Selective IRAK4 Degradation, Not Kinase Inhibition, Blocks TLR-activated NF-kB and p38 Signaling Leading to broad Cytokine Inhibition

Stephanie Skouras, **Virginia Massa**, Emily Lurier, Cedric Hubeau, Amy Wang, Xiaozhang Zheng, Kirti Sharma, Dirk Walther, Chris Browne, Scott Rusin, Michele Mayo, Michael Fitzgerald, Alice McDonald, Jared Gollob, Nello Mainolfi, Anthony Slavin, and Veronica Campbell

INTRODUCTION

IL-1R/TLR activation plays a central role in the pathophysiology of multiple autoimmune and inflammatory diseases via the Myddosome, that is dependent on both the kinase and scaffolding functions of Interleukin-1 receptor associated kinase 4 (IRAK4). Therefore, hetero-bifunctional molecules that selectively target IRAK4 for degradation and elimination by the ubiquitin proteasome pathway have the greatest potential to block IL-1R/TLR signaling, including NF-kB activation and cytokine production. To interrogate downstream signaling, phosphorylation events were monitored in monocytes and B cells following TLR7/8 or TLR9 activation, respectively. IRAK4 degradation, but not kinase inhibition, inhibited TLR activated NF-kB p65 activation and phosphorylation of p38 pathway members in both cell types. PBMCs pretreated with an IRAK4 degrader and then stimulated with the TLR7/8 agonist, R848, exhibited significantly broader inhibition of cytokines (IL-6, TNFa, IL-8 and IL-1b) compared to those pretreated with a selective IRAK4 kinase inhibitor. Compound washout experiments demonstrated a sustained effect of IRAK4 degrader on both target pharmacodynamics and cytokine inhibition that was differentiated from IRAK4 kinase inhibition. The data demonstrate that IRAK4 degradation was clearly more effective compared to kinase inhibition due to its impact on both kinase and scaffolding function.

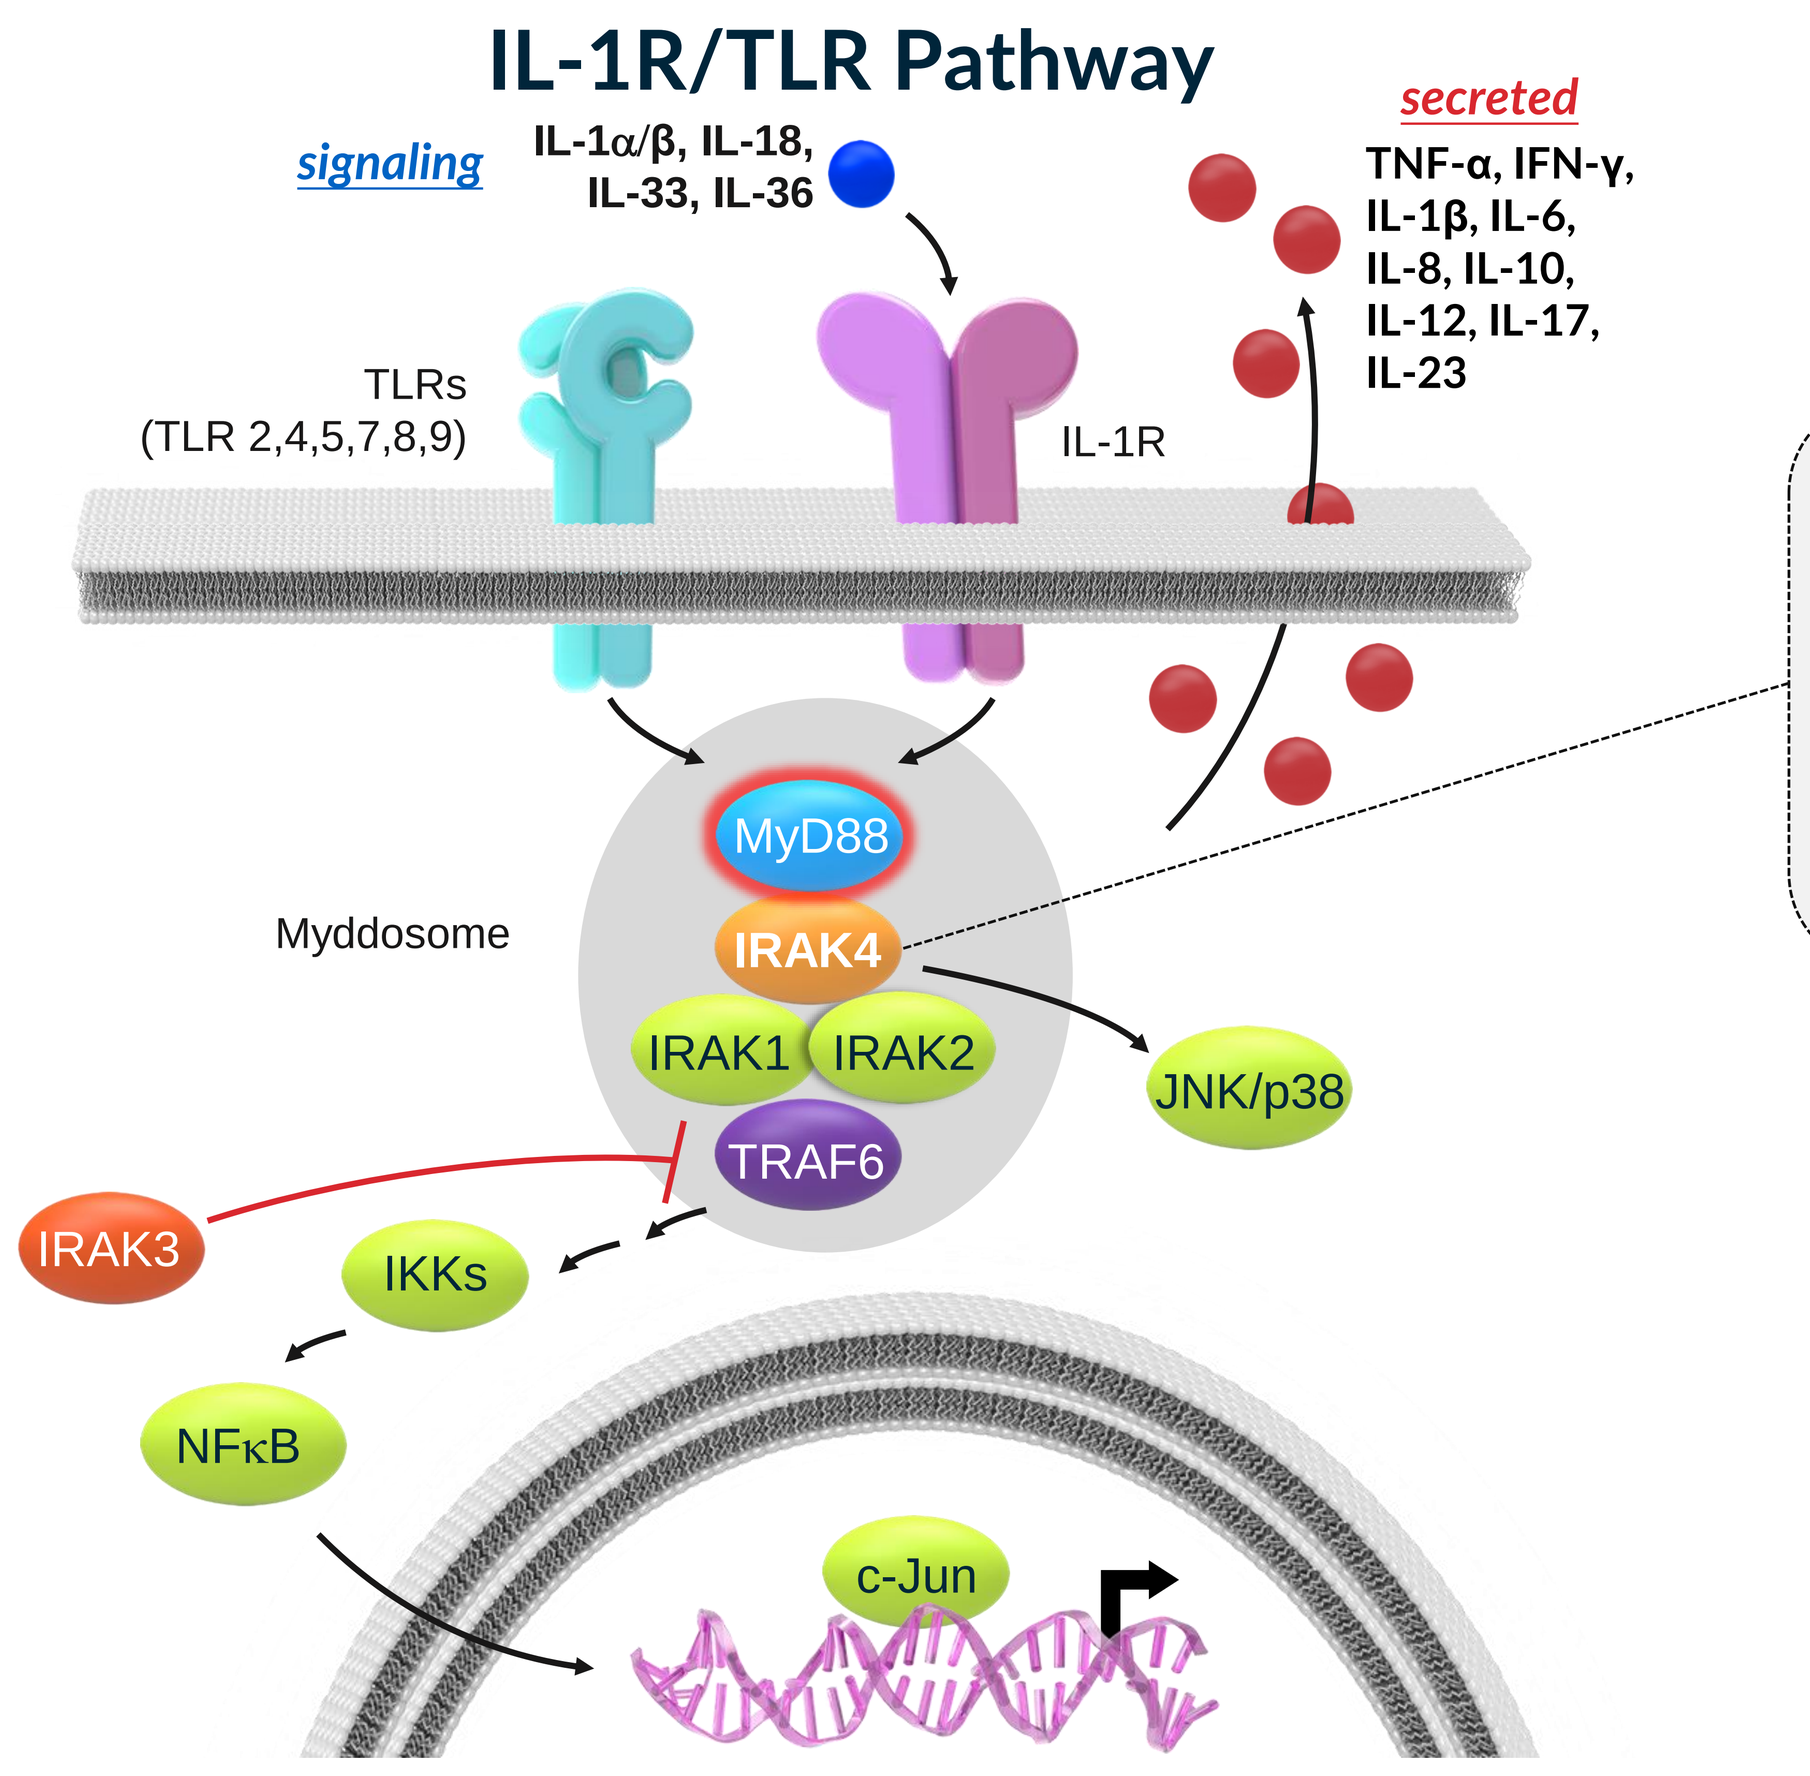
IL-1R/TLR Pathway
signaling
IL-1α/β, IL-18, IL-33, IL-36
secreted
TNF-α, IFN-γ, IL-1β, IL-6, IL-8, IL-10, IL-12, IL-17, IL-23
TLRs (TLR 2,4,5,7,8,9)
IL-1R
Myddosome
MyD88
IRAK4
IRAK1
IRAK2
TRAF6
JNK/p38
IRAK3
IKKs
NFκB
c-Jun

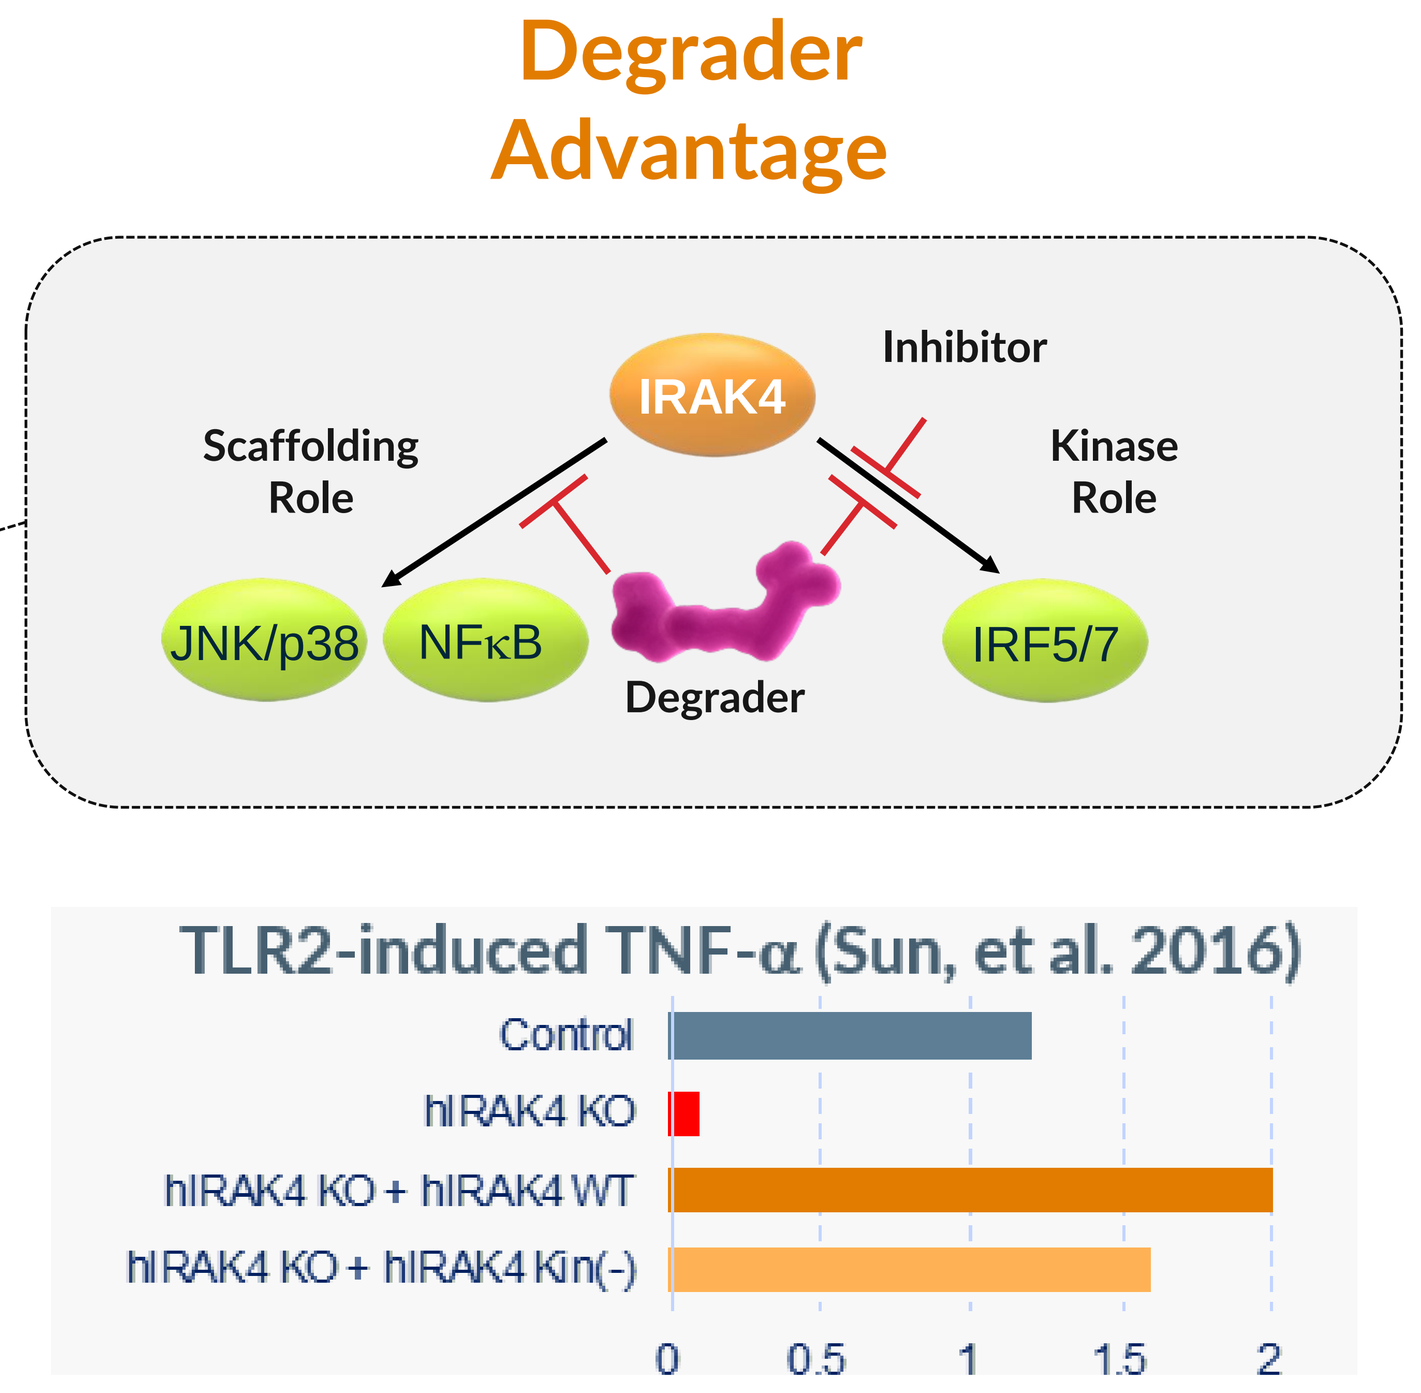
Degrader Advantage
Inhibitor
IRAK4
Scaffolding Role
Kinase Role
JNK/p38
NFκB
Degrader
IRF5/7
TLR2-induced TNF-α (Sun, et al. 2016)
Control
hIRAK4 KO
hIRAK4 KO + hIRAK4 WT
hIRAK4 KO + hIRAK4 Kin(-)
0
0.5
1
1.5
2

METHODS

All primary cell assays were conducted using cryopreserved PBMCs or B cells that were thawed and pre-treated with compounds for 24hours prior to collection or TLR stimulation.

Compounds: KTX-545 IRAK4 degrader, KTX-045 degrader negative control (no degrader function), PF-06550833- IRAK4 kinase inhibitor

Proteomics: TMT-labeled peptides from treated PBMCs were analyzed by LC-MS/MS.

IRAK4 Flow in Immune Subsets: Treated PBMC samples were fixed with 1.6% PFA, permeabilized and stained with IRAK4 and phenotypic markers CD4/CD8/CD16/CD19/CD14 to monitor degradation in different immune subsets

Washout Study: Pre-treated cells were washed 3x with PBS and then collected or re-seeded. Post wash out, cells were stimulated with TLR7/8 agonist, R848, at 10ug/mL for 4 hours. Samples were collected to measure IRAK4 and cytokine levels

Cytokine Release Studies: Treated PBMCs or B cells were stimulated with R848 (10ug/mL) or TLR9 agonist, CpG-B (2.5uM) for 5 hours, respectively. Supernatants were collected and cytokines quantified using Mesoscale Quickplex

Flow Phosphorylation Analysis: Treated CD14+ monocytes or B cells were stimulated with R848 15' or CpG-B for 60' fixed, permeabilized and stained with multiplex phospho panel or single stain for NF-kB p65

RESULTS AND CONCLUSIONS

KTX-545 is a potent and selective IRAK4 degrader and the cellular data generated demonstrate that IRAK4 degradation is clearly superior to kinase inhibition due to its removal of both scaffolding and kinase functions, leading to broader cytokine inhibition across multiple TLR stimuli and cell types. Phosphorylation analysis suggests this differentiated effect is driven in part by inhibition of NF-kB and MAPK signaling. These data highlight the potential for IRAK4 degraders to block multiple TLR signaling pathways across different immune cell types and thereby impact TLR/IL-1R-driven inflammatory and autoimmune diseases.

Figure 1: KTX-545 is a Selective and Potent IRAK4 Degrader Across Human Peripheral Mononuclear Subsets

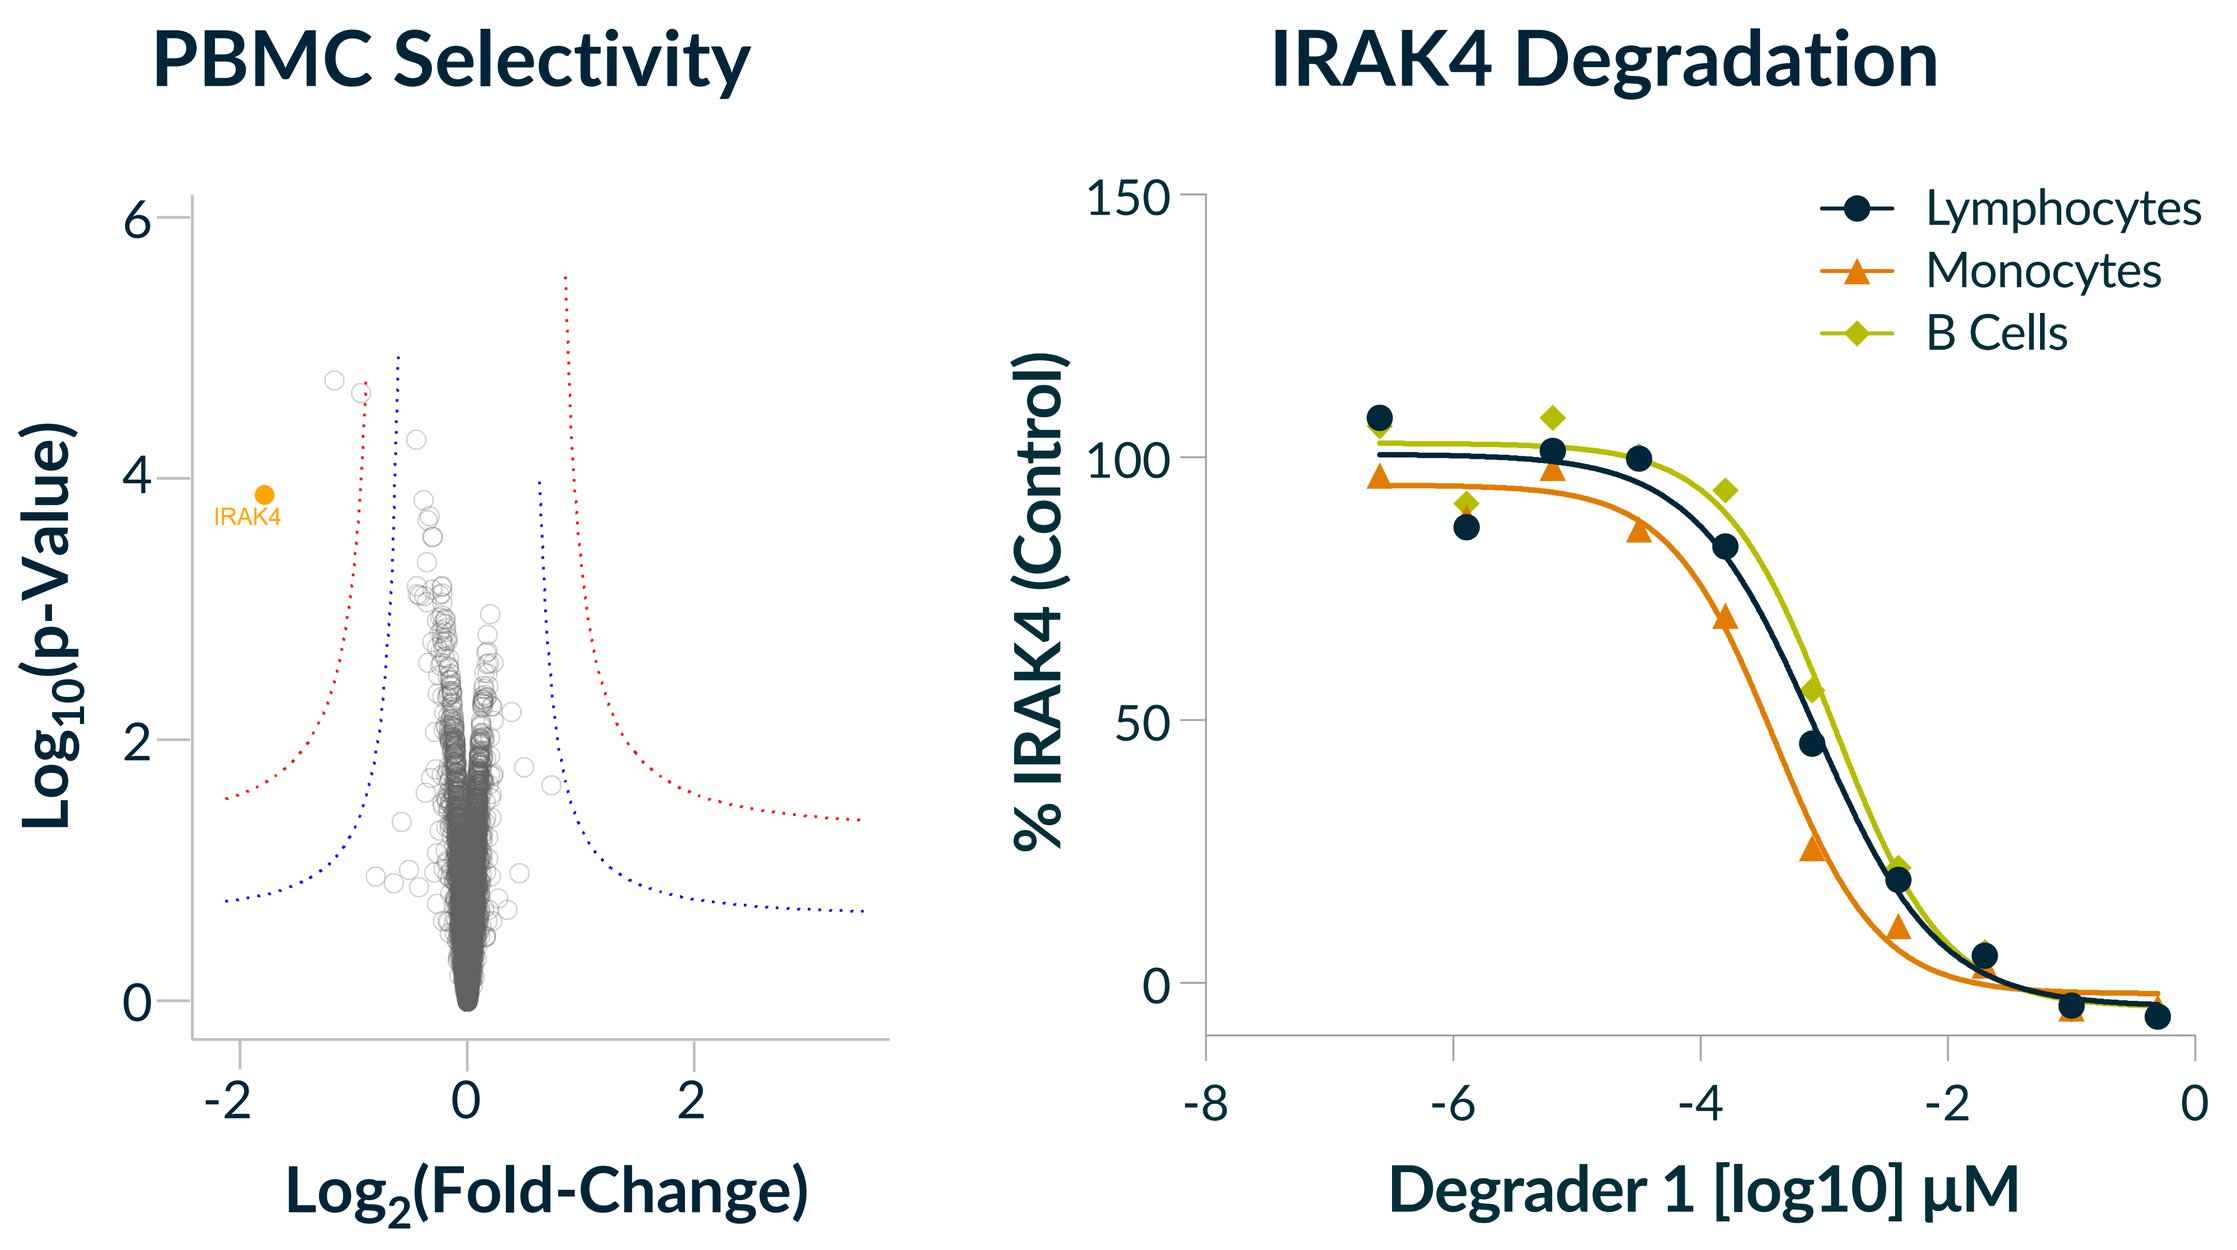
PBMC Selectivity
IRAK4
Log_{10}(p-Value)
Log_2(Fold-Change)
IRAK4 Degradation
Lymphocytes
Monocytes
B Cells
% IRAK4 (Control)
Degrader 1 [log10] µM

Following KTX-545 treatment and mass spectrometry analysis of over 8,000 proteins, IRAK4 was the only one significantly decreased at a dose of 10x DC90 (100nM).

Cell type	Lymphs	B cells	Mono(s)
IRAK4 DC_{50} (nM)	1.1 ± 0.53	1.9 ± 0.39	0.86 ± 0.68

Figure 2: Sustained Degradation and Cytokine Inhibition Following IRAK4 Degrader Washout Out to 48 Hours

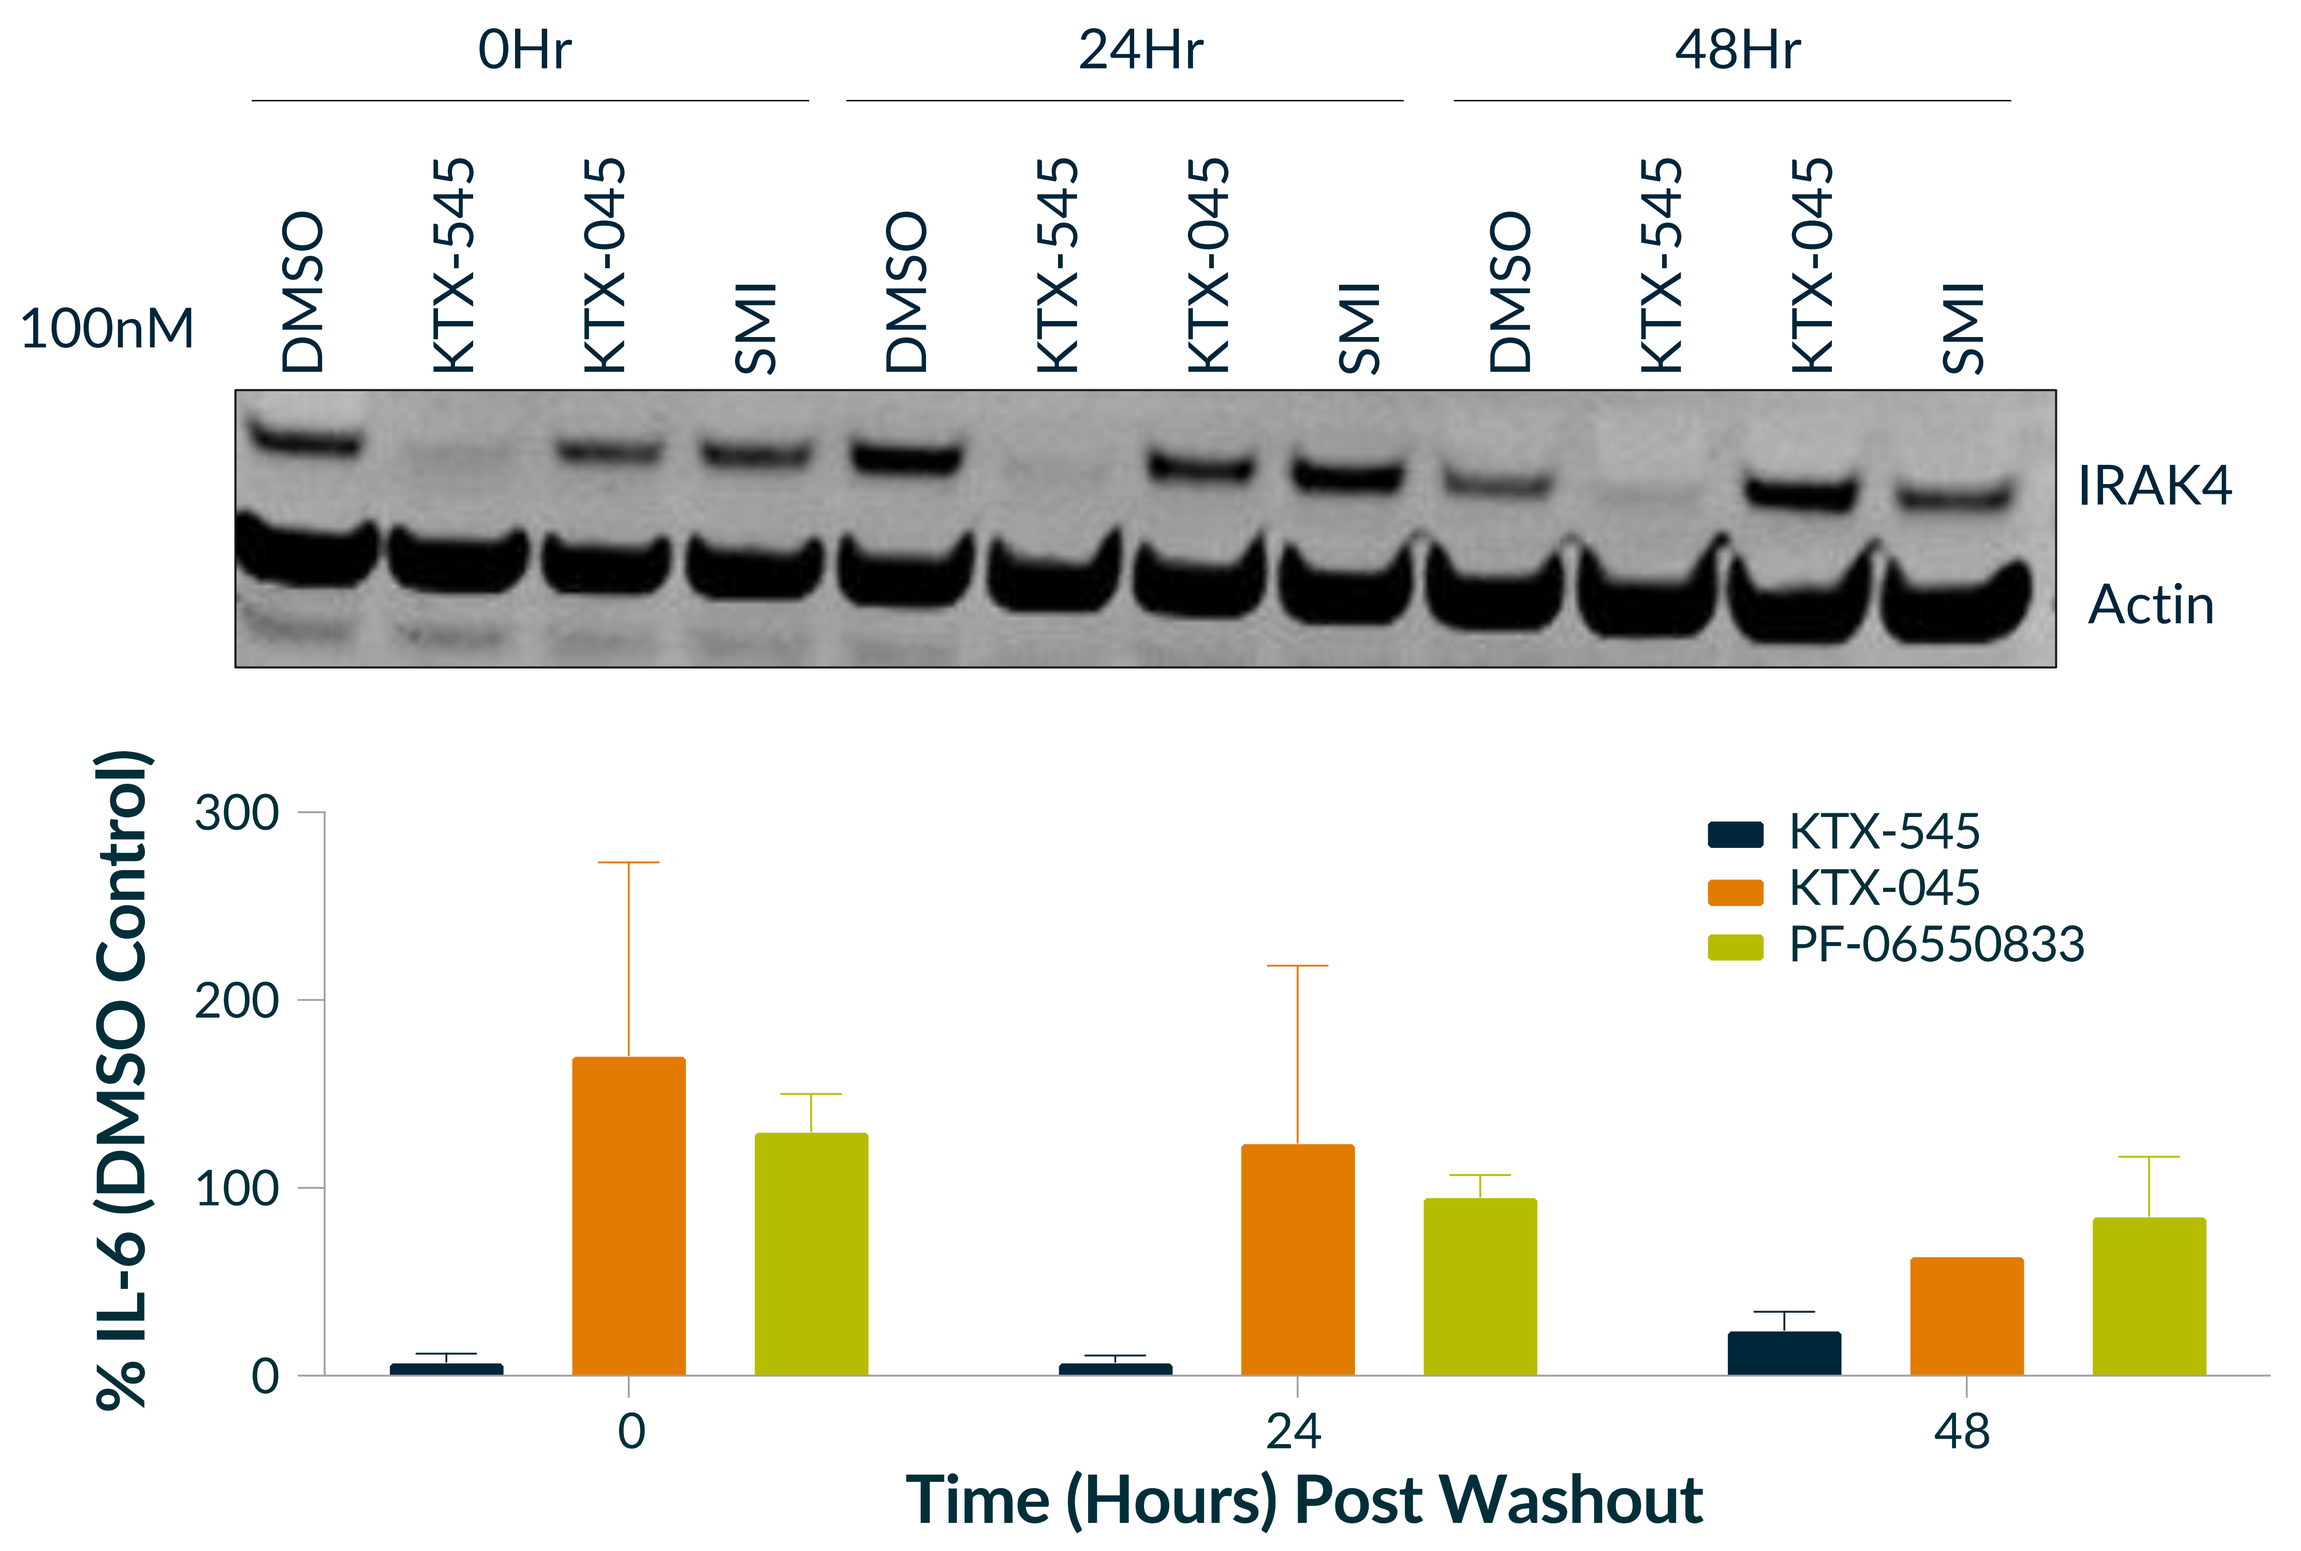
0Hr
24Hr
48Hr
100nM
DMSO
KTX-545
KTX-045
SMI
IRAK4
Actin
KTX-545
KTX-045
PF-06550833
% IL-6 (DMSO Control)
Time (Hours) Post Washout

Figure 3: IRAK4 Degradation Leads to More Potent Cytokine Inhibition that is Differentiated from Kinase Inhibition Following TLR7/8

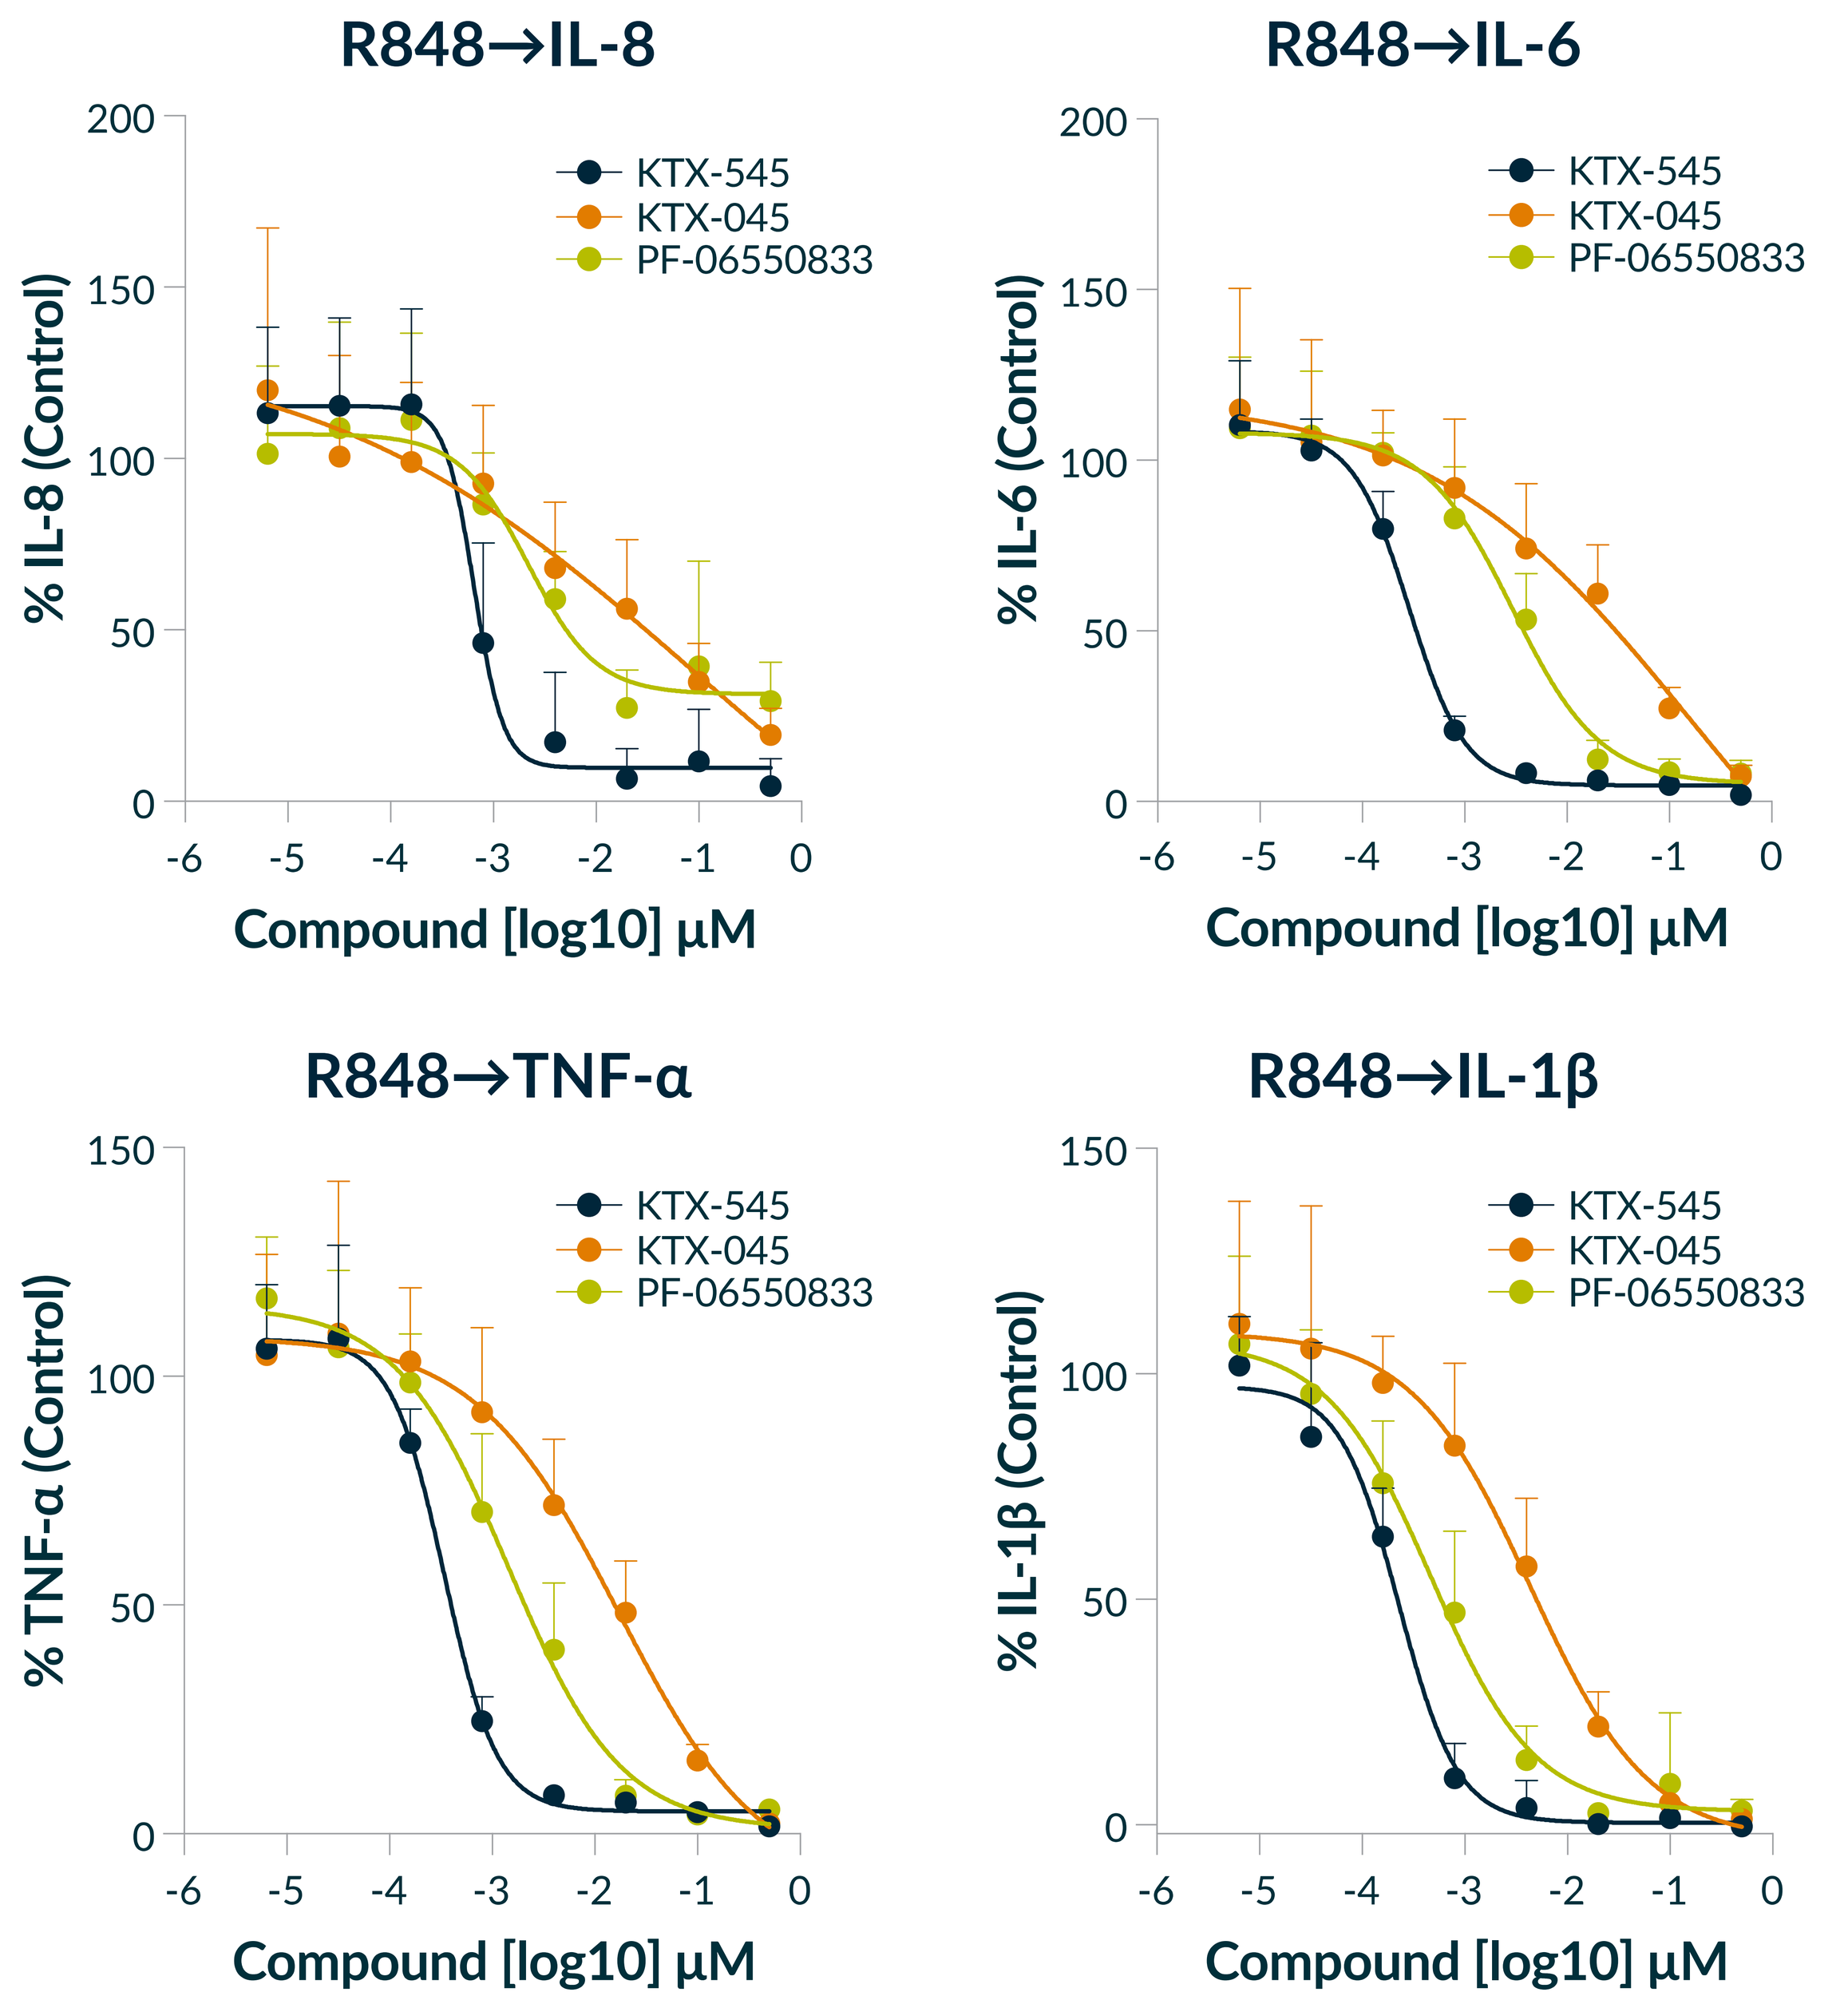
R848→IL-8
R848→IL-6
R848→TNF-α
R848→IL-1β
KTX-545
KTX-045
PF-06550833
% IL-8 (Control)
% IL-6 (Control)
% TNF-α (Control)
% IL-1β (Control)
Compound [log10] µM

Compound	IL-8 IC_{50} (nM)	IL-6 IC_{50} (nM)	TNF-α IC_{50} (nM)	IL-1β IC_{50} (nM)
KTX-545	0.90 ±0.16	0.34 ±0.03	0.40 ±0.03	0.22 ±0.04
KTX-045 (neg. control with no degrader function)	26.35 ±15.10	29.96 ±10.86	15.81 ±2.77	5.20 ±1.37
PF-06550833	6.22 ±2.11	3.88 ±1.00	2.28 ±0.82	0.66 ±0.31

N=3 donors

Figure 4: IRAK4 Degradation Leads to More Potent Inhibition of Phosphorylation Events Downstream of TLR7/8 Activation in Monocytes Compared to Kinase Inhibition Alone

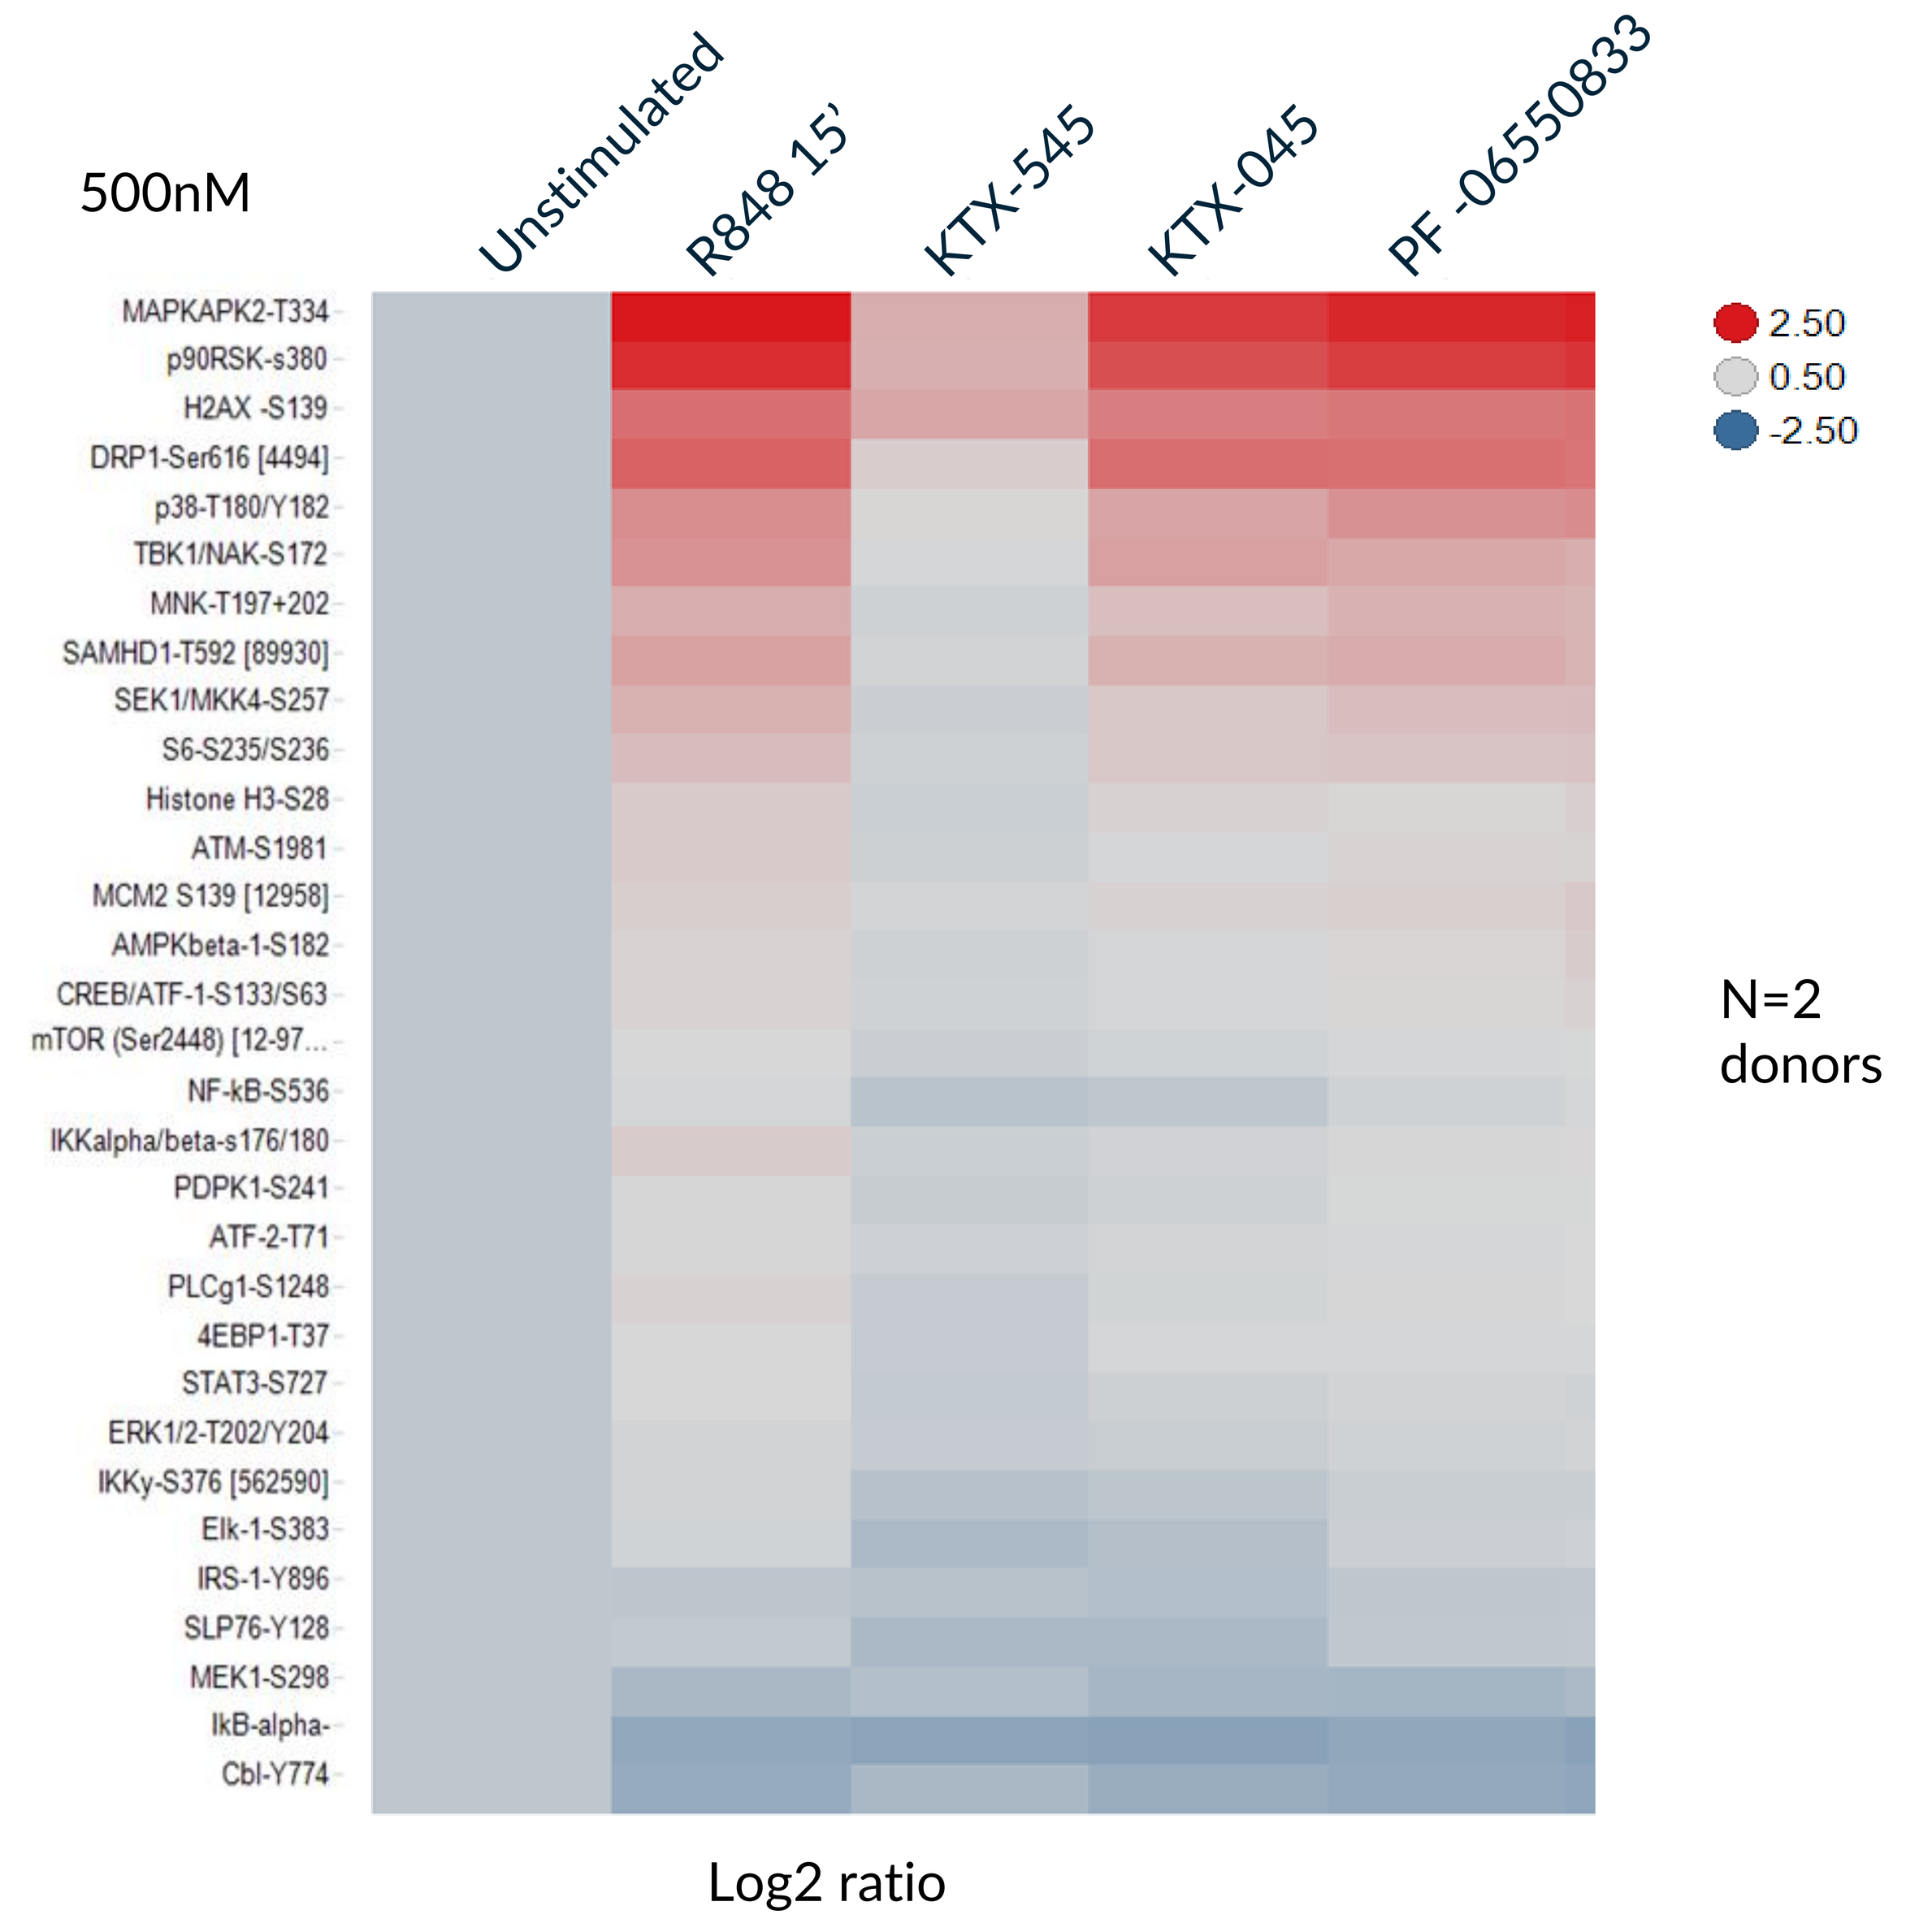
500nM
Unstimulated
R848 15'
KTX-545
KTX-045
PF -06550833
MAPKAPK2-T334
p90RSK-s380
H2AX -S139
DRP1-Ser616 [4494]
p38-T180/Y182
TBK1/NAK-S172
MNK-T197+202
SAMHD1-T592 [89930]
SEK1/MKK4-S257
S6-S235/S236
Histone H3-S28
ATM-S1981
MCM2 S139 [12958]
AMPKbeta-1-S182
CREB/ATF-1-S133/S63
mTOR (Ser2448) [12-97...
NF-kB-S536
IKKalpha/beta-s176/180
PDPK1-S241
ATF-2-T71
PLCg1-S1248
4EBP1-T37
STAT3-S727
ERK1/2-T202/Y204
IKKy-S376 [562590]
Elk-1-S383
IRS-1-Y896
SLP76-Y128
MEK1-S298
IkB-alpha-
Cbl-Y774
2.50
0.50
-2.50
N=2 donors
Log2 ratio

Figure 5: IRAK4 Degradation Inhibits TLR Activated NF-kB p65 and MAPK Signaling Better than Kinase Inhibition Alone in Both Monocytes and B Cells

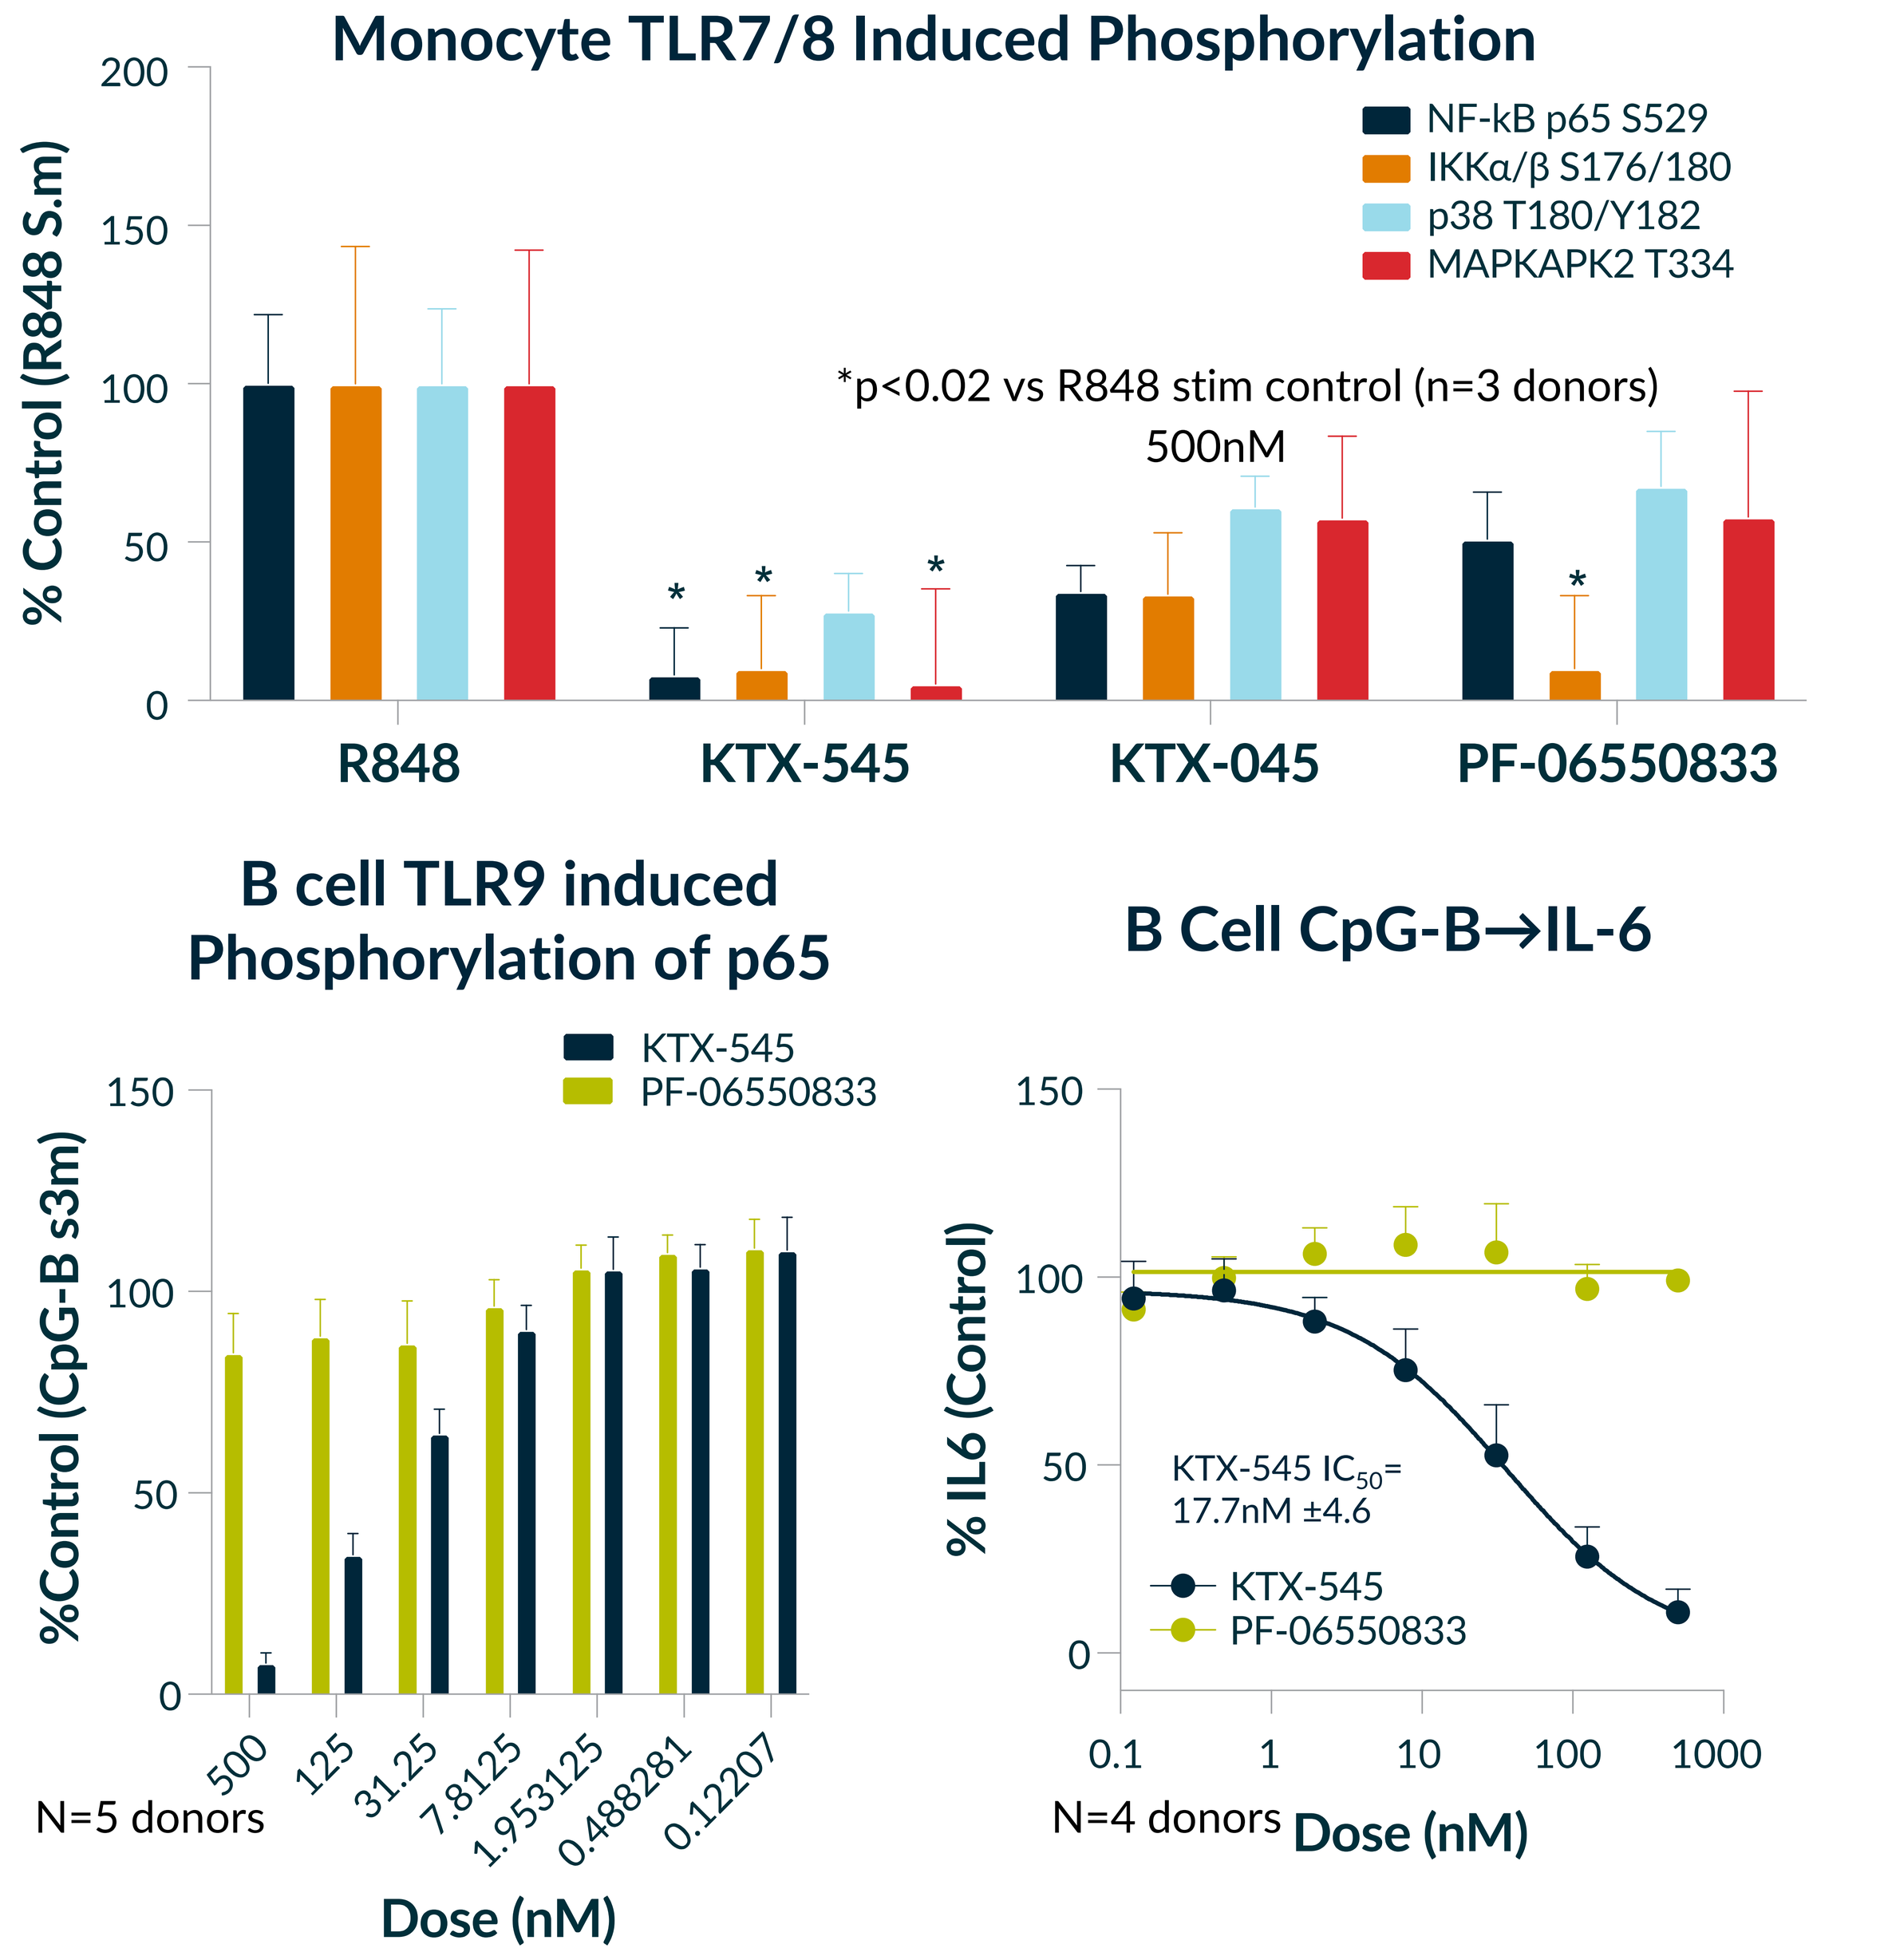
Monocyte TLR7/8 Induced Phosphorylation
NF-kB p65 S529
IKKα/β S176/180
p38 T180/Y182
MAPKAPK2 T334
% Control (R848 S.m)
*p<0.02 vs R848 stim control (n=3 donors)
500nM
R848
KTX-545
KTX-045
PF-06550833
B cell TLR9 induced Phosphorylation of p65
KTX-545
PF-06550833
%Control (CpG-B s3m)
N=5 donors
Dose (nM)
B Cell CpG-B→IL-6
% IL6 (Control)
KTX-545 IC_{50}= 17.7nM ±4.6
KTX-545
PF-06550833
N=4 donors
Dose (nM)

KYMERA
200 Arsenal Yards Blvd., Suite 230, Watertown, MA 02472, USA
+1 (857) 285-5300 - www.kymeratx.com
Contact: Virginia Massa / vmassa@kymeratx.com

References:
1. Sun, et al. *Sci Signal*, 2016

The work was done under collaboration agreement with Sanofi

The American Association of Immunologists • Immunology2022™ • Portland, Oregon • May 6-10, 2022