7536 KYM-001, first-in-class oral IRAK4 protein degraders, induce tumor regression in a xenograft model of MYD88-mutant ABC DLBCL alone and in combination with BTK inhibition LB-272

Joseph F. Kelleher, Veronica Campbell, Jesse Chen, Jared Gollob, Nan Ji, Hari Kamadurai, Christine Klaus, Henry Li, Christine Loh, Alice McDonald, Haojing Rong, Scott Rusin, Kirti Sharma, Dominico Vigil, Duncan Walker, Matt Weiss, Karen Yuan, Yi Zhang, Laurent Audoly and Nello Mainolfi,

Kymera Therapeutics, Inc. 300 Technology Square, Cambridge, MA 02139.

Contact: joe@kymeratx.com

Introduction:

- Recurrent mutations in MYD88 drive 30-40% of ABC-DLBCL; MYD88 L265P is the most prevalent
- IRAK4 is a critical component of the Myddosome; IRAK4 catalytic and scaffolding functions are essential for full signaling to NFκB and MAPK pathways
- Kymera Therapeutics is developing heterobifunctional small molecule degraders of IRAK4 with drug-like properties, typified here with series KYM-001 Compounds A and B.
- IRAK4 Degrader Compounds A and B are close analogs, differing mainly in PK properties
- Activating mutations in CD79A and CD79B cause BTK-dependent activation of NFκB signaling in ABC-DLBCL, and often co-occur with MYD88 mutation, prompting exploration of combinations of IRAK4 degradation with BTK inhibition

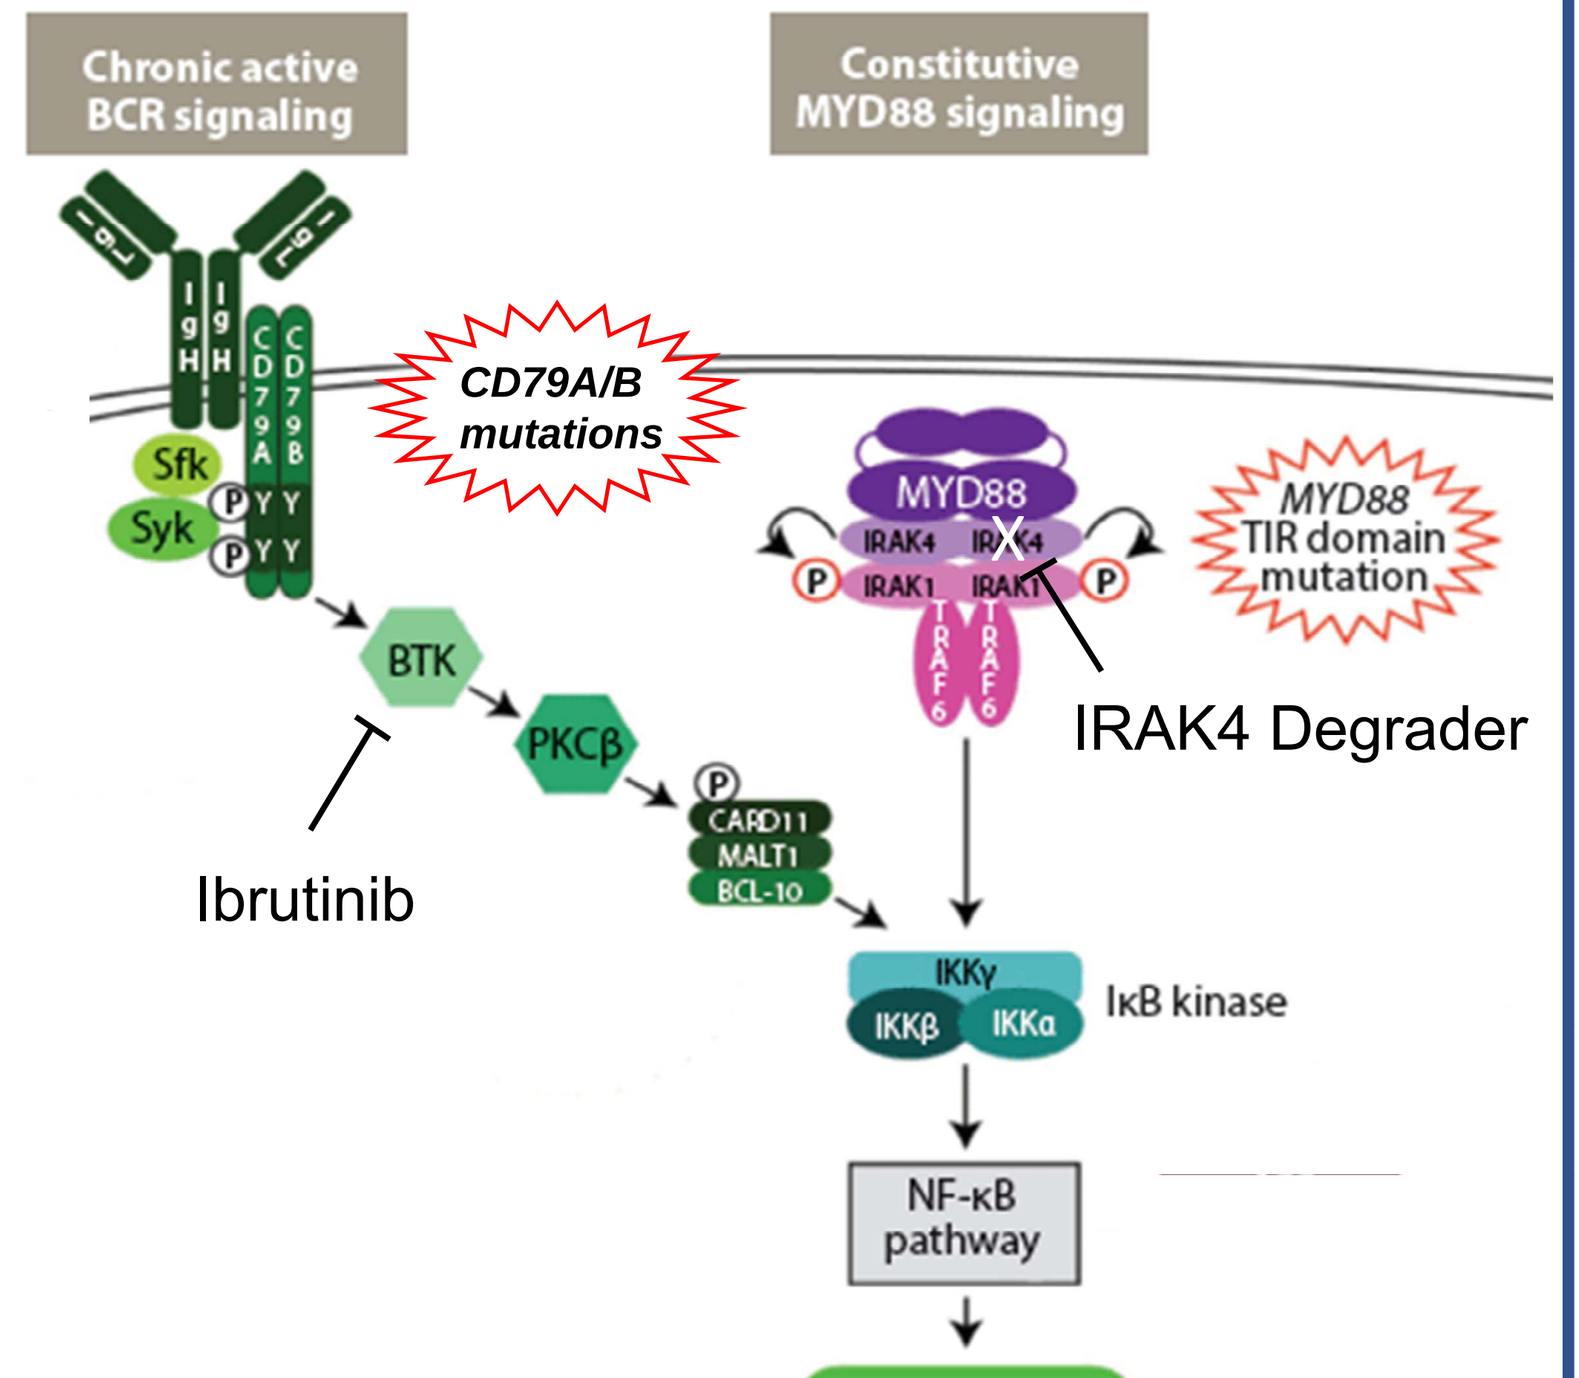
Chronic active BCR signaling
Constitutive MYD88 signaling
CD79A/B mutations
MYD88 TIR domain mutation
Sfk
Syk
BTK
PKCβ
CARD11 MALT1 BCL-10
MYD88
IRAK4
IRAK1
IRAK4 Degrader
Ibrutinib
IKKγ IKKβ IKKα
IκB kinase
NF-κB pathway
Survival

Modified from Yang, Cancer Cell 2012

IRAK4 is a key node in IL-1R and TLR signaling downstream of MYD88, with kinase-dependent and kinase-independent functions

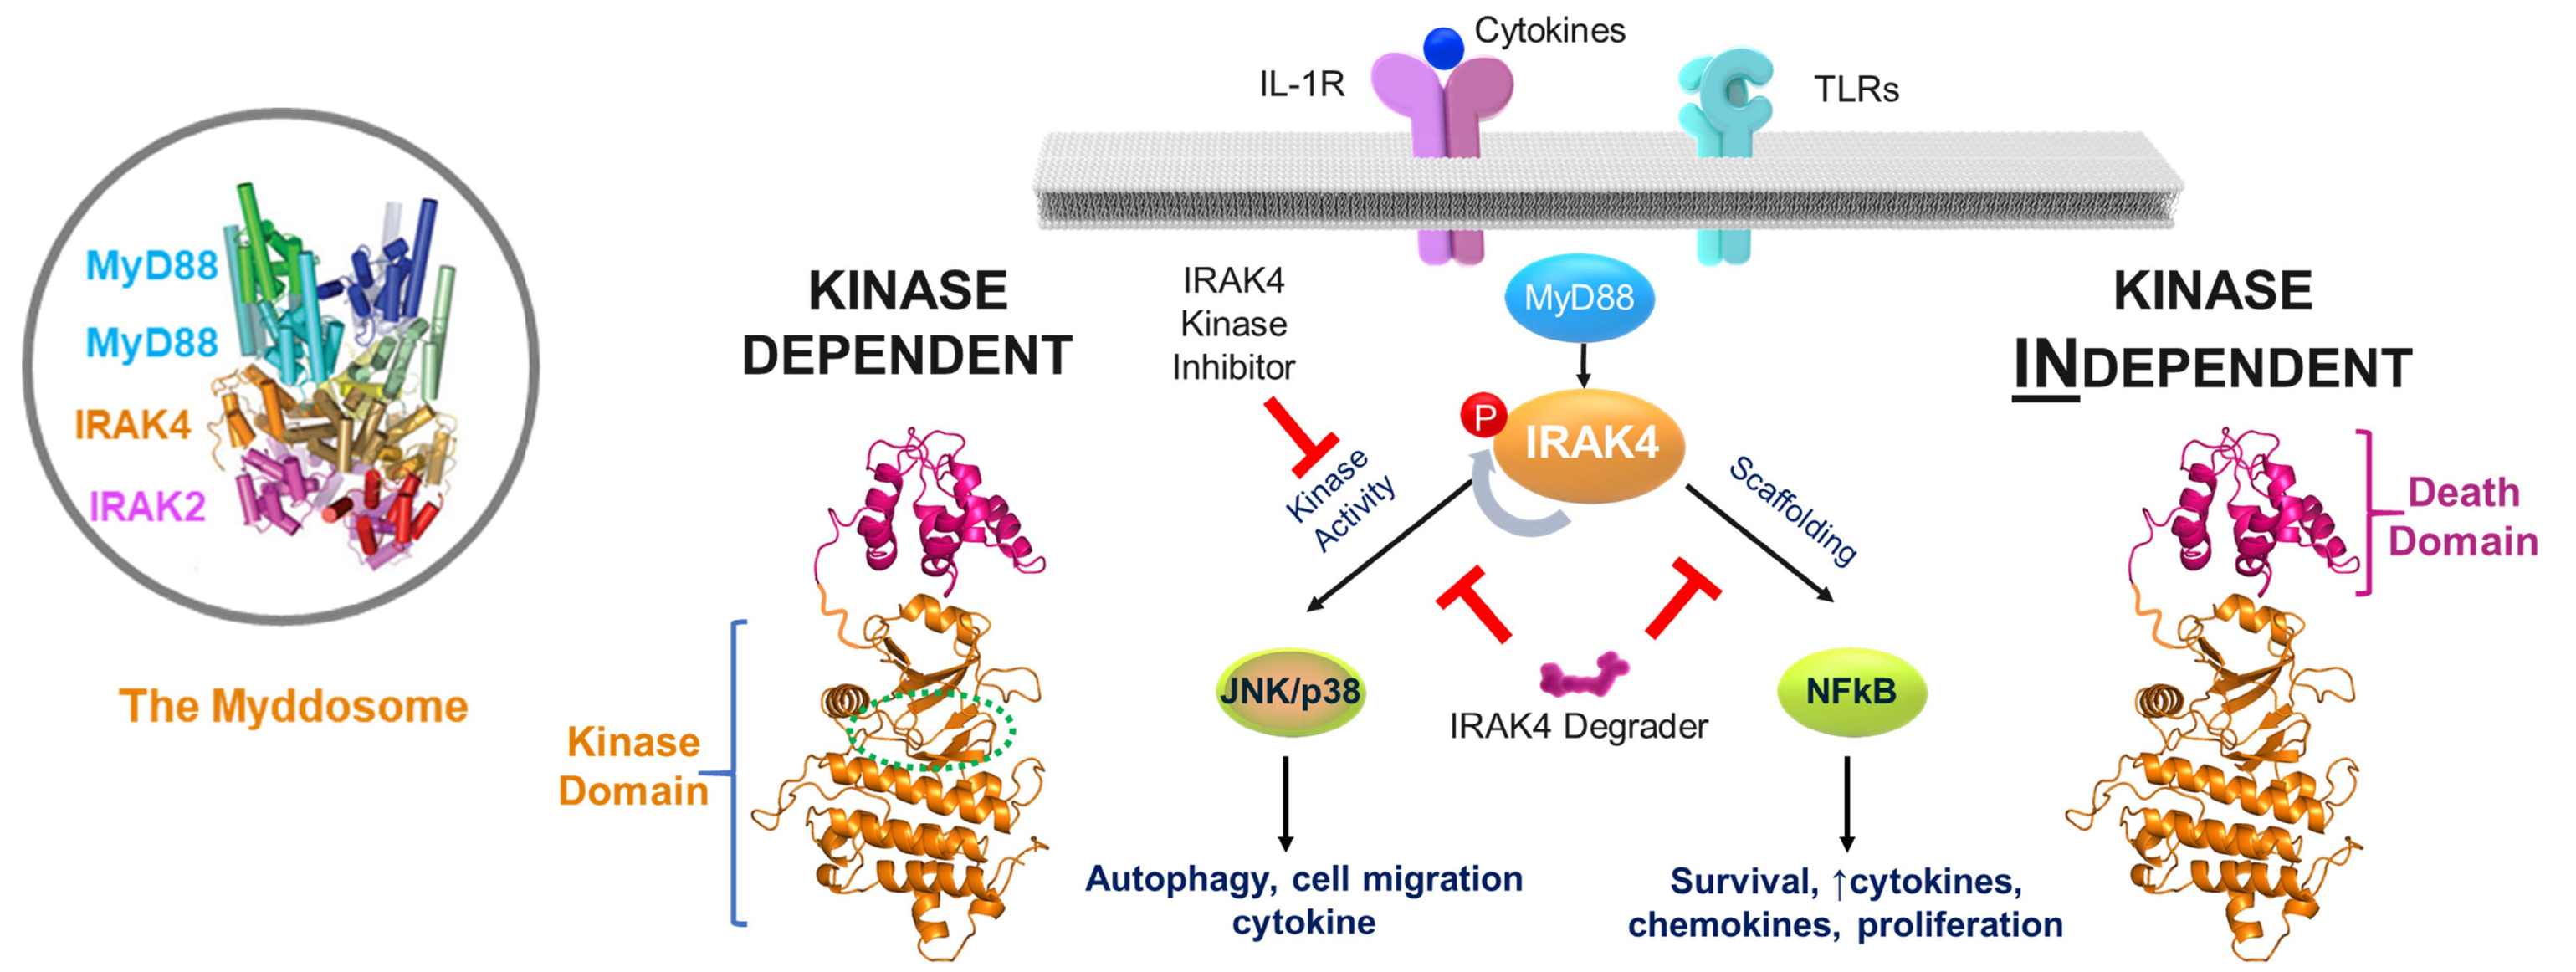
Cytokines
IL-1R
TLRs
MyD88
MyD88
IRAK4
IRAK2
The Myddosome
KINASE DEPENDENT
Kinase Domain
IRAK4 Kinase Inhibitor
Kinase Activity
IRAK4
Scaffolding
JNK/p38
IRAK4 Degrader
NFkB
Autophagy, cell migration cytokine
Survival, ↑cytokines, chemokines, proliferation
KINASE INDEPENDENT
Death Domain

Overview of targeted protein degradation with heterobifunctional small molecules

Mechanism of action:
- Bind simultaneously to Target Protein and to ubiquitin E3 ligase, bringing into physical proximity
- E3 ligase transfers ubiquitin to Target Protein
- Ubiquitinated Target is degraded by proteasome

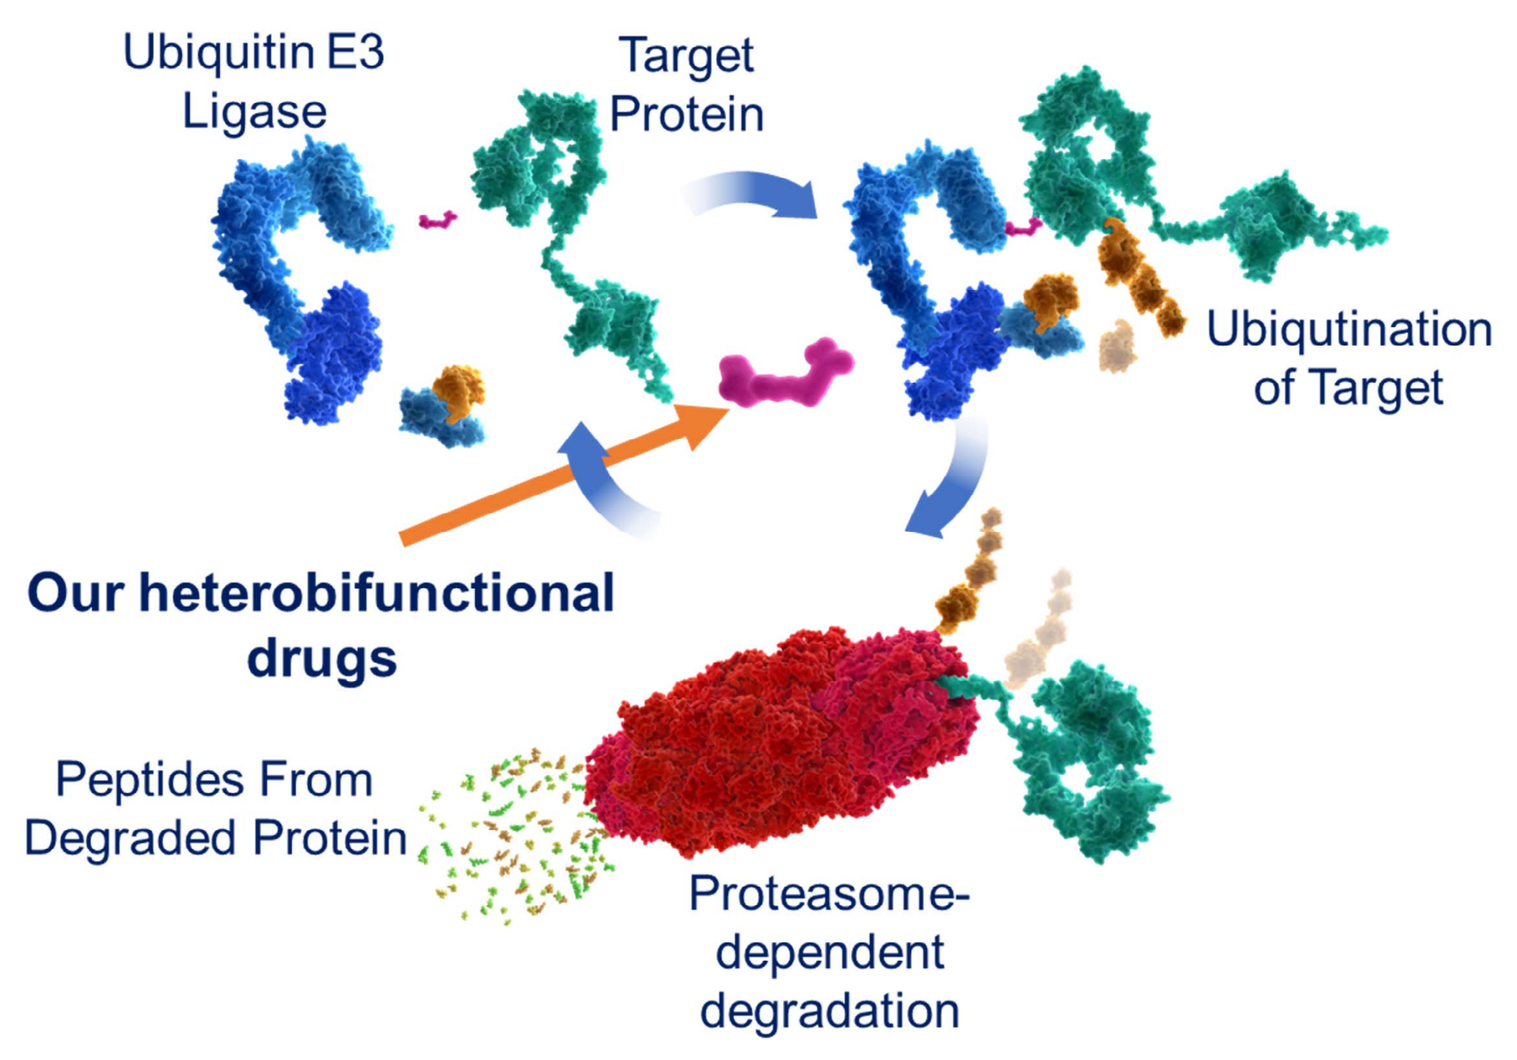
Ubiquitin E3 Ligase
Target Protein
Ubiquitination of Target
Our heterobifunctional drugs
Peptides From Degraded Protein
Proteasome-dependent degradation

Key features for developing therapeutics:
- Only a binding site is required
- Ternary complex-based selectivity
- Sub-stoichiometric, catalytic
- Protein re-synthesis rate limited
- Irreversible protein degradation

IRAK4 degradation is superior to IRAK4 kinase inhibition

TLR7/8 Induced Phosphoproteins in hPBMC

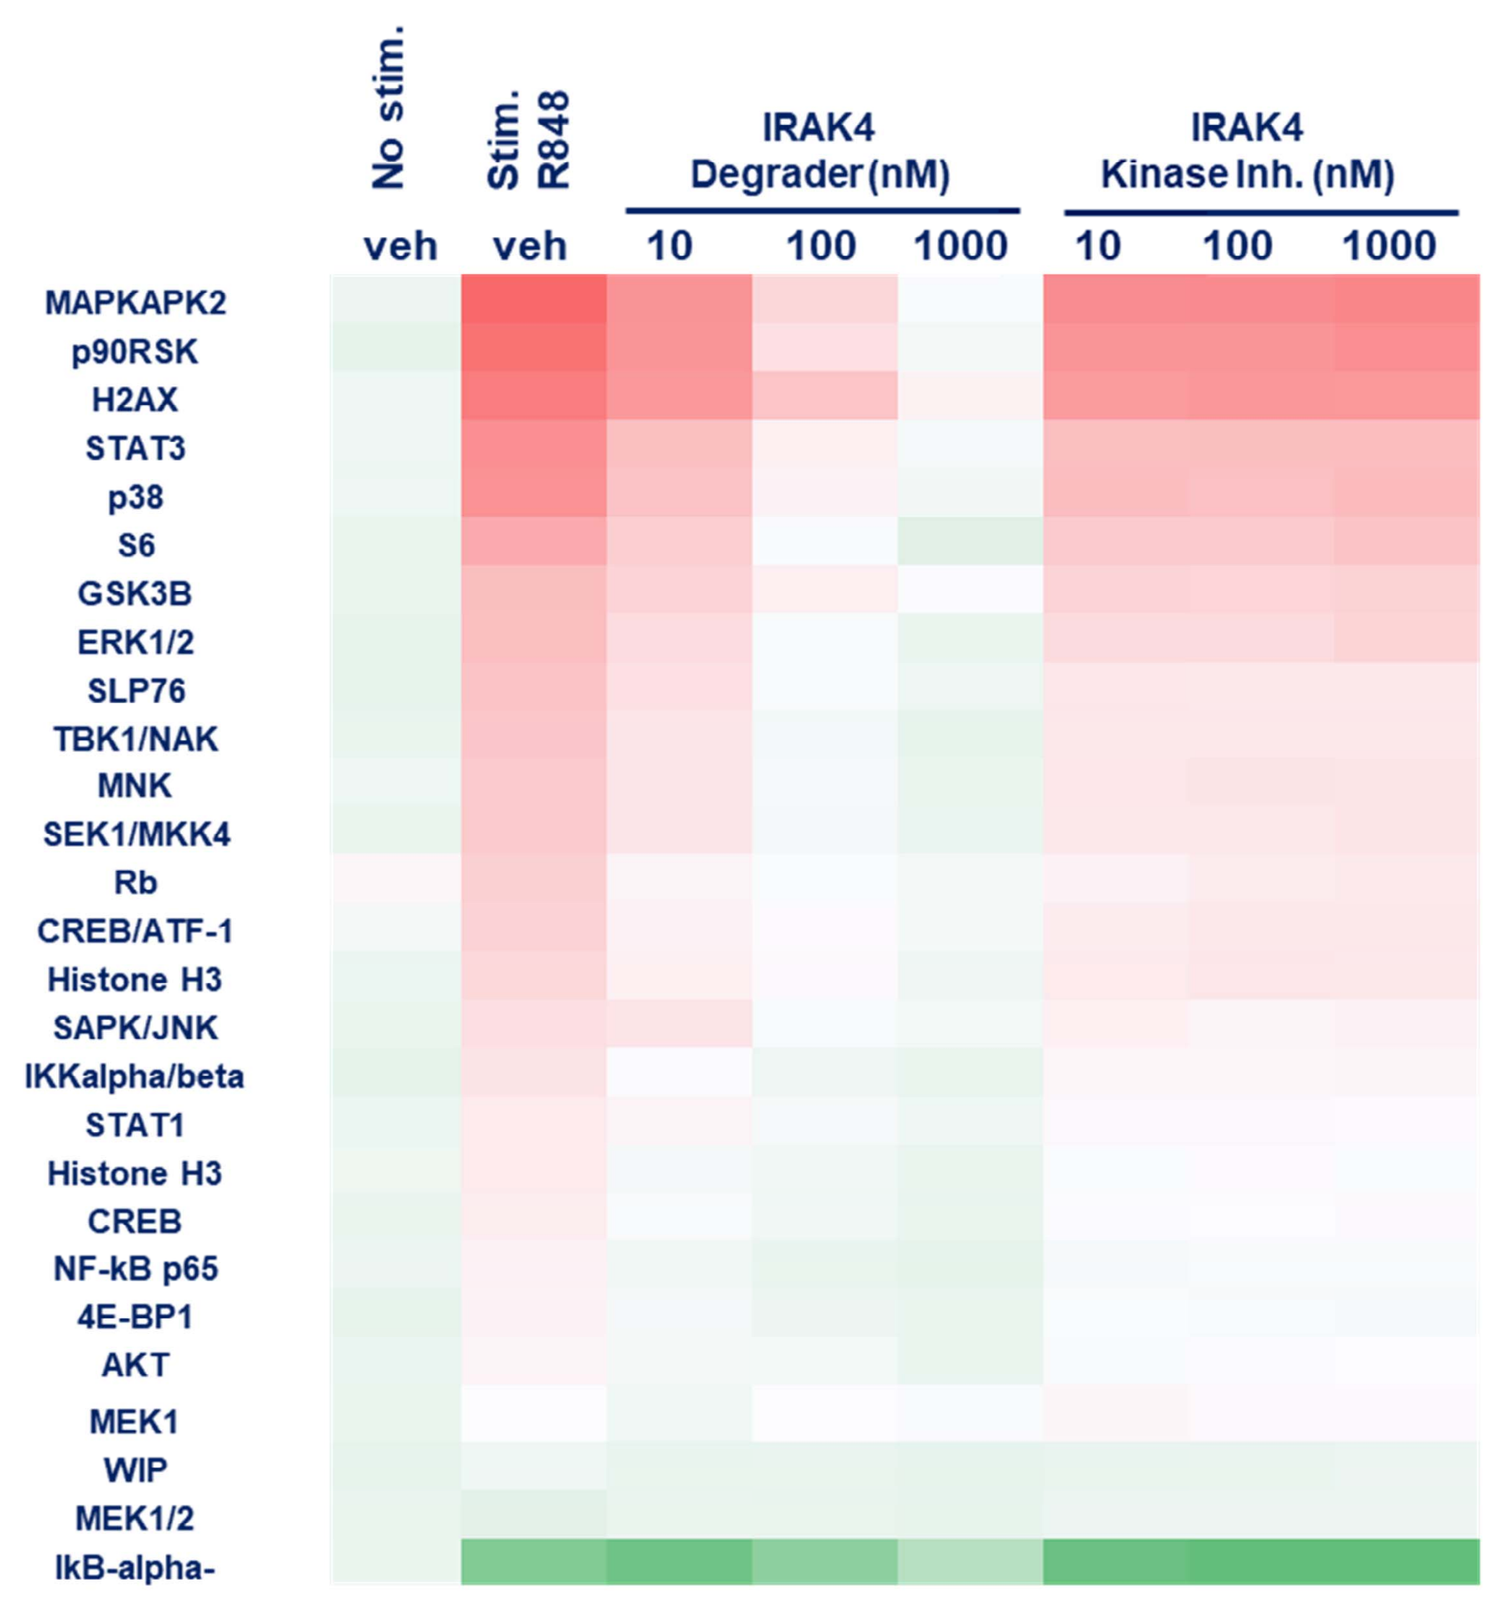
No stim.
Stim. R848
IRAK4 Degrader (nM)
IRAK4 Kinase Inh. (nM)
veh veh 10 100 1000 10 100 1000
MAPKAPK2
p90RSK
H2AX
STAT3
p38
S6
GSK3B
ERK1/2
SLP76
TBK1/NAK
MNK
SEK1/MKK4
Rb
CREB/ATF-1
Histone H3
SAPK/JNK
IKKalpha/beta
STAT1
Histone H3
CREB
NF-kB p65
4E-BP1
AKT
MEK1
WIP
MEK1/2
IkB-alpha-

IRAK4 Degrader causes potent and highly selective depletion of IRAK4 in MYD88/CD79 double mutant human ABC-DLBCL cell line OCI-LY10

Immunoblot in OCI-LY10 at 24 h

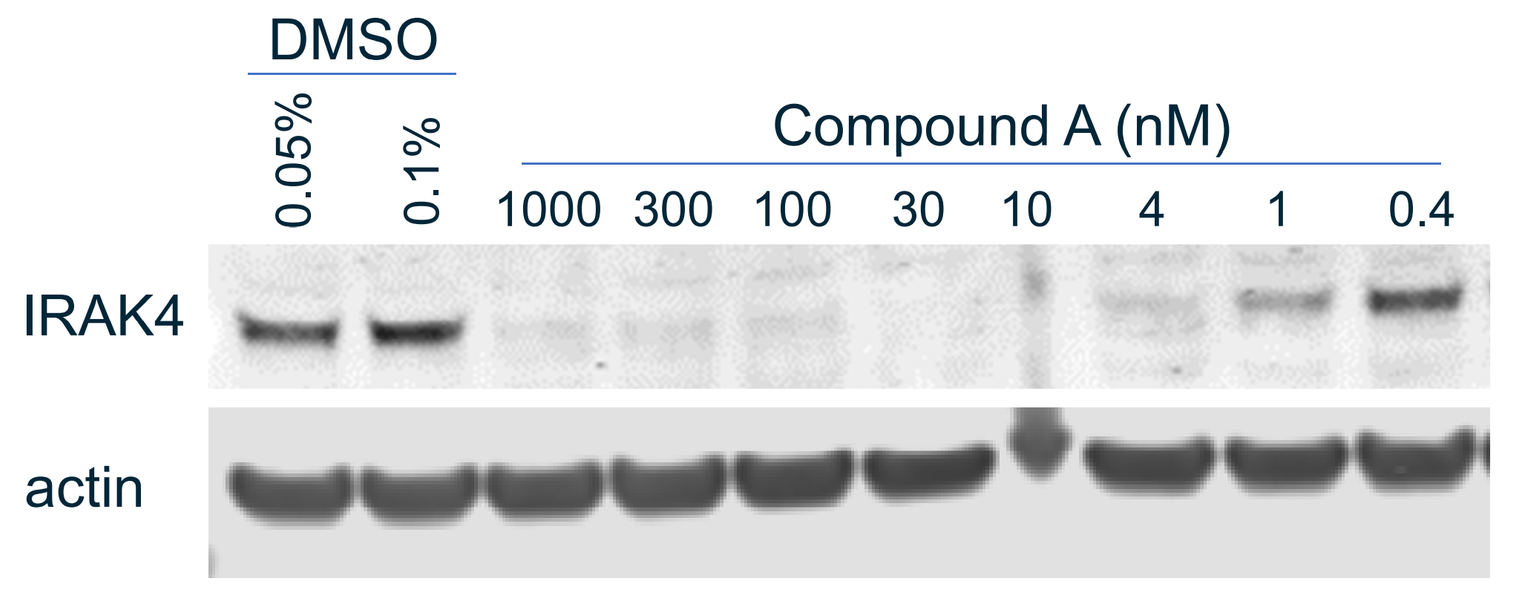
DMSO
0.05%
0.1%
Compound A (nM)
1000 300 100 30 10 4 1 0.4
IRAK4
actin

IRAK4 in OCI-LY10
MSD Assay at 4 h

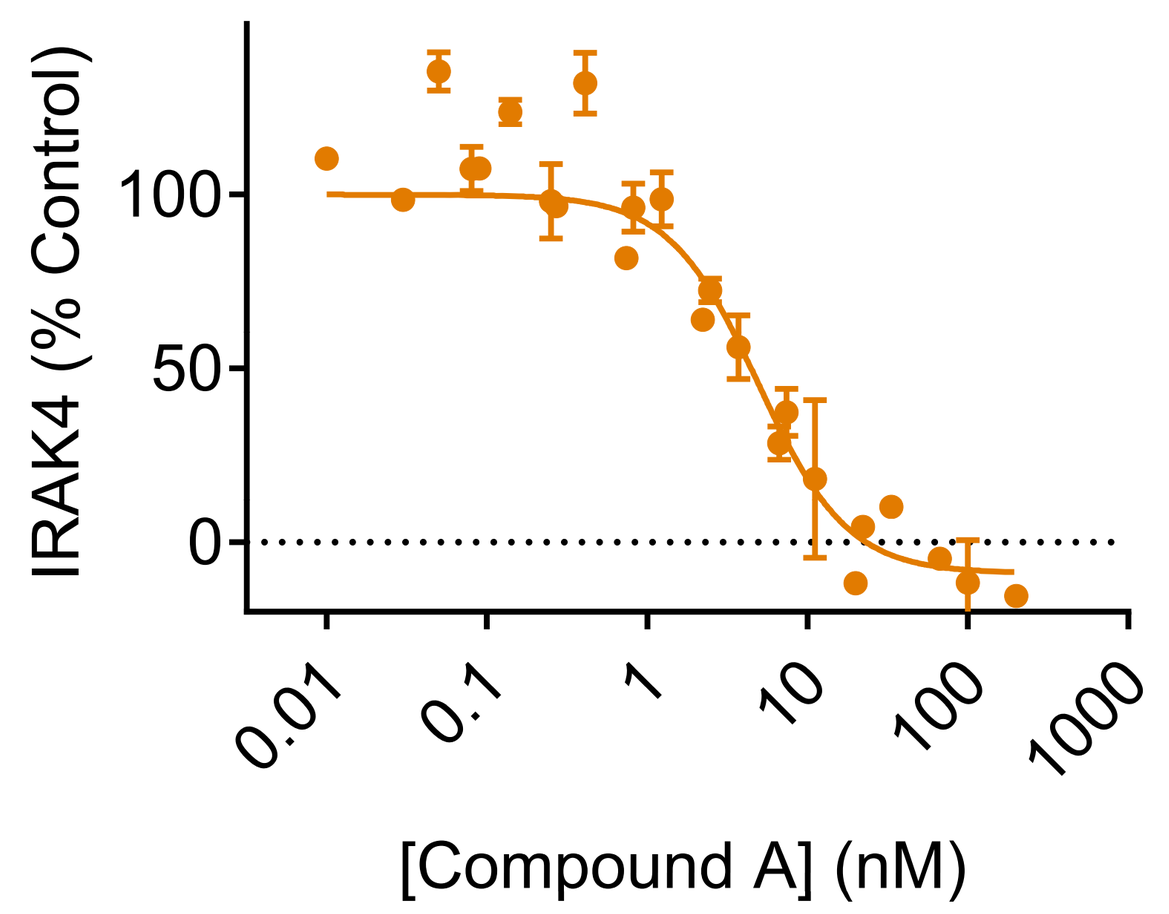
IRAK4 (% Control)
100
50
0
0.01
0.1
1
10
100
1000
[Compound A] (nM)

In Vitro Degradation (DC_{90}, μM)
OCI-LY10 at 4 h: 0.02

Similar results were obtained with Compound B

Deep TMT proteomics in OCI-LY10 at 8 h

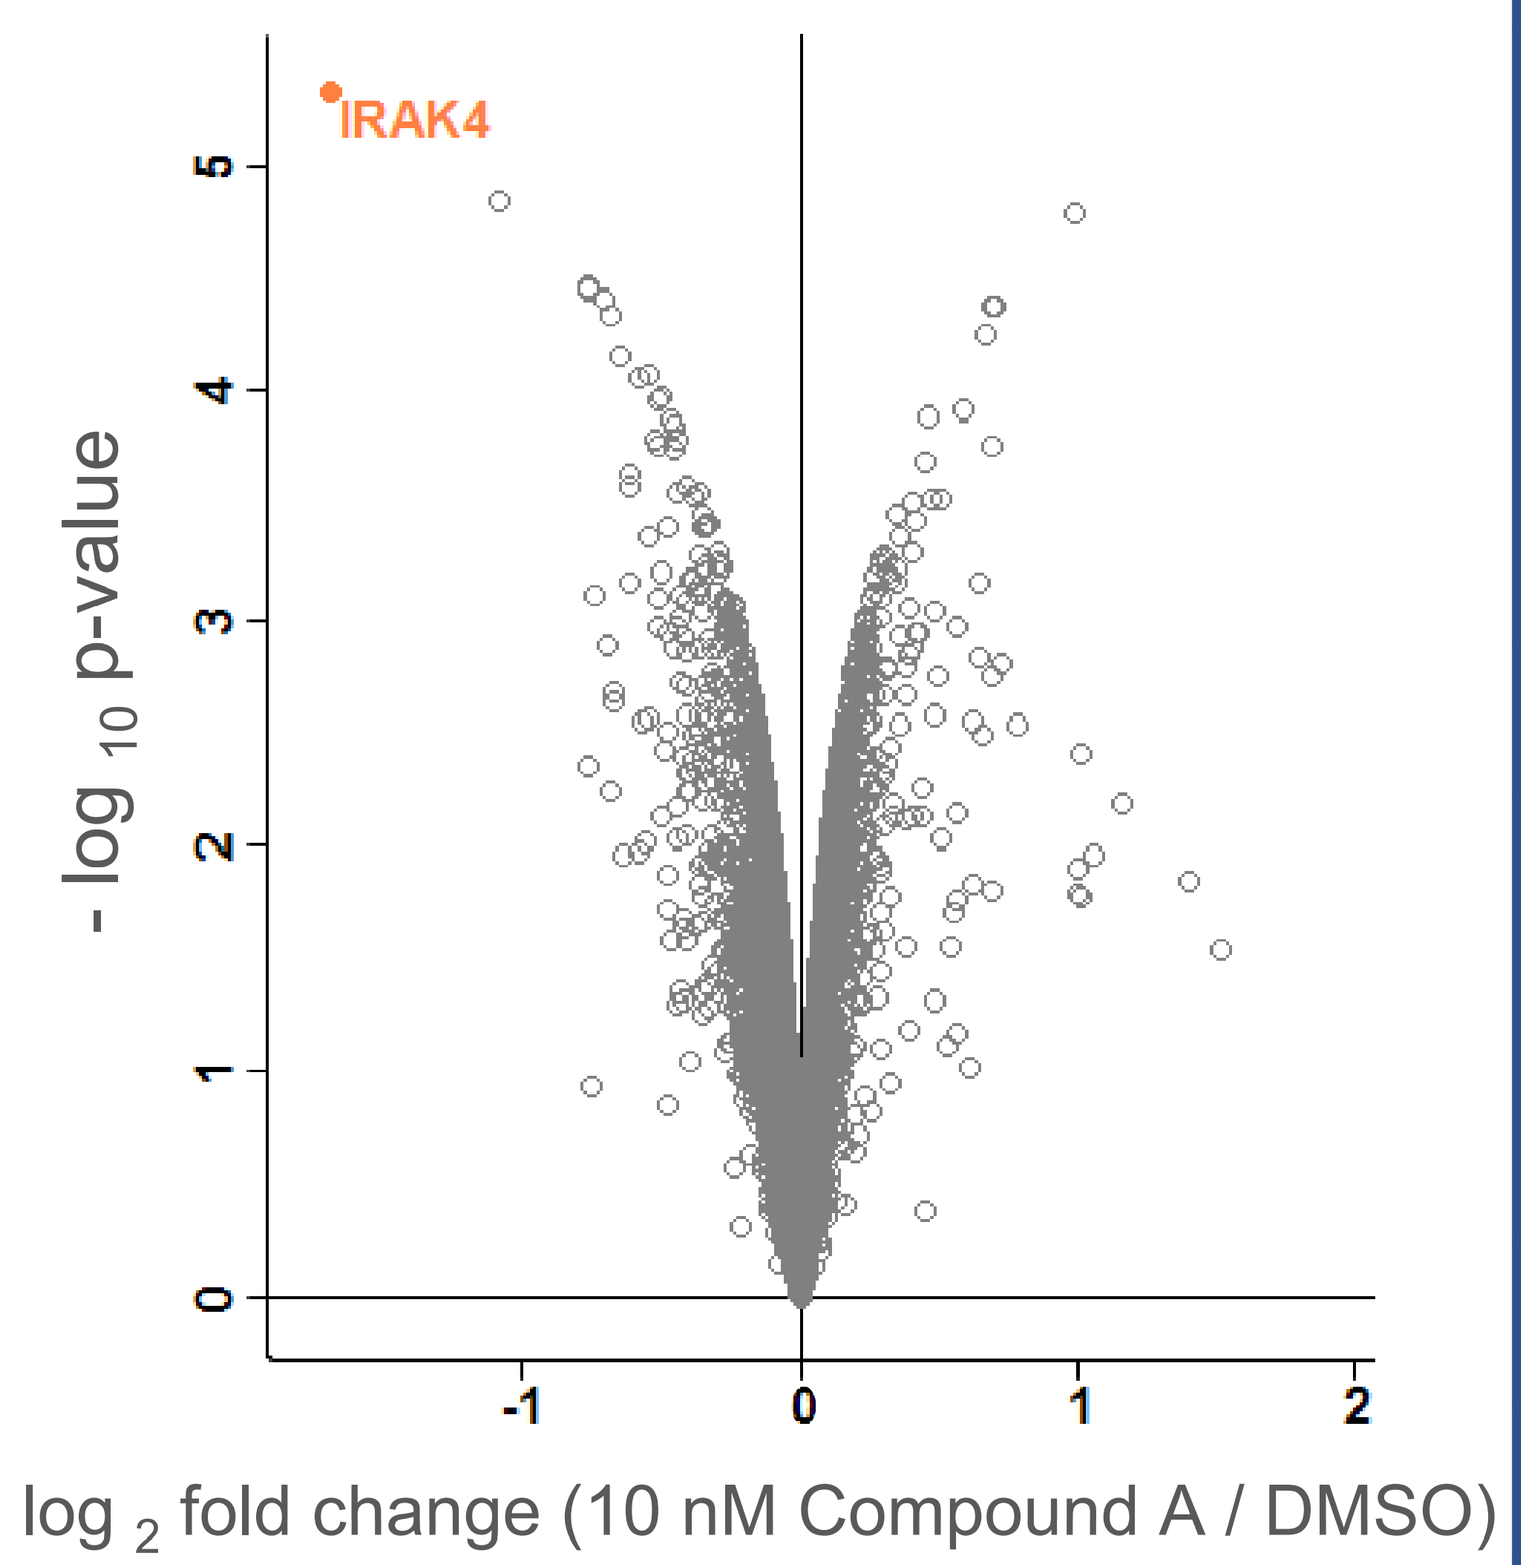
IRAK4
- log 10 p-value
log 2 fold change (10 nM Compound A / DMSO)

Number of unique proteins groups	11,231
Number of quantified proteins groups	10,992
Number of proteins with >1 unique peptides and quantified	9,526

TMT = Tandem Mass Tag

IRAK4 Degrader administered PO once daily causes tumor xenograft regression in association with ≥ 75% IRAK4 reduction in OCI-LY10

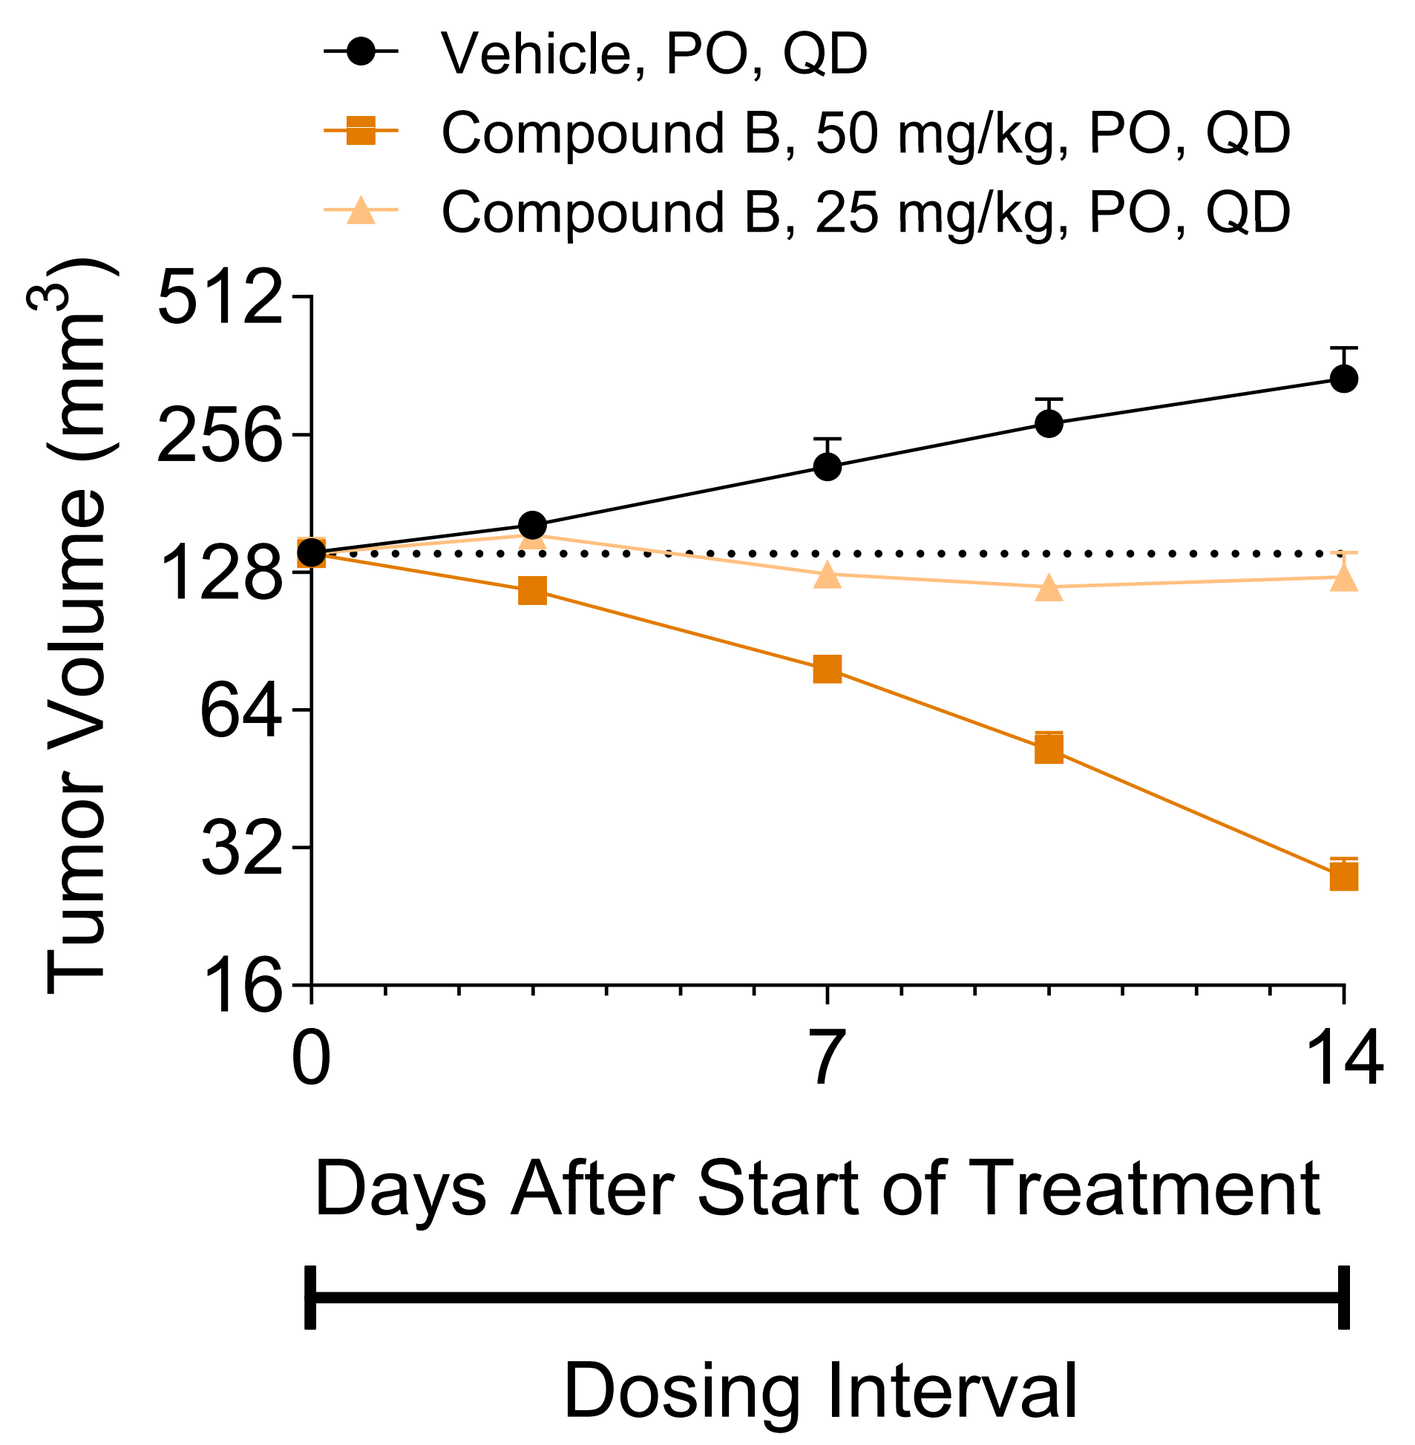
Vehicle, PO, QD
Compound B, 50 mg/kg, PO, QD
Compound B, 25 mg/kg, PO, QD
Tumor Volume (mm3)
512
256
128
64
32
16
0
7
14
Days After Start of Treatment
Dosing Interval

PO Dose (mg/kg)	Total Plasma Exposure (μM)	Tumor IRAK4↓ (% Control ± SD, day 5)
25	0.15	65.4 ± 8.2
50	0.34	75.3 ± 3.5

Similar results were obtained with Compound A

IRAK4 Degrader and ibrutinib show strong combination activity for induction of apoptosis in OCI-LY10 *in vitro* and induce tumor xenograft regression when used together *in vivo* at suboptimal dose of IRAK4 Degrader

Apoptosis in OCI-LY10 at 72 h *in vitro*

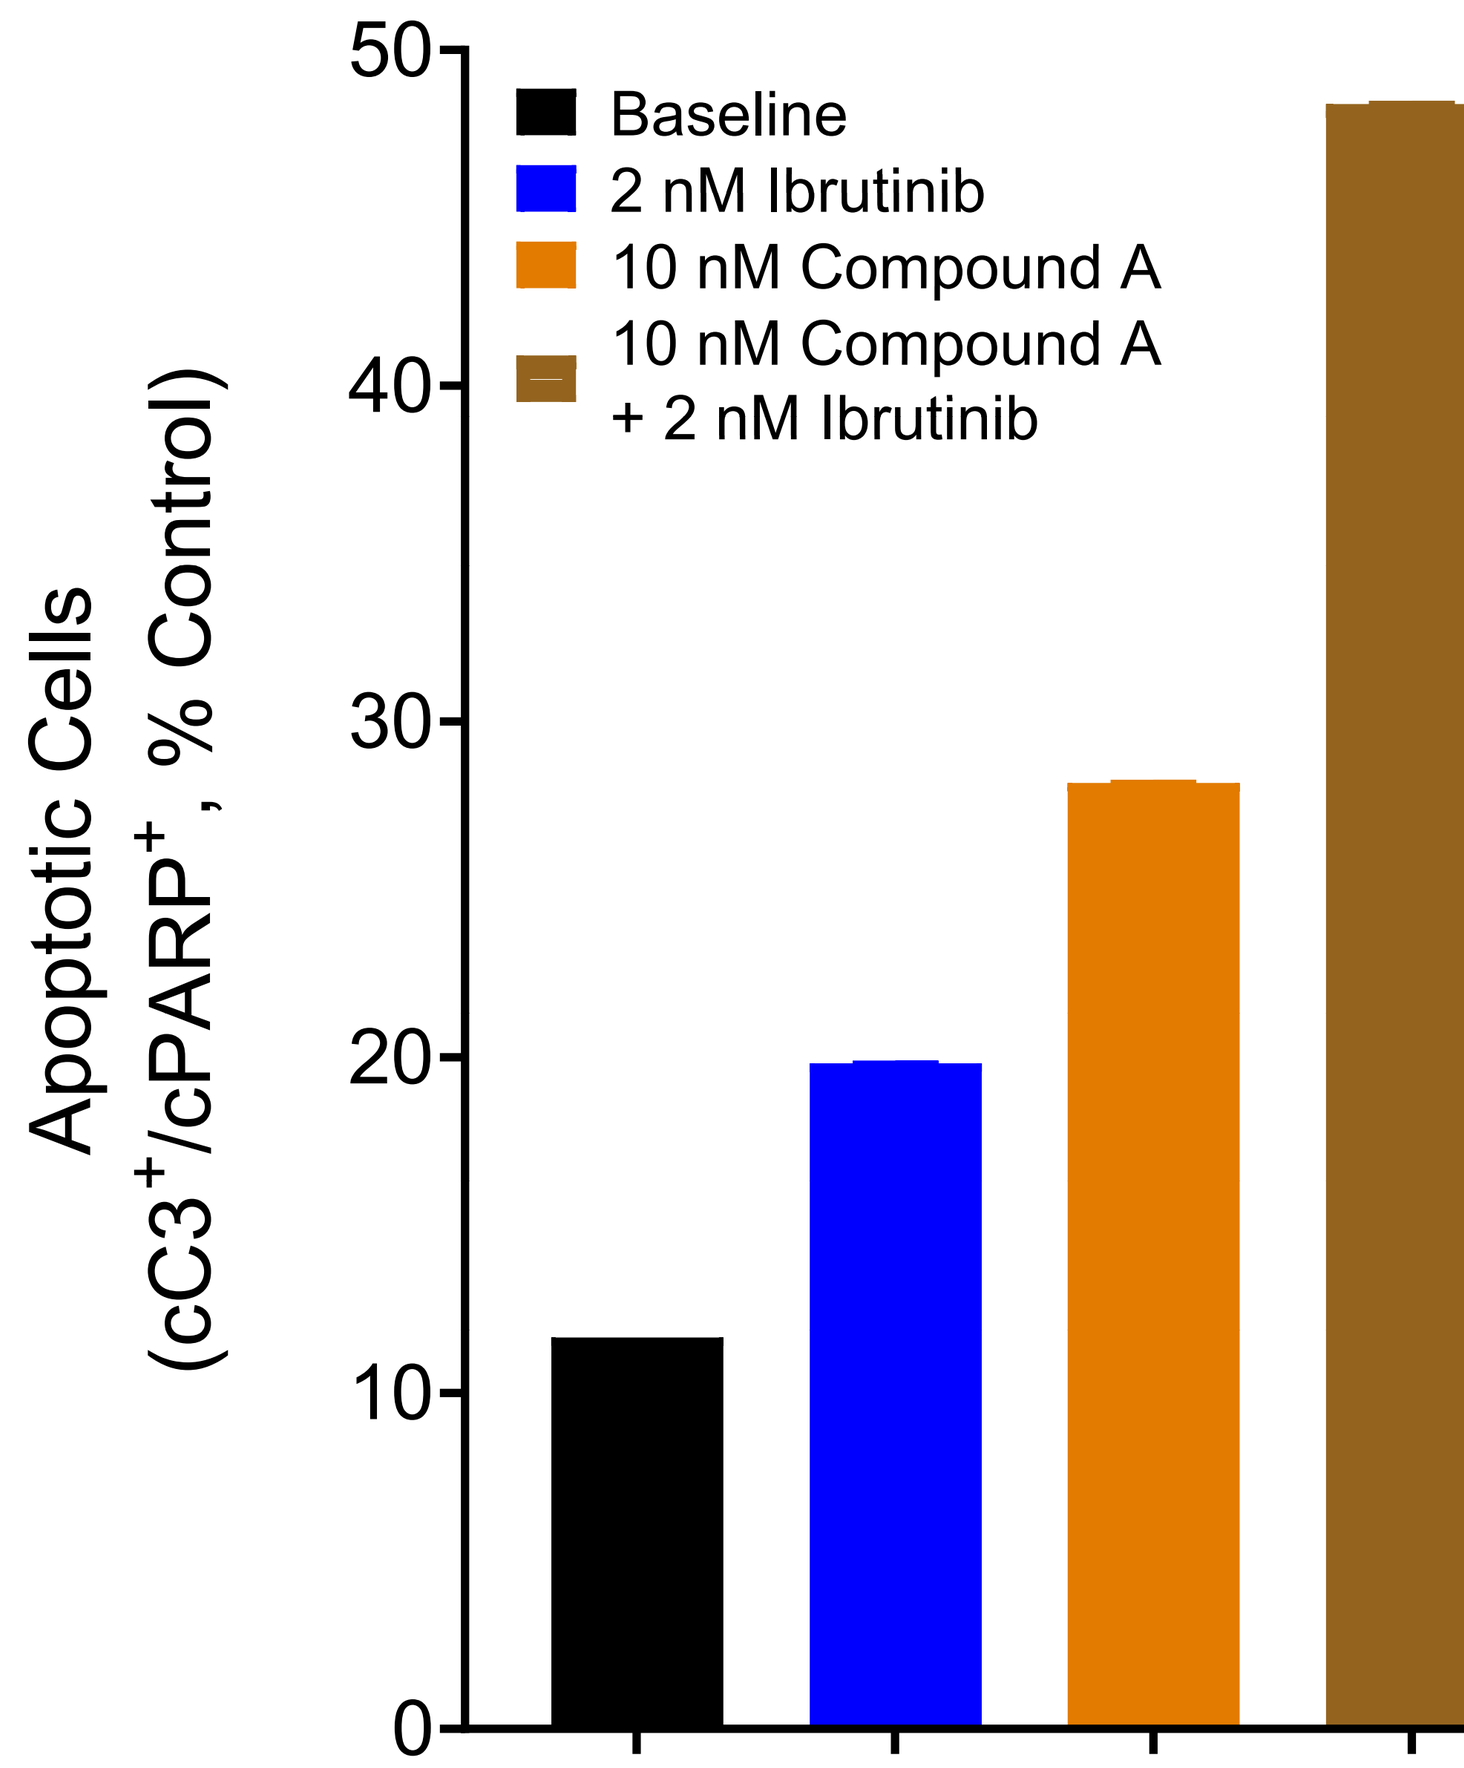
Baseline
2 nM Ibrutinib
10 nM Compound A
10 nM Compound A + 2 nM Ibrutinib
Apoptotic Cells (cC3+/cPARP+, % Control)
50
40
30
20
10
0

OCI-LY10 tumor xenograft growth *in vivo*

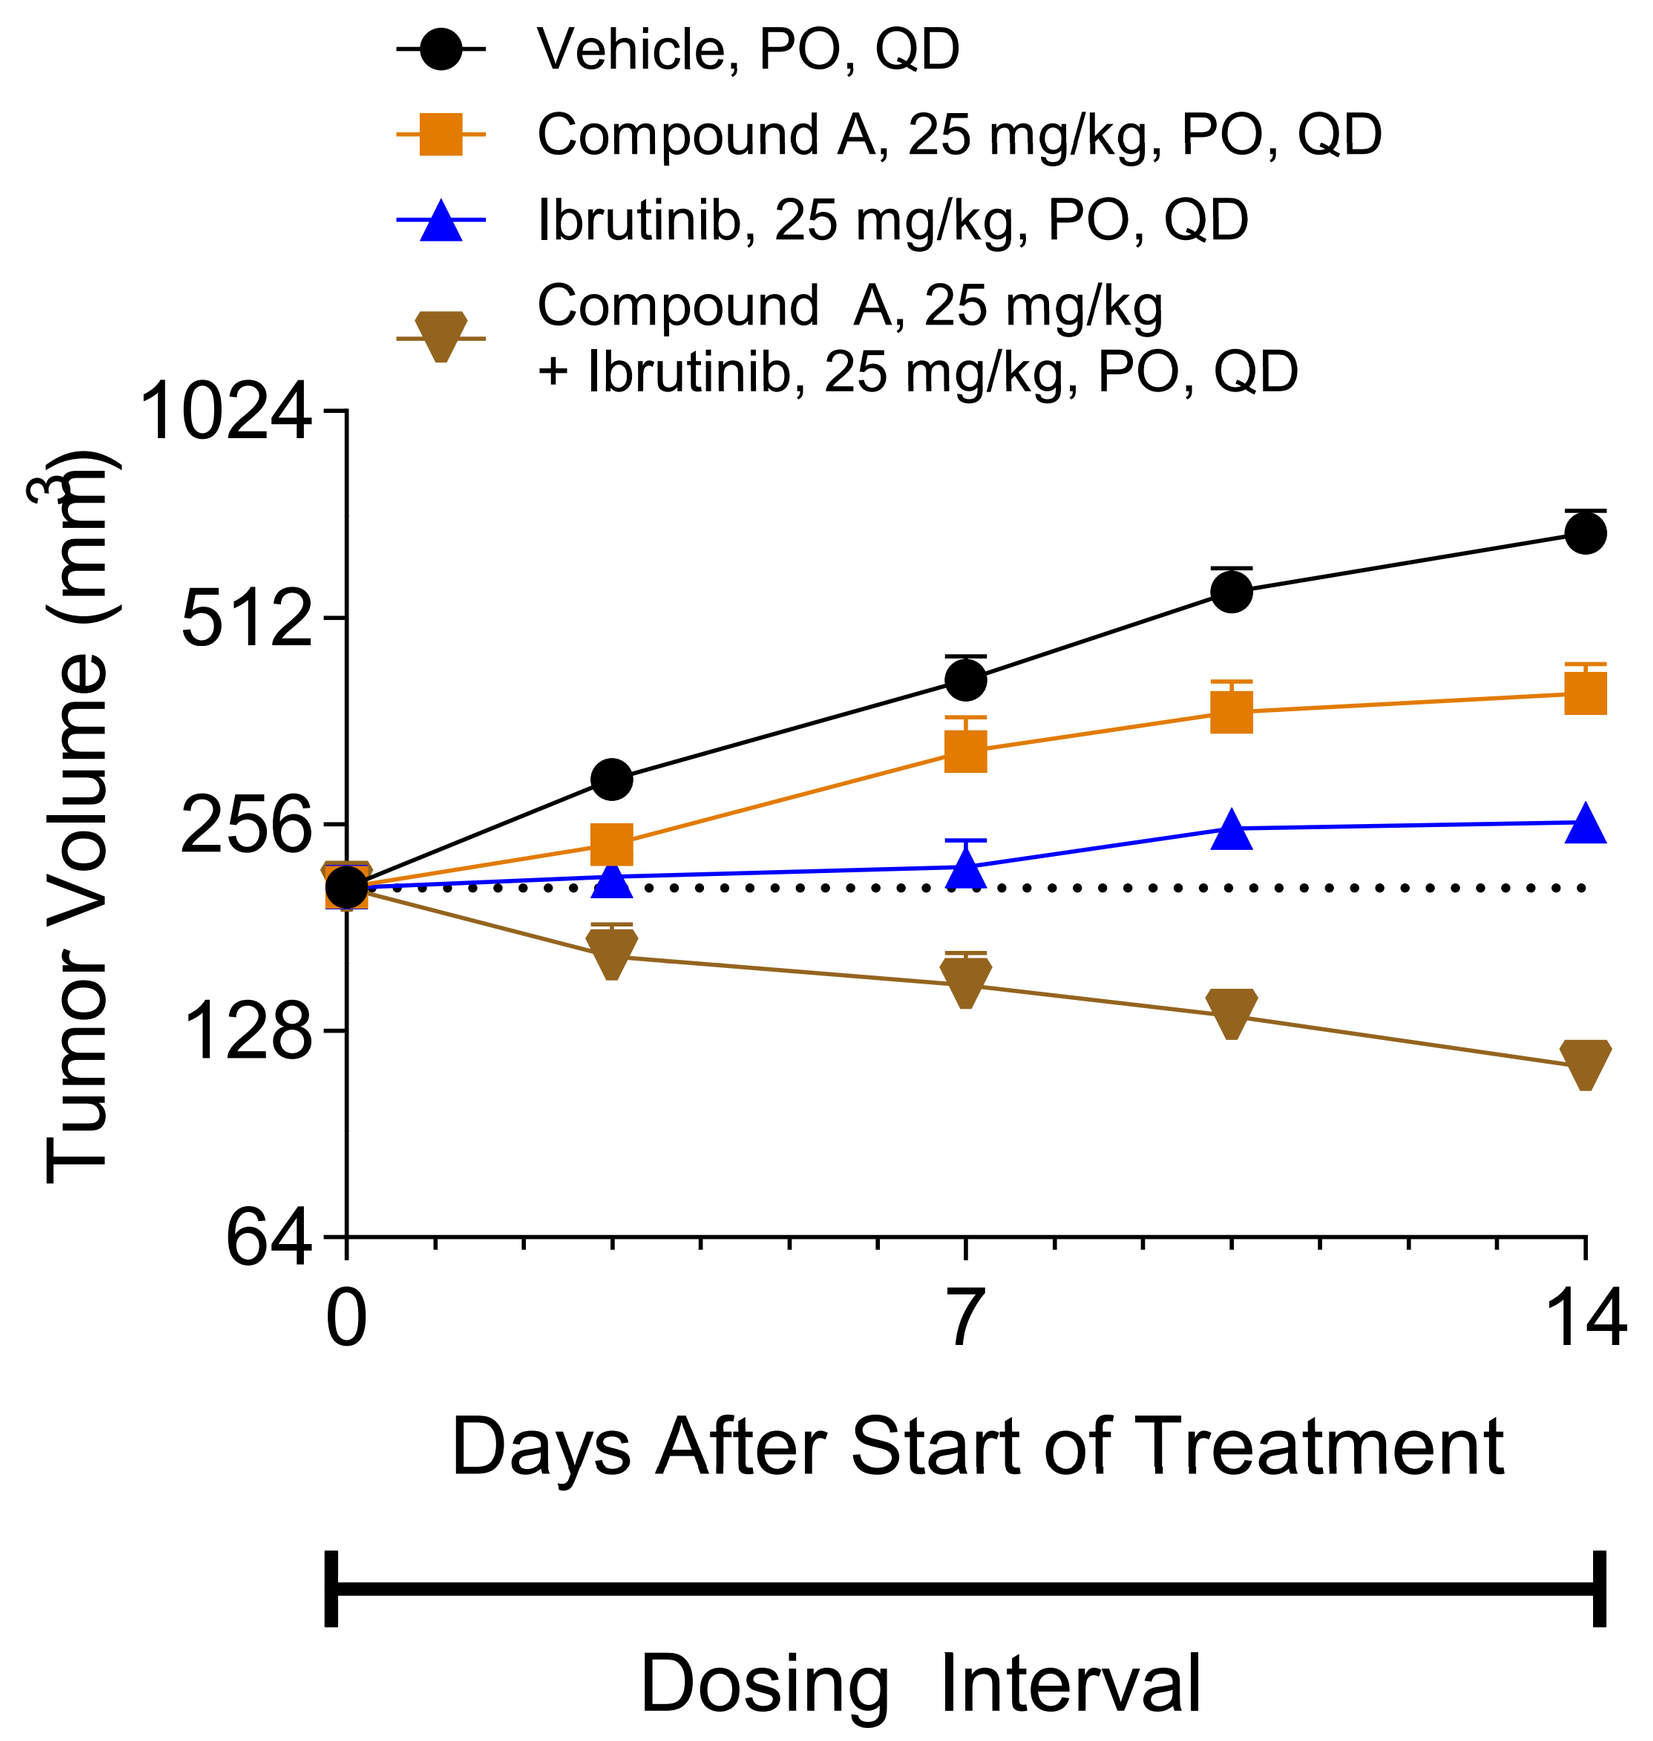
Vehicle, PO, QD
Compound A, 25 mg/kg, PO, QD
Ibrutinib, 25 mg/kg, PO, QD
Compound A, 25 mg/kg + Ibrutinib, 25 mg/kg, PO, QD
Tumor Volume (mm3)
1024
512
256
128
64
0
7
14
Days After Start of Treatment
Dosing Interval

Similar results were obtained with Compound B *in vitro*

IRAK4 Degrader shows dose proportional oral exposure in mouse and is orally bioavailable in rat, dog and monkey

Compound B dosed orally in mice shows dose-proportional exposure up to 300 mg/kg

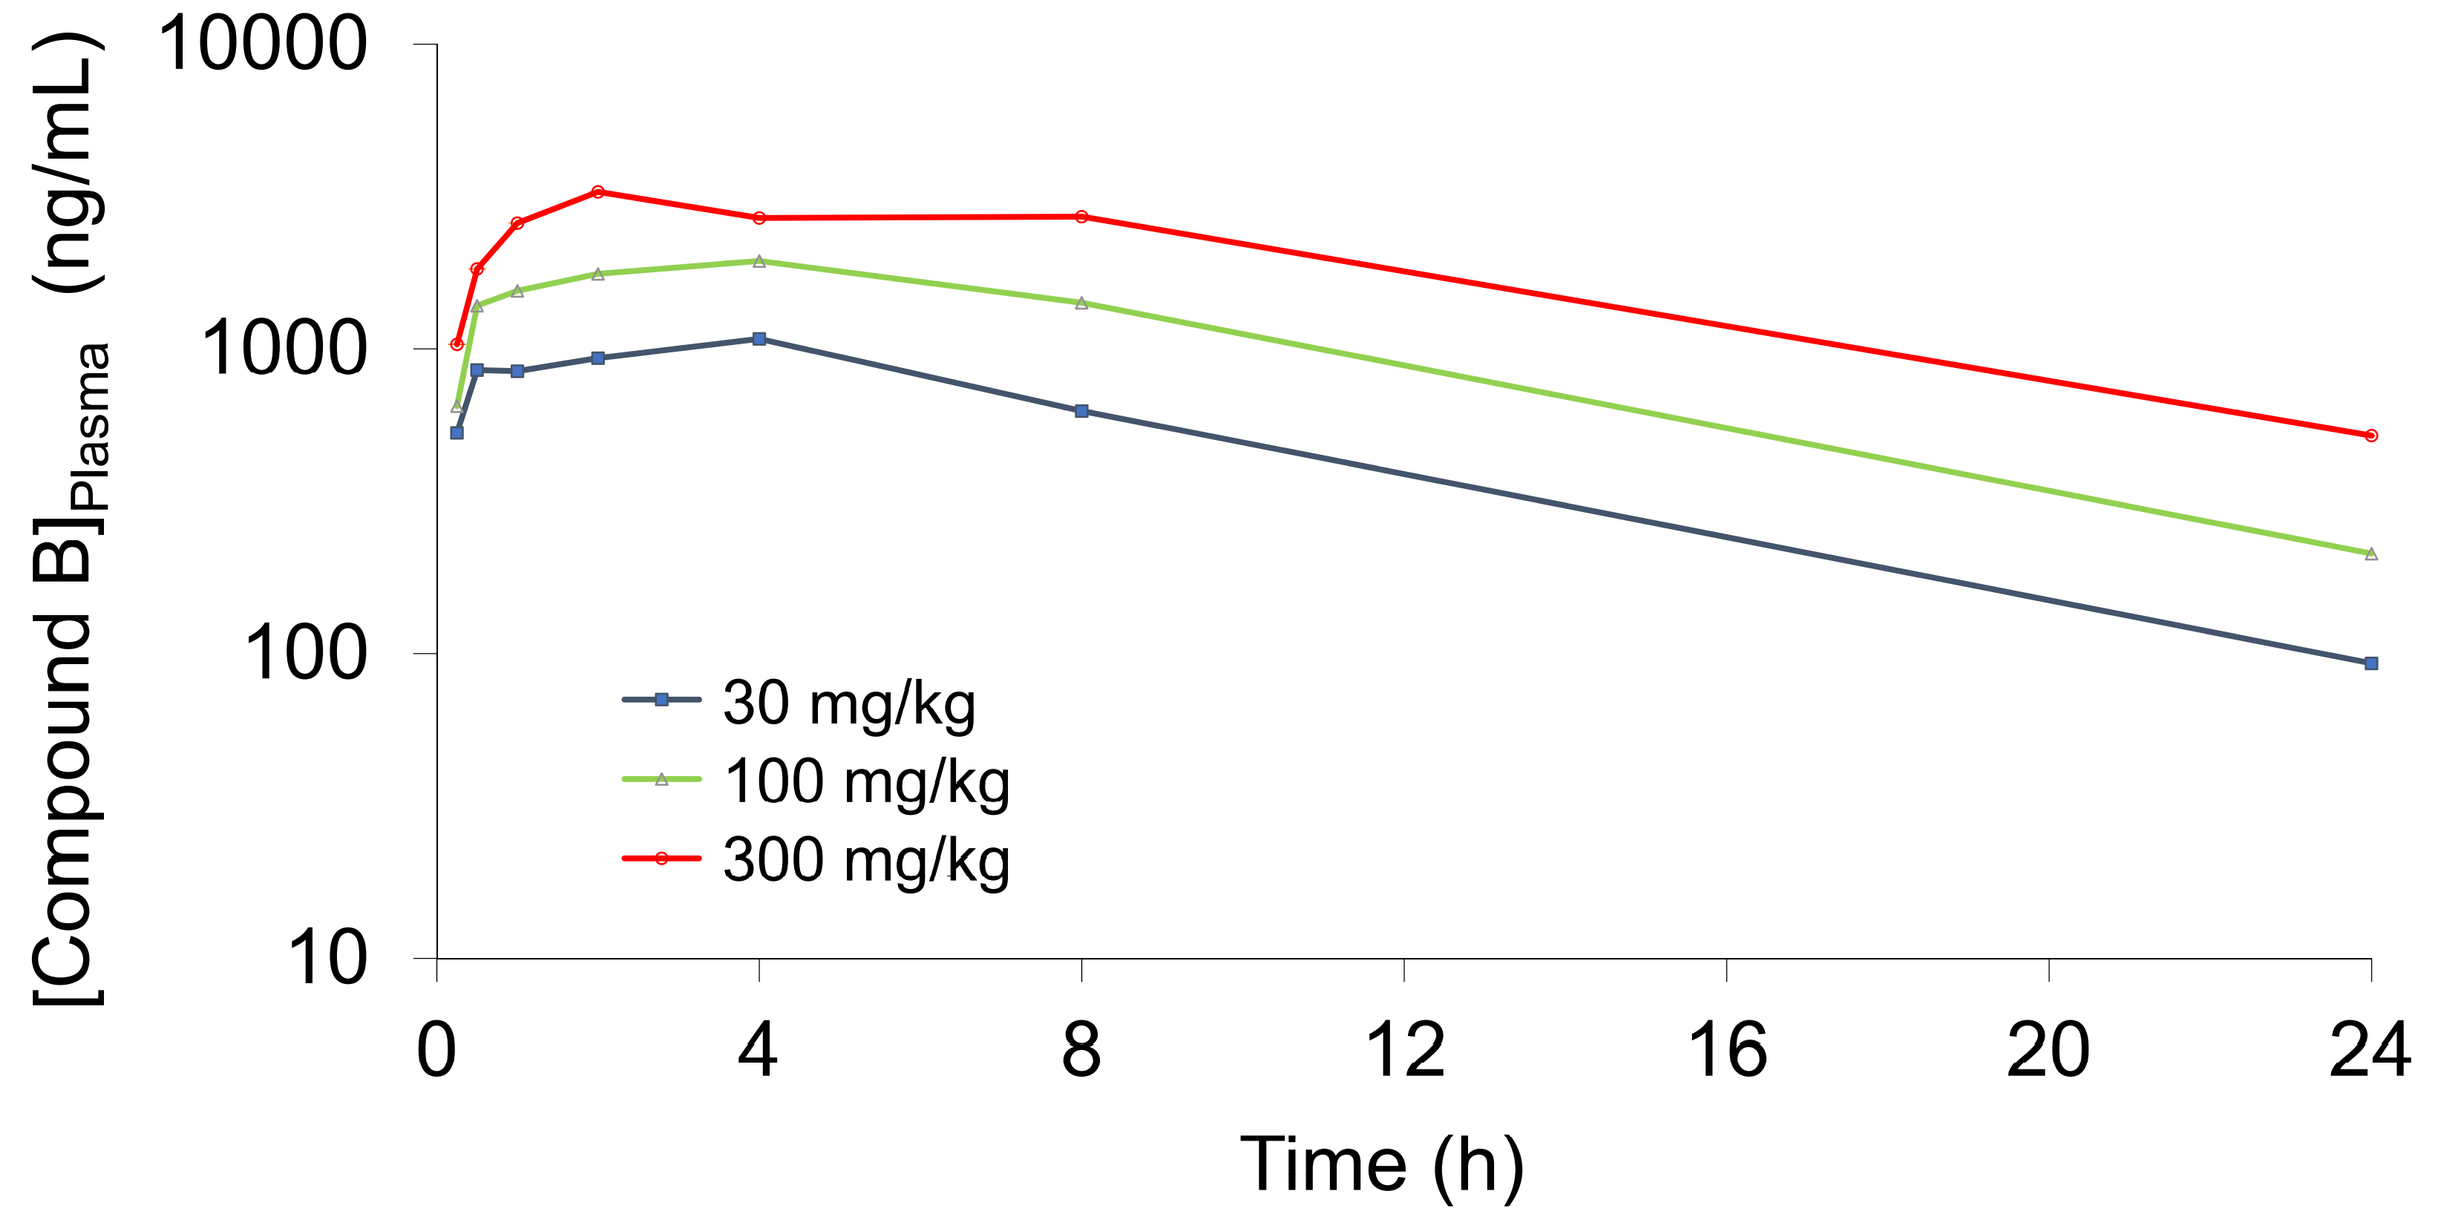
[Compound B]Plasma (ng/mL)
10000
1000
100
10
30 mg/kg
100 mg/kg
300 mg/kg
0
4
8
12
16
20
24
Time (h)

Compound B Shows promising PK characteristics in CD1 Mice

Species	Dose (mg/Kg)	Route	CI (mL/min/Kg)	T½ (h)	AUC_{last} (h*ng/mL)	%F
Mouse	2	IV	15	7.1	2050	-
	10	PO	-	5.4	2831	27

Compound B has oral bioavailability across species

Species	%F
Mouse	27
Rat	35
Dog	35
Monkey	14

Compound A also shows dose proportional oral bioavailability in mouse

IRAK4 degradation is synergistic with inhibition of BTK by ibrutinib in 4-day Cell Titer Glo *in vitro* cell viability assay in OCI-LY10

Compound A:Ibrutinib (144:1)

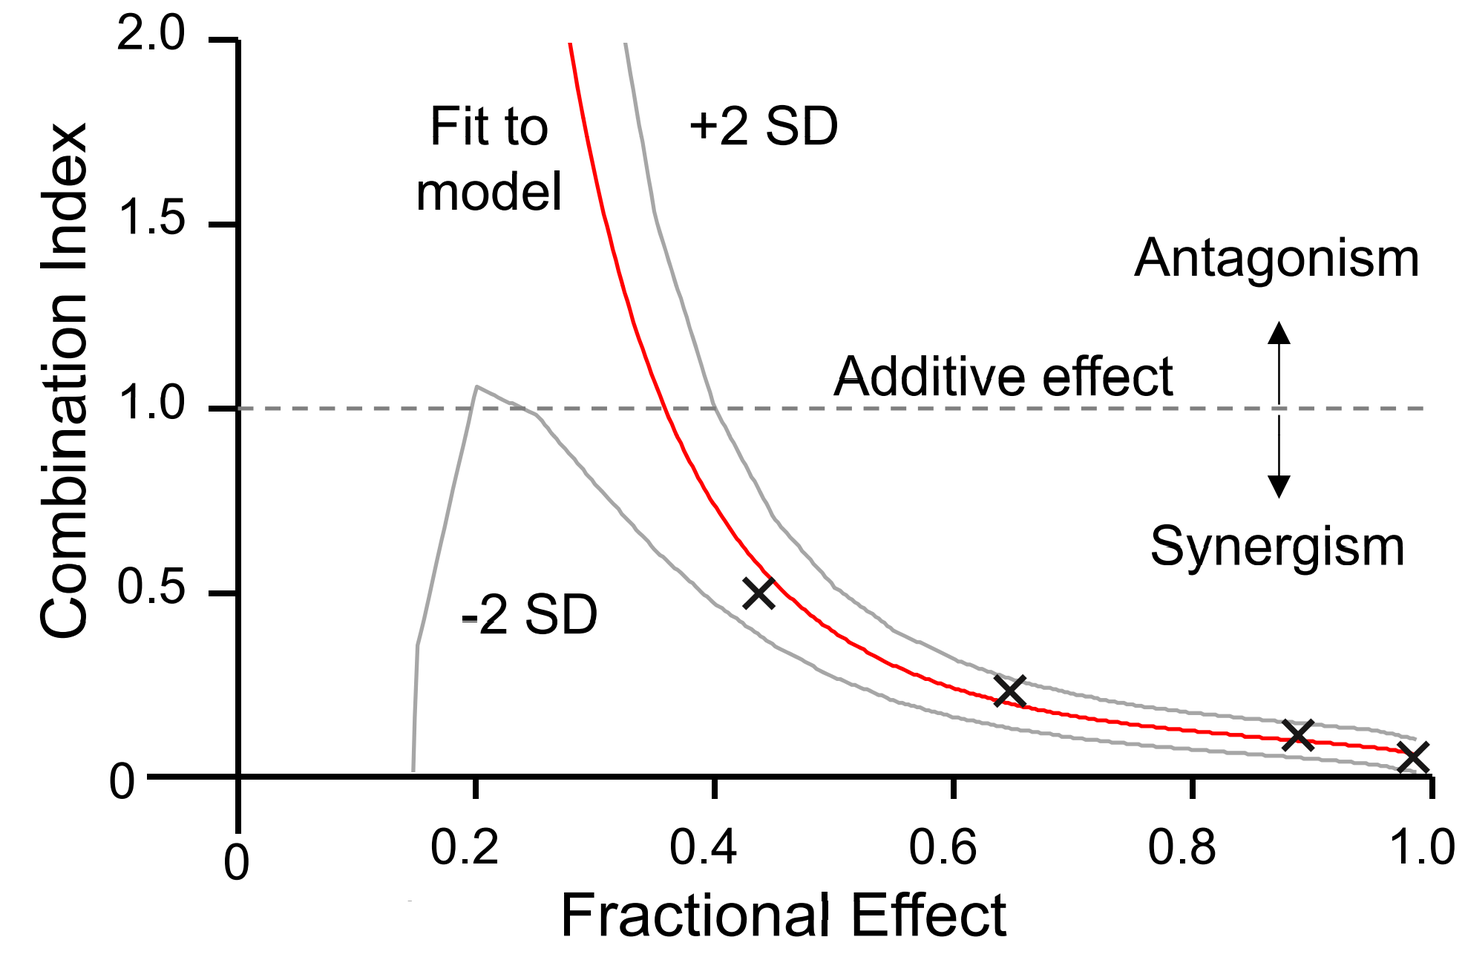
Combination Index
2.0
1.5
1.0
0.5
0
Fit to model
+2 SD
-2 SD
Antagonism
Additive effect
Synergism
0
0.2
0.4
0.6
0.8
1.0
Fractional Effect

Isobologram

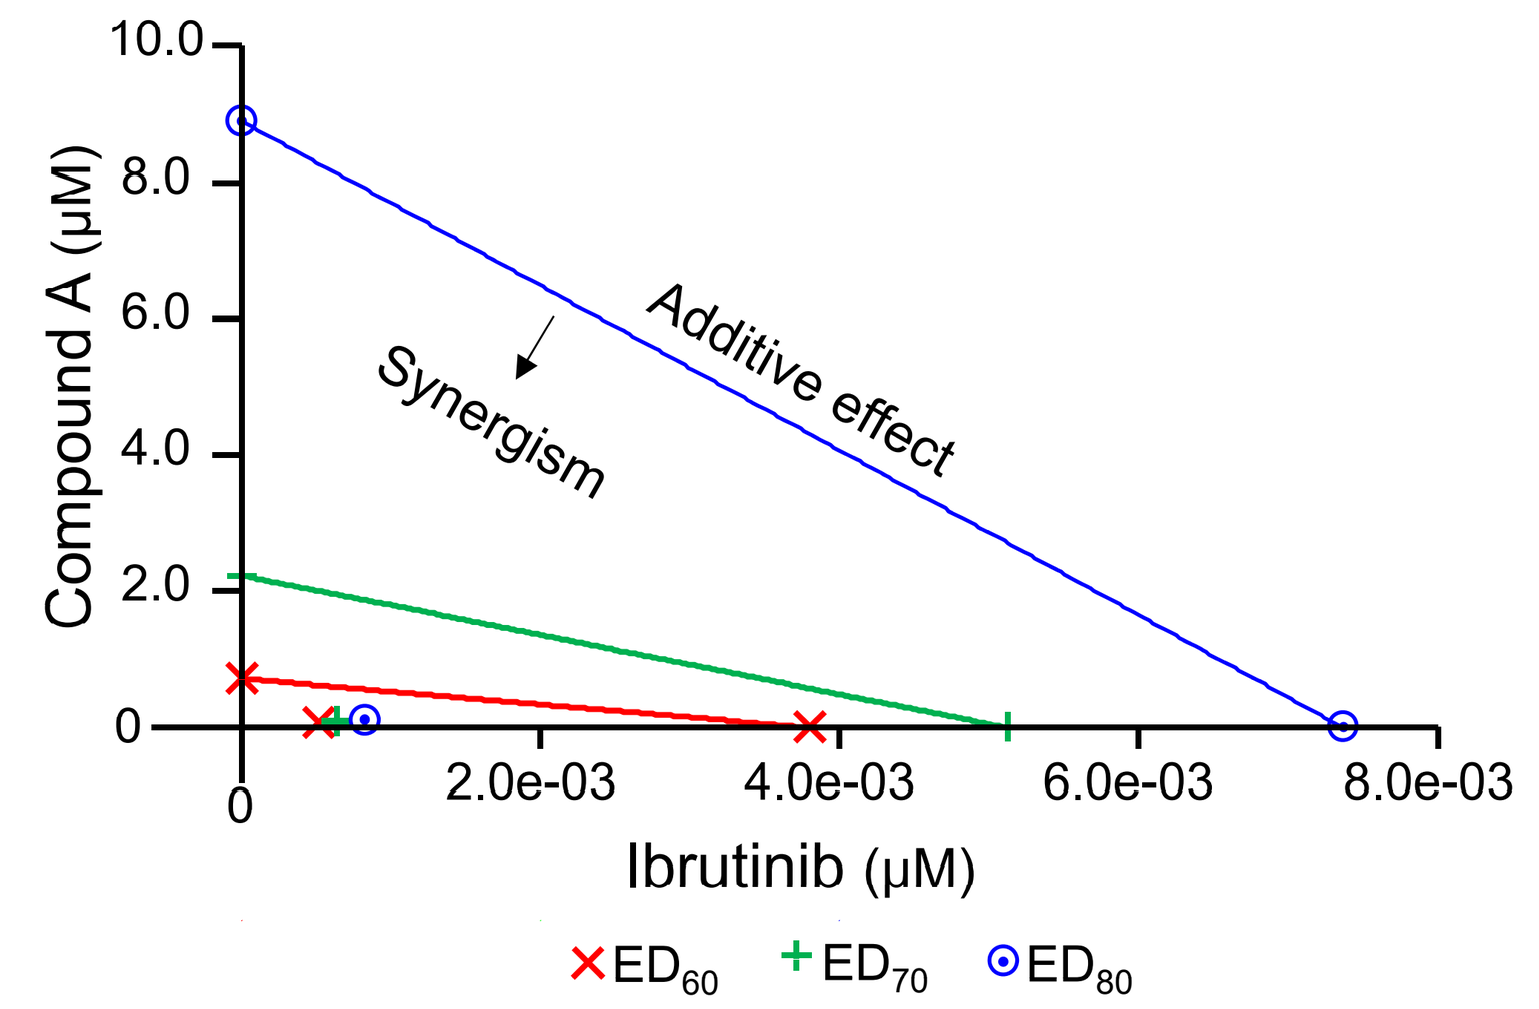
Compound A (μM)
10.0
8.0
6.0
4.0
2.0
0
Additive effect
Synergism
2.0e-03
4.0e-03
6.0e-03
8.0e-03
Ibrutinib (μM)
ED60
ED70
ED80

Similar results were obtained with Compound B

Conclusions

- IRAK4 Degrader induced potent and selective E3 ligase-dependent degradation of IRAK4, with 90% degradation at concentrations less than 100 nM.
- IRAK4 degradation was highly selective compared to >10,000 other proteins quantified by tandem mass tagged proteomics in MYD88 L265P/CD79 double mutant ABC DLBCL cell line OCI-LY10.
- IRAK4 Degrader has dose proportional oral exposure in plasma at doses up to 300 mg/kg in mice
- IRAK4 Degrader has oral bioavailability in rat, dog and monkey, enabling pre-clinical safety studies
- Oral dosing of an IRAK Degrader showed dose-dependent antitumor activity against OCI-LY10, with ≥75% degradation of IRAK4 correlating with tumor regression in xenograft-bearing mice.
- In OCI-LY10, IRAK Degrader synergized with BTK inhibition by ibrutinib to decrease cell viability *in vitro*, as measured using 4-day Cell Titer Glo assay
- *In vitro*, the combined activity of an IRAK4 Degrader and Ibrutinib resulted in a greater than additive increase in apoptosis within 72 h in OCI-LY10
- *In vivo*, the combined activity of an IRAK4 Degrader and ibrutinib drove regression of OCI-LY10 at concentration of Degrader that is suboptimal as single agent, supporting further exploration of combinations that target oncogenic NFκB signaling.
- These preclinical studies with IRAK4 Degraders provide guidance on tumor PK/PD projected to be effective in patients, and further support pre-clinical assessment and clinical development of KYM-001 Compound A, Compound B and related leads in MYD88-driven lymphomas via oral route.

Disclosures: Kelleher, Campbell, Chen, Gollob, Ji, Kamadurai, Klaus, Li, Loh, McDonald, Rong, Rusin, Sharma, Vigil, Walker, Weiss, Yuan, Zhang and Mainolfi: *Kymera Therapeutics:* Employment, Equity Ownership.
Audoly: *Kymera Therapeutics:* Employment, Equity Ownership, Membership on an entity's Board of Directors.