Targeted STAT3 Degradation Leads to Remodeling of an Immunosuppressive Tumor Microenvironment and Subsequent Sensitization to Immune Checkpoint Inhibition

#603

Joyoti Dey, Phillip CC Liu, Michele Mayo , Rahul Karnik , Bin Yang, Vaishali Dixit, Jieun Jeong, James Shaw, Yatao Shi, Susanne Breitkopf, Dirk Walther, Kirti Sharma, Jared Gollob and Chris De Savi

Kymera Therapeutics, Watertown, MA

ABSTRACT

Signal Transducer and Activator of Transcription 3 (STAT3), a multifaceted transcription factor, is aberrantly activated across a variety of malignancies; however, its selective targeting has to-date remained a therapeutic challenge.

STAT3 plays a pivotal role in shaping the tumor immune landscape through cancer cell-intrinsic mechanisms, direct regulation of immune cell function and cancer cell- tumor microenvironment (TME) crosstalk, which collectively result in an immunosuppressive TME.

Targeted protein degradation represents a novel therapeutic modality enabling direct targeting of previously undruggable oncoproteins. Here, we investigated the immunomodulatory impact of STAT3 degradation on tumorigenesis in syngeneic mouse models representing cancers with heterogeneous immune milieus.

Tumor cell intrinsic and extrinsic functions of STAT3

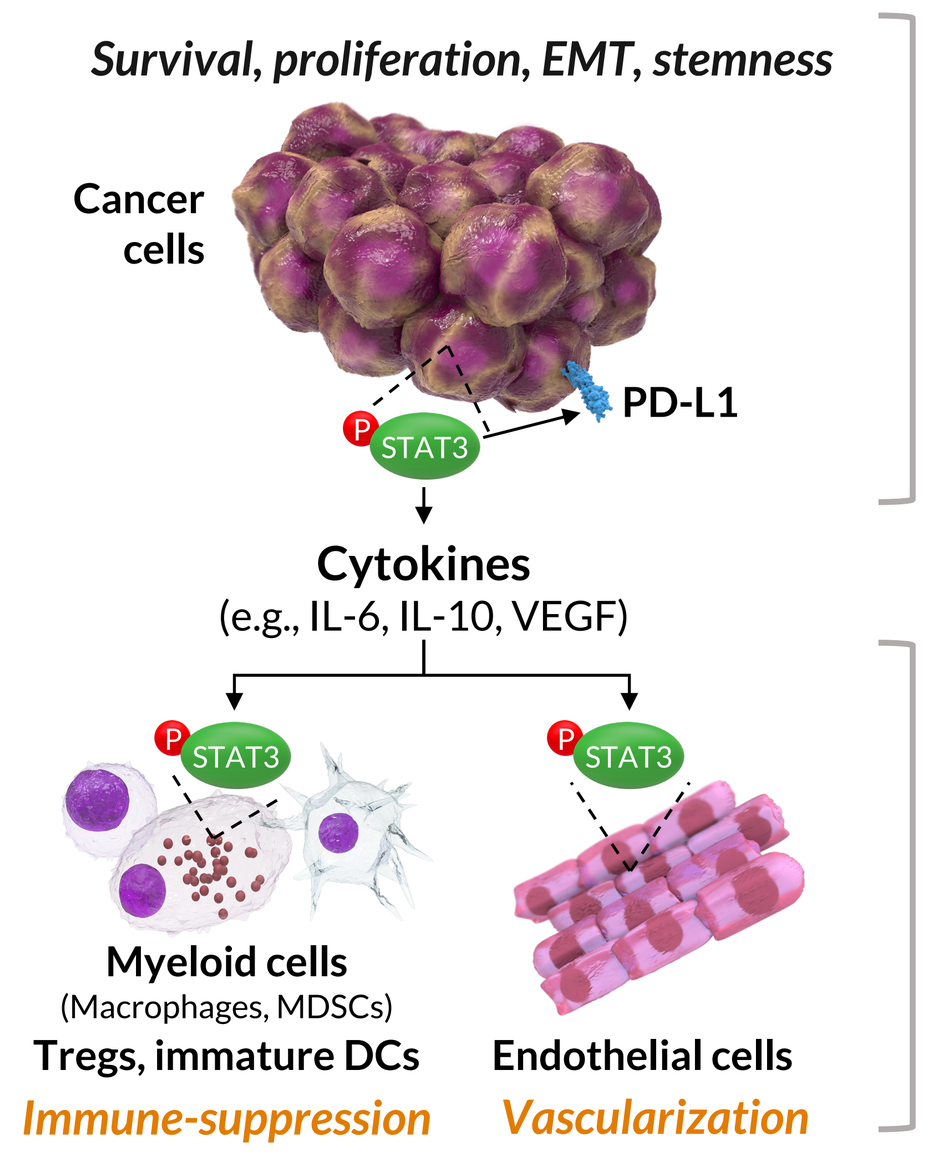
Survival, proliferation, EMT, stemness
Cancer cells
PD-L1
STAT3
Cytokines
(e.g., IL-6, IL-10, VEGF)
Myeloid cells
(Macrophages, MDSCs)
Tregs, immature DCs
Immune-suppression
Endothelial cells
Vascularization

Tumor intrinsic

- STAT3 promotes gene expression programs involved with survival, proliferation, stemness and metastasis of tumor cells
- In PTCL, activated STAT3 (phospho-STAT3) is detected in many subtypes due to: STAT3 mutation (ALK-ALCL, T-LGLL, NKTCL, AITL, PTCL-NOS), upstream kinase activation, ALK, JAK (ALK+ALCL, ALK-ALCL), and elevated inflammatory mediators (CTCL)[1-6]

Tumor extrinsic

- STAT3 promotes the differentiation and activity of immunosuppressive and endothelial cells, resulting in an immunosuppressive tumor microenvironment
- Degradation of STAT3 provides therapeutic opportunities in multiple heme and solid tumor indications that are not responsive to immune checkpoint inhibitors

Figure 1: Targeted protein degradation of STAT3

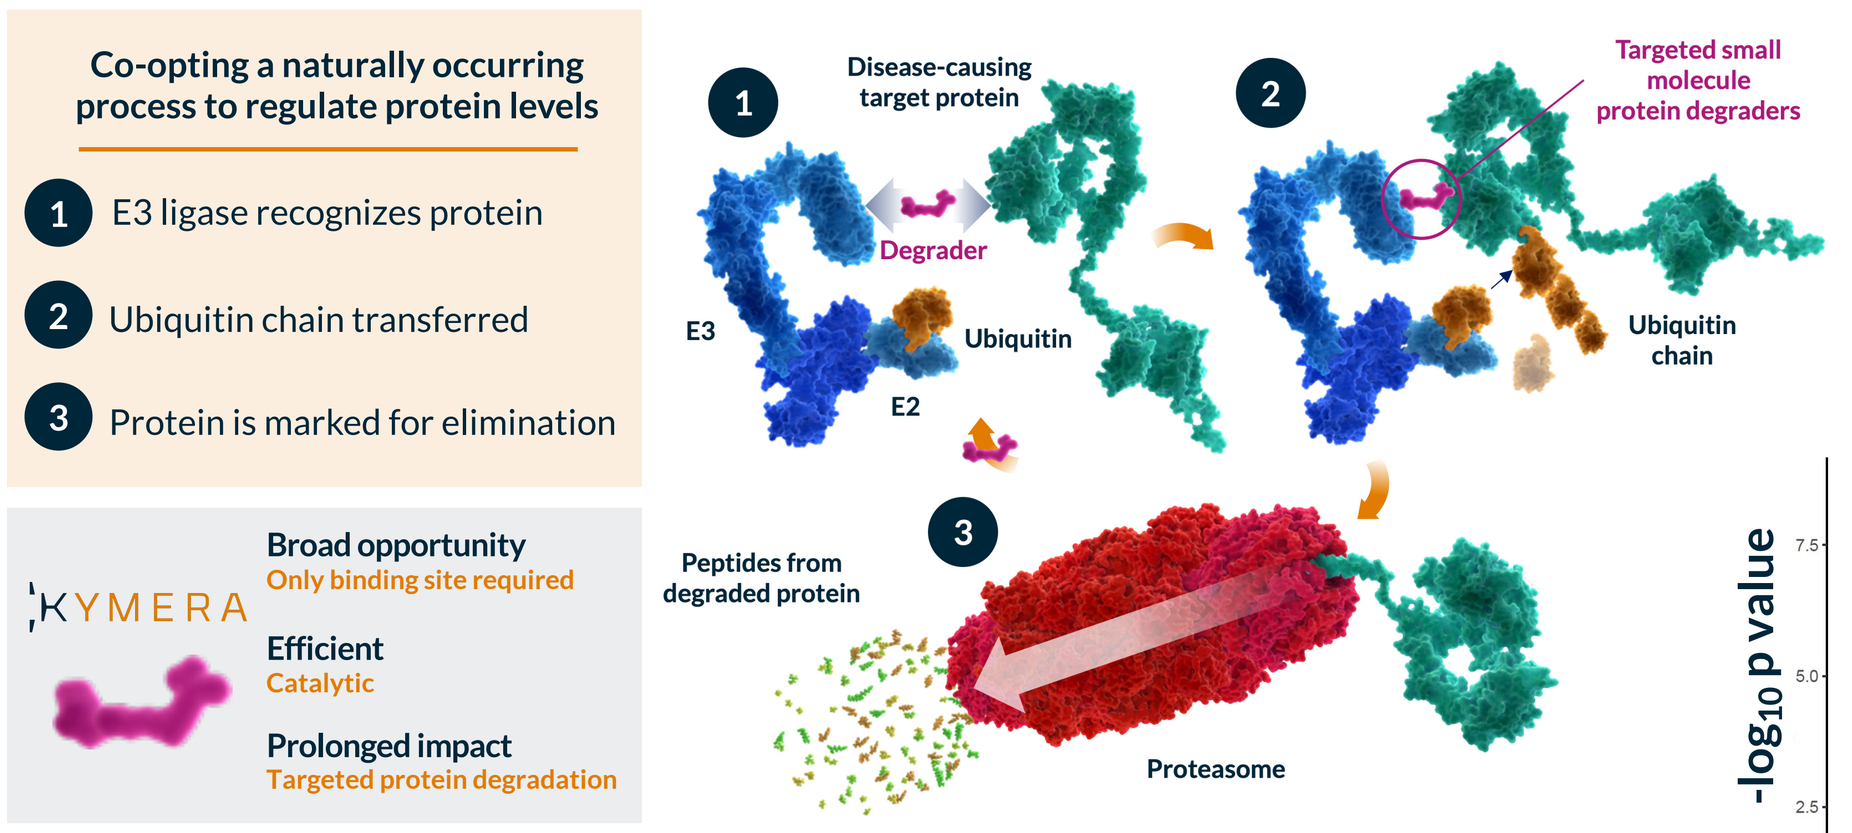
Co-opting a naturally occurring process to regulate protein levels
1 E3 ligase recognizes protein
2 Ubiquitin chain transferred
3 Protein is marked for elimination
KYMERA
Broad opportunity
Only binding site required
Efficient
Catalytic
Prolonged impact
Targeted protein degradation
Disease-causing target protein
Degrader
E3
E2
Ubiquitin
Targeted small molecule protein degraders
Ubiquitin chain
Peptides from degraded protein
Proteasome

KTX-201 is a potent and selective degrader of STAT3 as demonstrated in human immune cells and cancer cell lines

Cell Line	STAT3 DC_{50} (nM)
Monocyte	0.15
Lymphocyte	5.4
SU-DHL-1	11.7
SUP-M2	16.3

Selectivity analysis

Deep tandem mass tag proteomics (>10,000 proteins monitored)

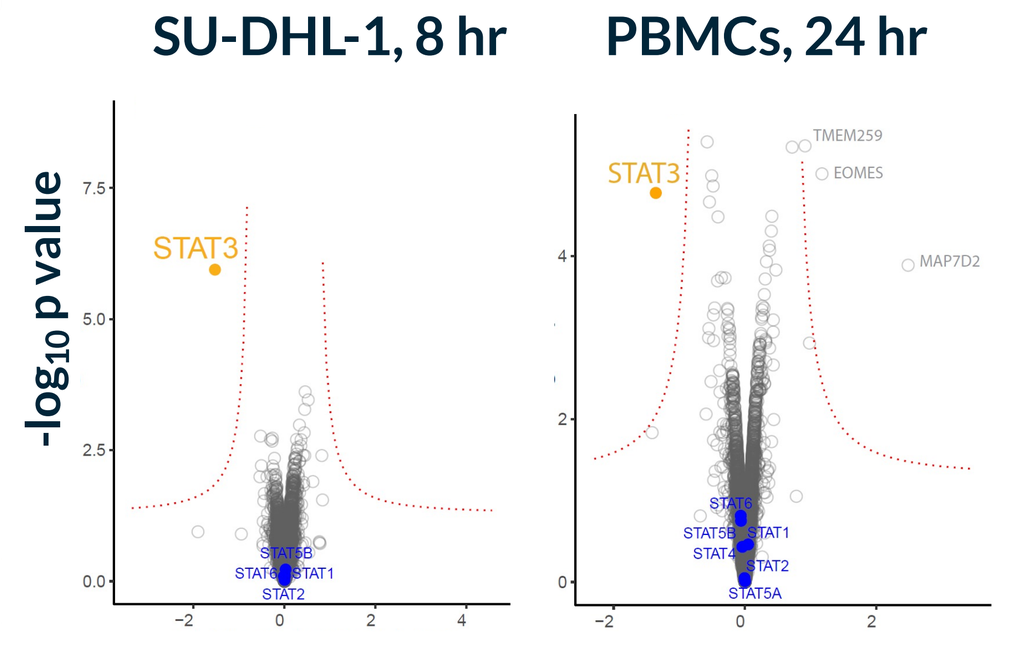
SU-DHL-1, 8 hr
PBMCs, 24 hr
$-\log_{10}$ p value
STAT3
KTX-201 DC90/DMSO
KTX-201 DC90/DMSO

Figure 2: STAT3 degraders have *in vivo* anti-tumor activity in syngeneic mouse models of cancer

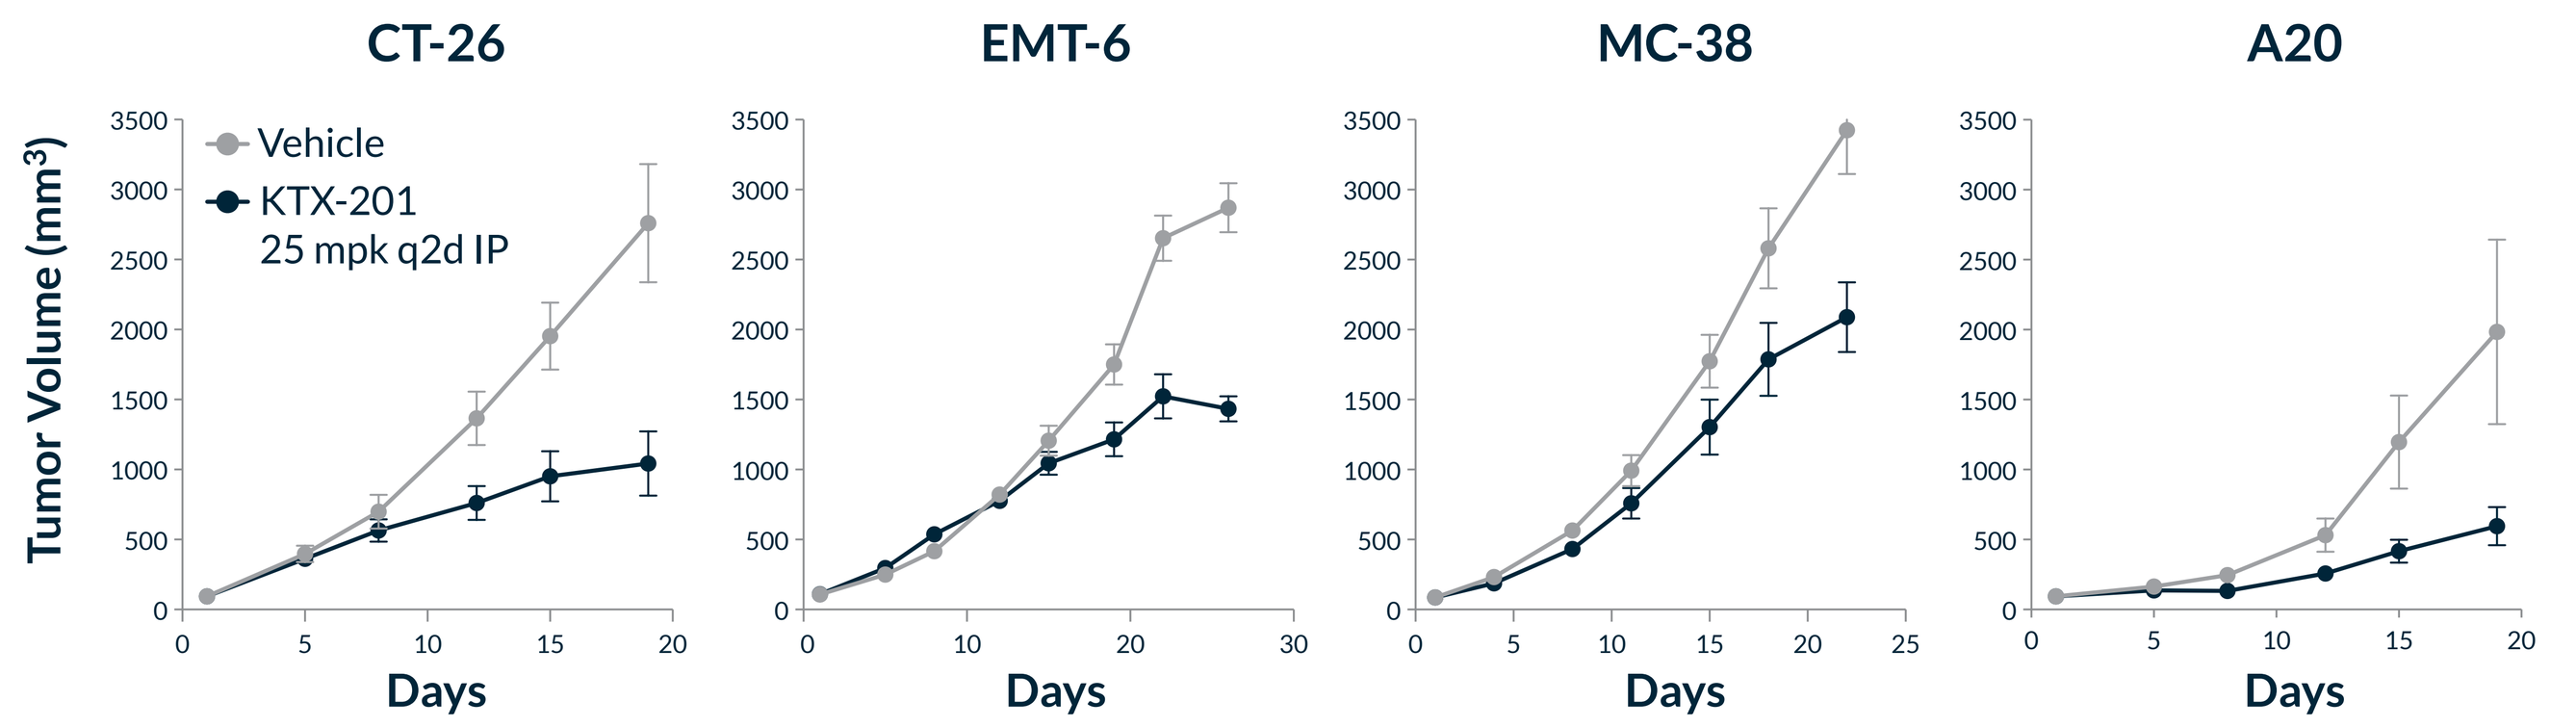
CT-26
EMT-6
MC-38
A20
Tumor Volume (mm³)
Vehicle
KTX-201
25 mpk q2d IP
Days

Representative IHC showing degradation of STAT3 in tumor and TME

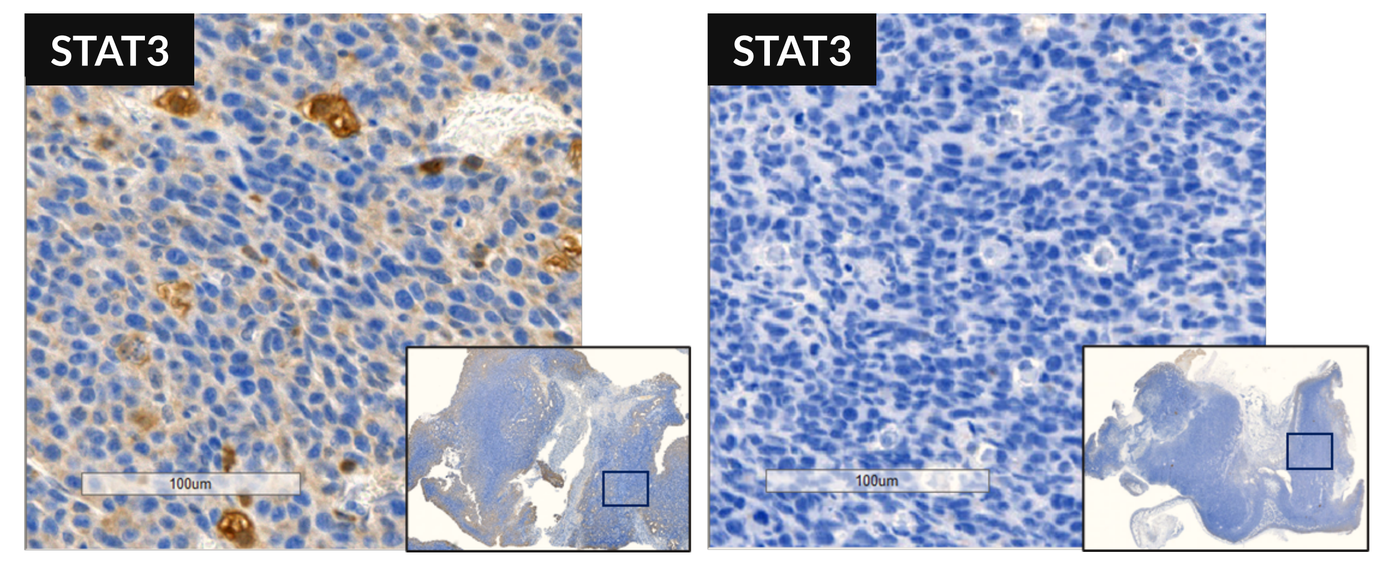
STAT3
STAT3

STAT3 is potently degraded *in vitro* but cell viability is not impacted in the absence of a TME

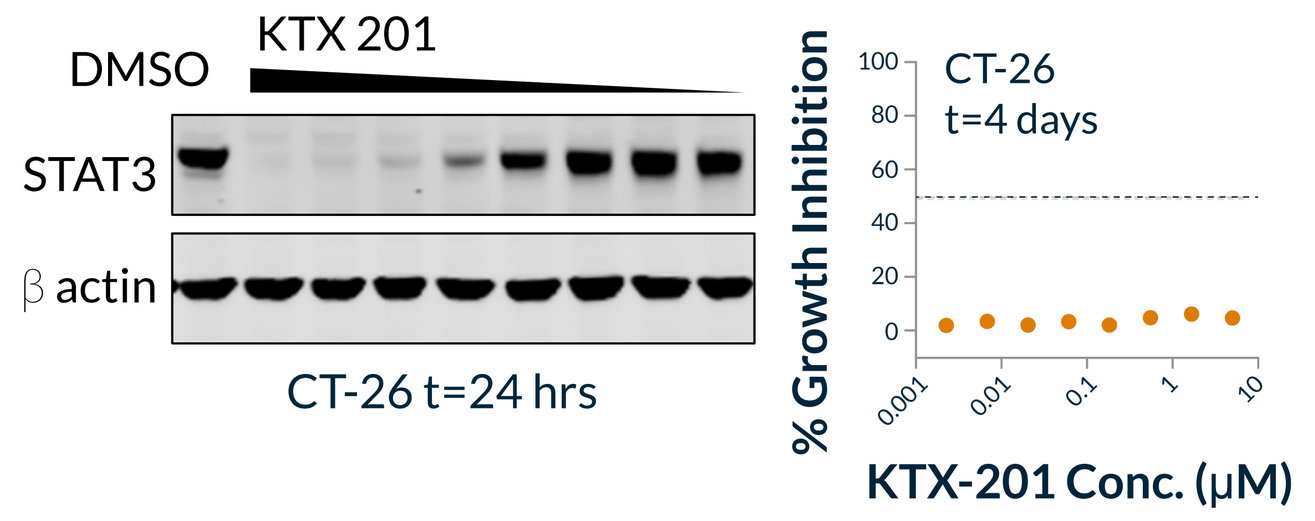
DMSO
KTX 201
STAT3
β actin
CT-26 t=24 hrs
% Growth Inhibition
CT-26
t=4 days
KTX-201 Conc. (µM)

Figure 3: Anti-tumor activity of STAT3 degradation is driven by immune-directed mechanisms

KTX-201 treatment results in increased M1/M2 ratio of tumor-associated macrophages and CD8+ T cells

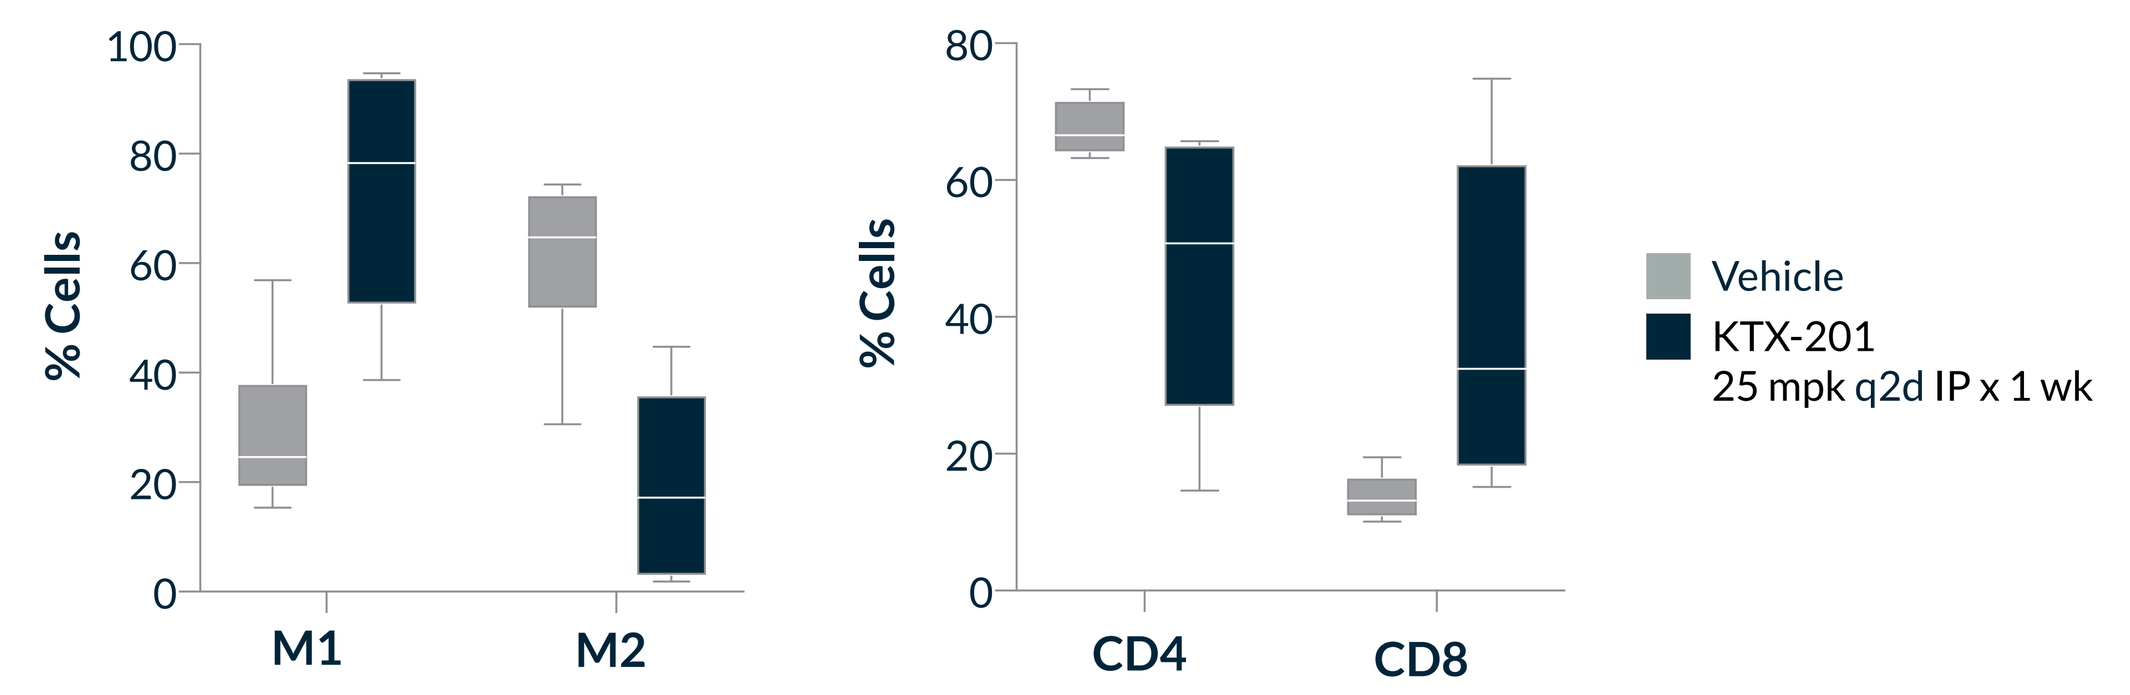
% Cells
M1
M2
CD4
CD8
Vehicle
KTX-201
25 mpk q2d IP x 1 wk

CD8+ T cell depletion abrogates KTX-201-mediated anti-tumor efficacy

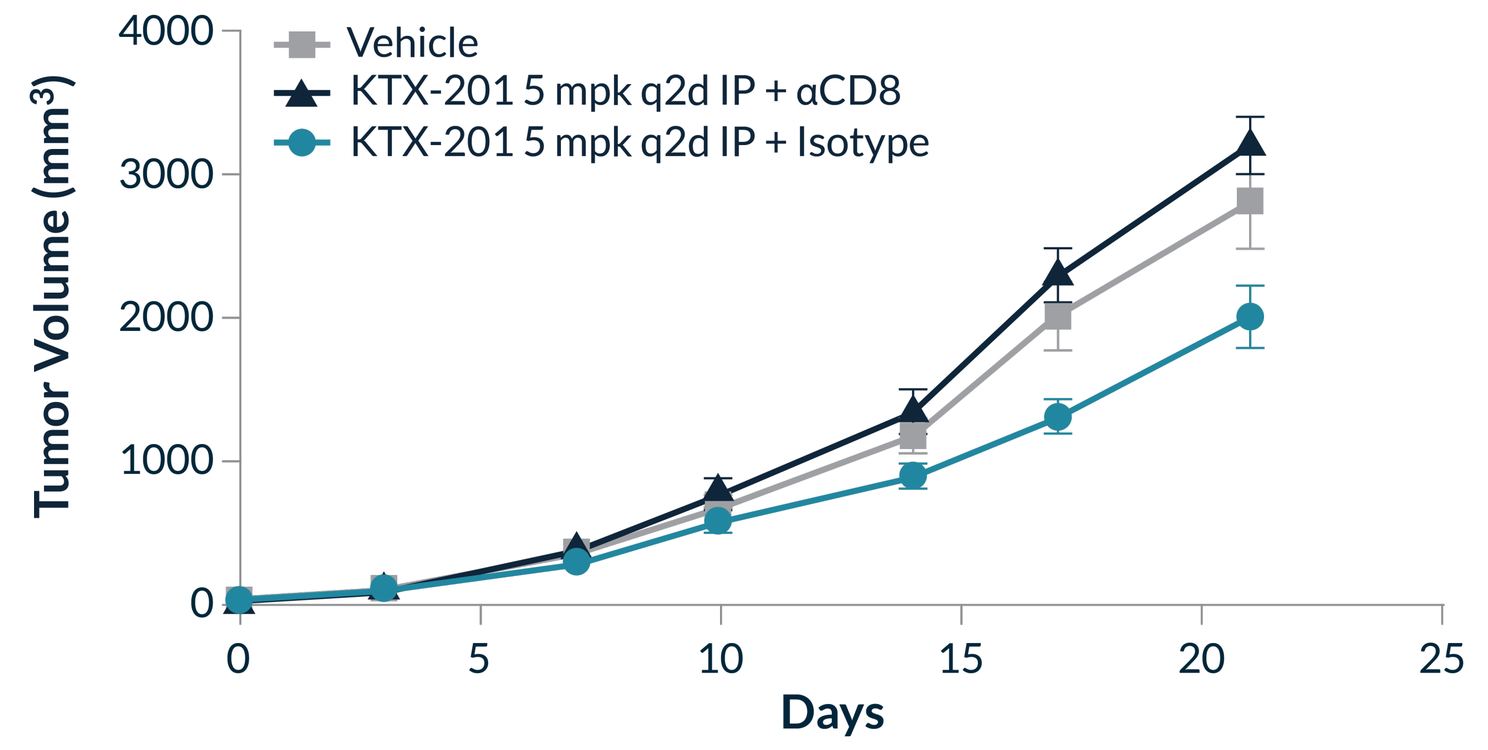
Tumor Volume (mm³)
Vehicle
KTX-201 5 mpk q2d IP + αCD8
KTX-201 5 mpk q2d IP + Isotype
Days

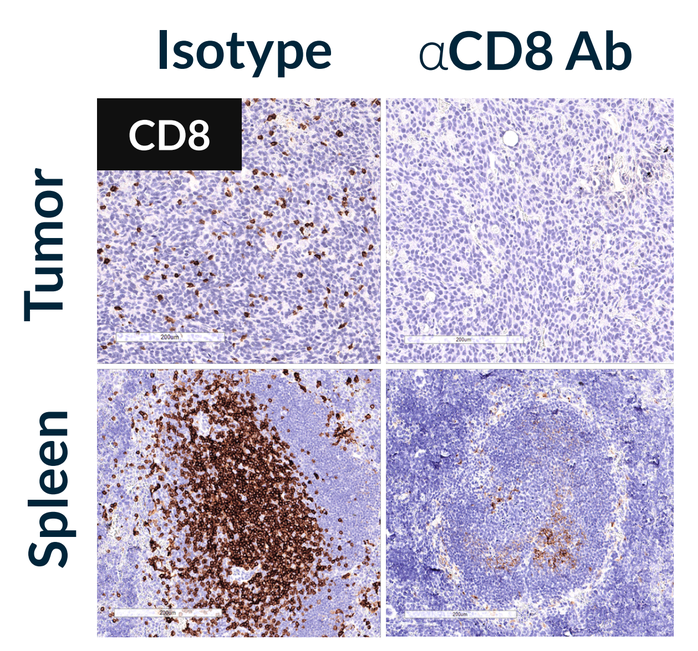
Isotype
αCD8 Ab
CD8
Tumor
Spleen

IHC confirmation of CD8+ T cell depletion

Figure 4: STAT3 degradation-induced remodeling results in an immune-favorable TME

Transcriptomic analysis shows STAT3 degradation induces anti-tumorigenic changes as well as compensatory mechanisms in the TME of CT-26 tumors

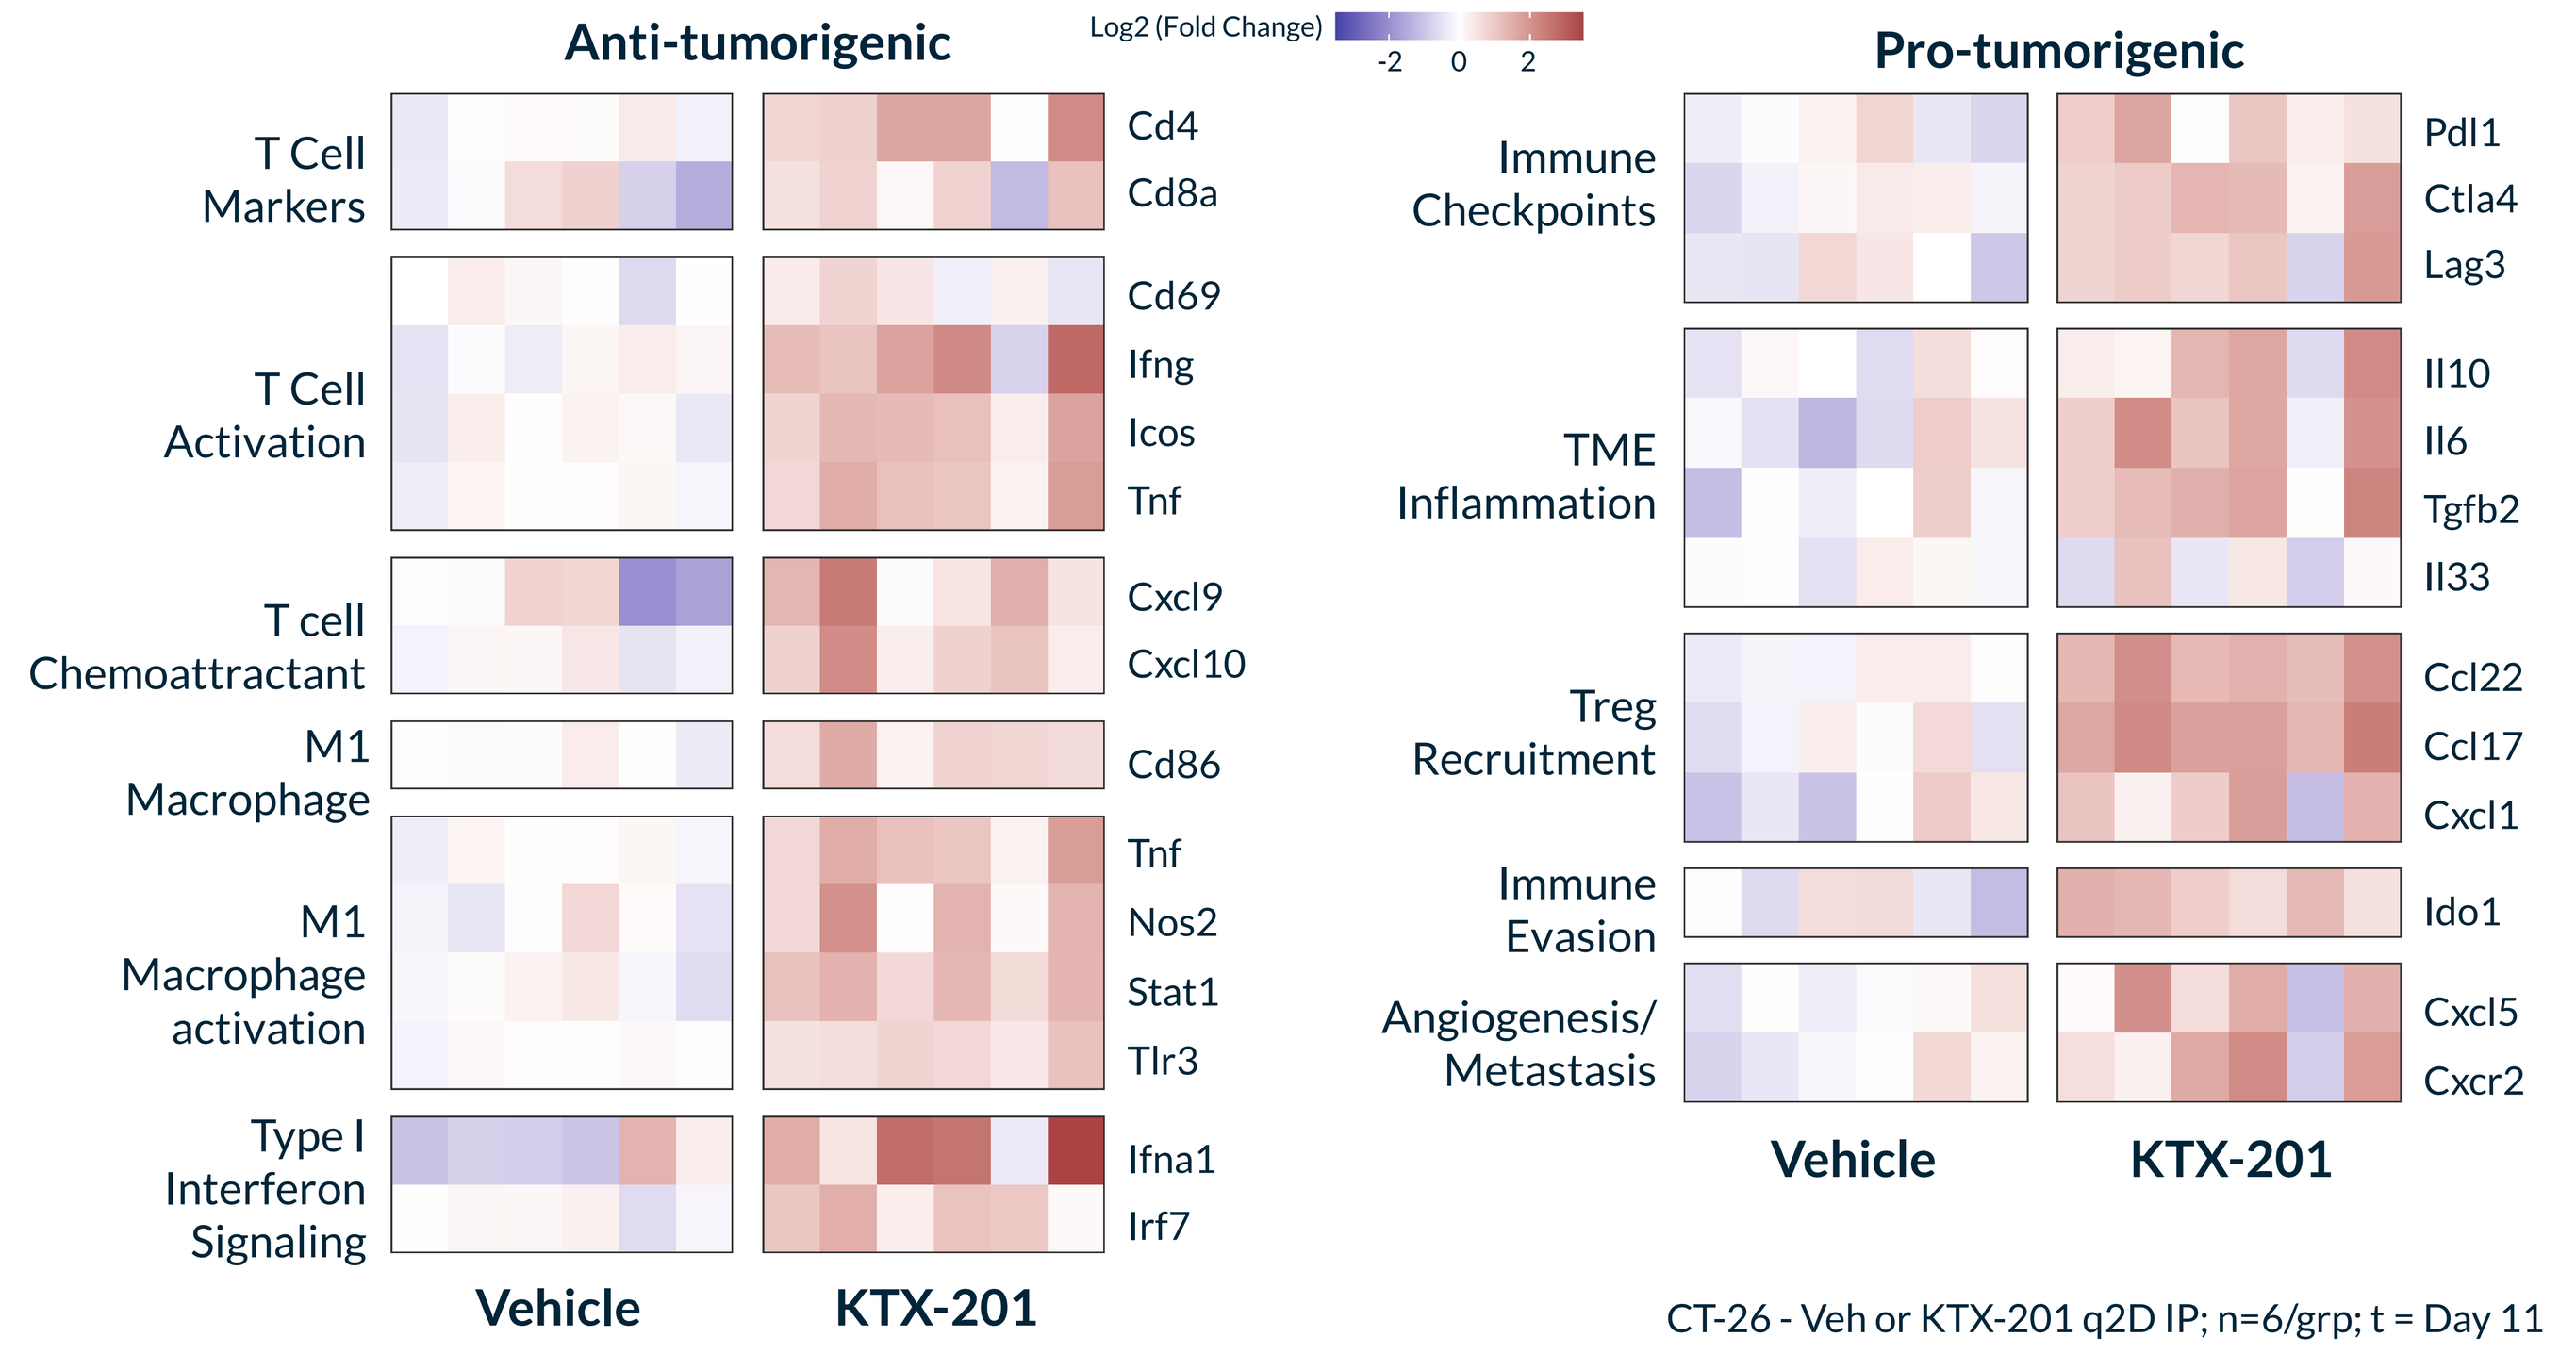
Anti-tumorigenic
Log2 (Fold Change)
Pro-tumorigenic
T Cell Markers
T Cell Activation
T cell Chemoattractant
M1 Macrophage
M1 Macrophage activation
Type I Interferon Signaling
Cd4
Cd8a
Cd69
Ifng
Icos
Tnf
Cxcl9
Cxcl10
Cd86
Tnf
Nos2
Stat1
Tlr3
Ifna1
Irf7
Immune Checkpoints
TME Inflammation
Treg Recruitment
Immune Evasion
Angiogenesis/ Metastasis
Pdl1
Ctla4
Lag3
Il10
Il6
Tgfb2
Il33
Ccl22
Ccl17
Cxcl1
Ido1
Cxcl5
Cxcr2
Vehicle
KTX-201

CT-26 - Veh or KTX-201 q2D IP; n=6/grp; t = Day 11

Figure 5: STAT3 degradation enriches for an IFNγ-dependent gene signature predictive of sensitivity to anti-PD-1[1,2]

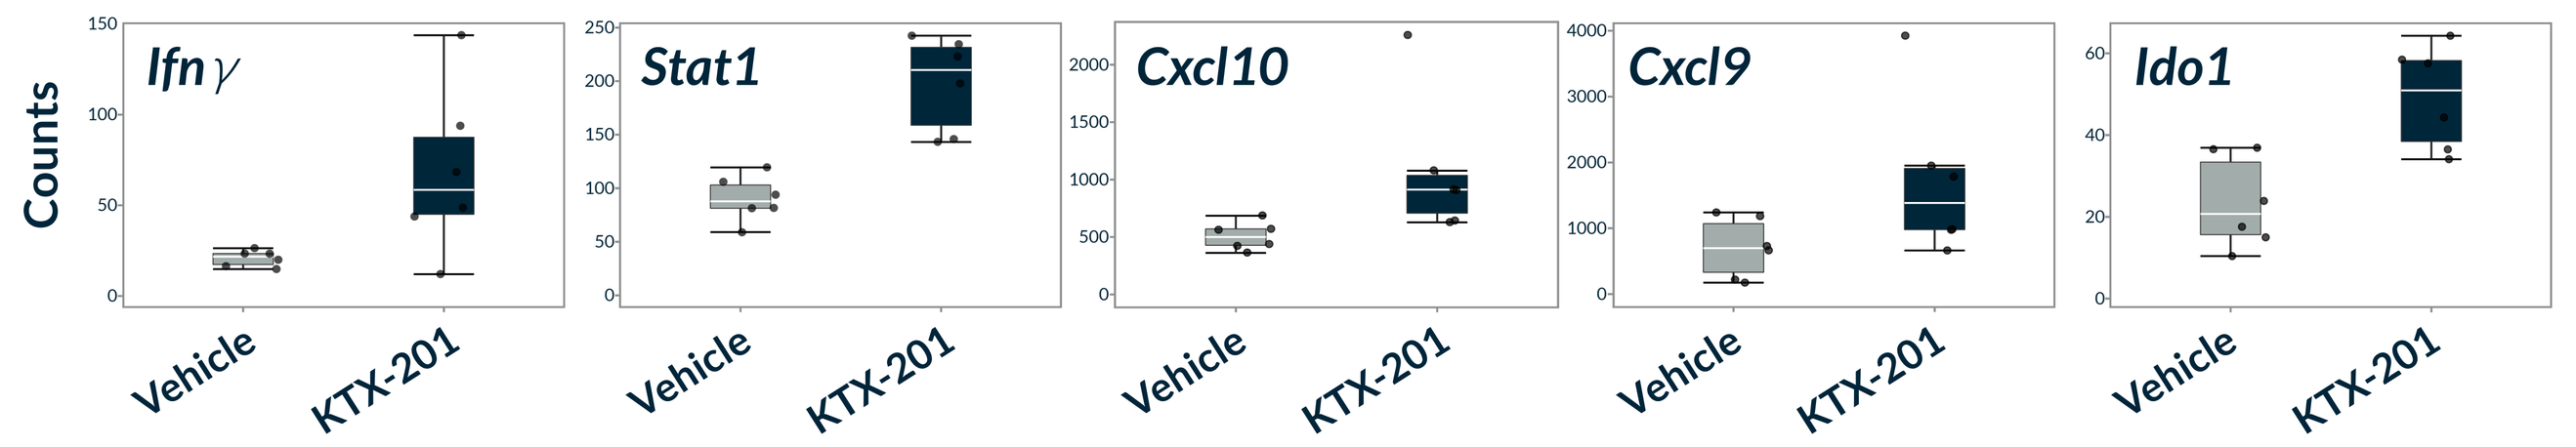
CT-26 - Veh or KTX 201 25 mpk q2D IP
n=6/grp; t = Day 11
Counts
Ifnγ
Stat1
Cxcl10
Cxcl9
Ido1
Vehicle
KTX-201

Increased *STAT1*+ TAMs in KTX-201-treated TME support IFN-primed M1 macrophage polarization

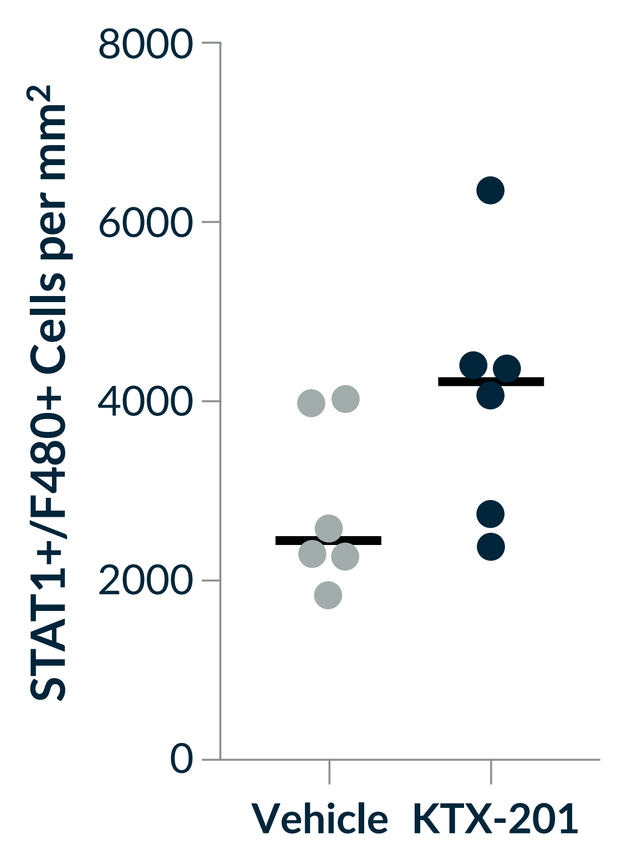
STAT1+/F480+ Cells per mm²
Vehicle
KTX-201
CT-26 - n=6/grp; t = Day 11

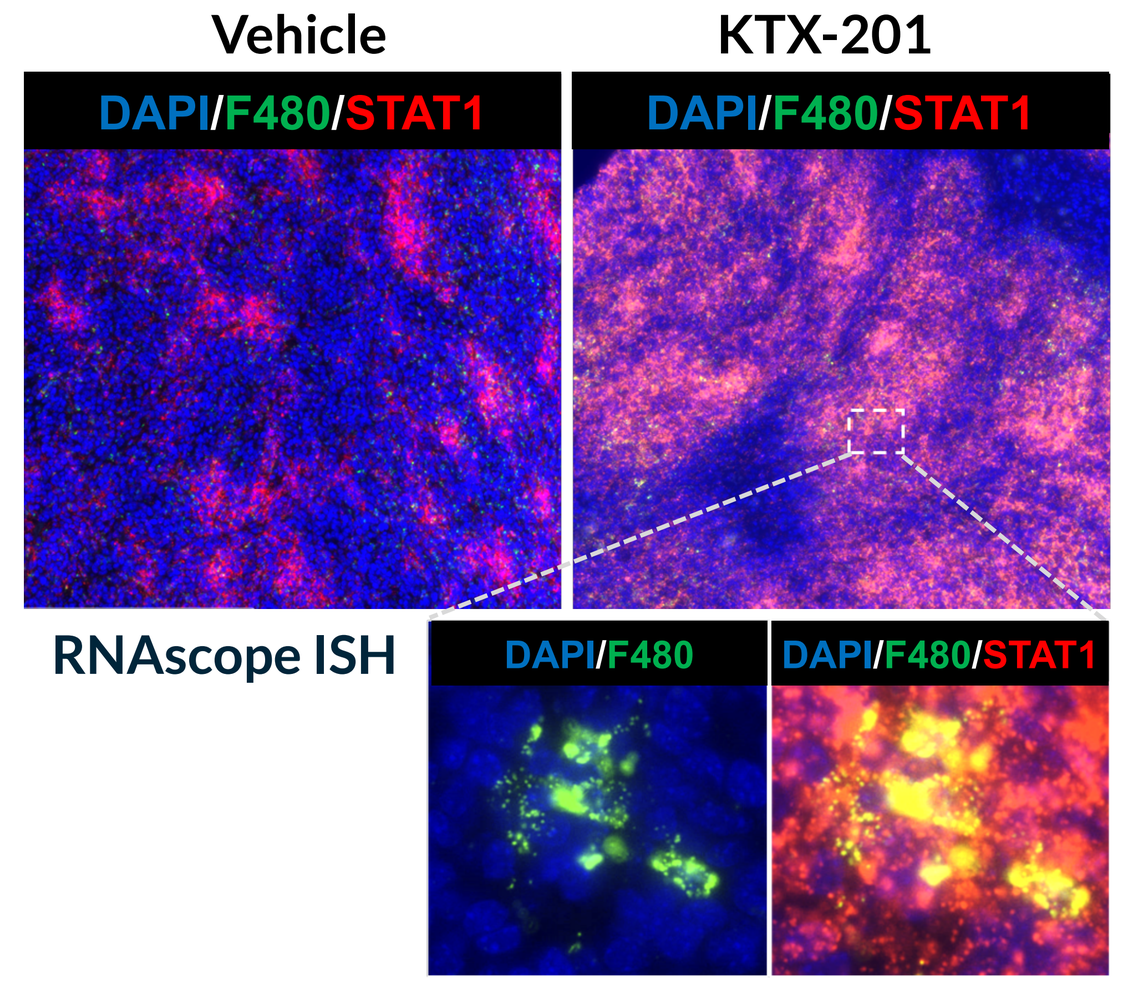
Vehicle
KTX-201
DAPI/F480/STAT1
DAPI/F480/STAT1
RNAscope ISH
DAPI/F480
DAPI/F480/STAT1

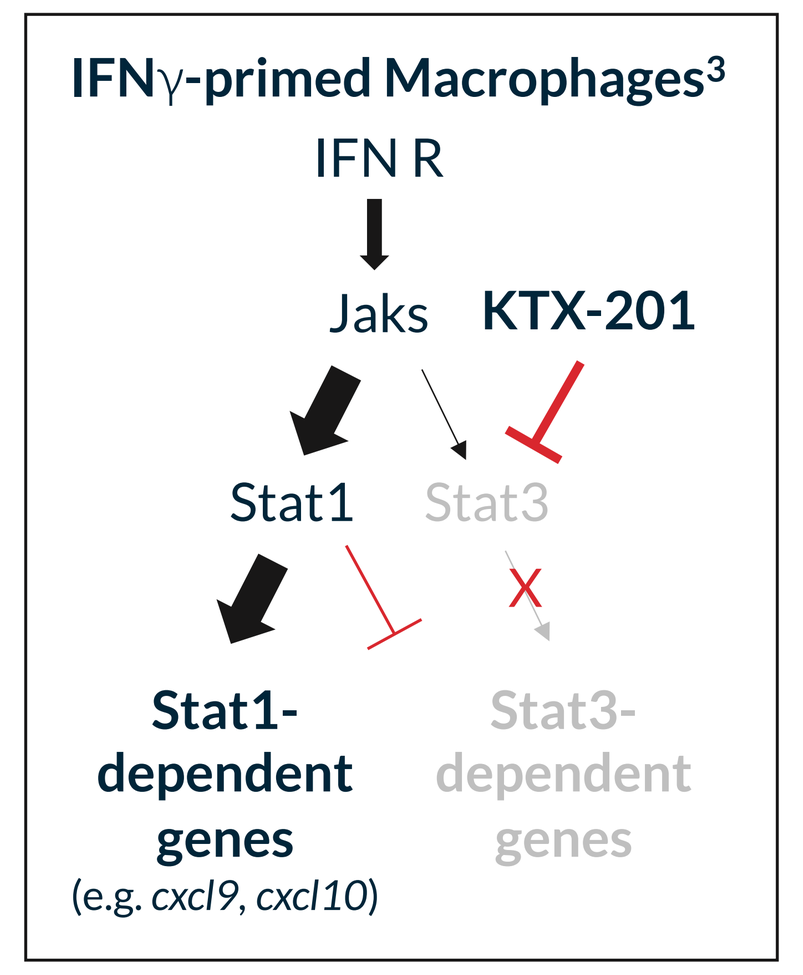
IFNγ-primed Macrophages³
IFN R
Jaks
KTX-201
Stat1
Stat3
Stat1-dependent genes
(e.g. cxcl9, cxcl10)
Stat3-dependent genes

Figure 6: STAT3 degradation enhances anti-PD-1 responses in mouse models of solid and hematologic cancers

KTX-201 synergizes with anti-PD-1 leading to 60% complete response and development of long-term immunological memory in CT-26 tumors

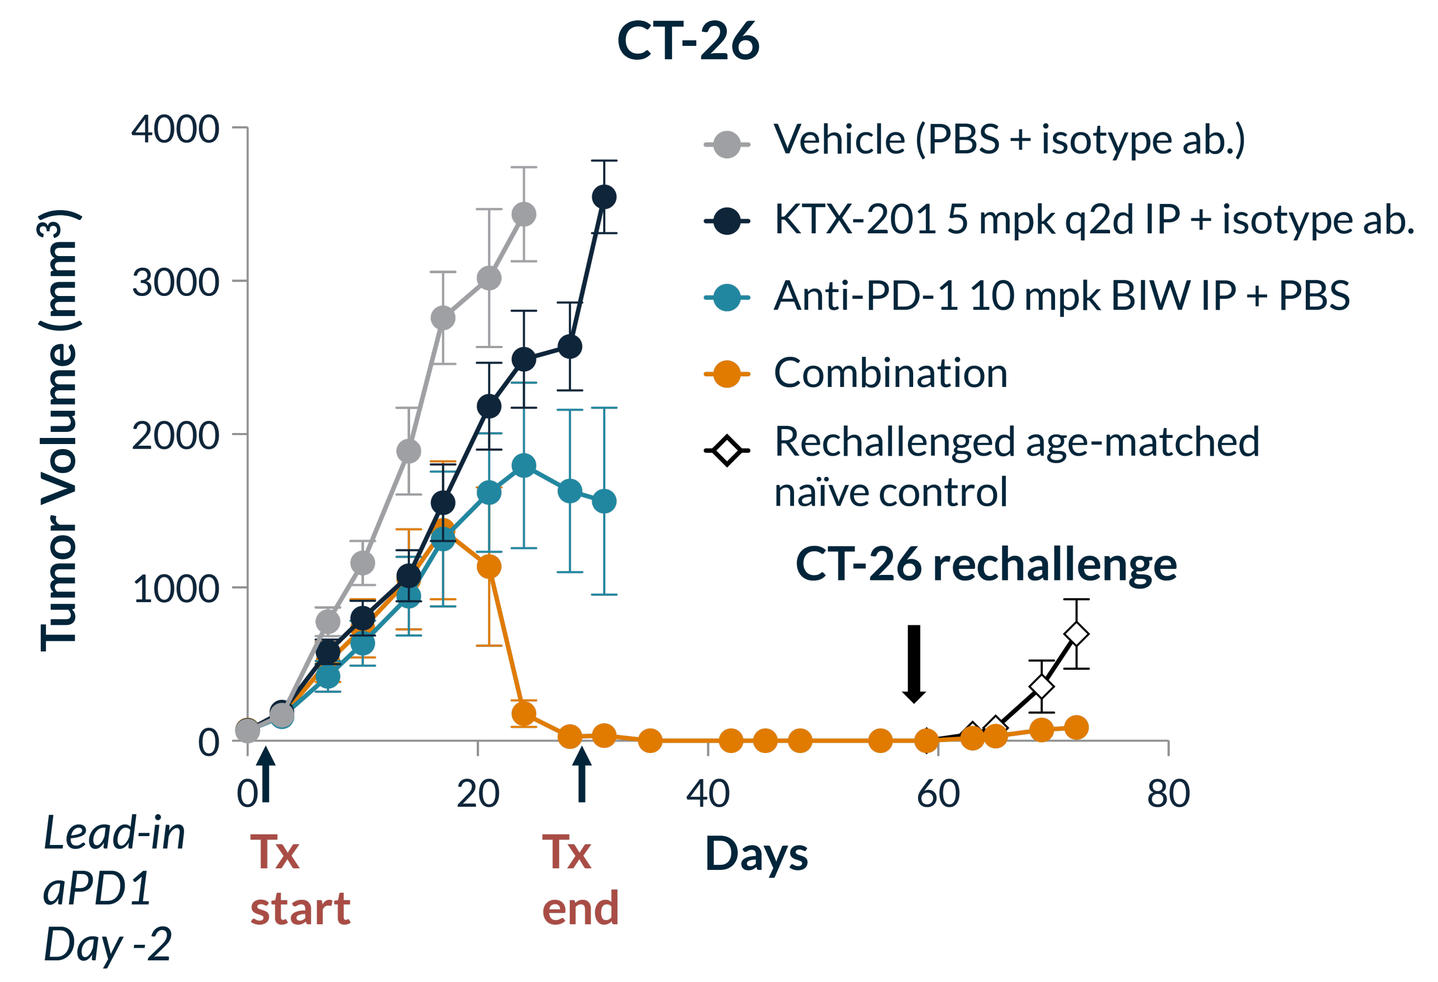
CT-26
Tumor Volume (mm³)
Vehicle (PBS + isotype ab.)
KTX-201 5 mpk q2d IP + isotype ab.
Anti-PD-1 10 mpk BIW IP + PBS
Combination
Rechallenged age-matched naïve control
CT-26 rechallenge
Lead-in aPD1 Day -2
Tx start
Tx end
Days

KTX-201 + anti-PD-1 trend towards a combination effect in A20 mouse lymphoma

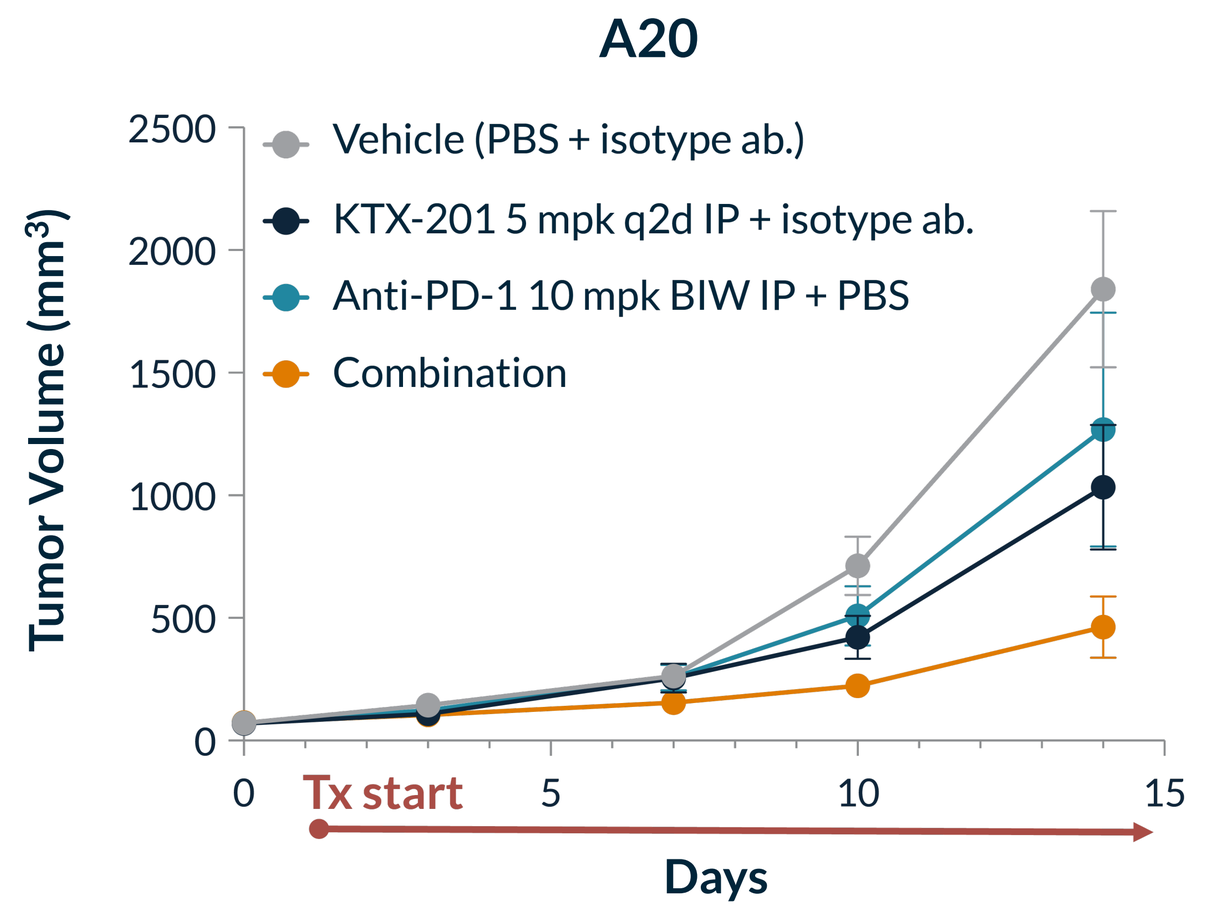
A20
Tumor Volume (mm³)
Vehicle (PBS + isotype ab.)
KTX-201 5 mpk q2d IP + isotype ab.
Anti-PD-1 10 mpk BIW IP + PBS
Combination
Tx start
Days

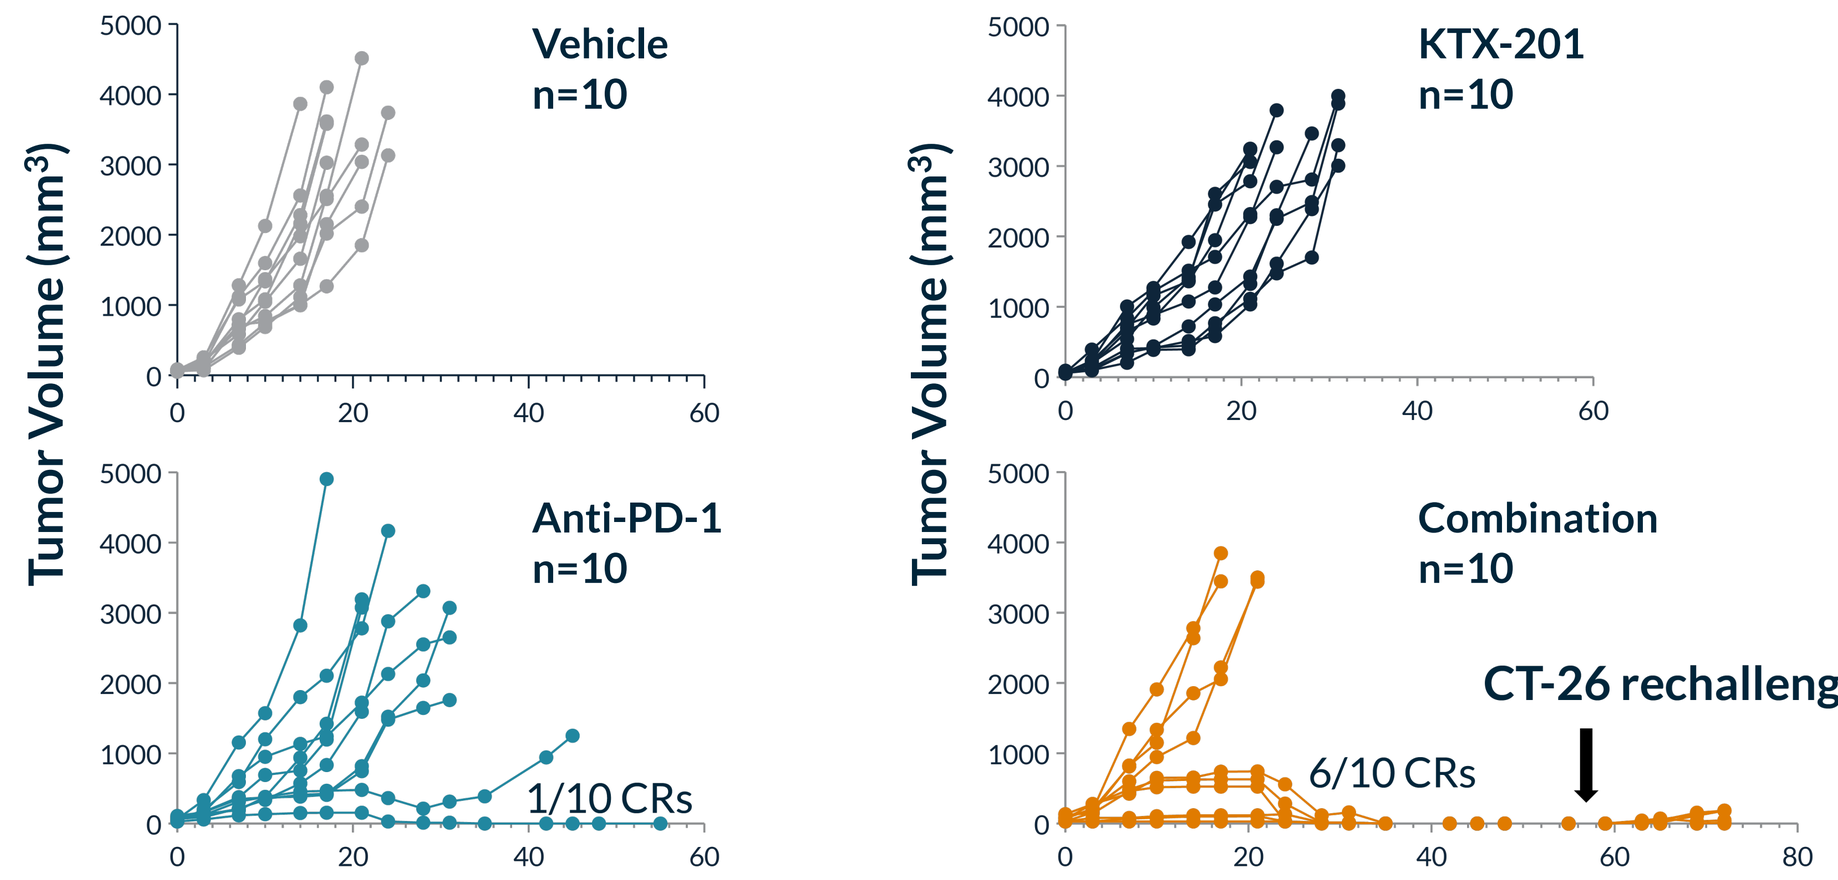
Tumor Volume (mm³)
Vehicle
n=10
KTX-201
n=10
Anti-PD-1
n=10
1/10 CRs
Combination
n=10
CT-26 rechallenge
6/10 CRs

Transcriptomic analyses of CT-26 tumors show upregulation of *STAT3* and other immunosuppressive genes with anti-PD-1 monotherapy, which are neutralized when combined with KTX-201

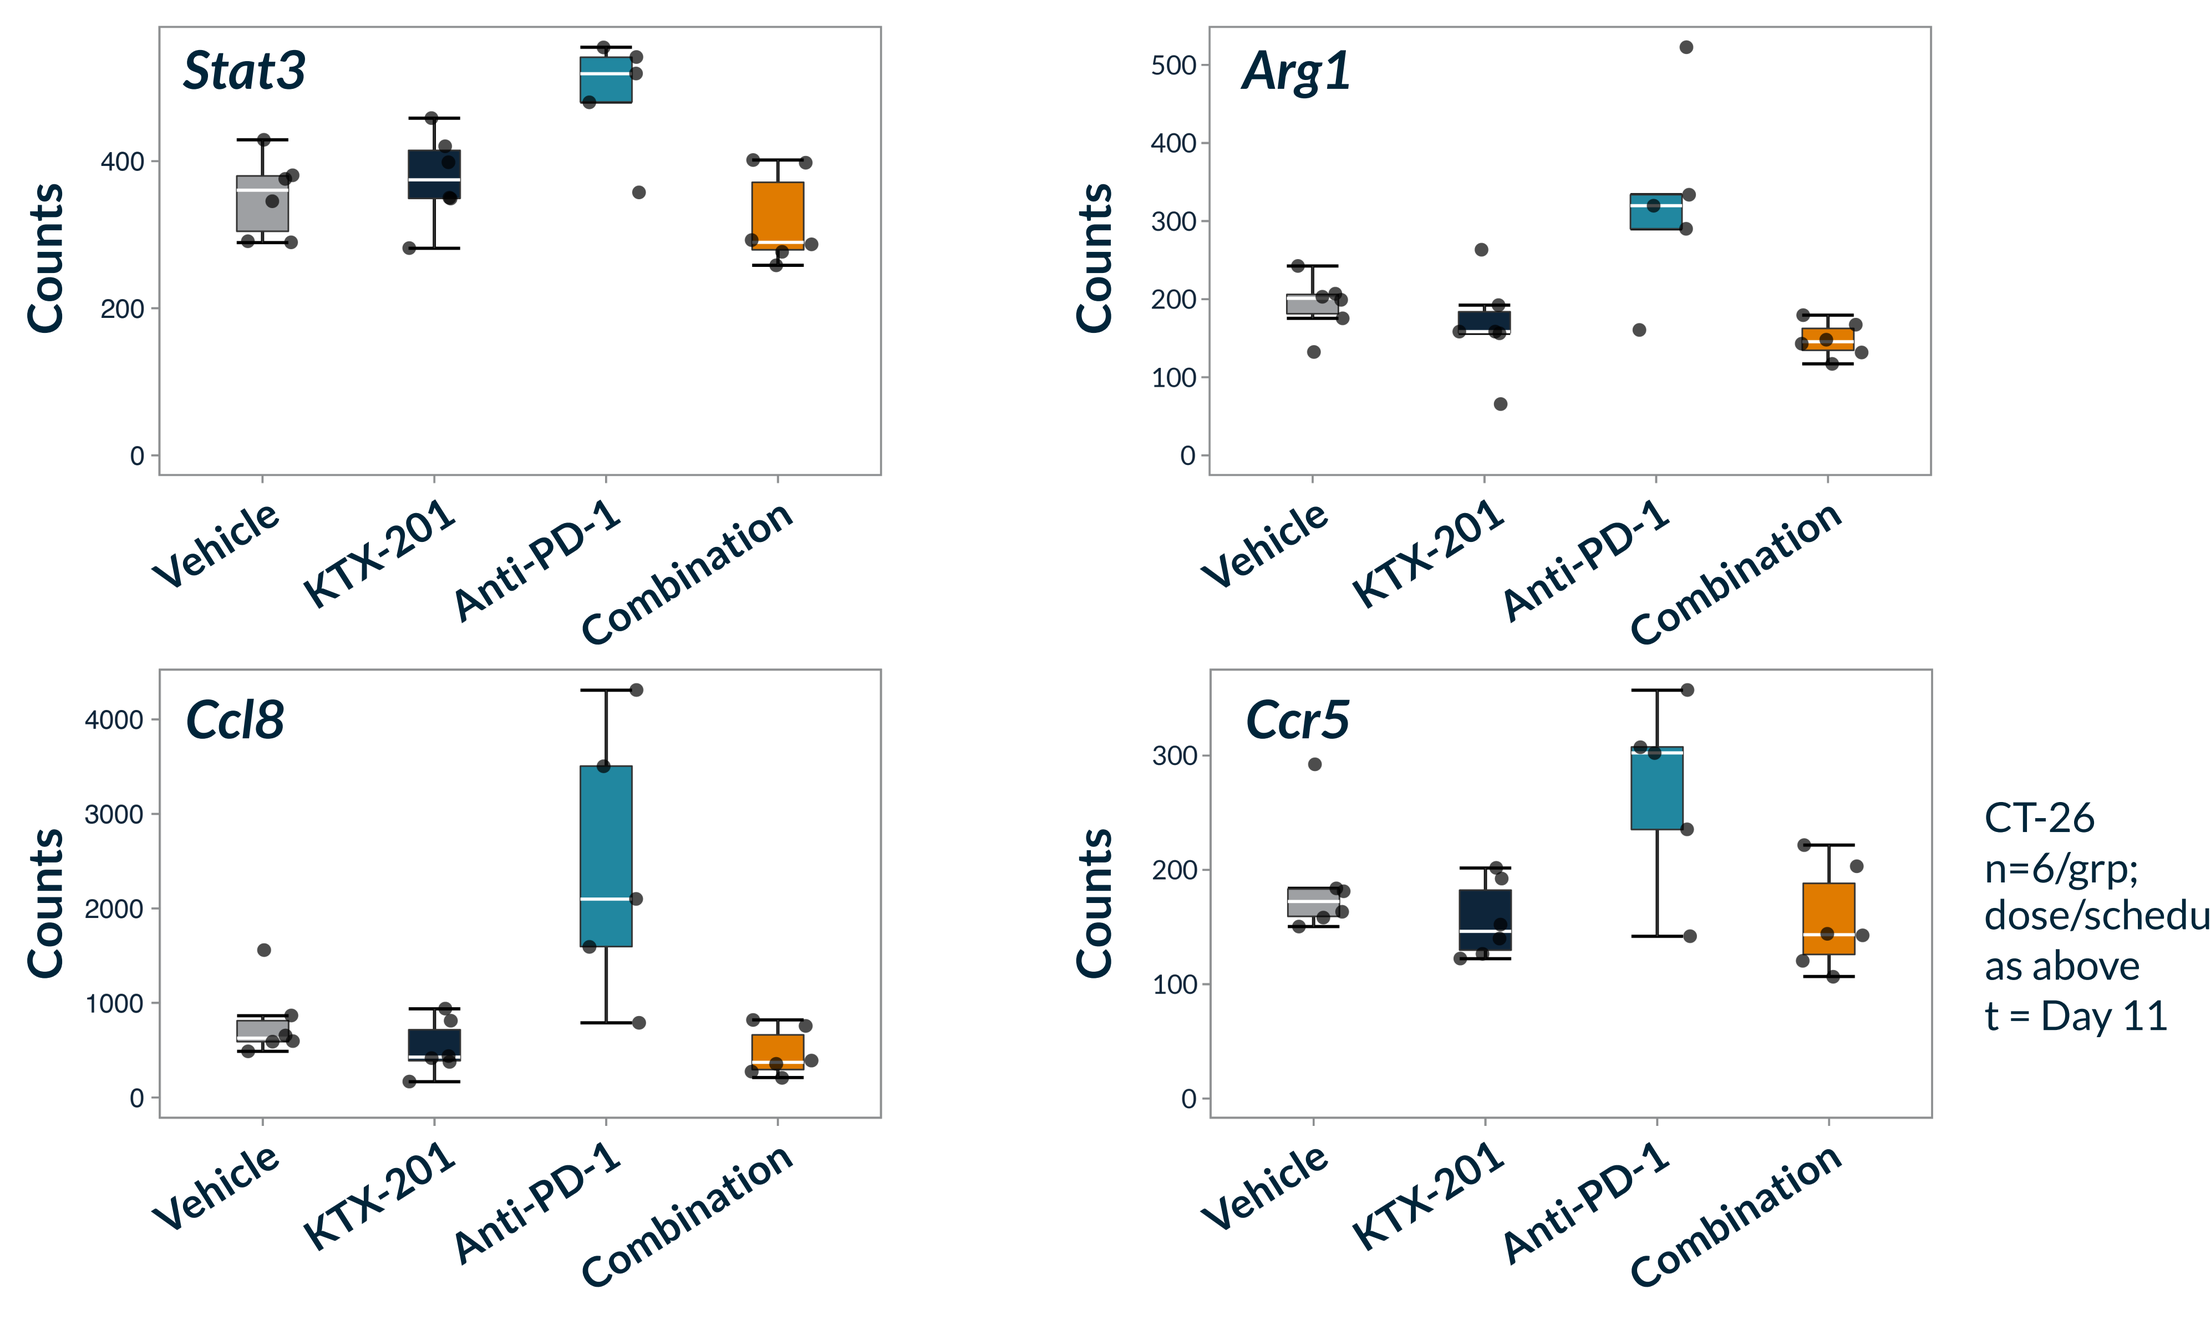
Counts
Stat3
Arg1
Ccl8
Ccr5
Vehicle
KTX-201
Anti-PD-1
Combination
CT-26
n=6/grp;
dose/schedule as above
t = Day 11

METHODS

In vitro Assays

All cell lines were cultured according to recommended procedures. For degradation assays, cells were treated with compounds for 24 h, and total STAT3 protein was assessed by western blotting. For DC50 determination, STAT3 levels were determined by MSD assay in cancer cell lines and by flow cytometry in human PBMCs. Viability was assessed by CTG assay using cells treated with KTX-201 for 4 days.

Proteomic Analysis

Human PBMCs and SU-DHL-1 cells were lyzed, digested and desalted, labeled with TMTpro 16plex, fractionated into 24 fractions on an Agilent off-line HPLC with an Extend300 C18 column and analyzed on Thermo Scientific Q Exactive HF-X Hybrid Quadrupole-Orbitrap Mass Spectrometer.

In vivo Experiments

Subcutaneous tumors were established in immunocompetent host mice strain using CT-26, A20, MC-38 or EMT-6 cell lines (ATCC). KTX-201 was formulated in buffered PBS and administered IP. In the CT-26 combination study, a single lead-in dose of anti-PD-1 was administered 2 days prior to administration of KTX-201 in animals with palpable tumors. Plots show mean tumor volume with error bars representing SEM. Immunophenotyping was carried out using FACS on day 7 after treatment. Tissue STAT3, CD8 levels were evaluated using immunohistochemistry. Anti-CD8 or isotype antibodies were administered 200 µg IP BIW for T cell depletion experiments.

Transcriptomic Analysis

Nanostring nCounter PanCancer IO360 panel was used for gene expression analysis using CT-26 tumors collected on Day 11 post treatment in respective cohorts. Normalization and differential expression was performed with DESeq2.[5]

SUMMARY

- Kymera's potent and selective STAT3 degraders have anti-tumor activity in models of solid and hematologic malignancies that are poorly sensitive to immunotherapies.
- STAT3 degradation results in a remodeled TME that is predictive of favorable responses to checkpoint inhibitors.
- STAT3 degrader is highly synergistic with anti-PD-1 in the CT-26 model leading to durable anti-tumor responses and development of long-term immunological memory.
- KT-333, a first-in-class selective STAT3 degrader, is expected to clear IND by end of 2021 and to be in Phase 1 clinical trial in liquid and solid tumors shortly thereafter.

REFERENCES

1. Ayers *et al.* JCI 2017
2. Proia *et al.* Clin Can Res 2020
3. Bagaev *et al* Cancer Cell 2021
4. Hu *et al.* J Immunol 2005
5. Love *et al.* Genome Biology 2015

ACKNOWLEDGEMENTS

We wish to thank the entire Kymera team including former colleagues Drs. Fred Csibi and Nan Ji for their valuable contributions, as well as our consultants, collaborators and CRO partners.

KYMERA 200 Arsenal Yards Blvd., Suite 230, Watertown, MA 02472, USA www.kymeratx.com Contact: Joyoti Dey, Ph.D., M.P.H / jdey@kymeratx.com

Society for Immunotherapy of Cancer (SITC) 36th Annual Meeting • November 10-14, 2021 • Washington, DC