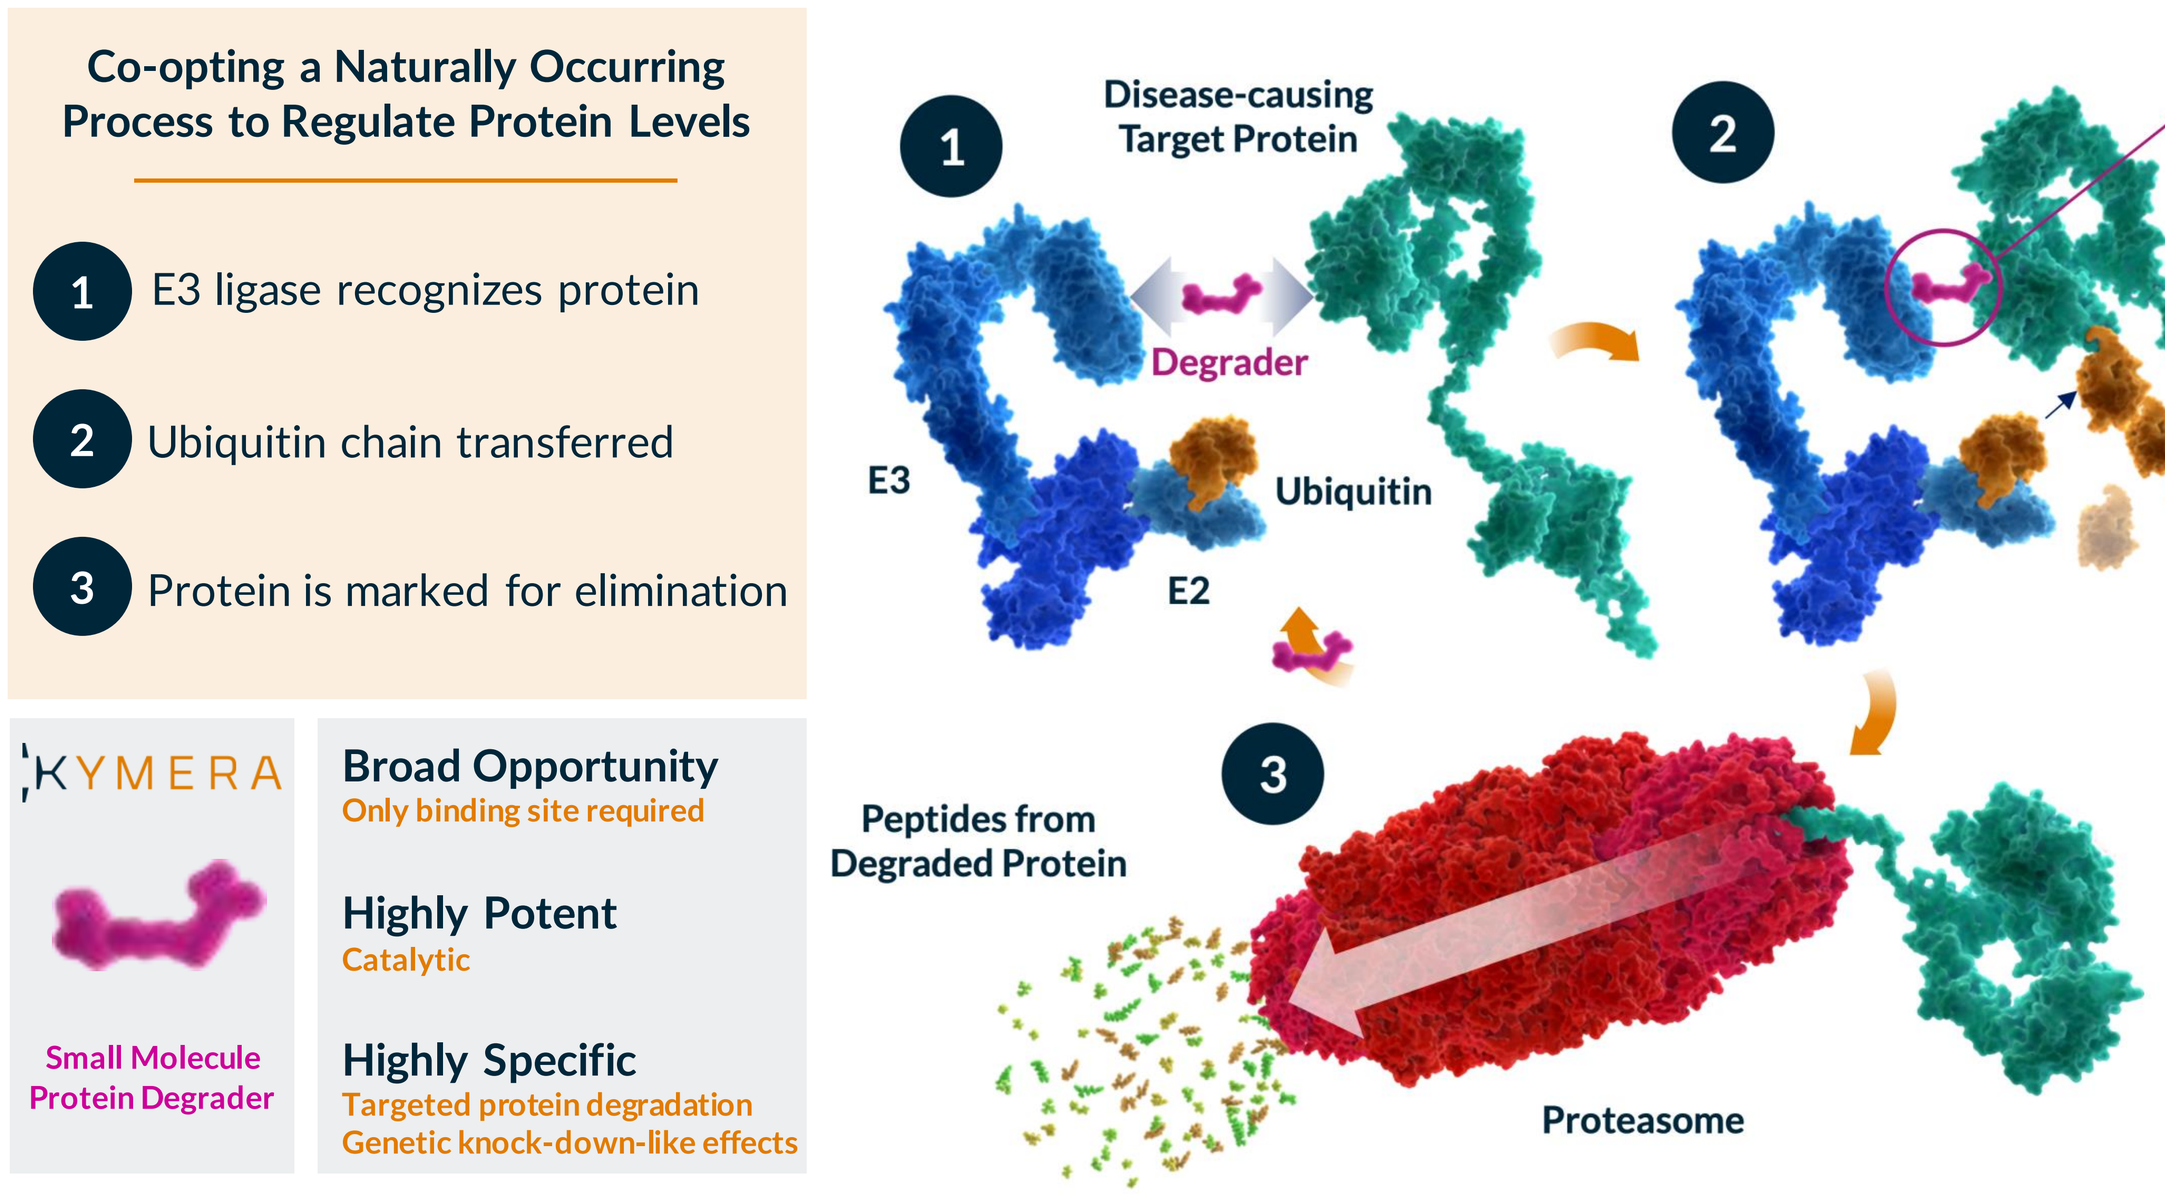
Co-opting a Naturally Occurring Process to Regulate Protein Levels
1 E3 ligase recognizes protein
2 Ubiquitin chain transferred
3 Protein is marked for elimination
KYMERA
Small Molecule Protein Degrader
Broad Opportunity
Only binding site required
Highly Potent
Catalytic
Highly Specific
Targeted protein degradation
Genetic knock-down-like effects
Disease-causing Target Protein
Degrader
E3
Ubiquitin
E2
Targeted Small Molecule Protein Degraders
Ubiquitin Chain
Peptides from Degraded Protein
Proteasome

Targeting IRAK4: De-risking Human Genetics, the Degrader Advantage and Clinical Opportunities

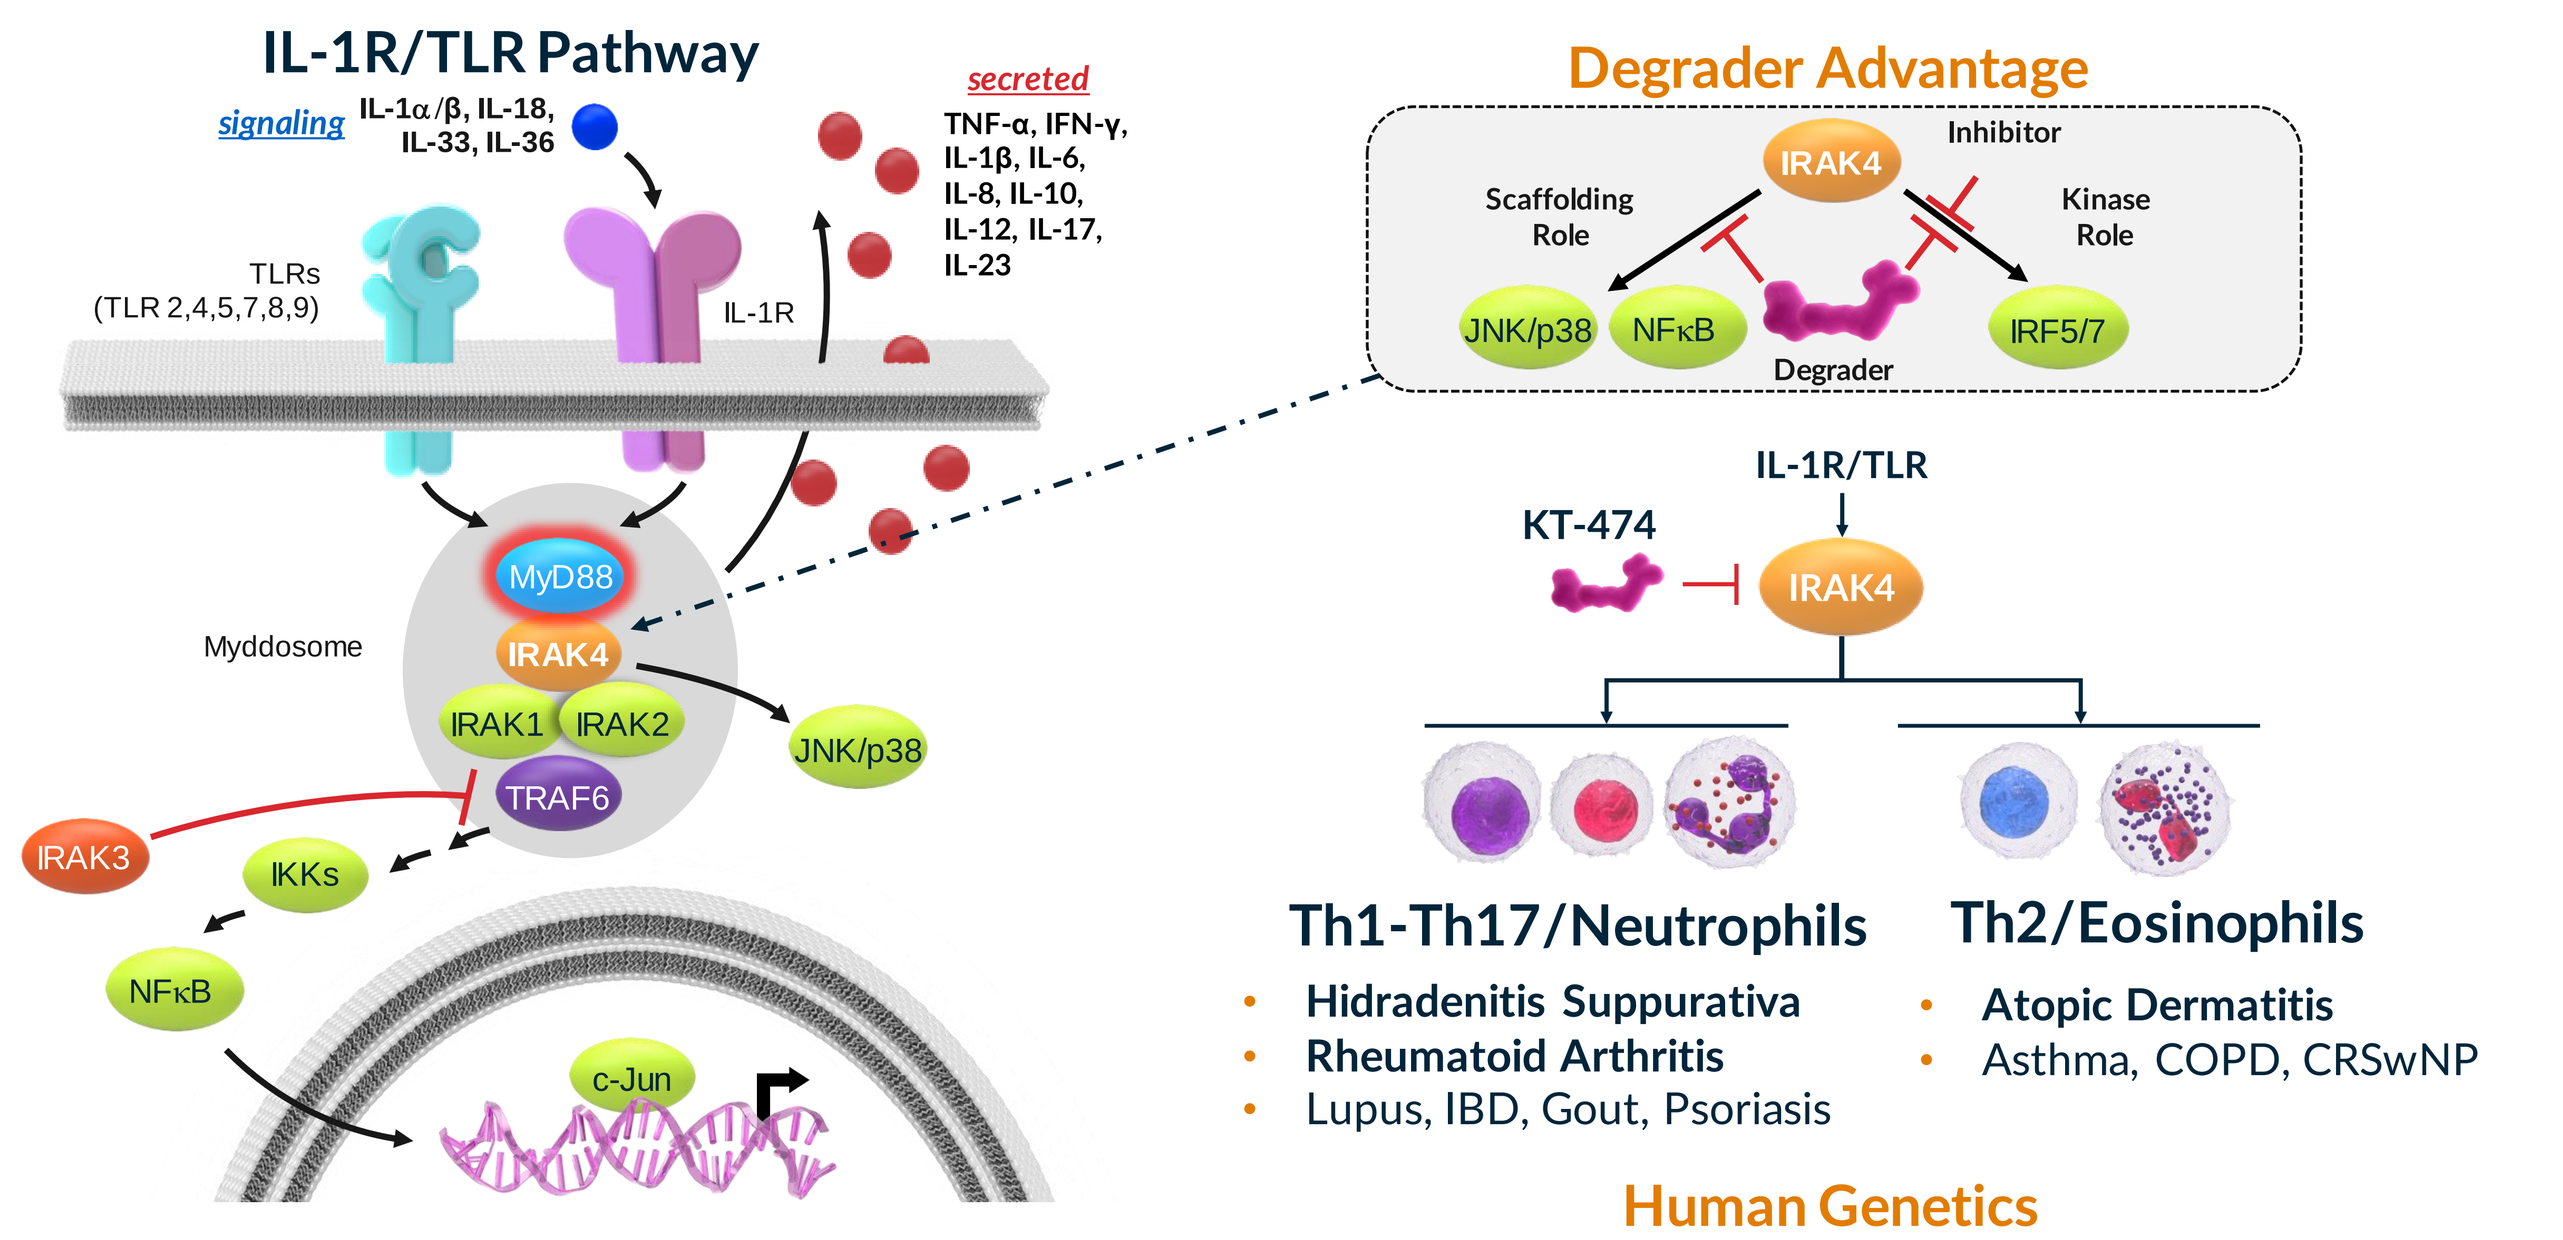
IL-1R/TLR Pathway
signaling
IL-1α/β, IL-18, IL-33, IL-36
secreted
TNF-α, IFN-γ, IL-1β, IL-6, IL-8, IL-10, IL-12, IL-17, IL-23
TLRs (TLR 2,4,5,7,8,9)
IL-1R
Myddosome
MyD88
IRAK4
IRAK1
IRAK2
TRAF6
JNK/p38
IRAK3
IKKs
NFκB
c-Jun
Degrader Advantage
Inhibitor
Scaffolding Role
Kinase Role
JNK/p38
NFκB
IRF5/7
Degrader
IL-1R/TLR
KT-474
IRAK4
Th1-Th17/Neutrophils
Hidradenitis Suppurativa
Rheumatoid Arthritis
Lupus, IBD, Gout, Psoriasis
Th2/Eosinophils
Atopic Dermatitis
Asthma, COPD, CRSwNP
Human Genetics
Adult humans with IRAK4 Null Mutation are healthy

IRAK4 degrader has potential to achieve a broad, well-tolerated anti-inflammatory effect, providing multiple development opportunities in chronic inflammatory diseases

Target Qualification | Hit to Lead | Lead Optimization | Preclinical Development | Clinical Development

Figure 1: Discovery and Targeted Proteomic workflows for proteomic profiling, mechanism of action, biomarker and pharmacodynamic analysis

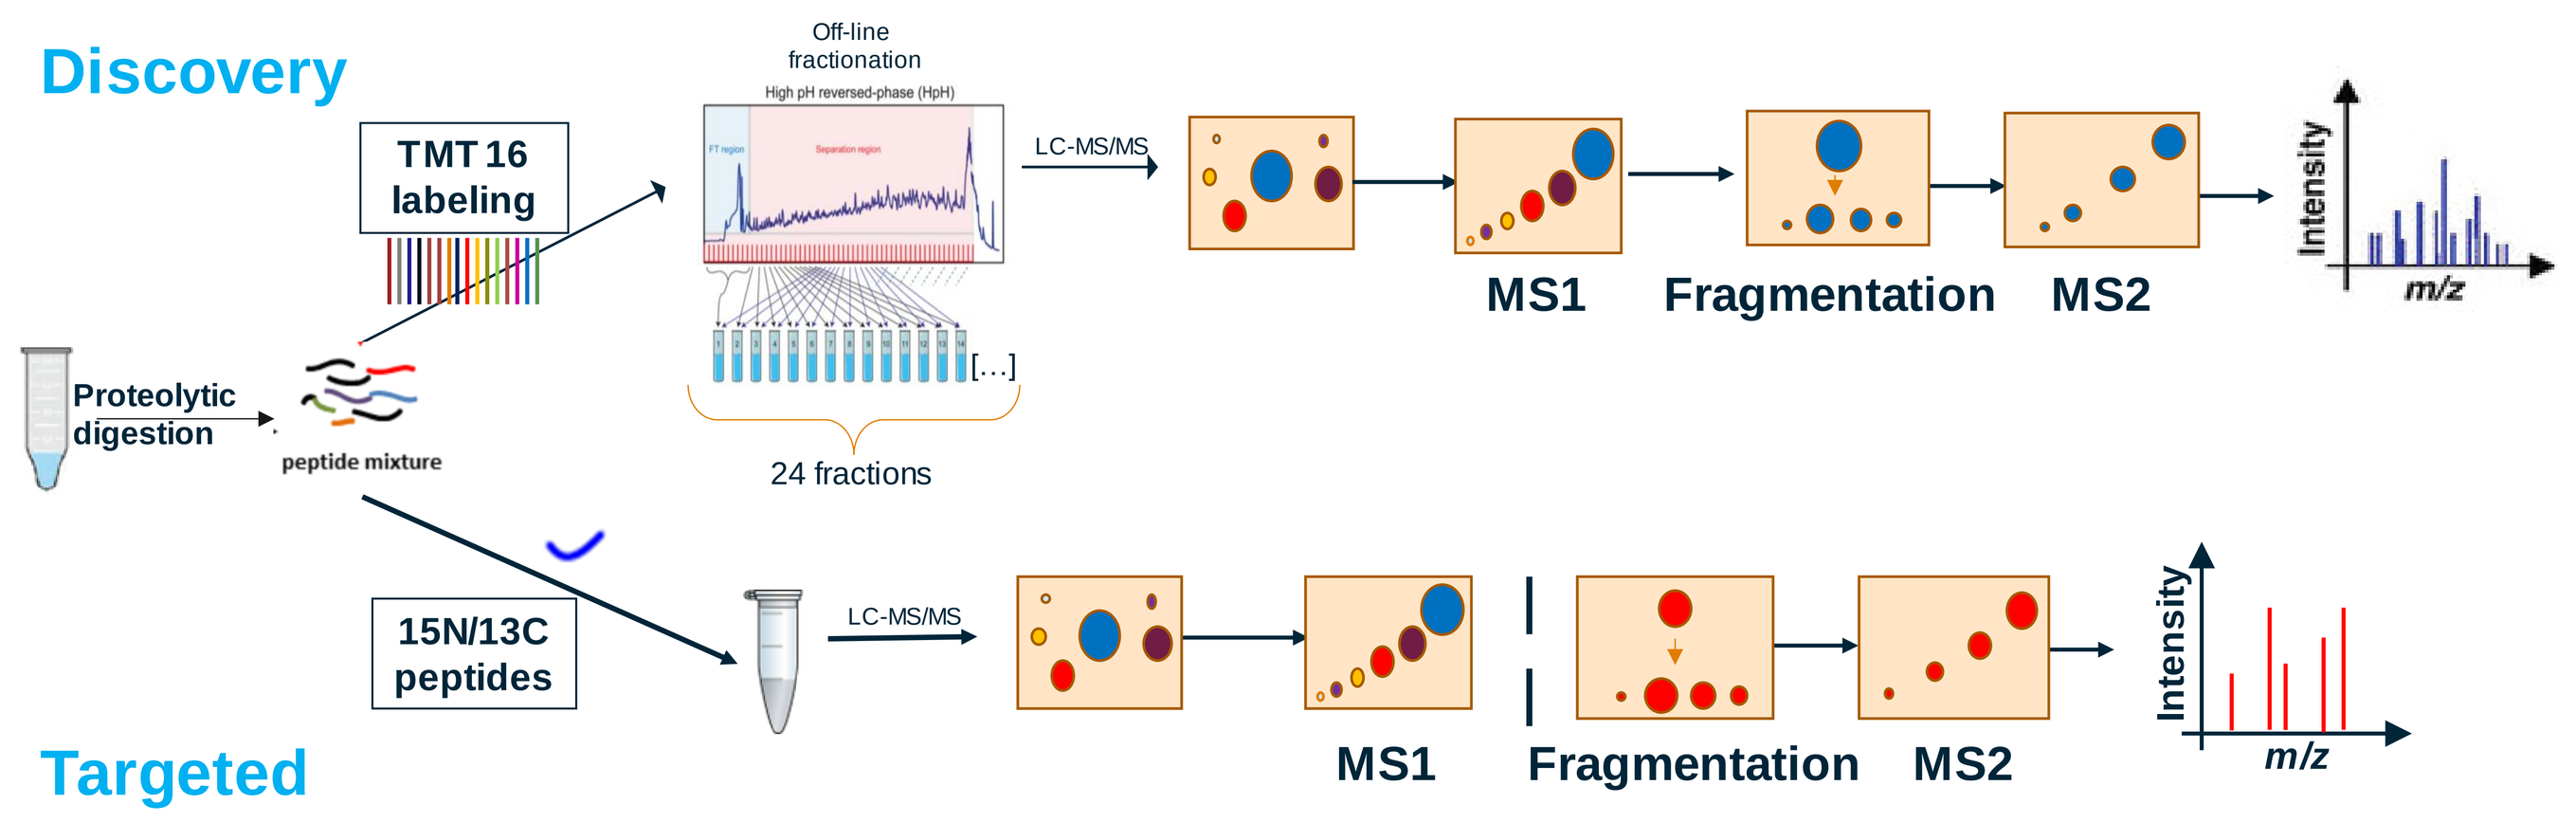
Discovery
TMT 16 labeling
Off-line fractionation
Proteolytic digestion
peptide mixture
24 fractions
LC-MS/MS
MS1
Fragmentation
MS2
Intensity
m/z
15N/13C peptides
Targeted

Discovery Proteomics: unbiased deep dive stable isotope labeling off-line fractionation process to quantify global proteome profiles during targeted protein degradation.

Targeted Proteomics: quantitative, precise, direct measurement of selected proteins with sensitivity and precision to accurately assess specific target protein degradation effects.

Figure 2: Discovery Proteomics identified E3 ligase related off-target activity

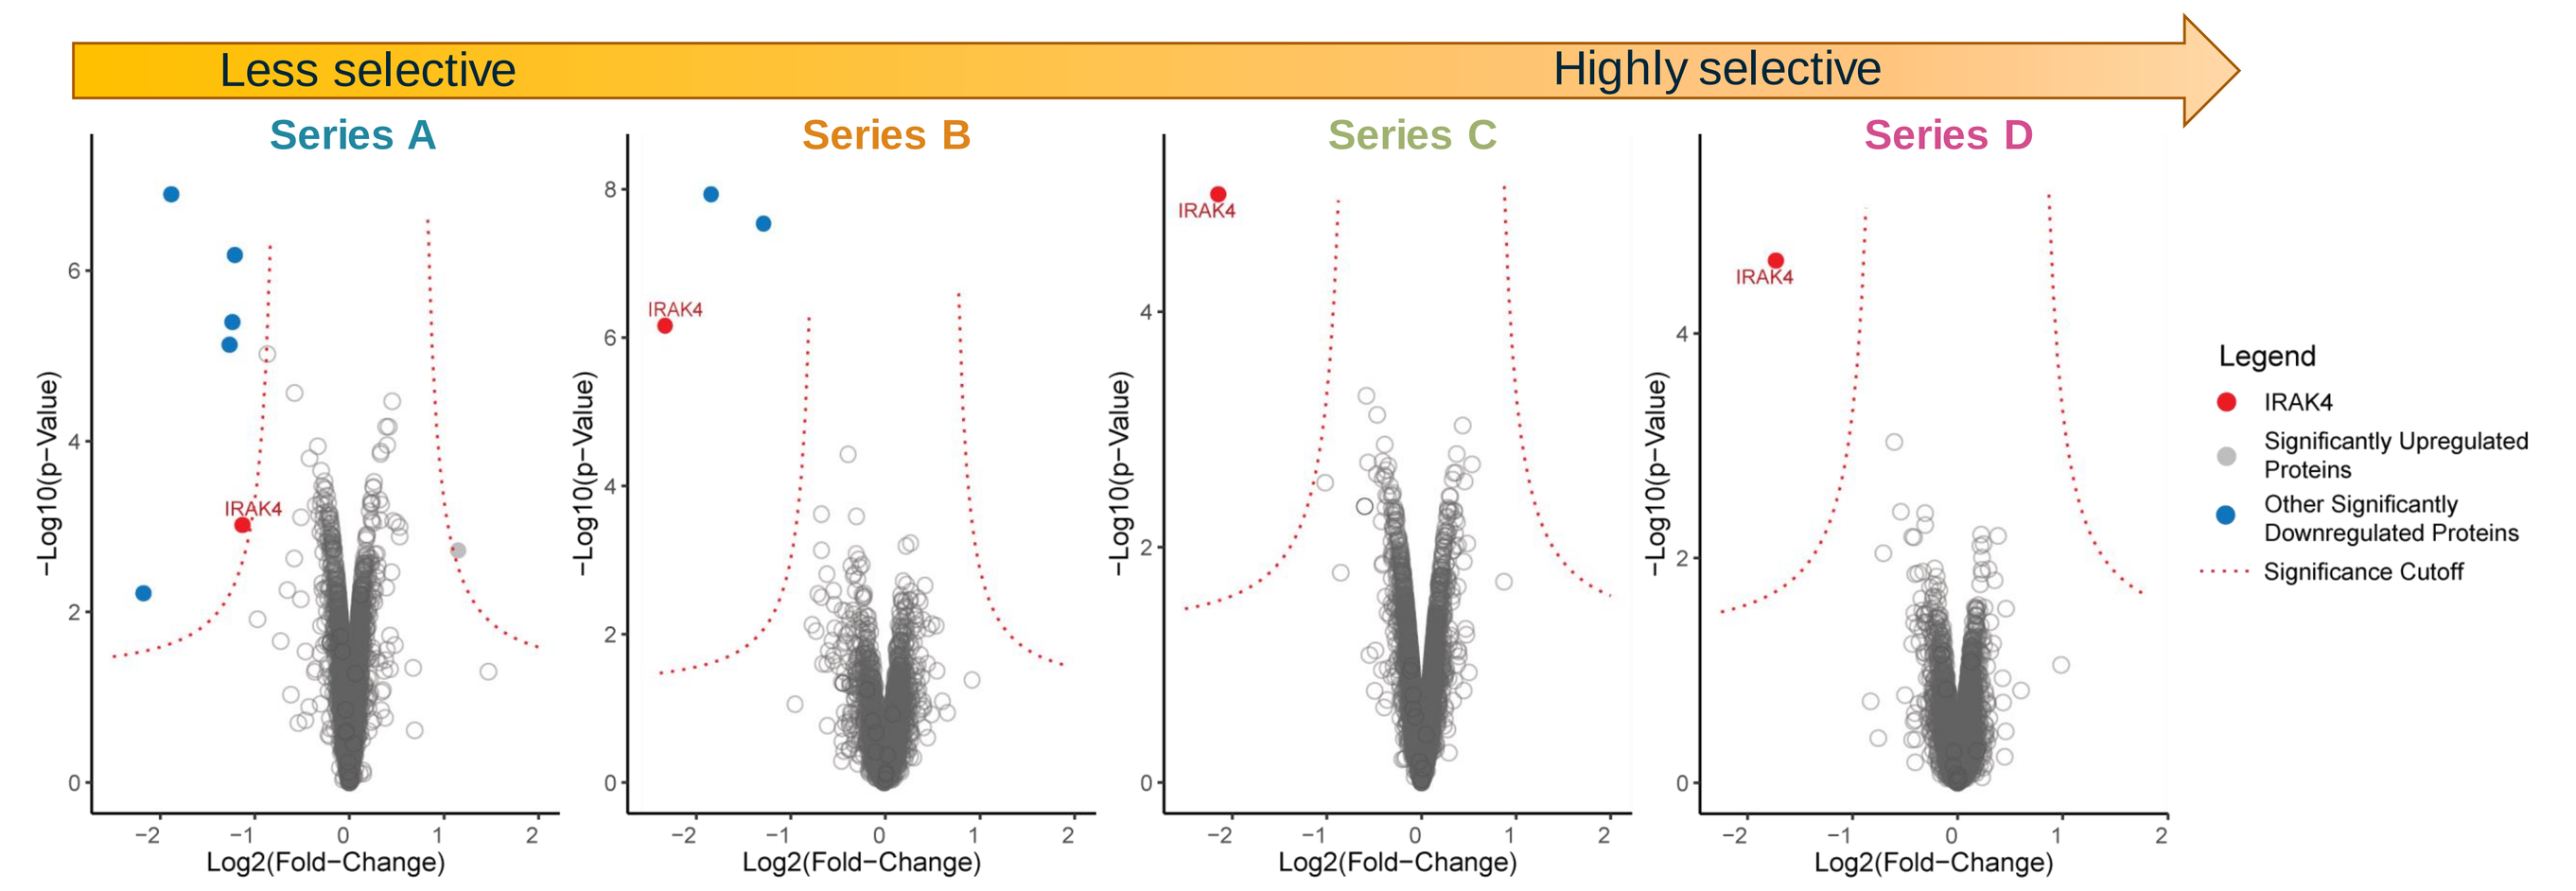
Less selective
Highly selective
Series A
Series B
Series C
Series D
−Log10(p−Value)
Log2(Fold−Change)
IRAK4
Legend
IRAK4
Significantly Upregulated Proteins
Other Significantly Downregulated Proteins
Significance Cutoff

Global Proteomics identified IRAK4 degraders that were highly selective for IRAK4 over the rest of the hPBMC proteome.

Figure 3: Targeted MS assays developed using stable isotope labeled peptides spiked into digested PBMC

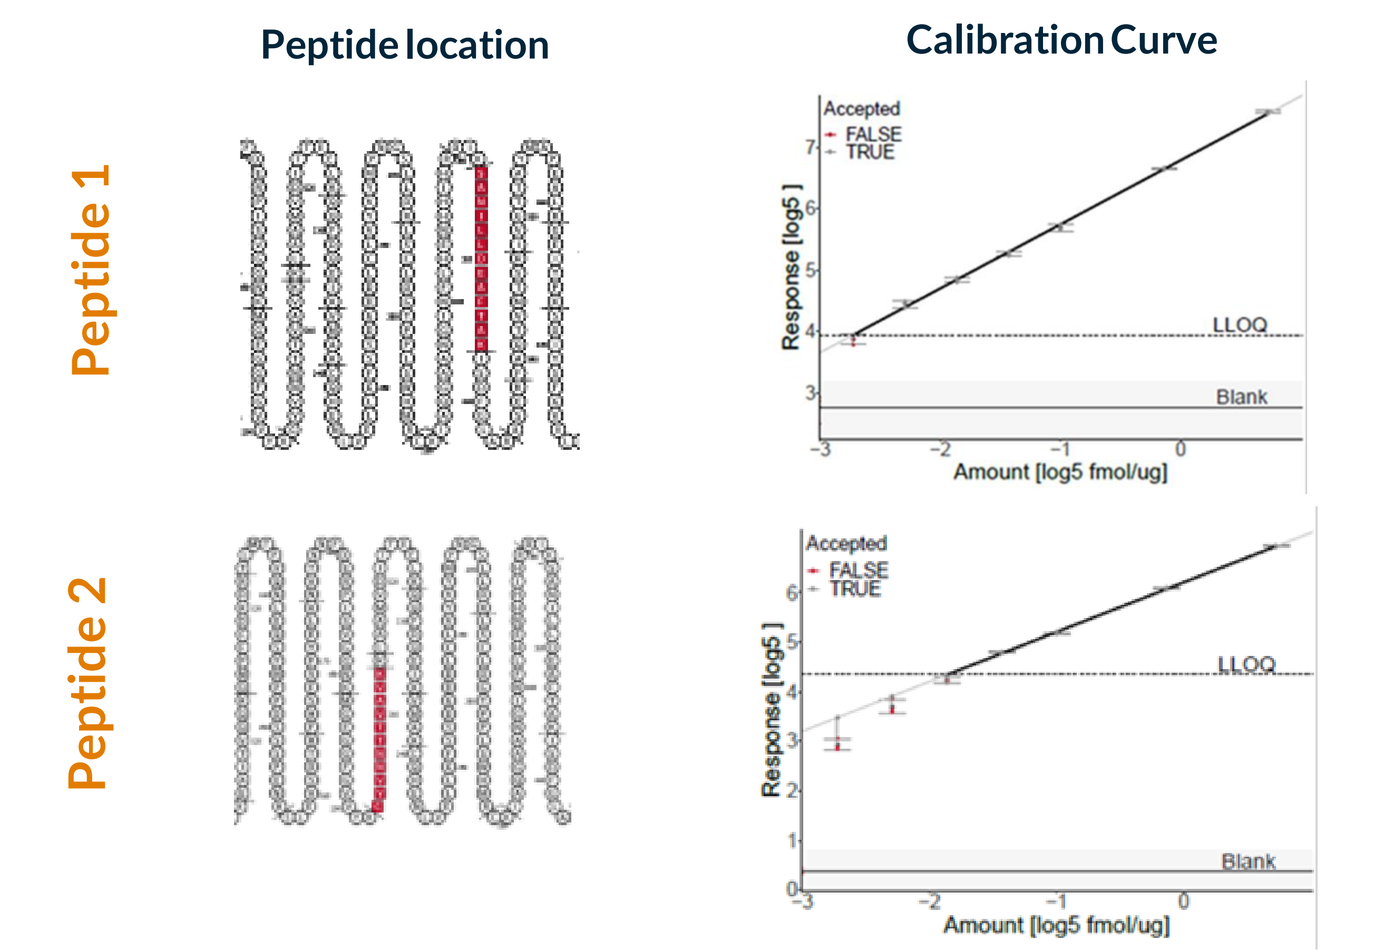
Peptide location
Calibration Curve
Peptide 1
Peptide 2
Response [log5]
Amount [log5 fmol/ug]
LLOQ
Blank
Accepted
FALSE
TRUE

Assay Characterization Results

Peptide 1

Parameter	Value	Unit
LLOQ	0.0125	fmol/ug
Average CV > LLOQ	6.94	%
Minimal Tested Concentration	0.0125	fmol/ug
Maximal Tested Concentration	3.2	fmol/ug
R^2	0.998	
Slope (Linear Range)	1.04	
Intercept (Linear Range)	6.79	

Peptide 2

Parameter	Value	Unit
LLOQ	0.05	fmol/ug
Average CV > LLOQ	3.87	%
Minimal Tested Concentration	0.0125	fmol/ug
Maximal Tested Concentration	3.2	fmol/ug
R^2	0.999	
Slope (Linear Range)	1	
Intercept (Linear Range)	6.21	

Figure 4: Targeted Proteomics enabled assessment of IRAK4 degradation in skin and PBMC for preclinical and clinical studies

A. Protocol established for PK & PD from a single sample of patient blood & skin

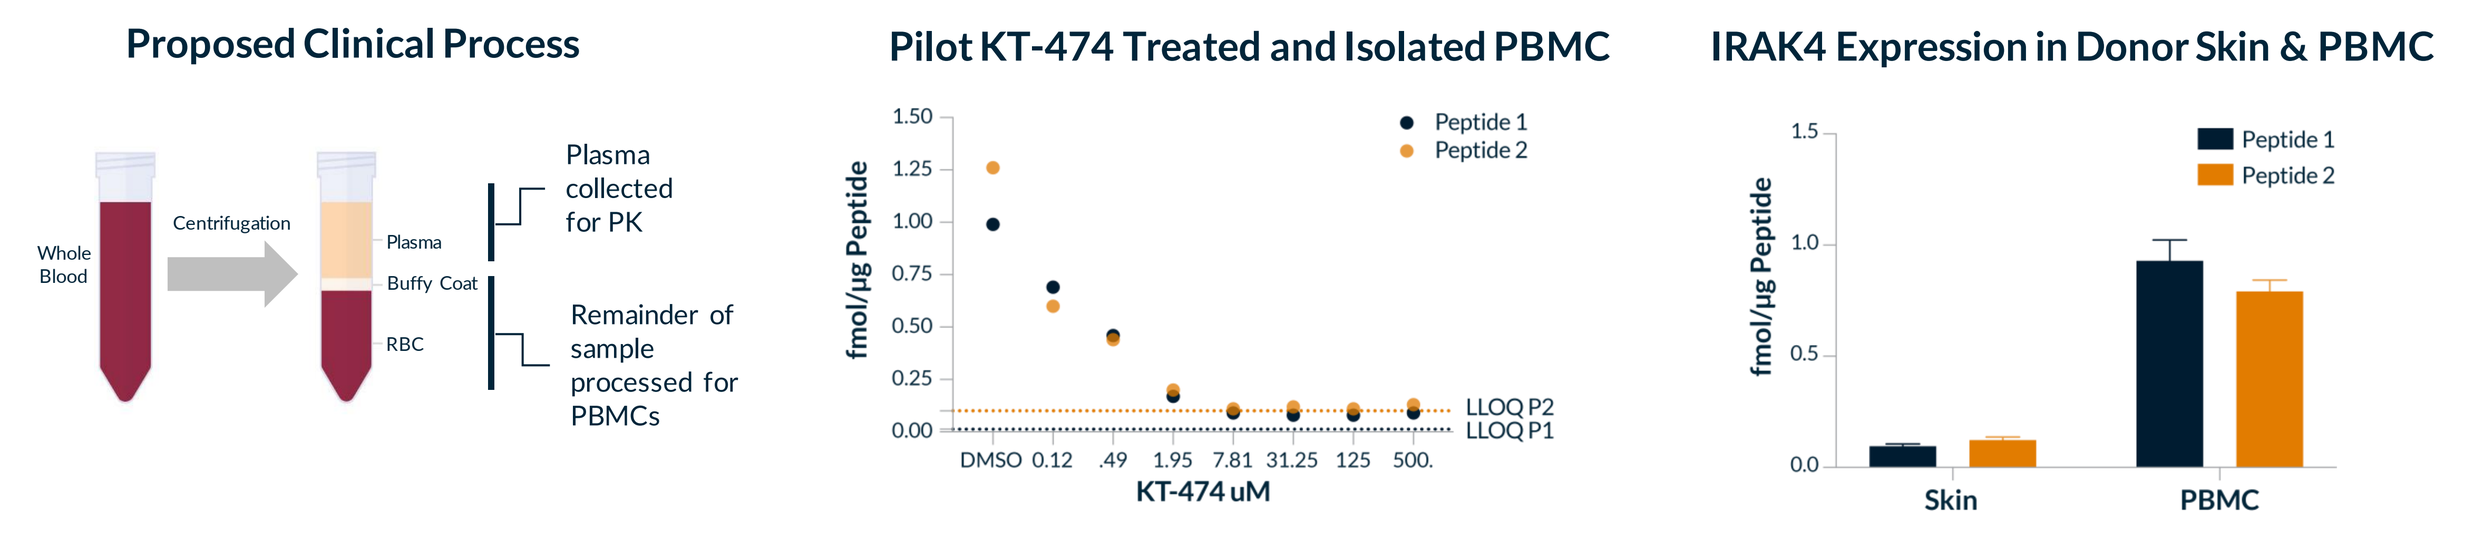
Proposed Clinical Process
Whole Blood
Centrifugation
Plasma
Buffy Coat
RBC
Plasma collected for PK
Remainder of sample processed for PBMCs
Pilot KT-474 Treated and Isolated PBMC
fmol/μg Peptide
KT-474 uM
Peptide 1
Peptide 2
LLOQ P2
LLOQ P1
IRAK4 Expression in Donor Skin & PBMC
Skin
PBMC

B. Upregulated IRAK4 expression in Hidradenitis Suppurativa patient skin lesions

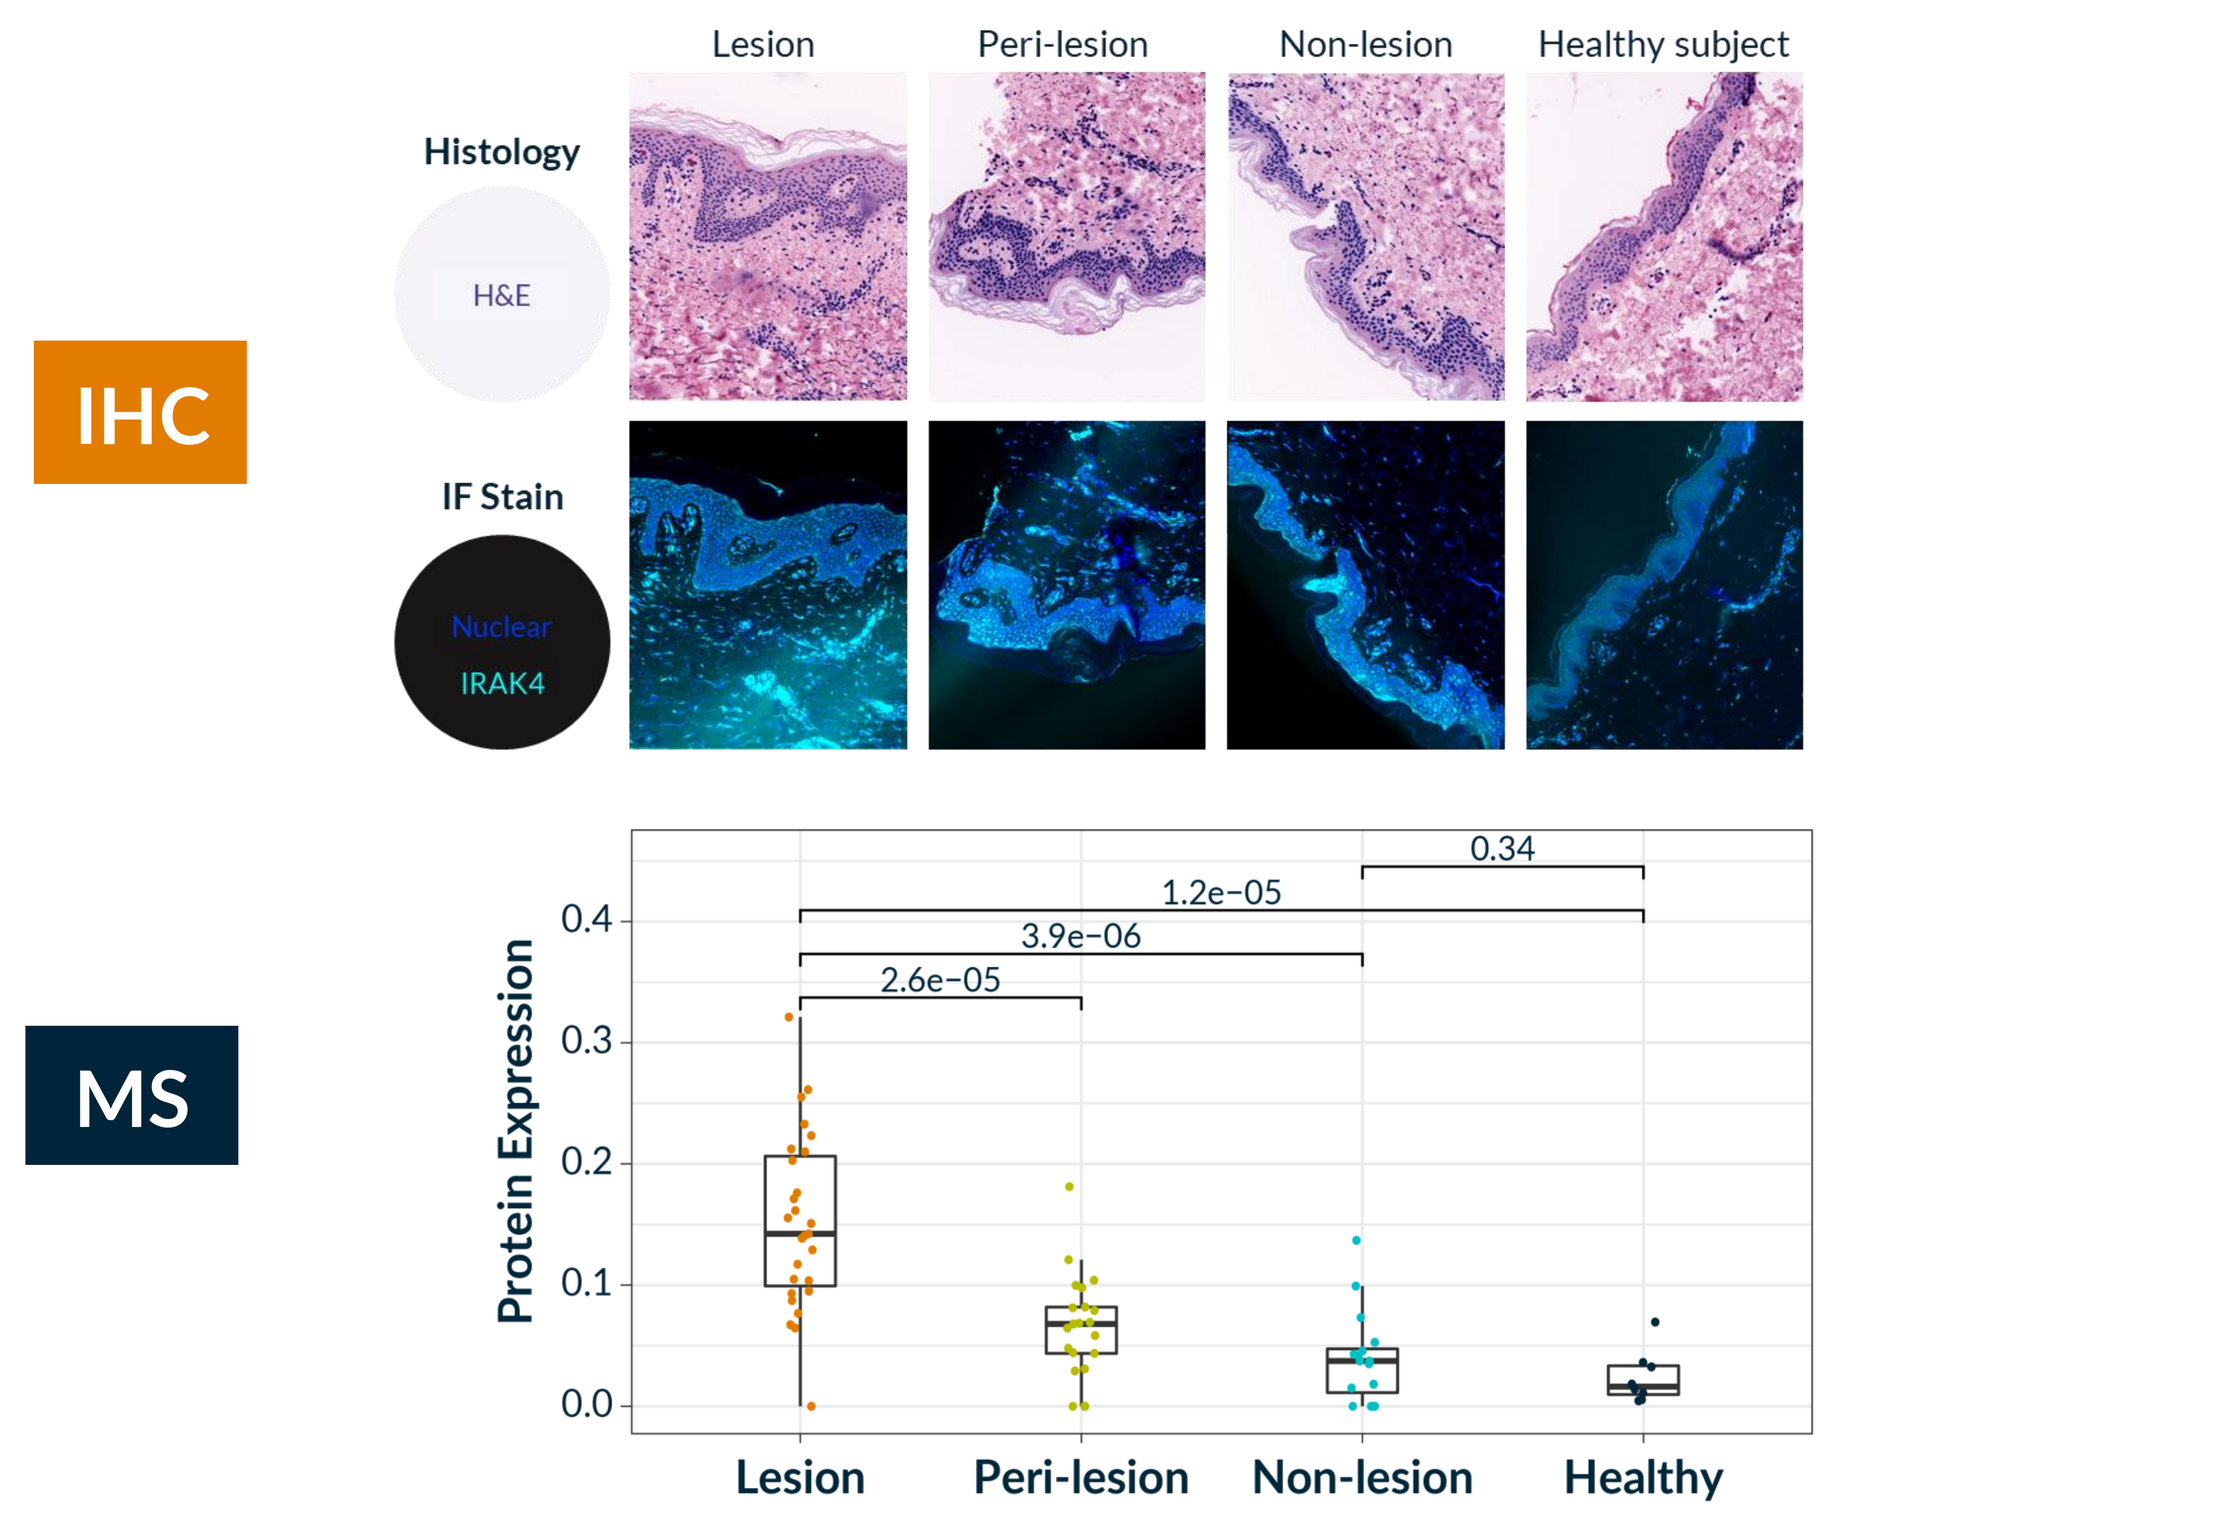
IHC
Lesion
Peri-lesion
Non-lesion
Healthy subject
Histology
H&E
IF Stain
Nuclear
IRAK4
MS
Protein Expression
2.6e−05
3.9e−06
1.2e−05
0.34
Lesion
Peri-lesion
Non-lesion
Healthy

C. Systematic IRAK4 degradation down to assay detection limits is well tolerated in preclinical non-rodent model → project doses in DRF and human studies

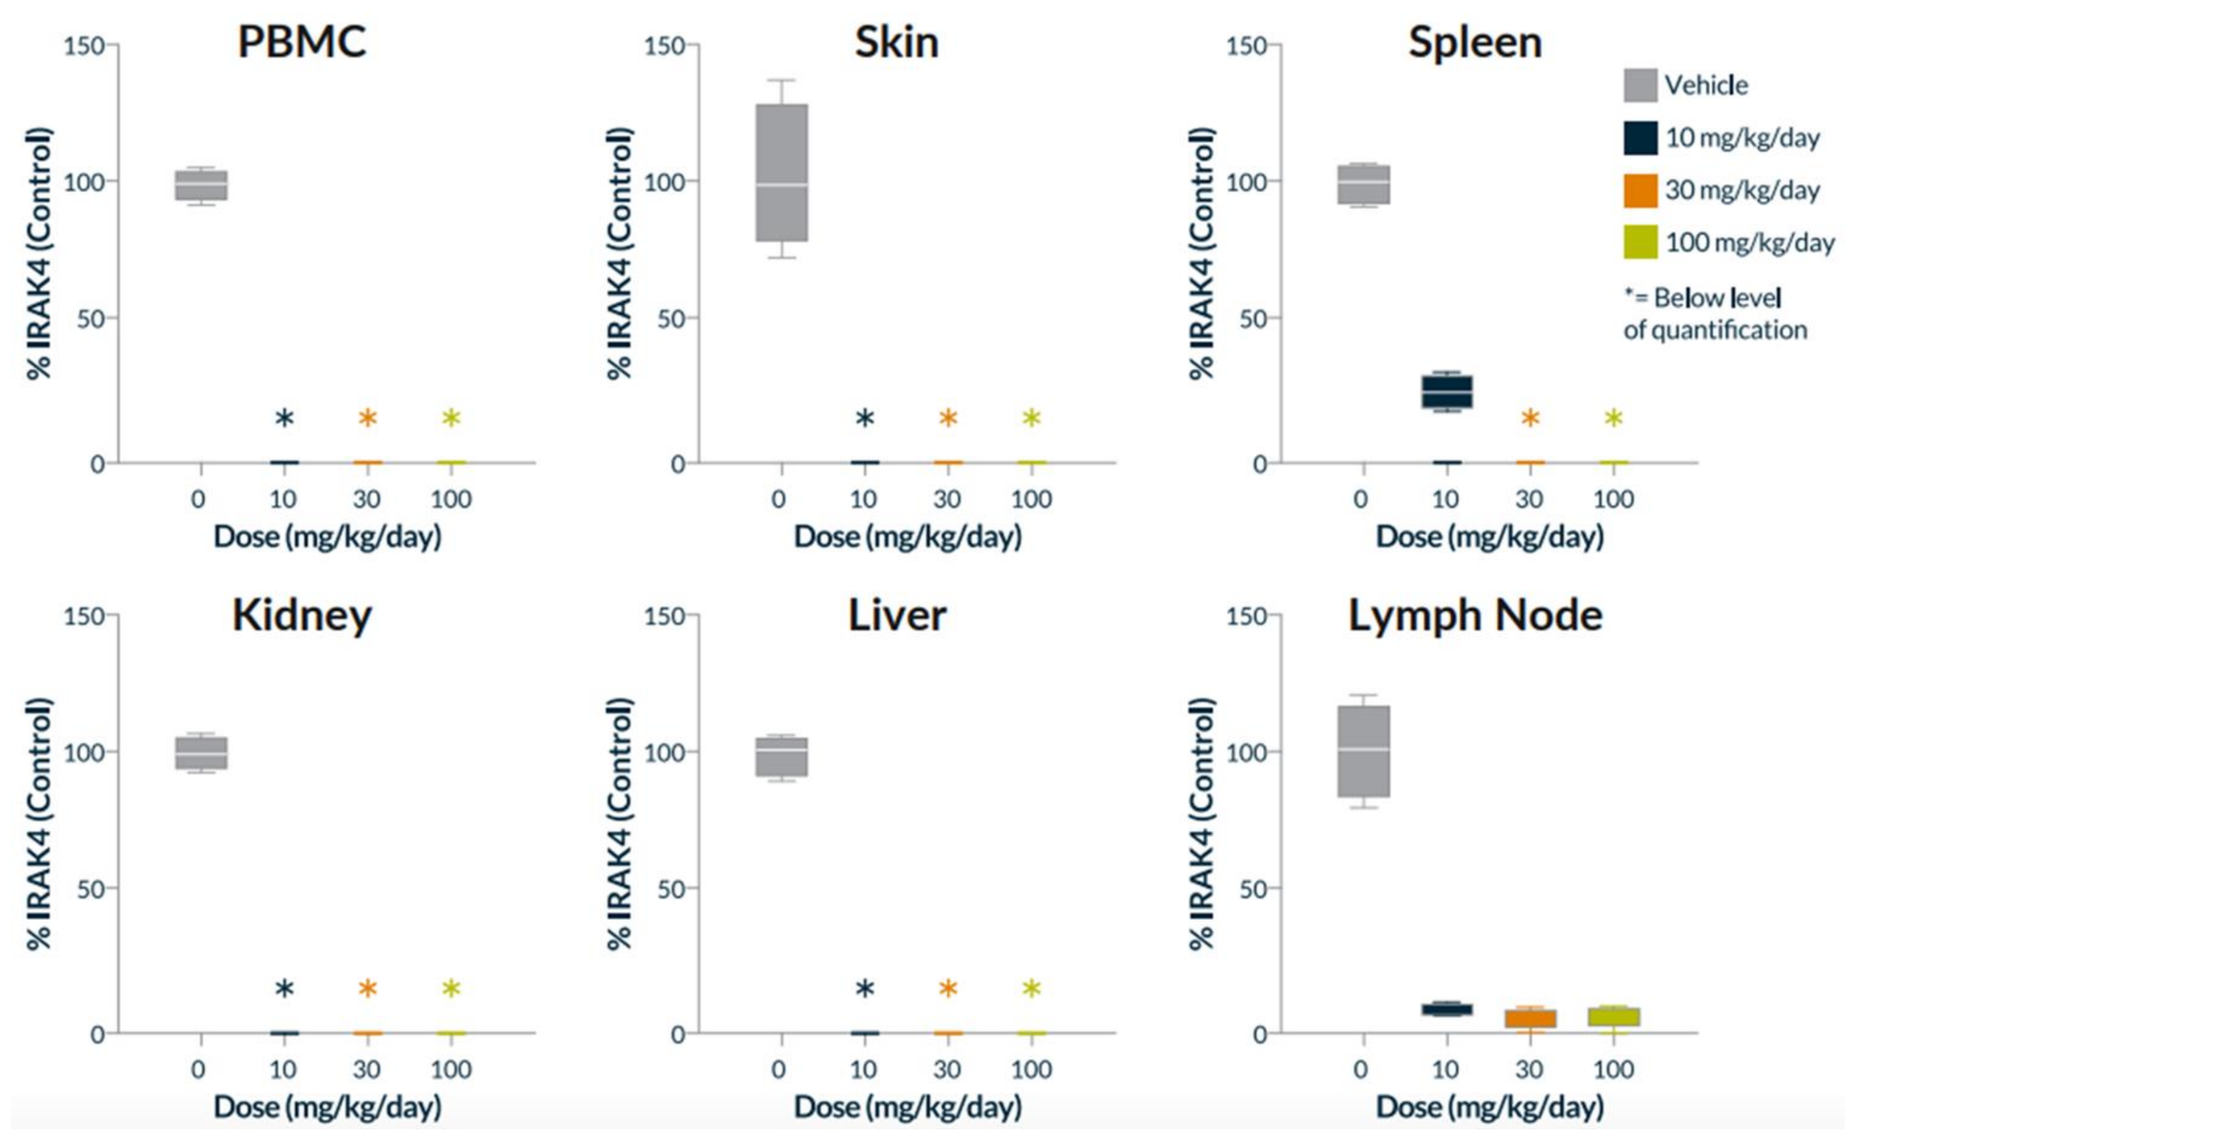
PBMC
Skin
Spleen
Kidney
Liver
Lymph Node
% IRAK4 (Control)
Dose (mg/kg/day)
Vehicle
10 mg/kg/day
30 mg/kg/day
100 mg/kg/day
*= Below level of quantification

Figure 5: KT-474 / SAR444656 Updated Phase 1 Trial Design Includes a 28 Day Patient Study

Double-blind, Placebo-controlled, Single Ascending Dose (SAD) and Multiple Ascending Dose (MAD) in HV; Open Label Patient Cohort in HS and AD Patients

Parts A & B — Healthy Volunteers SAD and MAD

7 SAD cohorts
- 8 subjects per cohort (6:2 randomization)
- 57 adult healthy subjects dosed

Single dose (25-1600 mg)

4 MAD cohorts
- 12 subjects per cohort (9:3 randomization)
- 48 adult healthy subjects dosed

14x daily doses (25-200 mg)

Primary
- Safety & tolerability

Secondary/ Exploratory
- Pharmacokinetic measures (half-life, bioavailability)
- IRAK4 knockdown in PBMC and skin (MAD only)
- *Ex vivo* response of whole blood to TLR agonists (SAD & MAD)

Part C — HS and AD Patients

1 cohort
Up to 20 HS and AD patients

Dose equiv. of 100 mg (fed state) *new*

Open-label

28x daily doses *new*

Primary
- Safety & tolerability

Secondary/ Exploratory
- Pharmacokinetic measures (half-life, bioavailability)
- IRAK4 knockdown in PBMC and skin
- Change in systemic inflammatory biomarkers and proinflammatory gene transcripts in skin
- *Ex vivo* response of whole blood to TLR agonists
- Clinical endpoints: EASI (AD), Total AN Count (HS), symptom scores and global assessments *new*

ClinicalTrials.gov Identifier: NCT04772885

Figure 6: KT-474 / SAR444656 Achieved Deep and Dose-Dependent IRAK4 Degradation after Single Oral Doses Lasting for at Least 6 Days

1 SAD Portion Healthy Volunteers

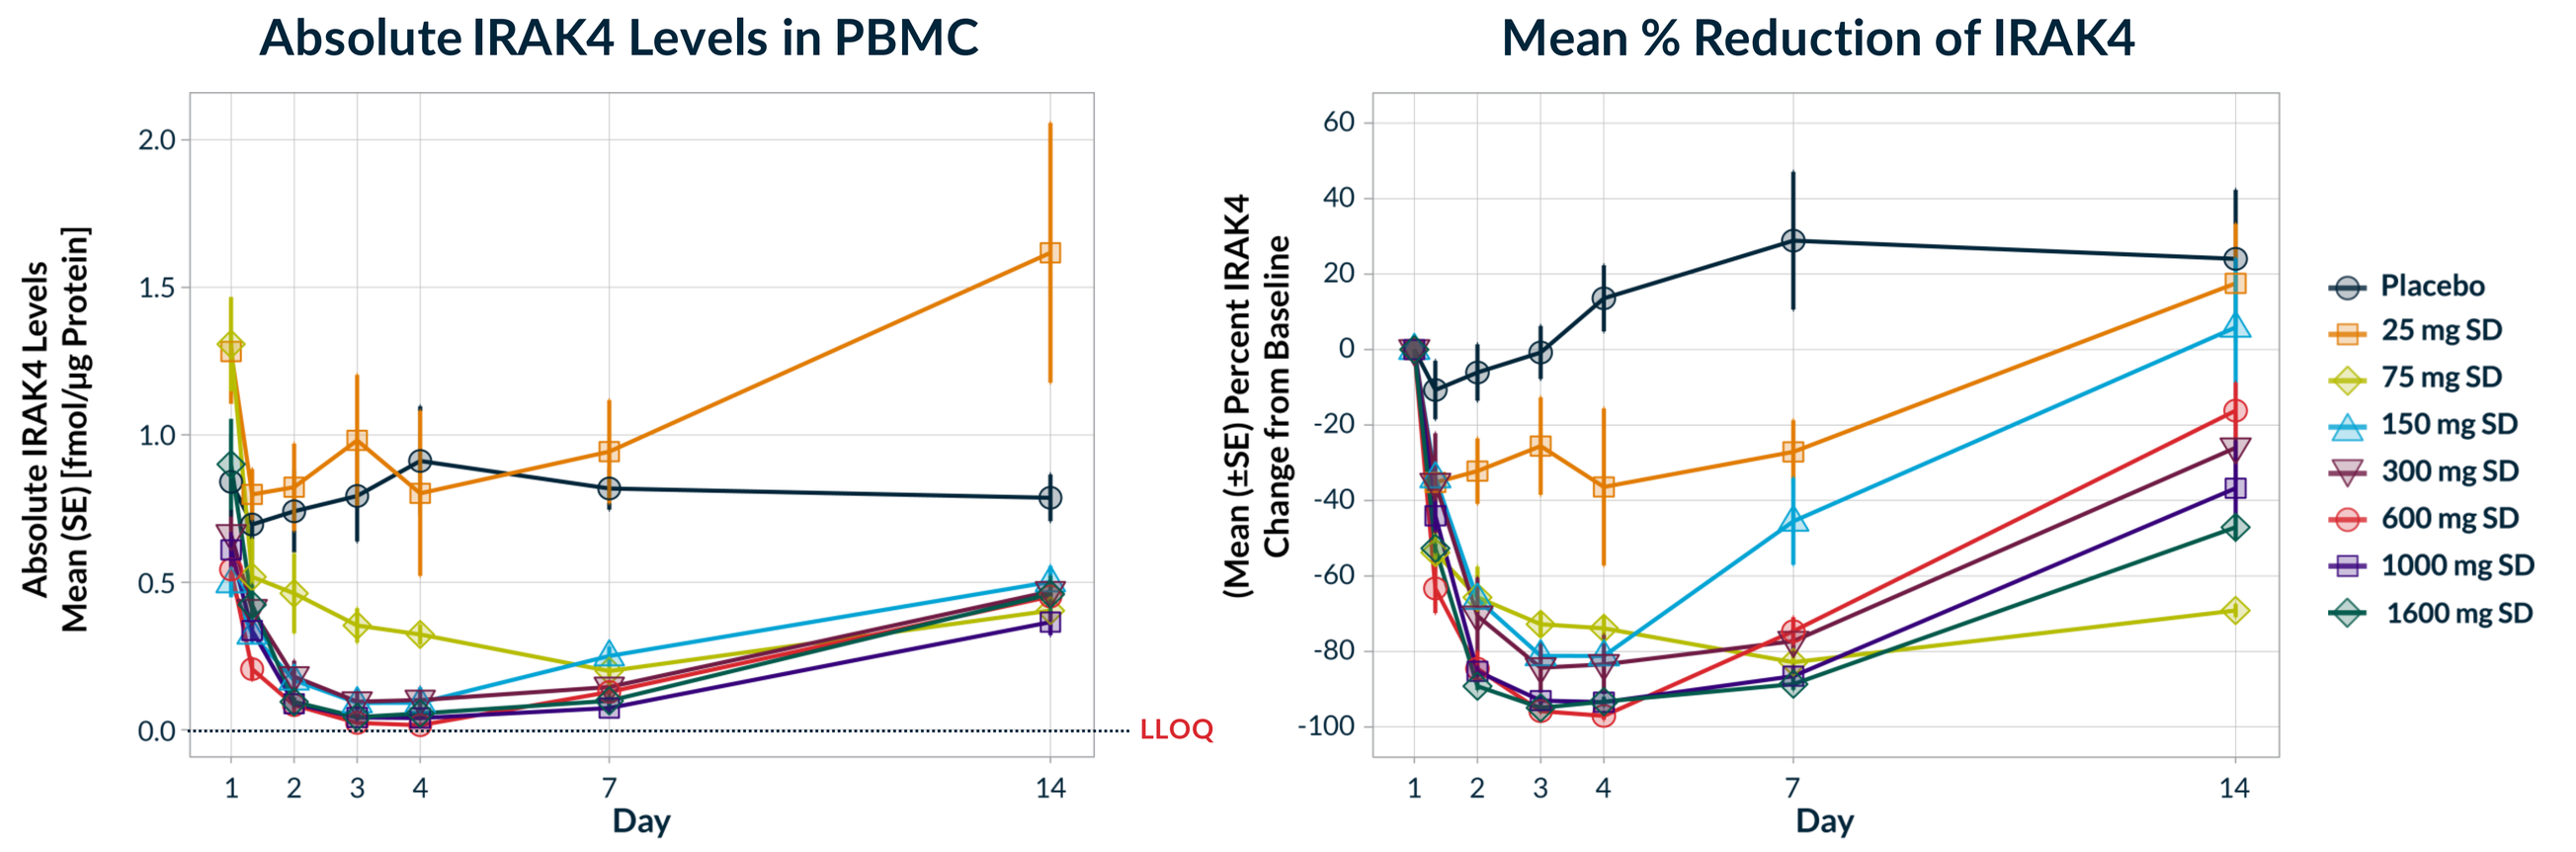
Absolute IRAK4 Levels in PBMC
Absolute IRAK4 Levels Mean (SE) [fmol/μg Protein]
LLOQ
Day
Mean % Reduction of IRAK4
(Mean (±SE) Percent IRAK4 Change from Baseline
Placebo
25 mg SD
75 mg SD
150 mg SD
300 mg SD
600 mg SD
1000 mg SD
1600 mg SD

- IRAK4 levels nadired at 48-72 hours (Day 3-4)
- SAD 5 through 7 approached or exceeded Lower Limit of Quantitation (LLOQ)

Figure 7: KT-474 Achieved Robust and Sustained IRAK4 Degradation with Multiple Daily Oral Doses (14 Days)

2 MAD Portion Healthy Volunteers

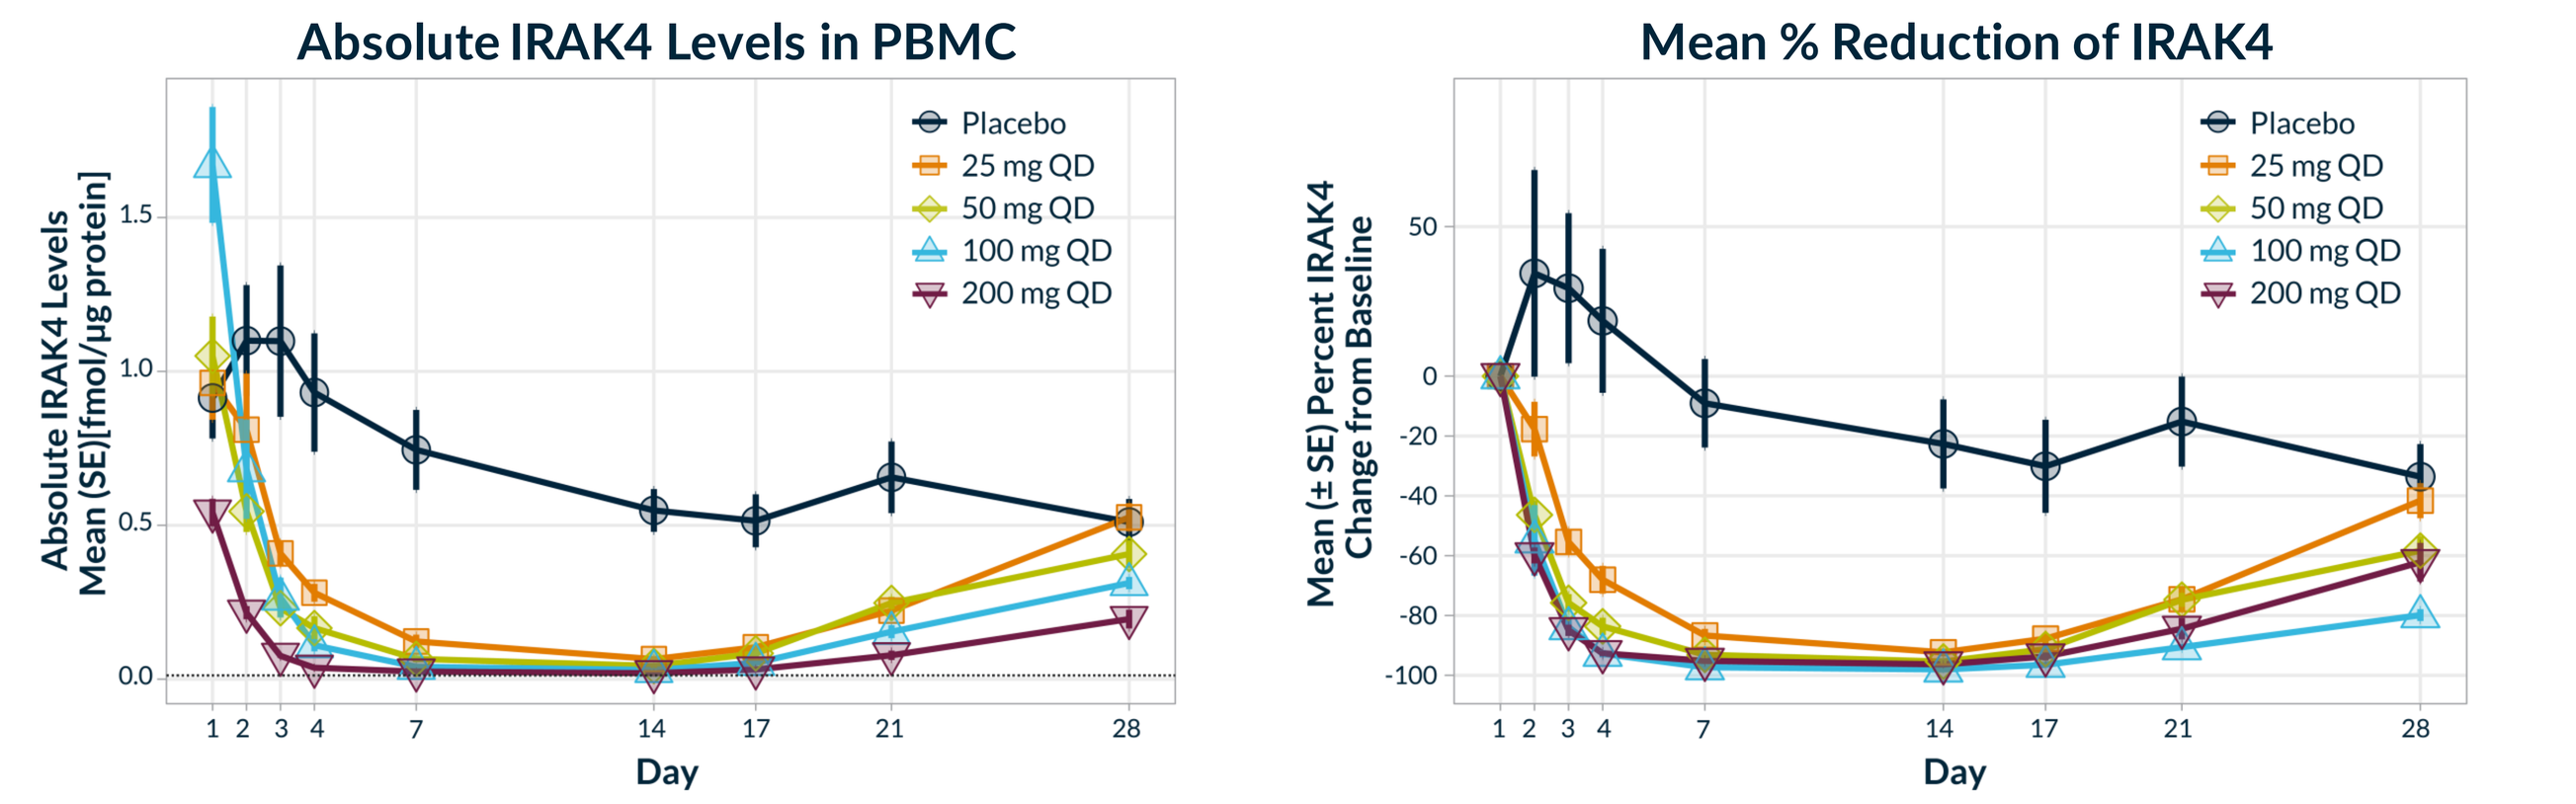
Absolute IRAK4 Levels in PBMC
Absolute IRAK4 Levels Mean (SE)[fmol/μg protein]
Day
Mean % Reduction of IRAK4
Mean (± SE) Percent IRAK4 Change from Baseline
Placebo
25 mg QD
50 mg QD
100 mg QD
200 mg QD

- Steady state IRAK4 reduction achieved between Days 7 and 14
- Recovery towards baseline by Day 28 (2 weeks after last dose)
- MAD 2 through 4 approached Lower Limit of Quantitation (LLOQ)

METHODS

PBMC treatment and preparation

Human peripheral blood mononuclear cells (PBMC) from three or more healthy donors were collected and treated with lead series IRAK4 degrader compounds at 10x DC90 for 24 h and analyzed by global proteomics TMT workflow.

Discovery Proteomics

Tandem Mass Tag discovery proteomics was performed on PBMC treated with KT-474 at 10x DC90 (300nM). Concatenated basic pH fractions were separated with a 1200 nanoLC using over a 3 h reversed-phase gradient and analyzed by LC-MS/MS on an Orbitrap mass spectrometer. Collected spectra were searched against a human database using MaxQuant to identify to a depth of ~9,000 proteins. Statistical analysis was carried out using the Limma statistical package. A weighted cutoff between statistical significance and fold-change was applied.

Targeted Proteomics

Canine or human PBMC or skin tissues were collected after compound exposure in vivo and analyzed by stable isotope dilution mass spectrometry monitoring peptides unique to IRAK4. Peak areas integrated using Skyline or SpectroDive™ software and further processed to calculate protein concentration and plot relative to controls.

CONCLUSIONS

A combination of highly sensitive and reproducible nanoLC MS Discovery and Targeted Proteomic Workflows characterize, quantify and understand the activity of novel IRAK4 degrader *in vitro, in vivo* preclinically and in the clinic

- Discovery Proteomics enabled the selection of the compounds with the fewest off-target liabilities and further identified compounds with specific IRAK4 degradation activity.
- Targeted Proteomic assays were developed for two peptides unique to IRAK4 with concentrations as low as 0.01 fmol/ug protein isolated and digested from PBMC or skin. Assay sensitivity enabled calculation of protein degradation to greater than 95%. Peptides were selected that were unique to human, rodent and non-rodent species that permitted assay transfer across the preclinical species tissue cohorts enabling cross-species comparison of degrader compounds.
- KT-474 degraded IRAK4 to greater than 95% in both single ascending dose (SAD) and multiple ascending dose (MAD) portions of Phase 1 study in healthy volunteers.

REFERENCES

1. Meissner, F et al. The emerging role of mass spectrometry-based proteomics in drug discovery. *Nature Reviews Drug Discovery* 2022
2. Gillette, MA and Carr, SA. Quantitative analysis of peptides and proteins in biomedicine by targeted mass spectrometry *Nat Methods 2013 Jan;10(1):28-34. doi: 10.1038/nmeth.2309.*

DISCLOSURES

Kuhn, Feeney, Breitkopf, Browne, Shi, Walther, Chen, Zheng, Skouras, Campbell, Rong, Agarwal, McDonald, Gollob, Slavin, Weiss, De Savi, Mainolfi, Sharma are Kymera Therapeutics employees and equity owners. The work was done under collaboration agreement with Sanofi. The work is funded by Kymera.

Kymera Therapeutics • 200 Arsenal Yards Blvd., Suite 230, Watertown, MA 02472, USA www.kymeratx.com Contact: Eric Kuhn / ekuhn@kymeratx.com ASMS Annual Meeting 2022 • June 5 - 9, 2022 • Minneapolis, MN