Kinetics of IRAK4 Degradation and Impact on Functional Response in Circulating Immune Cells and Skin Cell Subsets

Emily Lurier, Jeffrey Sullivan, Stephanie Skouras, Virginia Massa, Michael Fitzgerald, Amy Wang, Xiaozhang Zheng, Dirk Walther, Chris Browne, Joyoti Dey, Alice McDonald, Jared Gollob, Nello Mainolfi, Anthony Slavin, Veronica Campbell

KYMERA

INTRODUCTION

Kymera has developed selective IRAK4 degraders for the treatment of autoimmune and inflammatory diseases where IL-1R/TLR activation plays a central role in driving the inflammatory response. In autoimmune diseases of the skin, like hidradenitis suppurativa, keratinocytes are active inflammatory mediators that are triggered by IL-1R signaling. IL-1R signals through the myddosome which is dependent on the kinase and scaffold functions of Interleukin-1 receptor associated kinase 4 (IRAK4) to induce downstream production of proinflammatory cytokines like IL-6 and IL-8 (Fig. 1). Selective IRAK4 degraders can achieve maximal degradation of >90% in several immune cell types in vitro after 24 hours of treatment that is associated with maximum inhibition of pro-inflammatory cytokine production following IL-1R stimulation.

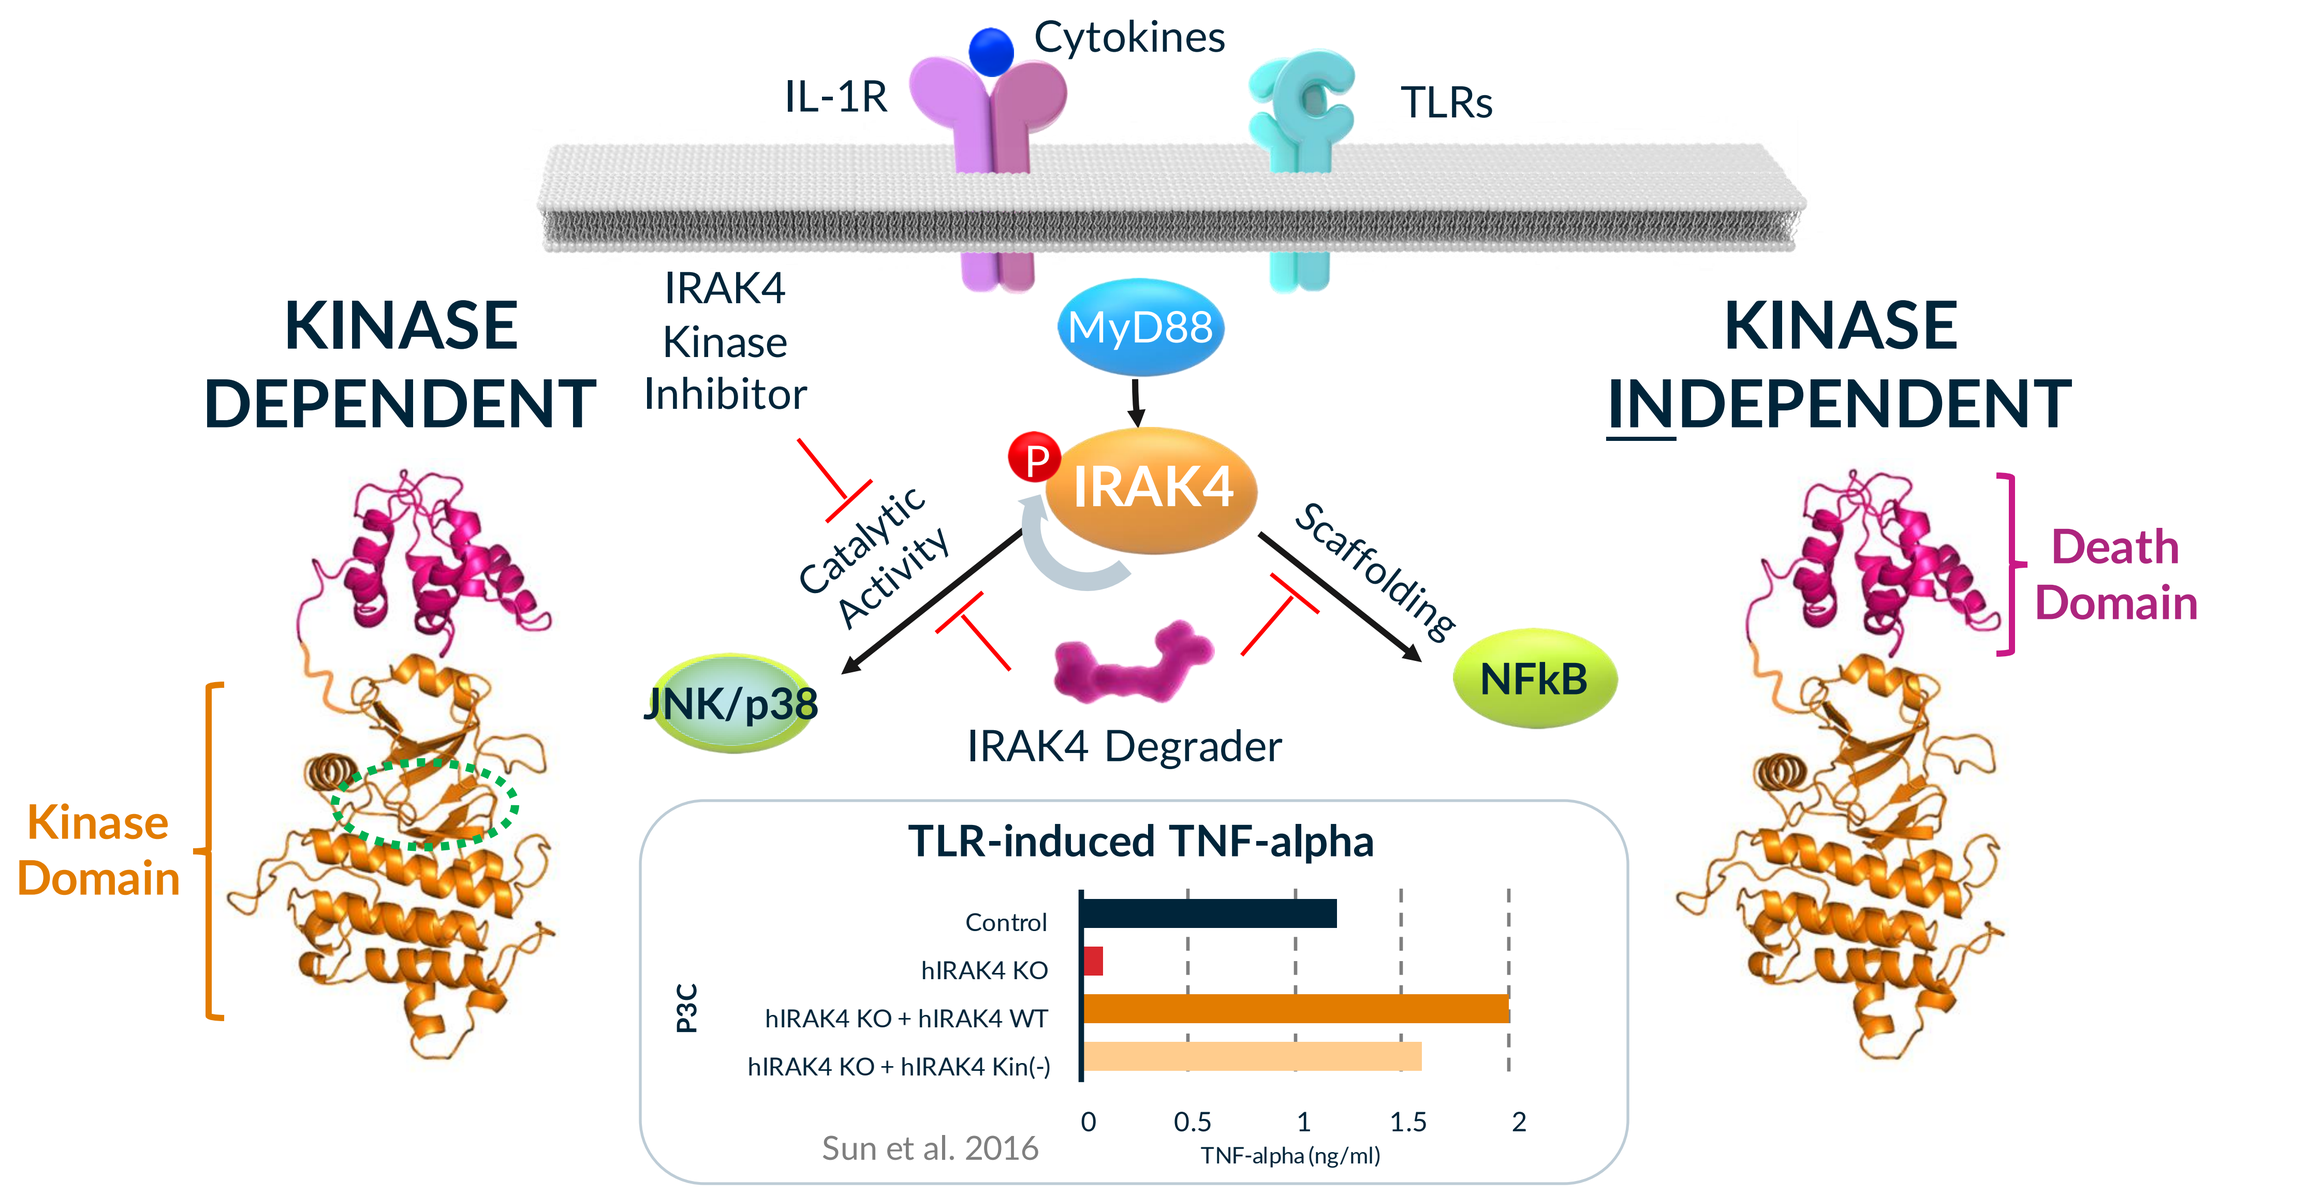
Cytokines
IL-1R
TLRs
KINASE DEPENDENT
IRAK4 Kinase Inhibitor
MyD88
KINASE INDEPENDENT
IRAK4
Catalytic Activity
Scaffolding
JNK/p38
NFkB
IRAK4 Degrader
Death Domain
Kinase Domain
TLR-induced TNF-alpha
Control
hIRAK4 KO
hIRAK4 KO + hIRAK4 WT
hIRAK4 KO + hIRAK4 Kin(-)
P3C
TNF-alpha (ng/ml)
Sun et al. 2016

Figure 1: Schematic of IRAK4 signaling through myddosomal activation

The kinetics and maximal degradation of IRAK4 in relevant skin cell types were characterized in this study. The effects of IRAK4 degradation on inhibiting cytokine production under IL-1β stimulation was also quantified. In a separate Phase 1 clinical study, IRAK4 degradation data were collected in healthy volunteers dosed with KT-474, where changes in IRAK4 levels in systemic immune cells was compared to the skin.

METHODS

In vitro Degradation Assay

Peripheral blood mononuclear cells (PBMCs), keratinocytes, and fibroblasts were incubated with degrader for 24, 48, 72 and 96 hours. The degrader (KT-474) and E3-inactive control (KTX-653) were dosed at the known concentrations that result in 50% (DC50 = 1nM) or 90% (DC90 = 30nM) IRAK4 degradation in an in vitro PBMC assay. Cell lysate was collected and IRAK4 levels were quantified via western blot.

IL-1β Stimulation Assay

PBMCs, keratinocytes, and fibroblasts were incubated with degrader for 24 or 72 hours followed by human recombinant IL-1β (12.5ng/mL) for an additional 24 hours. IL-6 and IL-8 were quantified using an MSD electrochemiluminescence assay. Quantification was completed using Graphpad prism.

Clinical Data

Healthy volunteer subjects enrolled in the multiple ascending dose (MAD) portion of the KT-474 Phase 1 study were dosed daily for 14 days with oral KT-474 or placebo (randomized 9:3). PBMC and skin biopsies were serially collected for targeted mass spectrometry and multiplexed immunofluorescence (skin only), including on Day 14 (ClinicalTrials.org identifier: NCT04772885).

Targeted Protein Degraders

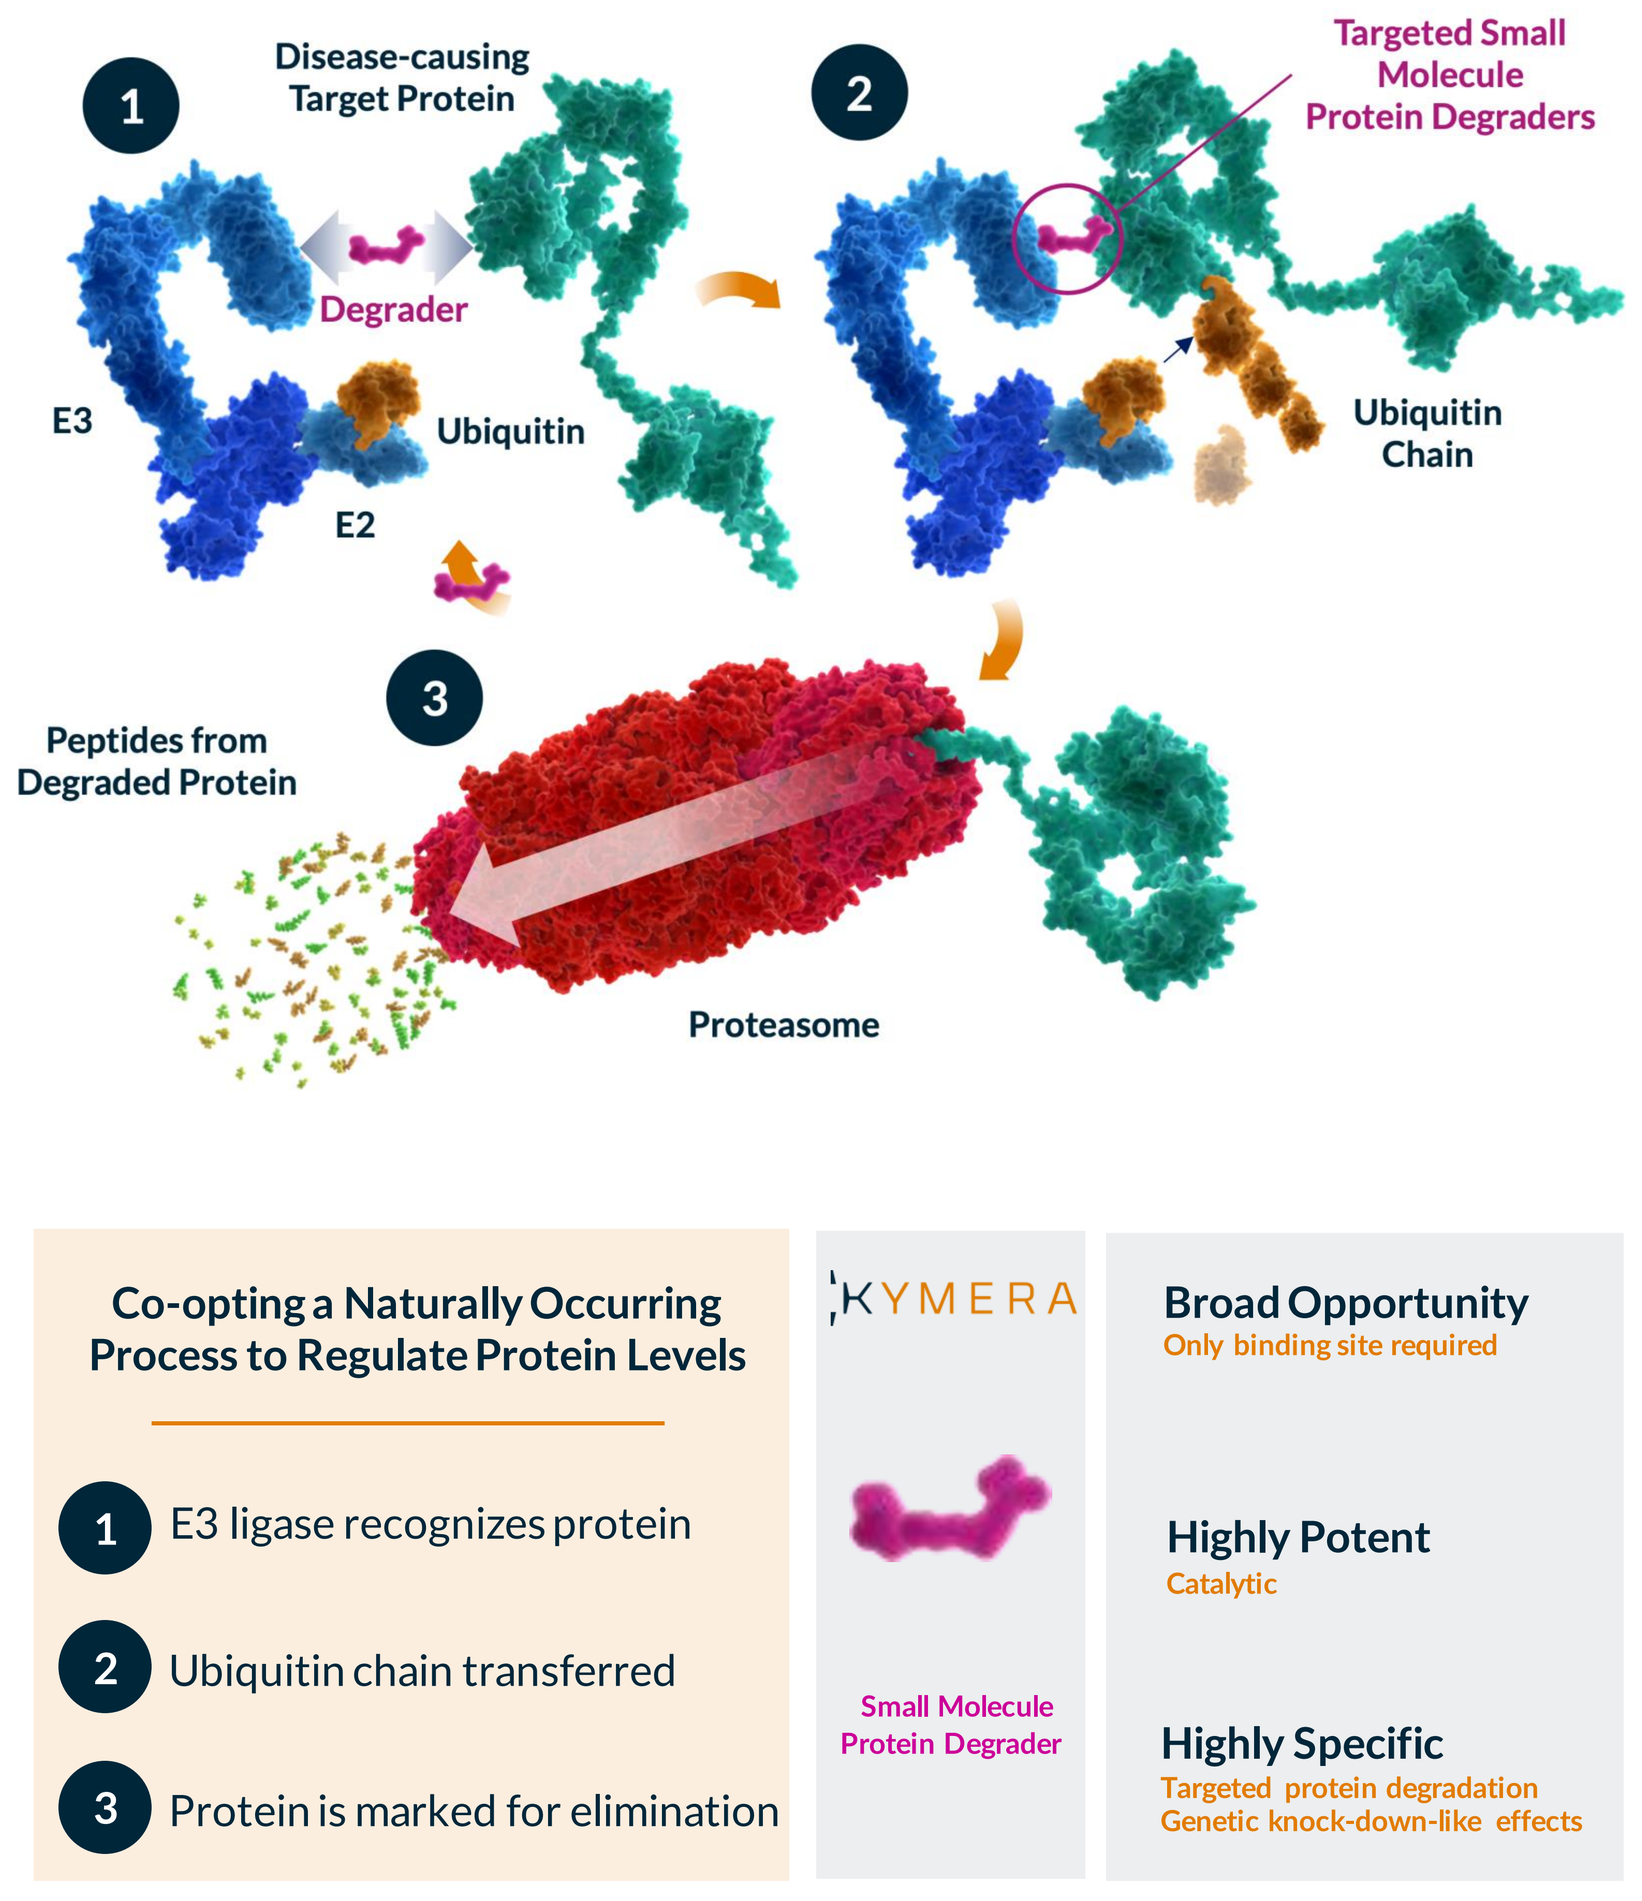
Disease-causing Target Protein
Targeted Small Molecule Protein Degraders
Degrader
E3
Ubiquitin
E2
Ubiquitin Chain
Peptides from Degraded Protein
Proteasome
Co-opting a Naturally Occurring Process to Regulate Protein Levels
1 E3 ligase recognizes protein
2 Ubiquitin chain transferred
3 Protein is marked for elimination
KYMERA
Small Molecule Protein Degrader
Broad Opportunity
Only binding site required
Highly Potent
Catalytic
Highly Specific
Targeted protein degradation
Genetic knock-down-like effects

Figure 2: Schematic of Targeted Protein Degrader function

In vitro Kinetics of IRAK4 Degradation in Immune and Cutaneous Cell Types

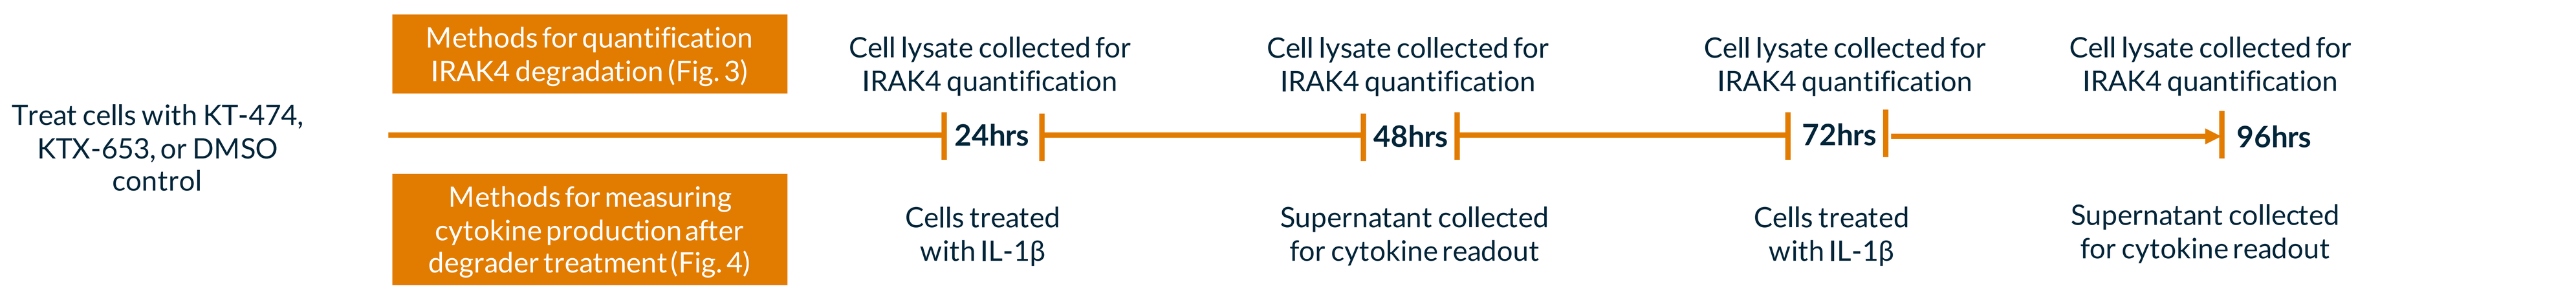
In vitro Experimental Design
Treat cells with KT-474, KTX-653, or DMSO control
Methods for quantification IRAK4 degradation (Fig. 3)
Methods for measuring cytokine production after degrader treatment (Fig. 4)
Cell lysate collected for IRAK4 quantification
24hrs
48hrs
72hrs
96hrs
Cells treated with IL-1β
Supernatant collected for cytokine readout

Figure 3: Quantification of IRAK4 degradation via Western blot in PBMCs, Fibroblasts, and Keratinocytes treated with IRAK4 degraders for 24, 48, 72, and 96hrs (n-=2-3 human donors)

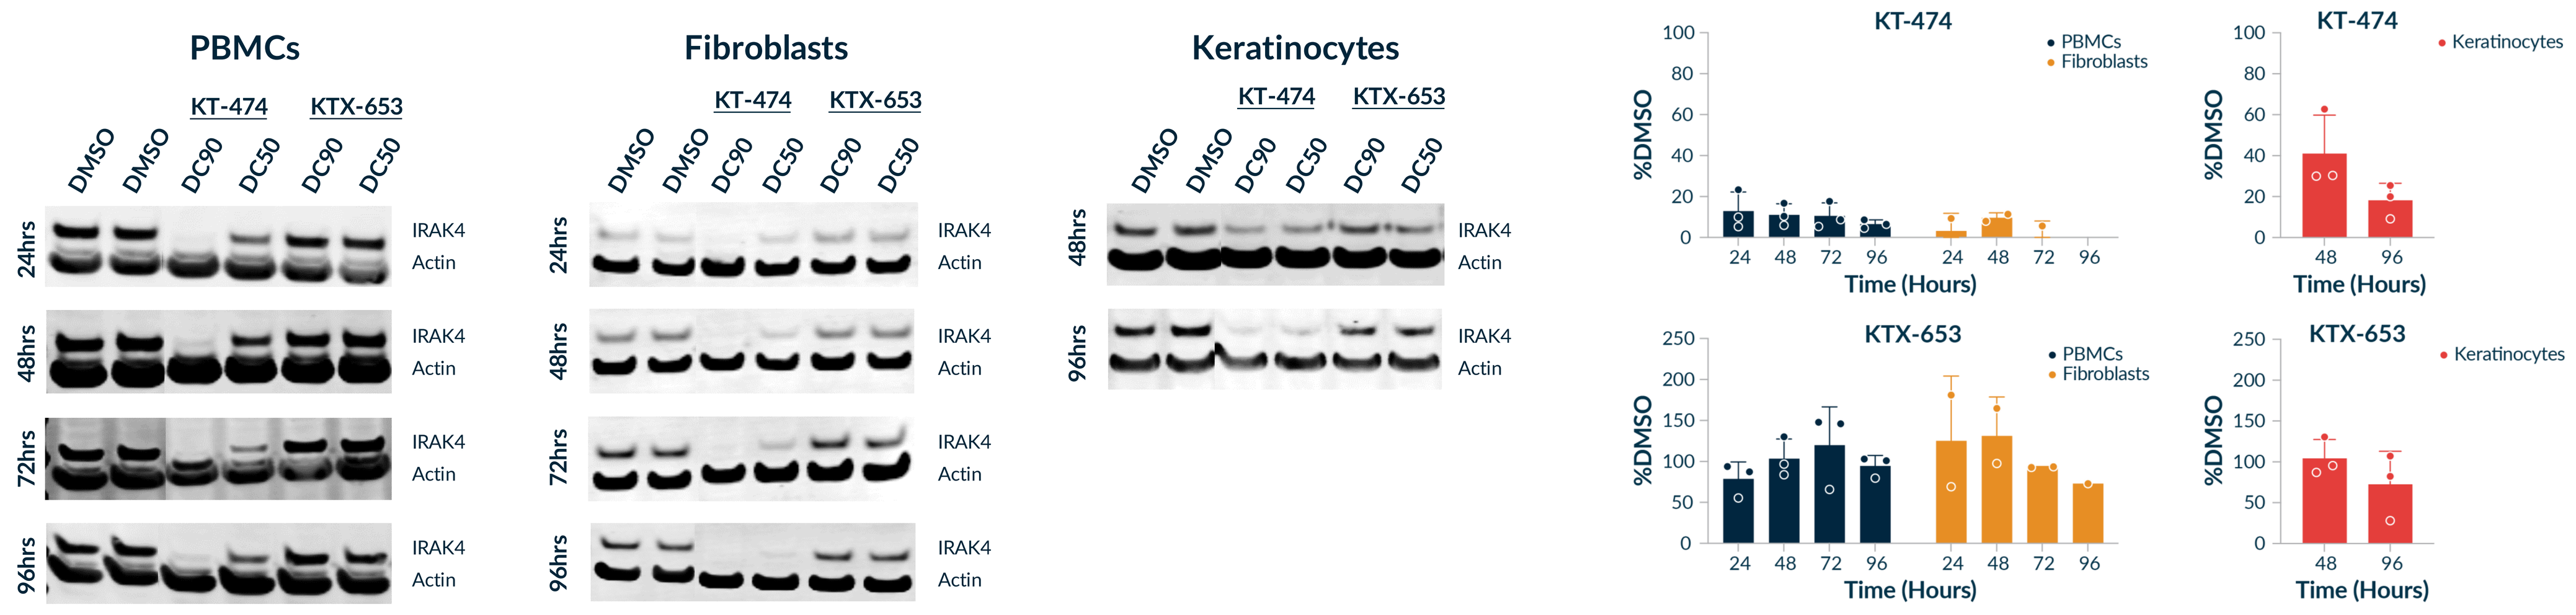
PBMCs
Fibroblasts
Keratinocytes
KT-474
KTX-653
DMSO
DC90
DC50
24hrs
48hrs
72hrs
96hrs
IRAK4
Actin
%DMSO
Time (Hours)
KTX-653

- Maximal degradation of IRAK4 in PBMCs is achieved by 24 hours, with sustained degradation over 96 hours (Max. degradation ~90% compared to DMSO control)
- Fibroblasts treated with KT-474 demonstrate similar kinetics to PBMCs, reaching maximal degradation of IRAK4 by 24 hours following degrader exposure and maintained through 96 hours (Max. degradation ~90%)
- Degradation of IRAK4 in Keratinocytes treated with KT-474 displays slower kinetics, maximal degradation at the dosed concentration occurring after 96 hours
- No degradation observed with the negative control compound, KTX-653 (inactive E3 ligase)

IRAK4 Degraders Inhibit Cytokine Production in IL-1β Stimulated Immune and Cutaneous Cells Types

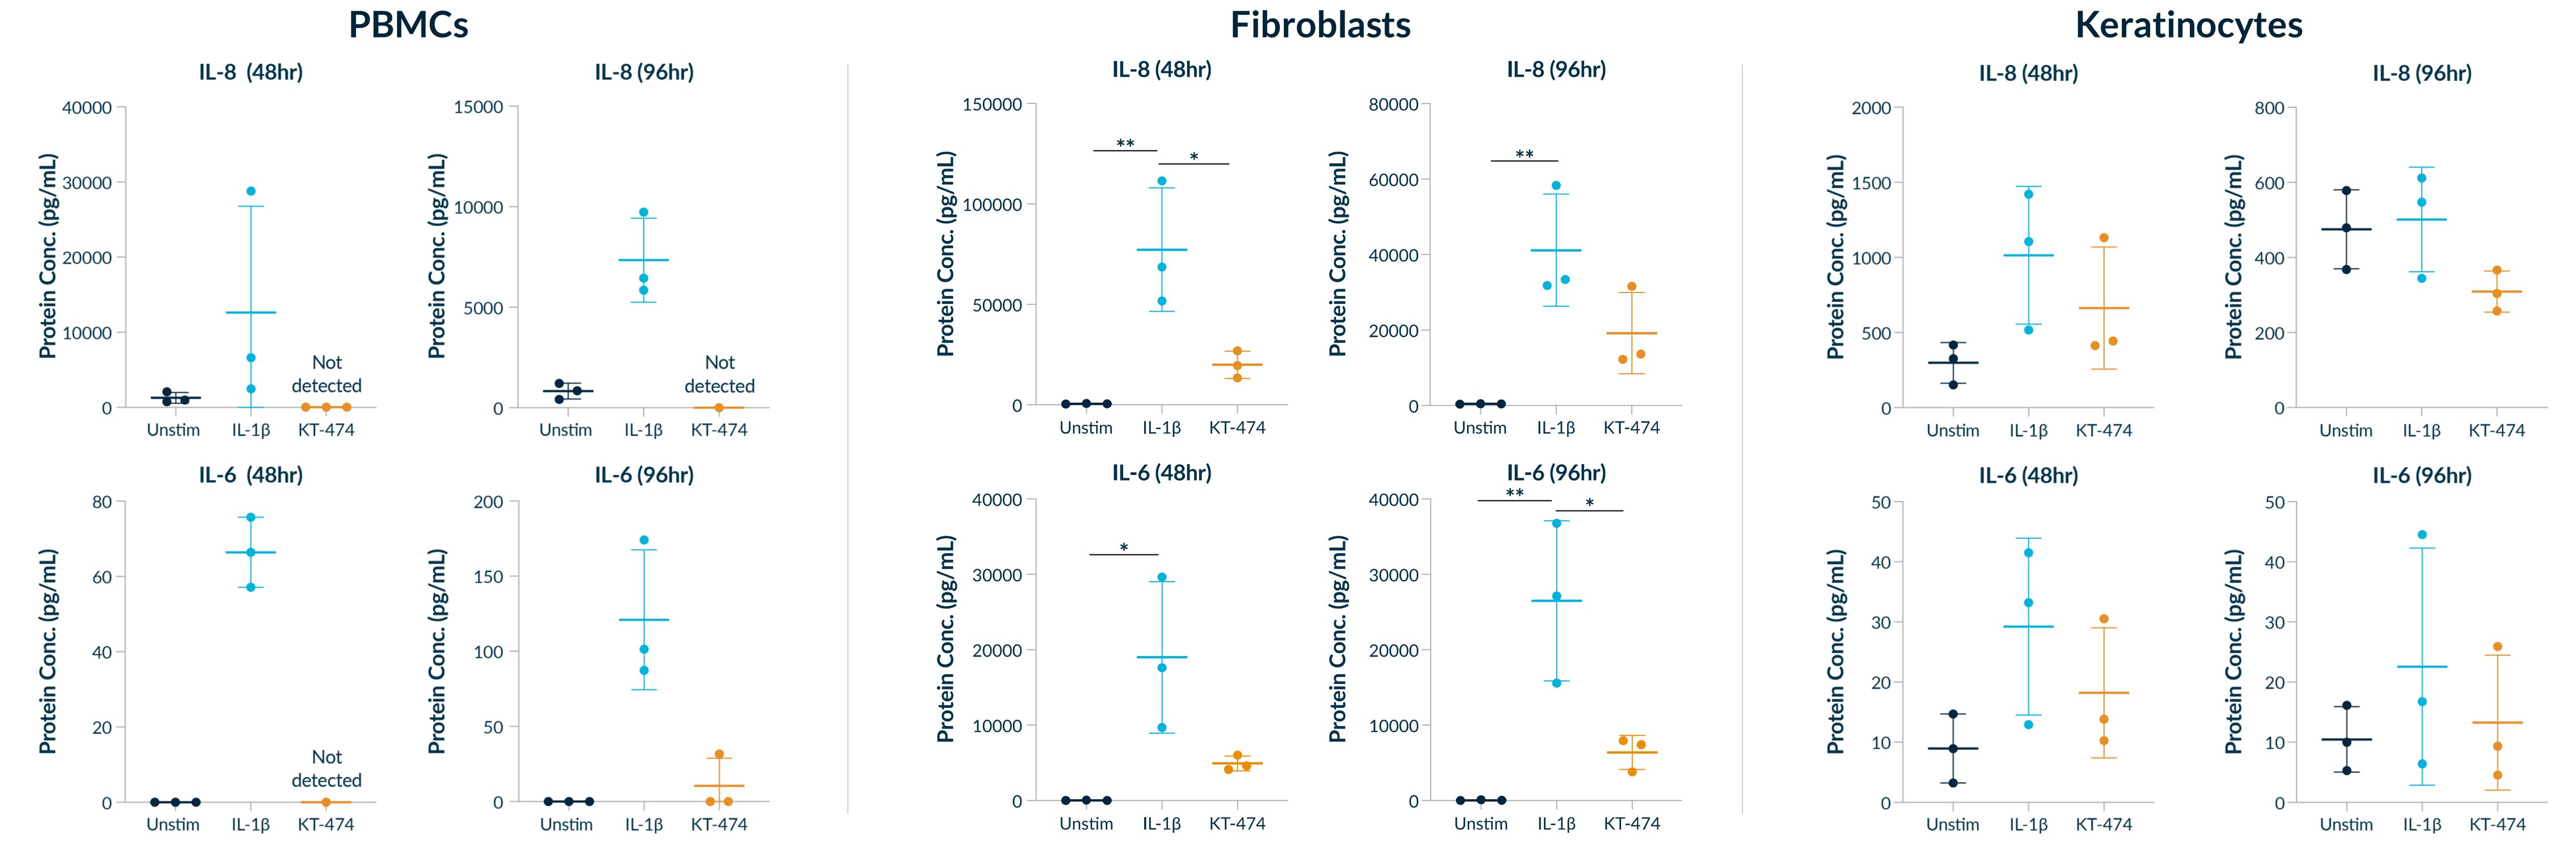
PBMCs
Fibroblasts
Keratinocytes
IL-8 (48hr)
IL-8 (96hr)
IL-6 (48hr)
IL-6 (96hr)
Protein Conc. (pg/mL)
Unstim
IL-1β
KT-474
Not detected

- Induction of cytokine production at baseline in keratinocytes is weaker compared to PBMCs and fibroblasts under IL-1β stimulation
- Strong inhibition is observed in PBMCs at all time points, with cytokine levels returning to baseline with KT-474 treatment
- Cytokine inhibition in fibroblasts with KT-474 treatment is observed across time points, achieving strong inhibition compared to the IL-1β untreated control
- In keratinocytes, KT-474 achieves strong inhibition across time points
- IL-1β induced IL-8 production was weaker at 72 hours, but KT-474 continues to achieve inhibition

Figure 4: Quantification of cytokine production after 24hr or 72hr of IRAK4 degradation followed by an additional 24hrs of IL-1β stimulation (media collected after 48hr or 96hr in culture). One-way ANOVA was performed (n=3 human donors), *p<0.05, **p<0.01

IRAK4 Degradation in Skin and PBMCs in KT-474 Phase 1 Trial

Similar IRAK4 Levels in Skin and PBMC Post KT-474 Treatment at Day 14

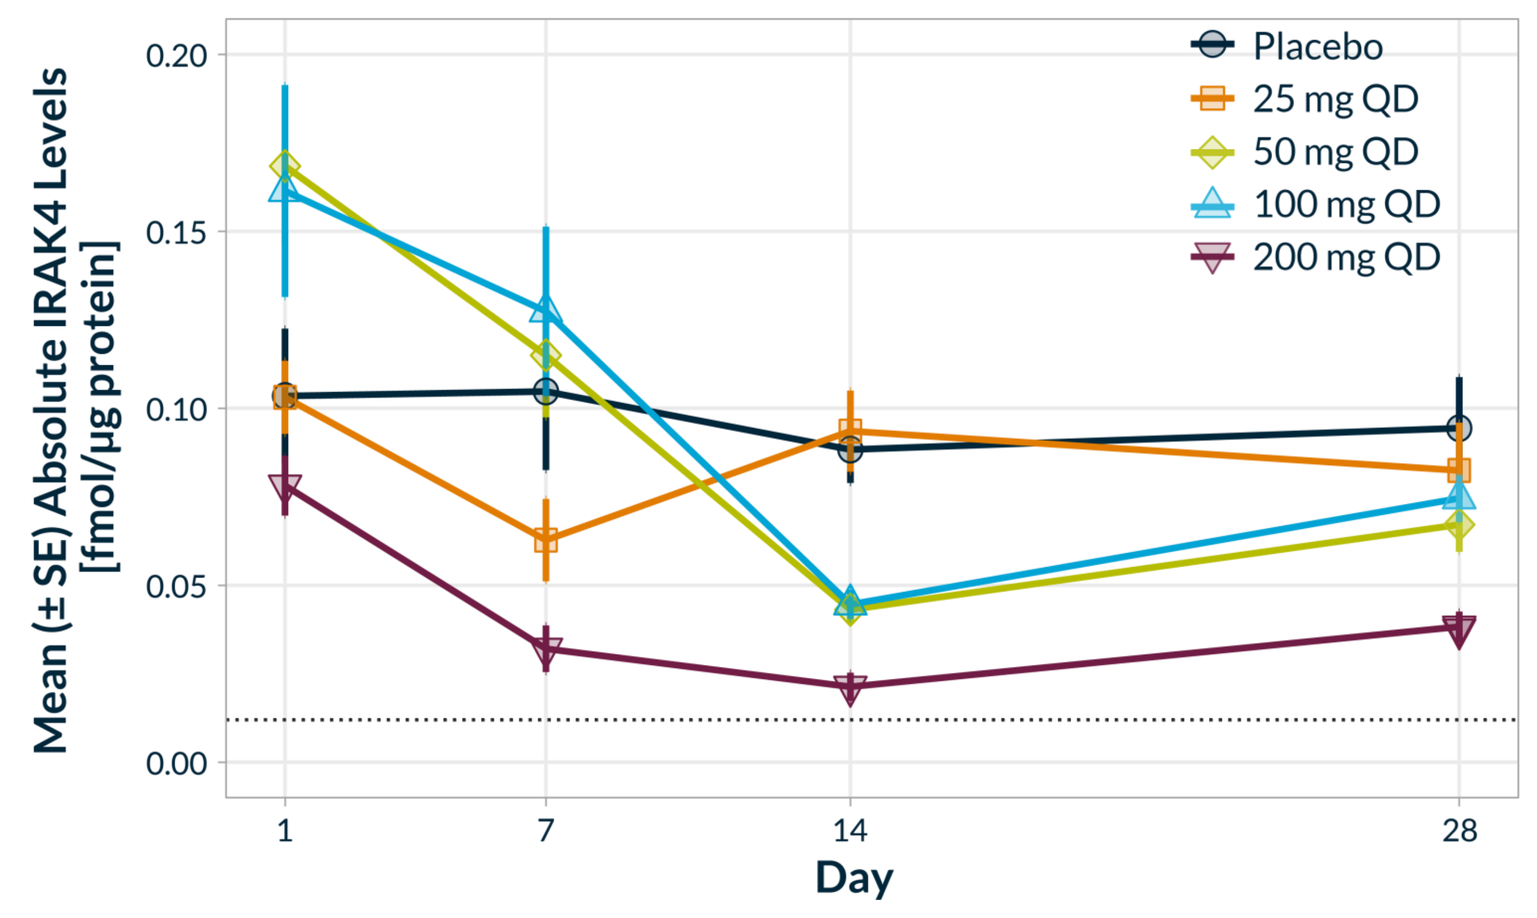
Placebo
25 mg QD
50 mg QD
100 mg QD
200 mg QD
Mean (± SE) Absolute IRAK4 Levels [fmol/μg protein]
Day

Reduction of Absolute IRAK4 Levels in Skin

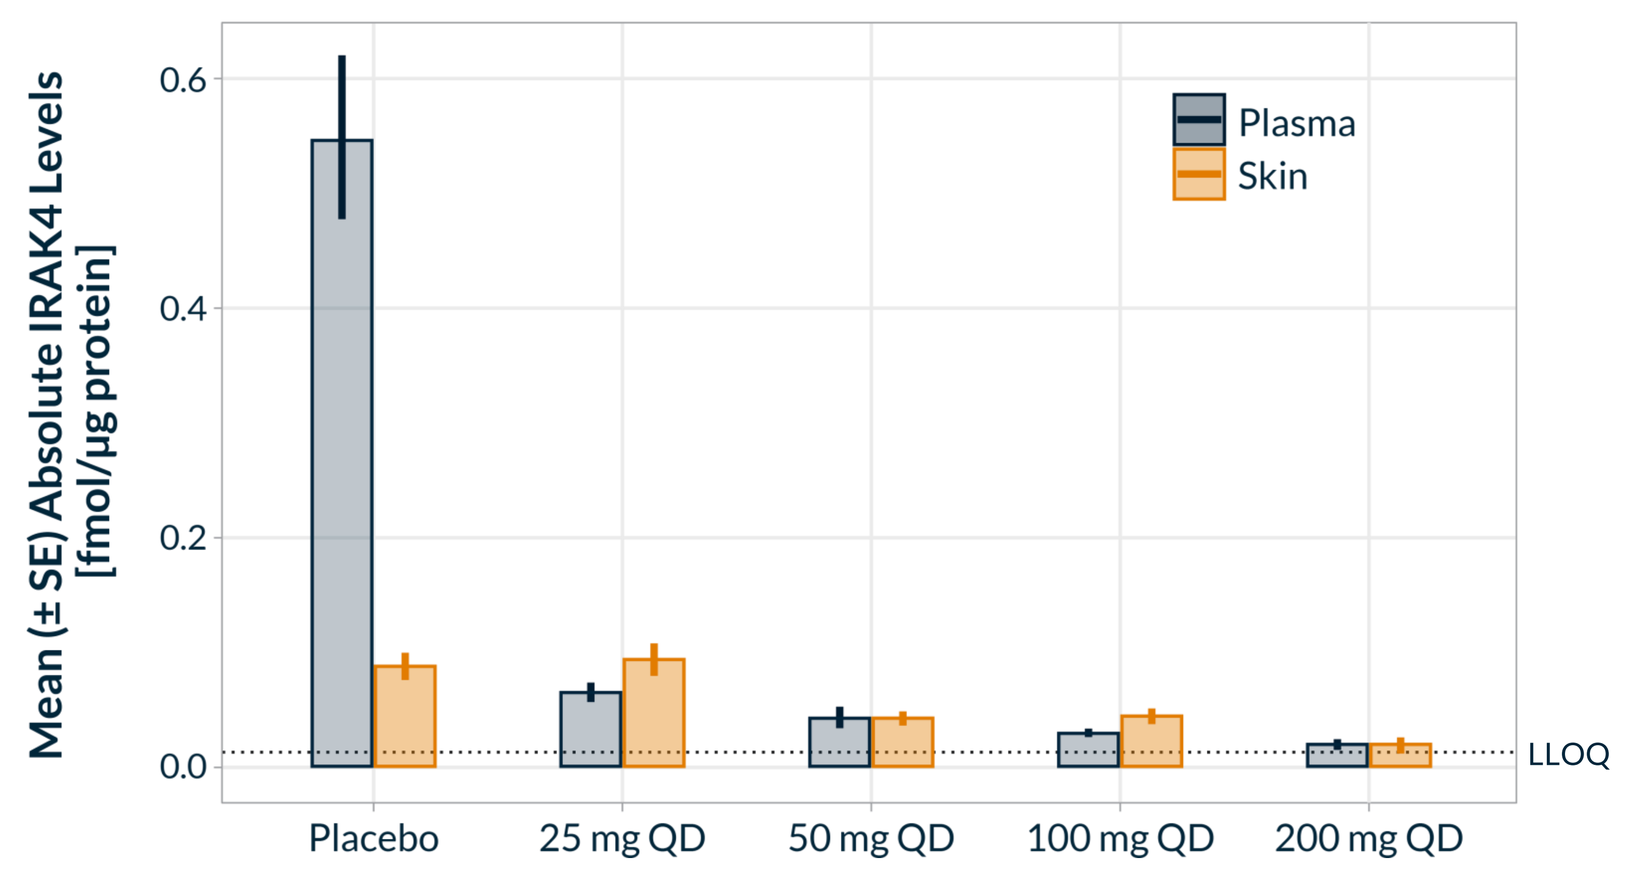
Plasma
Skin
Mean (± SE) Absolute IRAK4 Levels [fmol/μg protein]
LLOQ
Placebo
25 mg QD
50 mg QD
100 mg QD
200 mg QD

Substantial IRAK4 Degradation in Skin Observed in Dermis and Epidermis

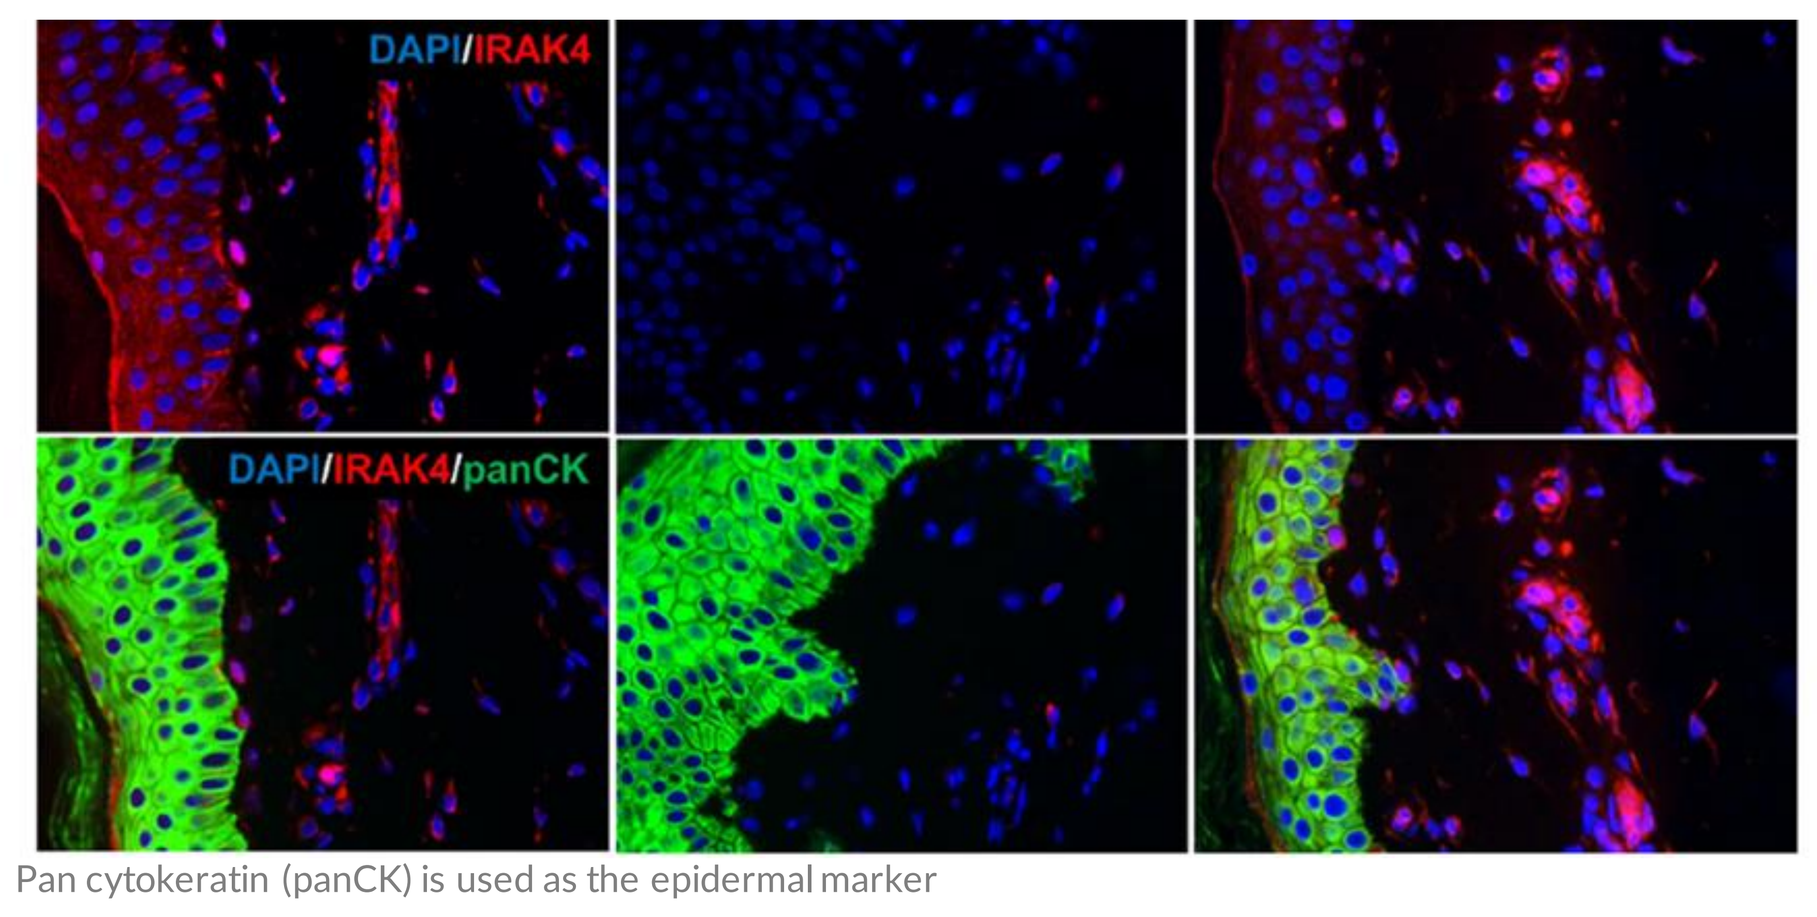
Predose
Day 14
Day 28 (Recovery)
DAPI/IRAK4
DAPI/IRAK4/panCK

Pan cytokeratin (panCK) is used as the epidermal marker

Representative image from patient in 50mg cohort

- Baseline IRAK4 levels in skin substantially lower compared to PBMC
- Mean IRAK4 levels at 200 mg dose nearing the lower limit of quantification (LLOQ) by Day 14
- Robust IRAK4 knockdown also seen at doses of 50-100 mg, with steady state degradation not reached at Day 14
- Comparable degradation in PBMC shows that effect of KT-474 is independent of baseline expression level

CONCLUSIONS

- Collectively, these data demonstrate that KT-474 has potent and broad activity across immune and cutaneous cell types and support development in autoimmune diseases of the skin where IL-1R/TLR plays a central role
- KT-474 induces IRAK4 degradation in Fibroblasts and Keratinocytes, similar to PBMCs, although the kinetics vary across cell types
 - Maximal degradation is observed after 24 hrs of treatment in PBMCs and Fibroblasts *in vitro*, while maximum degradation occurs in keratinocytes after 96hr degrader treatment
- IRAK4 degradation in immune and cutaneous cell types leads to inhibition of pro-inflammatory cytokine production (IL-6 and IL-8) following IL-1β stimulation
- In a Phase 1 trial in healthy volunteers, KT-474 achieved near complete degradation in both PBMC and skin following 14 days of daily dosing

REFERENCES

1. Frew, John W. "Hidradenitis suppurativa is an autoinflammatory keratinization disease: A review of the clinical, histologic, and molecular evidence." JAAD international 1.1 (2020): 62-72.
2. Cai, Yihua, et al. "A critical role of the IL-1β–IL-1R signaling pathway in skin inflammation and psoriasis pathogenesis." Journal of investigative Dermatology 139.1 (2019): 146-156.
3. ClinicalTrials.gov

ACKNOWLEDGEMENTS

Kymera thanks those participating in the Phase 1 trial and contributions of many colleagues who have enabled this work.

The work was done under collaboration agreement with Sanofi.

KYMERA
200 Arsenal Yards Blvd., Suite 230, Watertown, MA 02472, USA
+1 (857) 285-5300 - www.kymera.com

Contact: Emily Lurier / elurier@kymeratx.com

Society for Investigative Dermatology Annual Meeting • May 18-21, 2022 • Portland, OR